Effect of Omecamtiv Mecarbil in Patients With Heart Failure With Reduced Ejection Fraction and Atrial Fibrillation/Flutter: Results from COSMIC-HF

John R. Teerlink,[1] G. Michael Felker,[2] John J. V. McMurray,[3] Scott D. Solomon,[4] John GF Cleland,[5] Steven R. Goldsmith,[6] Christopher E. Kurtz,[7] Gustavo Buchele,[7] Jason Legg,[7] Fady I. Malik,[8] Narimon Honarpour[7]

[1]UCSF and SF Veterans Affairs Medical Center, San Francisco, CA, USA; [2]Duke University, Durham, NC, USA; [3]University of Glasgow, Glasgow, UK; [4]Brigham and Women's Hospital and Harvard Medical School, Boston, MA, USA; [5]Castle Hill Hospital, East Yorkshire, UK; [6]Hennepin County Medical Center and the University of Minnesota, Minneapolis, Minnesota, USA; [7]Amgen Inc, Thousand Oaks, CA, USA; [8]Cytokinetics, South San Francisco, CA, USA

INTRODUCTION

- Omecamtiv mecarbil (OM) is a novel selective cardiac myosin activator under investigation as a potential treatment for heart failure with reduced ejection fraction

Mechanochemical Cycle of Myosin[1,2]

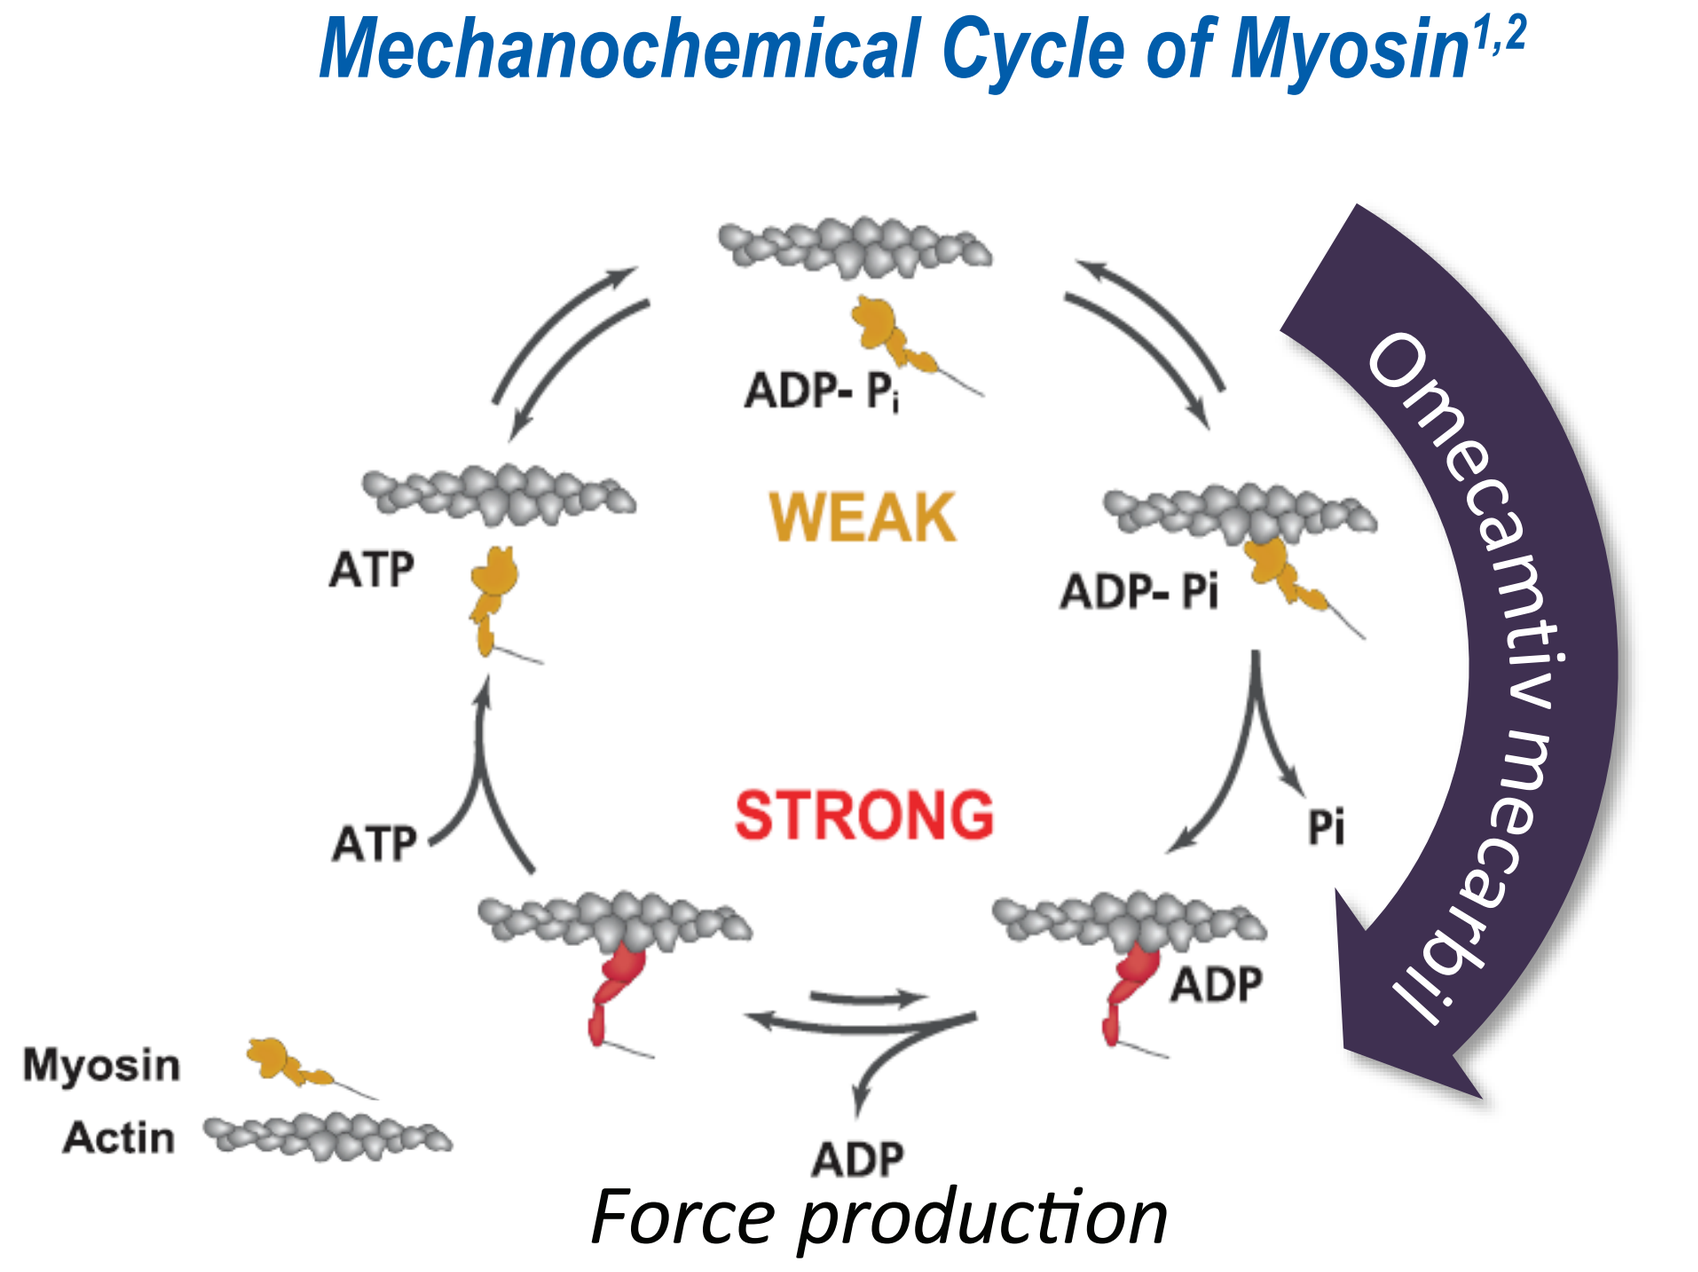
ADP- Pi
WEAK
ATP
ADP- Pi
STRONG
Pi
ATP
ADP
ADP
Myosin
Actin
Force production
Omecamtiv mecarbil

OM increases the entry rate of myosin into the tightly-bound, force-producing state with actin[3]

"More hands pulling on the rope"

- Increases duration of systole
- Increases stroke volume
- No increase in myocyte calcium
- No change in dP/dt_{max}
- No increase in MVO_2

MVO_2 = myocardial oxygen consumption

- Atrial fibrillation/flutter (AF) is a common comorbidity in patients with heart failure (HF)[4]
 - Prevalence increases by HF severity (4% in NYHA Class I to 40% in Class IV)

OBJECTIVES

- To characterize, in patients with and without atrial fibrillation or flutter, the effects of omecamtiv mecarbil (OM) at 20 weeks on change from baseline in:
 - Systolic ejection time (SET)
 - Stroke volume
 - Left ventricular end-diastolic (systolic) diameter (LVEDD, LVESD)
 - Heart rate
 - N-terminal pro-B-type natriuretic peptide (NT-proBNP)

METHODS

Study Design

- COSMIC-HF (NCT01786512) was a multicenter, randomized, placebo-controlled, double-blind phase 2 study that enrolled patients with:
 - Stable, optimally-treated chronic heart failure (HF)
 - New York Heart Association (NYHA) class II or III
 - Left ventricular ejection fraction (LVEF) ≤ 40%
 - N-terminal pro B-type natriuretic peptide (NT-proBNP) ≥ 200 pg/mL (≥ 1200 pg/mL with atrial fibrillation or flutter)
- Patients receiving chronic antiarrhythmics (except amiodarone) were excluded
- OM and placebo groups were compared on the basis of presence/absence of AF
 - Here we present data for the OM PK-guided titration group versus placebo group

Figure 1. Study Schema[5]

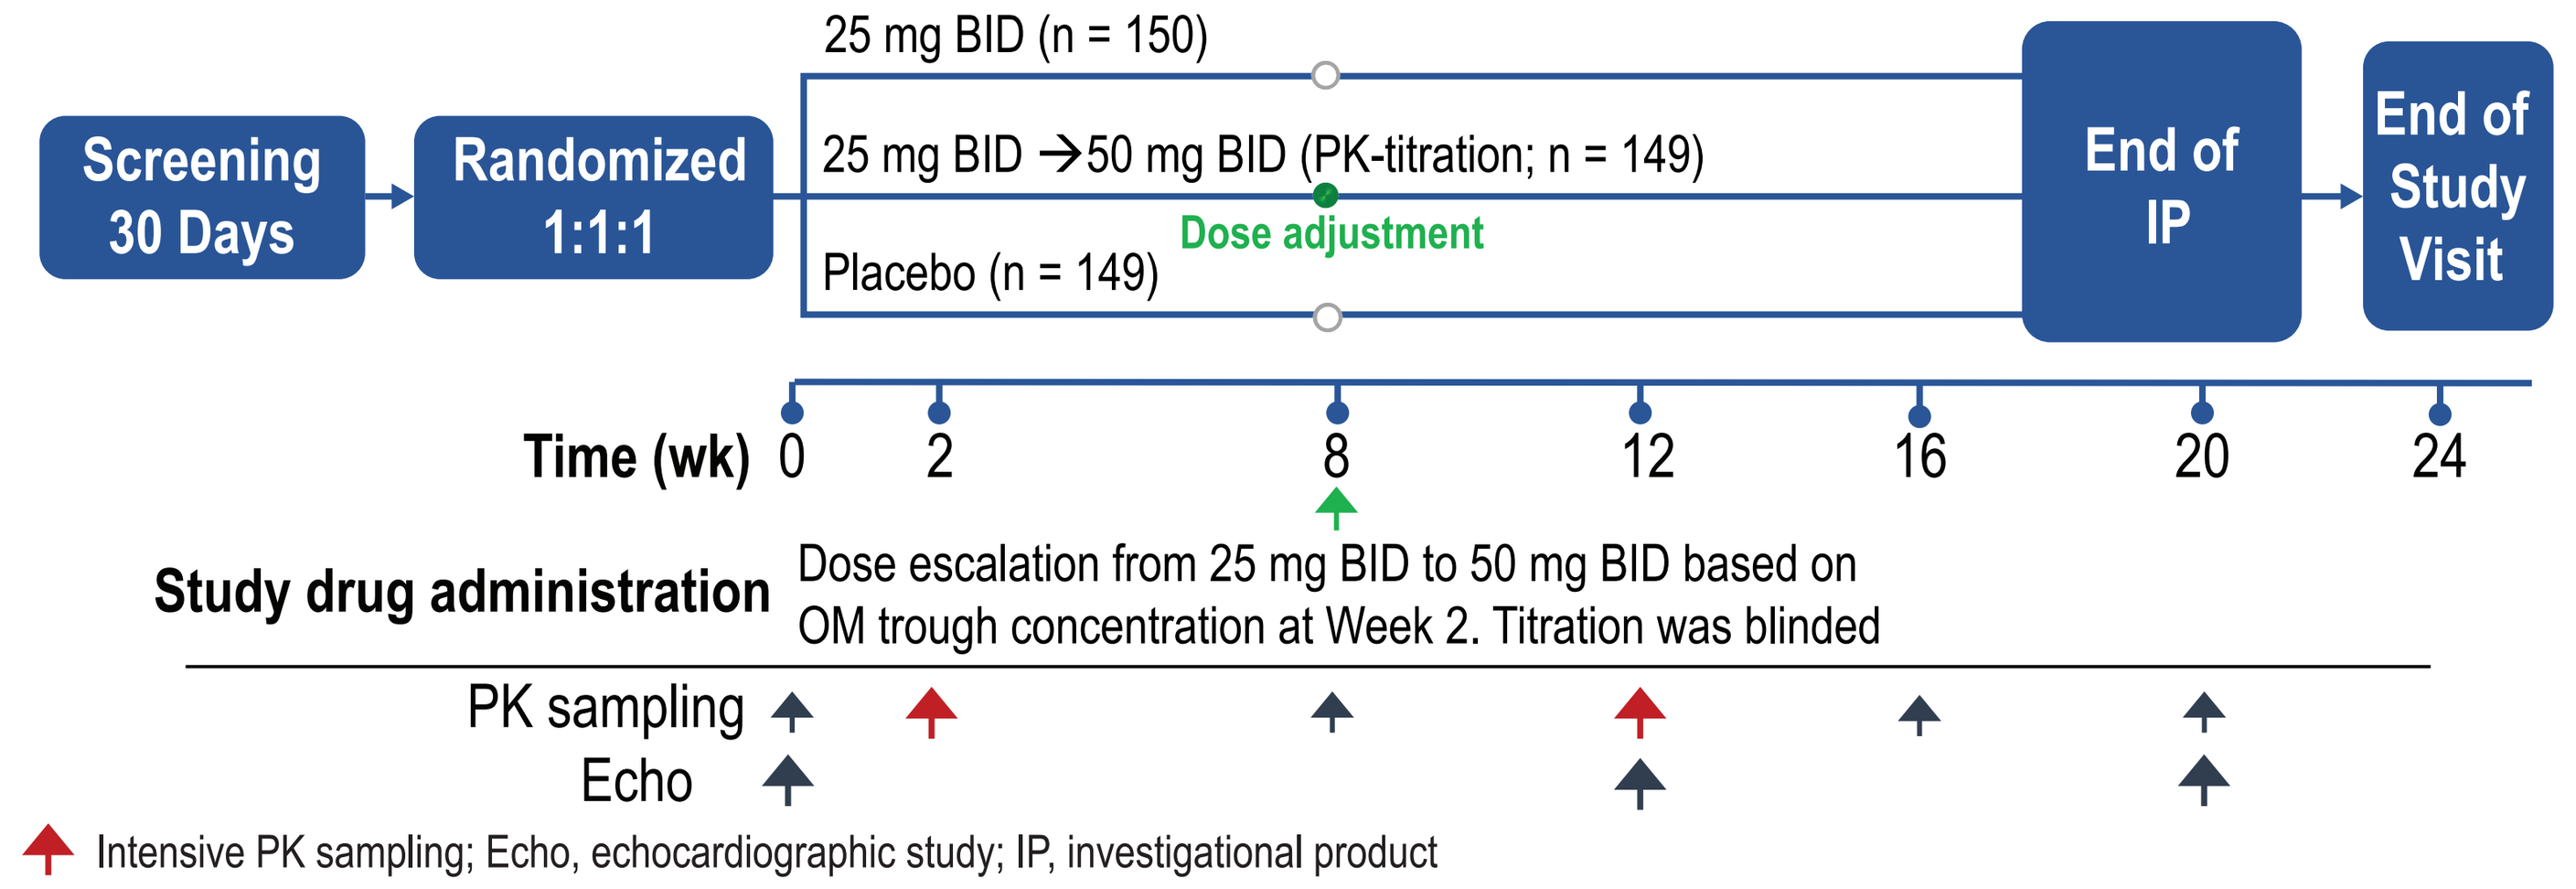
Screening
30 Days
Randomized
1:1:1
25 mg BID (n = 150)
25 mg BID →50 mg BID (PK-titration; n = 149)
Dose adjustment
Placebo (n = 149)
End of
IP
End of
Study
Visit
Time (wk) 0 2 8 12 16 20 24
Study drug administration
Dose escalation from 25 mg BID to 50 mg BID based on
OM trough concentration at Week 2. Titration was blinded
PK sampling
Echo

Intensive PK sampling; Echo, echocardiographic study; IP, investigational product

A dose escalation phase preceded the expansion phase described above by the study schema

RESULTS

Table 1. Baseline Demographics

- 21% of patients had atrial fibrillation/flutter (AF) at randomization, which was used as a stratification factor

	AF		No AF	
	Placebo n = 32	OM 25 – >50 mg PK-based titration n = 32	Placebo n = 117	OM 25 – >50 mg PK-based titration n = 117
Age, years, mean (SD)	65.8 (9.9)	70.3 (8.8)	63.1 (9.6)	60.6 (11.5)
Sex, male, %	71.9	87.5	82.1	82.9
Systolic blood pressure (mmHg), mean (SD)	117.4 (13.1)	116.5 (14.3)	119.9 (14.9)	118.9 (16.5)
Heart rate (bpm), mean (SD)	72.9 (12.3)	69.2 (11.5)	67.6 (9.7)	69.6 (12.1)
HF Characteristics				
NYHA Class II, %	65.6	71.9	71.8	71.8
Ischemic heart disease, %	56.3	84.4	60.7	63.2
Years from HF diagnosis, mean (SD)	9.5 (9.2)	9.3 (6.7)	7.6 (6.4)	7.2 (6.4)
HF hospitalization in prior year, %	31.3	21.9	23.9	26.5
Comorbidities, %				
Myocardial infarction	50.0	62.5	56.4	53.0
Percutaneous coronary intervention	40.6	46.9	41.9	41.0
Coronary artery bypass grafting	15.6	50.0	19.7	20.5
Hypertension	71.9	78.1	66.7	71.8
Stroke	18.8	9.4	6.8	9.4
Diabetes	46.9	28.1	39.3	39.3
HF Medications, %				
Angiotensin converting enzyme inhibitor (ACEI)	65.6	65.6	72.6	65.0
Angiotensin II receptor blocker (ARB)	34.4	21.9	21.4	28.2
Beta blocker (BB)	100.0	90.6	97.4	98.3
Mineralocorticoid receptor antagonist (MRA)	65.6	40.6	57.3	69.2
Diuretics	90.6	87.5	82.1	90.6
Echocardiographic Variables				
Stroke volume, mL, mean (SD)	51.3 (13.3)	51.0 (14.0)	52.4 (15.4)	52.8 (15.1)
SET, msec, mean (SD)	280.7 (30.7)	297.7 (28.5)	303.5 (37.2)	299.1 (33.9)
LVEDD, cm, mean (SD)	6.3 (0.9)	6.0 (0.8)	6.2 (1.0)	6.4 (0.9)
LVESD, cm, mean (SD)	5.4 (0.9)	5.1 (0.7)	5.3 (1.0)	5.5 (0.9)
NT-proBNP, pg/mL, median (Q1,Q3)	2941 (1717, 5394)	2998 (1687, 4356)	1322 (647, 2483)	1564 (687, 2457)

Abbreviations: LVEDD, left ventricular end-diastolic diameter; LVESD, left ventricular end-systolic diameter; NT-proBNP, N-terminal pro-B-type natriuretic peptide; SD, standard deviation; SET, systolic ejection time

Figure 1. COSMIC-HF: Overall Effects of Omecamtiv Mecarbil

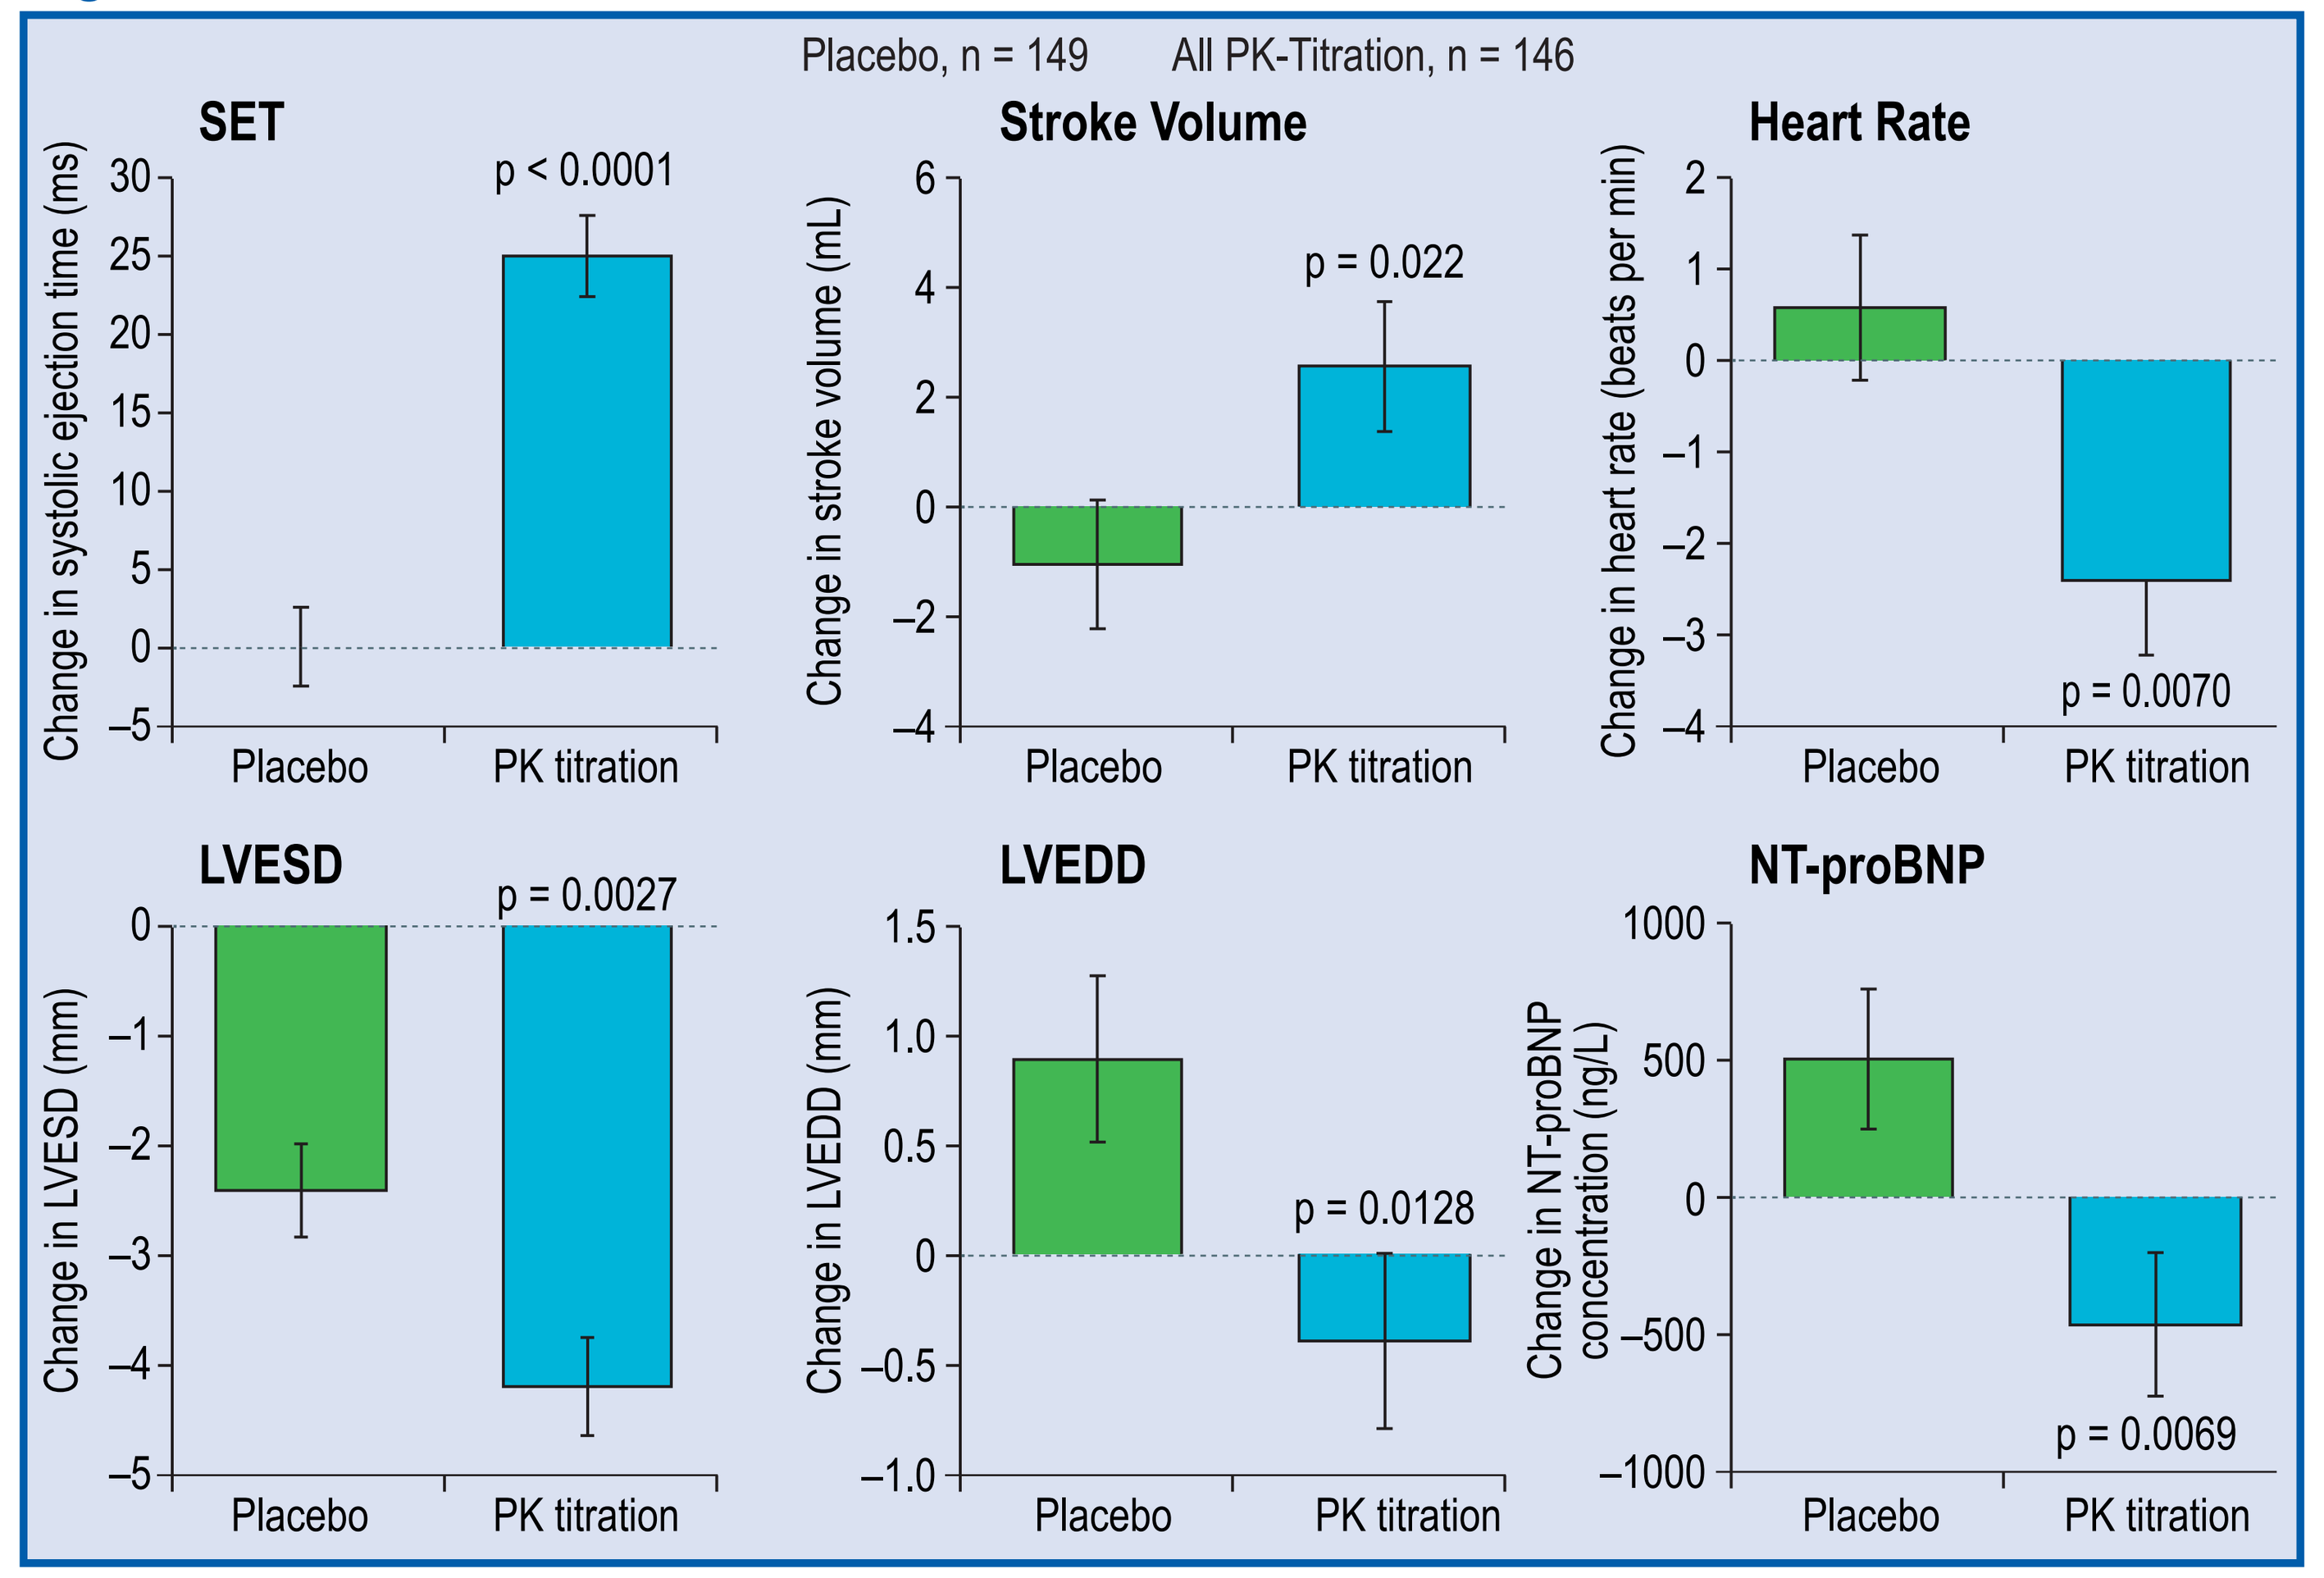
Placebo, n = 149 All PK-Titration, n = 146
SET
p < 0.0001
Change in systolic ejection time (ms)
Stroke Volume
p = 0.022
Change in stroke volume (mL)
Heart Rate
p = 0.0070
Change in heart rate (beats per min)
LVESD
p = 0.0027
Change in LVESD (mm)
LVEDD
p = 0.0128
Change in LVEDD (mm)
NT-proBNP
p = 0.0069
Change in NT-proBNP concentration (ng/L)
Placebo
PK titration

LS, Least squares; SE, standard error

Table 2. Effects of Omecamtiv Mecarbil in Patients With HFrEF, With and Without AF

Prespecified Secondary Endpoints	Placebo-corrected change from baseline at 20 weeks in OM PK-titration group: AF, n = 32 LS mean (95% CI)	No AF, n = 117 LS mean (95% CI)	Interaction (p-value)
SET (msec)	27 (13, 41)	24 (17, 32)	0.69
Stroke volume (mL)	6.6 (–0.6, 13.8)	3.0 (–0.4, 6.5)	0.38
LVEDD (mm)	–1.1 (–3.3, 1.1)	–1.3 (–2.4, –0.1)	0.90
LVESD (mm)	–2.1 (–4.5, 0.4)	–1.6 (–2.9, –0.3)	0.75
HR (bpm)	–7.8 (–13.8, –1.7)	–2.0 (–4.2, 0.1)	0.08
NT-proBNP (pg/mL)	–187 (–2216, 1842)	–1022 (–1597, –448)	0.43

P-values are post-hoc, nominal and unadjusted for multiple comparisons; Doppler-derived stroke volume

Abbreviations: CI, confidence interval; HR, heart rate.

Figure 2. Effects of Omecamtiv Mecarbil on Change in Left Ventricular Diameter, Cardiac Function, Heart Rate, and NT-proBNP in Patients With and Without AF

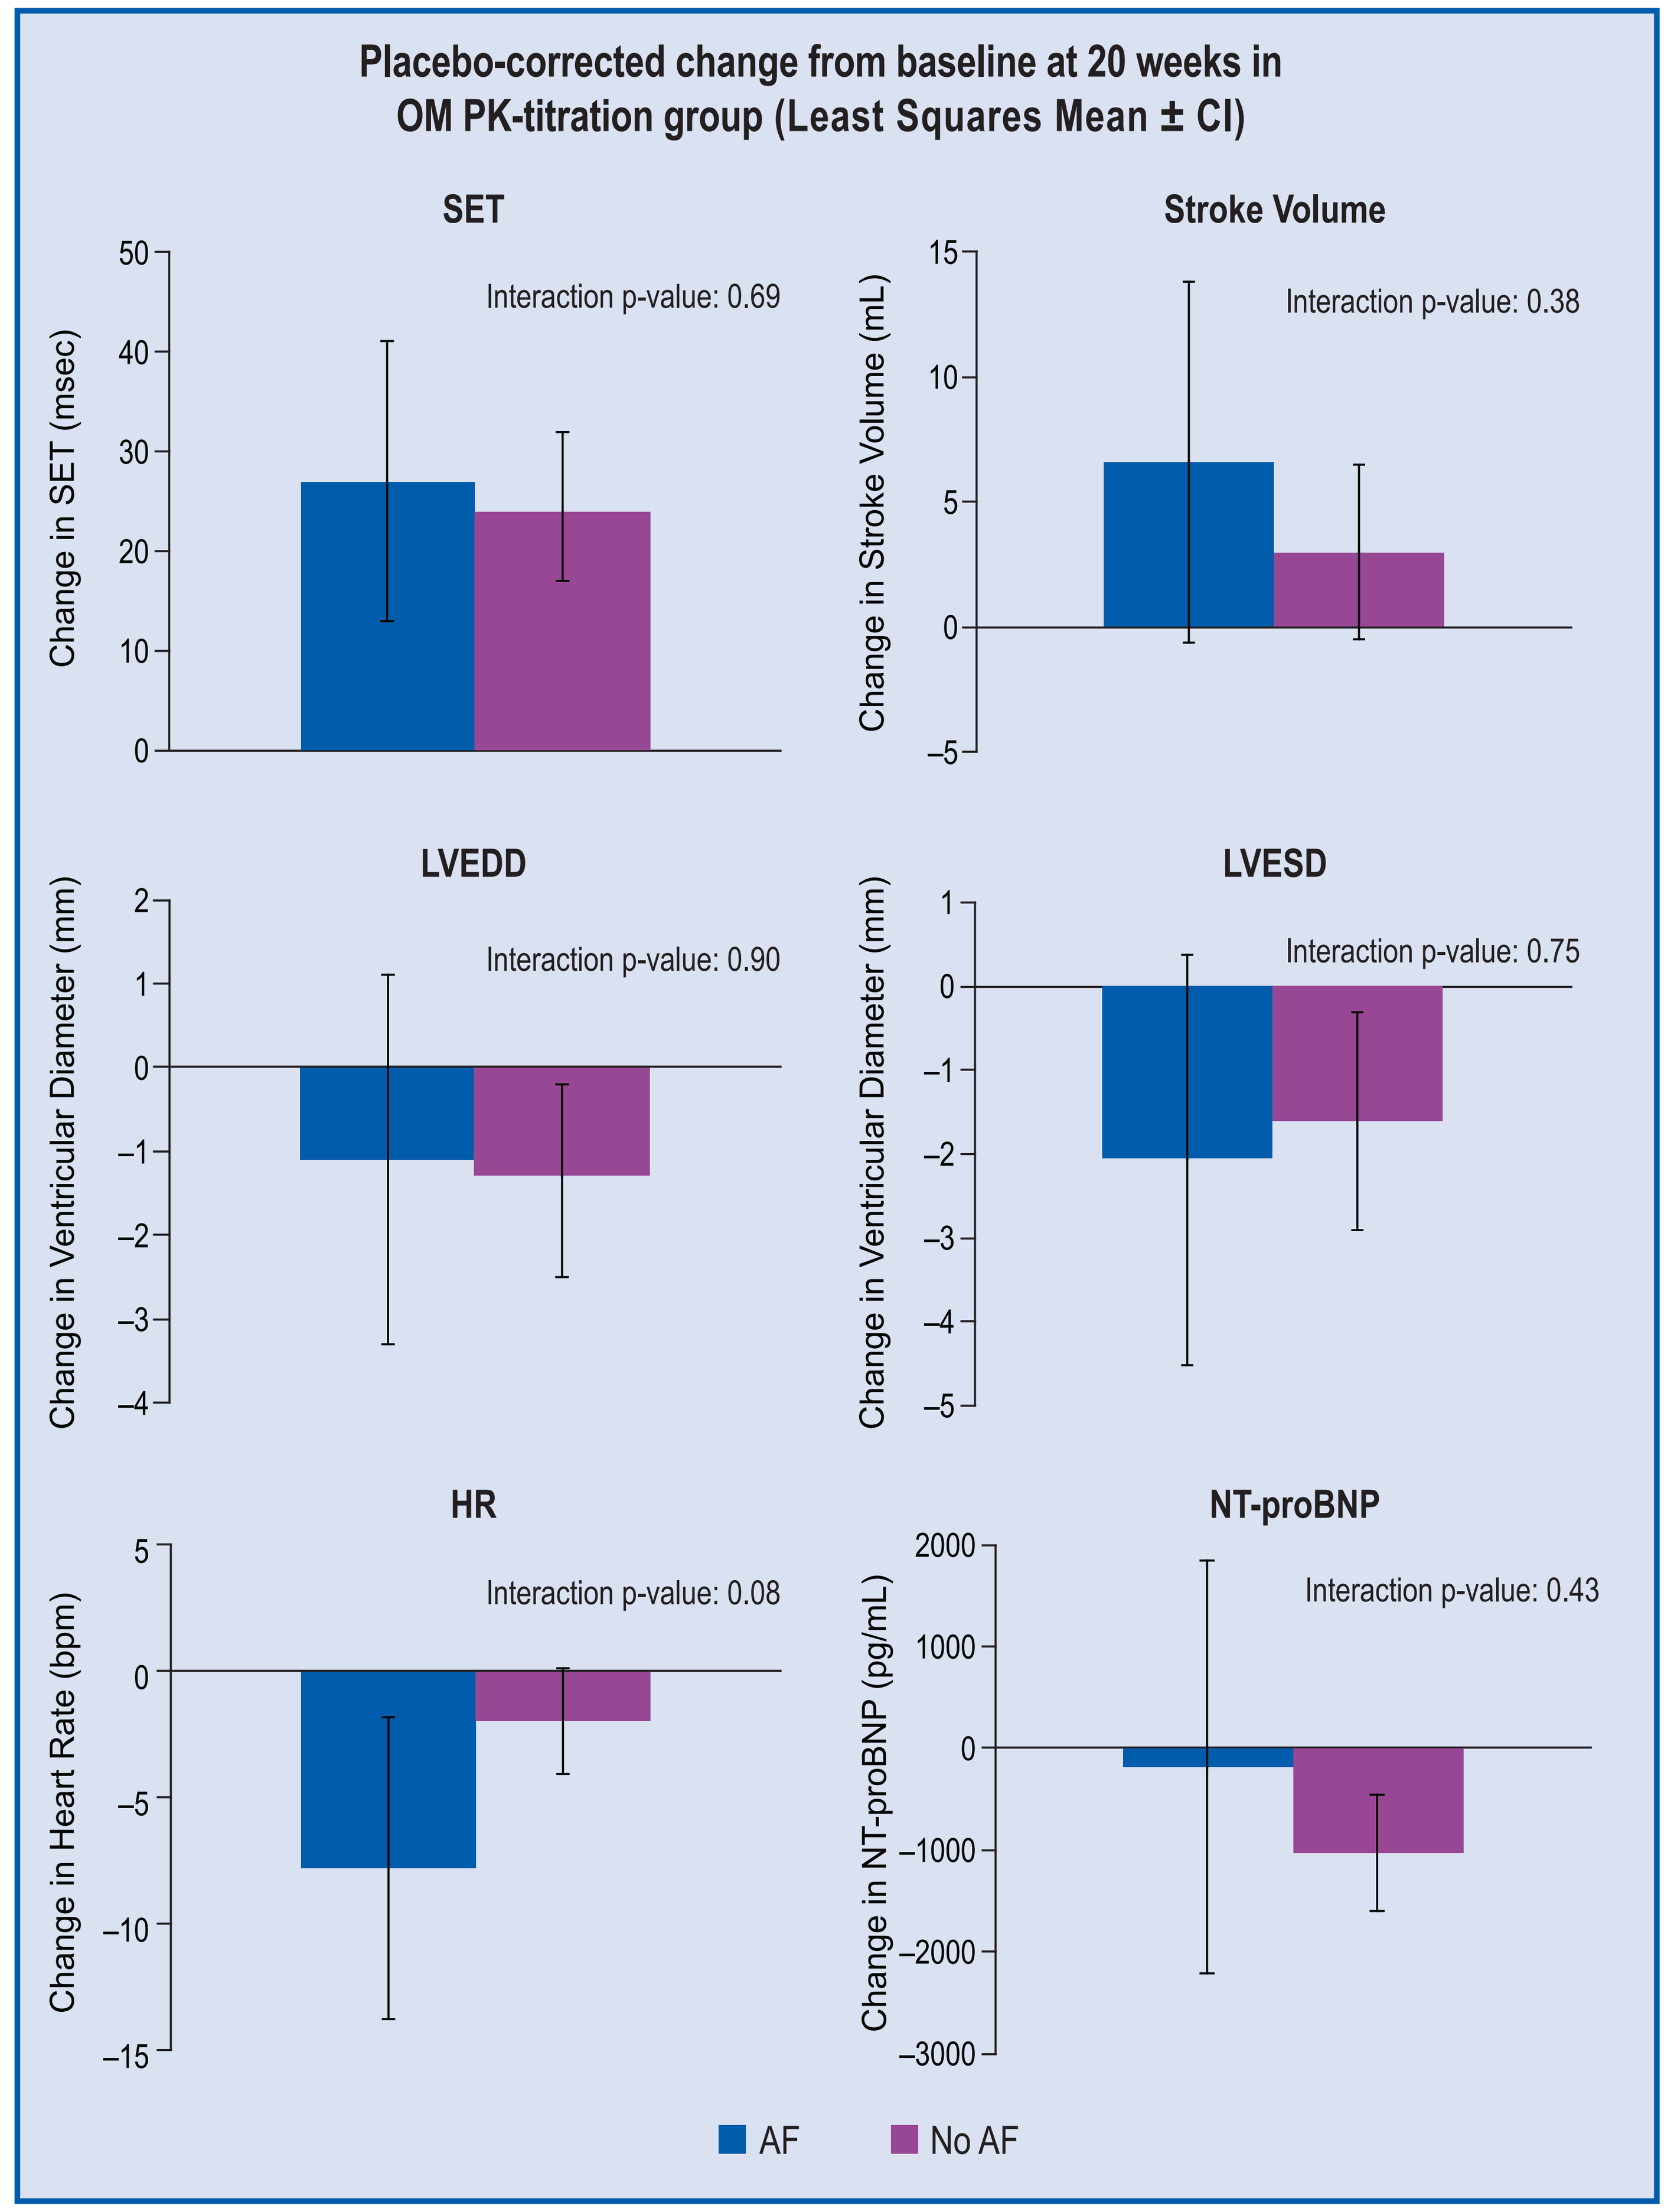
Placebo-corrected change from baseline at 20 weeks in
OM PK-titration group (Least Squares Mean ± CI)
SET
Interaction p-value: 0.69
Change in SET (msec)
Stroke Volume
Interaction p-value: 0.38
Change in Stroke Volume (mL)
LVEDD
Interaction p-value: 0.90
Change in Ventricular Diameter (mm)
LVESD
Interaction p-value: 0.75
Change in Ventricular Diameter (mm)
HR
Interaction p-value: 0.08
Change in Heart Rate (bpm)
NT-proBNP
Interaction p-value: 0.43
Change in NT-proBNP (pg/mL)
AF
No AF

Table 3. Summary of Adverse Events

	AF		No AF	
	Placebo (n = 32)	OM 25 – >50 mg PK-based titration (n = 30)	Placebo (n = 117)	OM 25 – >50 mg PK-based titration (n = 116)
Treatment-emergent AEs, n (%)	21 (65.6)	19 (63.3)	70 (59.8)	76 (65.5)
Serious AEs	4 (12.5)	8 (26.7)	26 (22.2)	24 (20.7)
Leading to discontinuation	1 (3.1)	5 (16.7)	11 (9.4)	7 (6.0)
Cardiac AEs, n (%)	4 (12.5)	8 (26.7)	32 (27.4)	23 (19.8)
Cardiac failure	3 (9.4)	3 (10.0)	10 (8.5)	5 (4.3)
Acute cardiac failure	0 (0)	0 (0)	1 (0.9)	3 (2.6)
Congestive cardiac failure	0 (0)	3 (10.0)	3 (2.6)	1 (0.9)
Angina pectoris	0 (0)	0 (0)	3 (2.6)	1 (0.9)
Ventricular tachycardia	0 (0)	1 (3.3)	2 (1.7)	3 (2.6)
Adjudicated events, n	2	8	23	19
Death	0	0	4*	3*
MI	0	0	1	0
Hospitalization for HF	2	5	9	5
Troponin I (ng/mL), change from baseline to week 20, median (Q1, Q3)	0.00 (–0.01, 0.01)	0.02 (0.01, 0.03)	0.00 (-0.01, 0.00)	0.00 (0.00, 0.02)

*In the placebo group, 2 of the 4 deaths were cardiovascular (2 sudden cardiac deaths); in the OM group, 2 of the 3 deaths were cardiovascular (1 sudden cardiac death, 1 due to other CV cause)

CONCLUSION

- Overall, COSMIC-HF showed that omecamtiv mecarbil, a novel selective cardiac myosin activator, improved measures of cardiac systolic function and decreased ventricular volumes, heart rate, and NT-proBNP over 20 weeks of treatment
 - Results for the OM fixed-dose and PK-guided titration groups were qualitatively similar
- The effects of omecamtiv mecarbil on the above parameters did not differ statistically according to the presence or absence of AF at baseline
- Overall, the pattern of AEs did not appear markedly different between treatment arms, regardless of the presence of AF at baseline
- Findings are limited by relatively small subgroup size and differences
- The phase 3 study GALACTIC-HF (NCT02929329) will examine the effects of omecamtiv mecarbil on CV death and HF hospitalization, including approximately 25% of patients with AF at baseline
 - Chronic HF patients on standard-of-care therapy with LVEF ≤ 35%, NYHA II-IV, HF hospitalization or ED visit within 12 months, elevated natriuretic peptide
 - N≈8,000 patients, primary endpoint of CV death and HF hospitalization; powered for CV death

REFERENCES

1. Malik FI, et al. *Science.* 2011;331:1439-43.
2. Shen YT, et al. *Circ Heart Fail*. 2010;3:522-527.
3. Planelles-Herrero VJ, et al. *Nat Commun*. 2017;8:190
4. Yancy CW, et al. *JACC*. 2013;62(16):147-239.
5. Teerlink JR, et al. *Lancet*. 2016;388:2895-903.

DISCLOSURES

JRT has received research grants and consulting fees from Amgen, Astra Zeneca, Bayer, BMS, Celyad, Cytokinetics, Medtronic, Merck, Novartis, St. Jude, and Trevena and is a consultant for Stealth.

JL, CK and HN are Amgen employees and own Amgen stock/stock options.

FIM is an employee of Cytokinetics and owns Cytokinetics stock/ stock options.

Funding: Amgen Inc. provided funding for this study

Editorial support for the development of this presentation was provided by Amgen Inc.

© 2019 Amgen Inc.

American College of Cardiology, New Orleans, LA; March 16–18, 2019