615

The Cardiac Myosin Inhibitor, CK-3773274, Reduces Contractility in the R403Q Mouse Model of Hypertrophic Cardiomyopathy

Darren T. Hwee, Yangsong Wu, Peadar Cremin, Bradley P. Morgan, Fady I. Malik, Eva R. Chin

Cytokinetics, Inc., South San Francisco, CA

ABSTRACT

Cardiac sarcomere hypercontractility appears to underlie pathological hypertrophy and fibrosis in select genetic hypertrophic cardiomyopathies. The small molecule CK-3773274 is a novel cardiac myosin inhibitor that decreases contractility in vitro and in healthy animals in vivo. The objective of this study was to evaluate the effect of CK-3773274 in the genetic R403Q mouse model of hypertrophic cardiomyopathy. At approximately 40 weeks of age, left ventricular wall dimensions were determined by echocardiography in male wild type (WT) and heterozygous R403Q mice. As an indicator of cardiac hypertrophy, R403Q mice had significantly greater septal and posterior wall thickness than WT mice (septal wall WT: 0.93 ± 0.03 mm vs. R403Q: 1.22 ± 0.08 mm; posterior wall WT: 0.84 ± 0.04 mm vs. R403Q: 1.09 ± 0.04 mm; mean ± SEM, *p* < 0.05). R403Q mice were treated with single oral doses of CK-3773274 ranging from 0.25 to 1.5 mg/kg, and fractional shortening (FS) and heart rate were assessed at select time points over a 24-hour period. One hour after dose administration, CK-3773274 significantly reduced FS in a dose-related fashion relative to pre-dose baseline values (FS % R403Q baseline: 55.5 ± 2%; 0.25 mg/kg: 43.9 ± 2%; 1.5 mg/kg: 13.7 ± 1%; mean ± SEM, *p* < 0.05 baseline vs. all doses) without any changes to heart rate. At all dose levels, FS returned to baseline values by 24 hours. The plasma concentration at 10% and 50% reduction of FS relative to baseline (IC_{10} and IC_{50}) was 0.11 and 0.78 μM, respectively. In summary, single oral dose administration of CK-3773274 reduced FS in a dose and concentration-dependent manner in the genetic R403Q mouse model of hypertrophic cardiomyopathy. Cardiac myosin inhibition may be a viable approach to reduce underlying hypercontractility of the cardiac sarcomere in hypertrophic cardiomyopathies.

INTRODUCTION

- Hypercontractility of the cardiac sarcomere may be essential for the underlying pathological hypertrophy and fibrosis seen in many hypertrophic cardiomyopathy (HCM) patients[1]
- Direct modulation of the sarcomere is a novel approach to potentially treat conditions with maladaptive changes in cardiac contractility[1,2]
- The objective of this study was to evaluate the effect of the novel small molecule cardiac myosin inhibitor CK-3773274 in the genetic R403Q mouse model of HCM

METHODS

- To model the human R403Q β-myosin heavy chain mutation, a mouse model with an R404Q point mutation on α-myosin heavy chain was generated using CRISPR-Cas9 technology by the Jackson Laboratory (Bar Harbor, Maine)
- 40- to 48-week-old male wild type (WT) and heterozygote R403Q mice were dosed with CK-3773274 (0.25, 0.5,1, 1.25, or 1.5 mg/kg, PO) in a 0.5% (w/v) hydroxypropylmethylcellulose/0.1% (v/v) Tween 80 suspension at a dose volume of 5 mL/kg
- Echocardiographic assessment of left ventricular contractility was performed before dosing and 1, 4, 8, and 24 hours post-dose
 - A Vevo3100 ultrasound system and software (Toronto, Ontario, Canada) were used
 - A 55 MHz probe was placed at the level of the papillary muscles and 2D M-mode images of the left ventricle were captured in short axis view
 - Diastolic and systolic wall dimensions, heart rate, and cardiac fractional shortening (FS) was determined by analysis of the M-mode images
- Blood samples were collected following echocardiographic assessment for compound plasma concentration analysis

RESULTS

Table 1. At 40 to 48 weeks of age, R403Q mice have significantly greater wall thickness than WT mice

	Body Mass (g)	Heart Mass (mg): Tibia Length (cm)	Interventricular Septal Wall Thickness (mm)	Left Ventricular Posterior Wall Thickness (mm)
Wild Type (n=10)	31.1 ± 0.64	6.7 ± 0.18	0.93 ± 0.03	0.84 ± 0.04
R403Q (n=9)	33.7 ± 1.7	7.4 ± 0.137	1.22 ± 0.08*	1.09 ± 0.04**

Values are expressed as mean ± standard error of the mean.
*p < 0.001, **p < 0.0001 R403Q vs. WT.

Figure 1. Single doses of CK-3773274 do not affect heart rate in either WT (A) or R403Q (B) mice

A

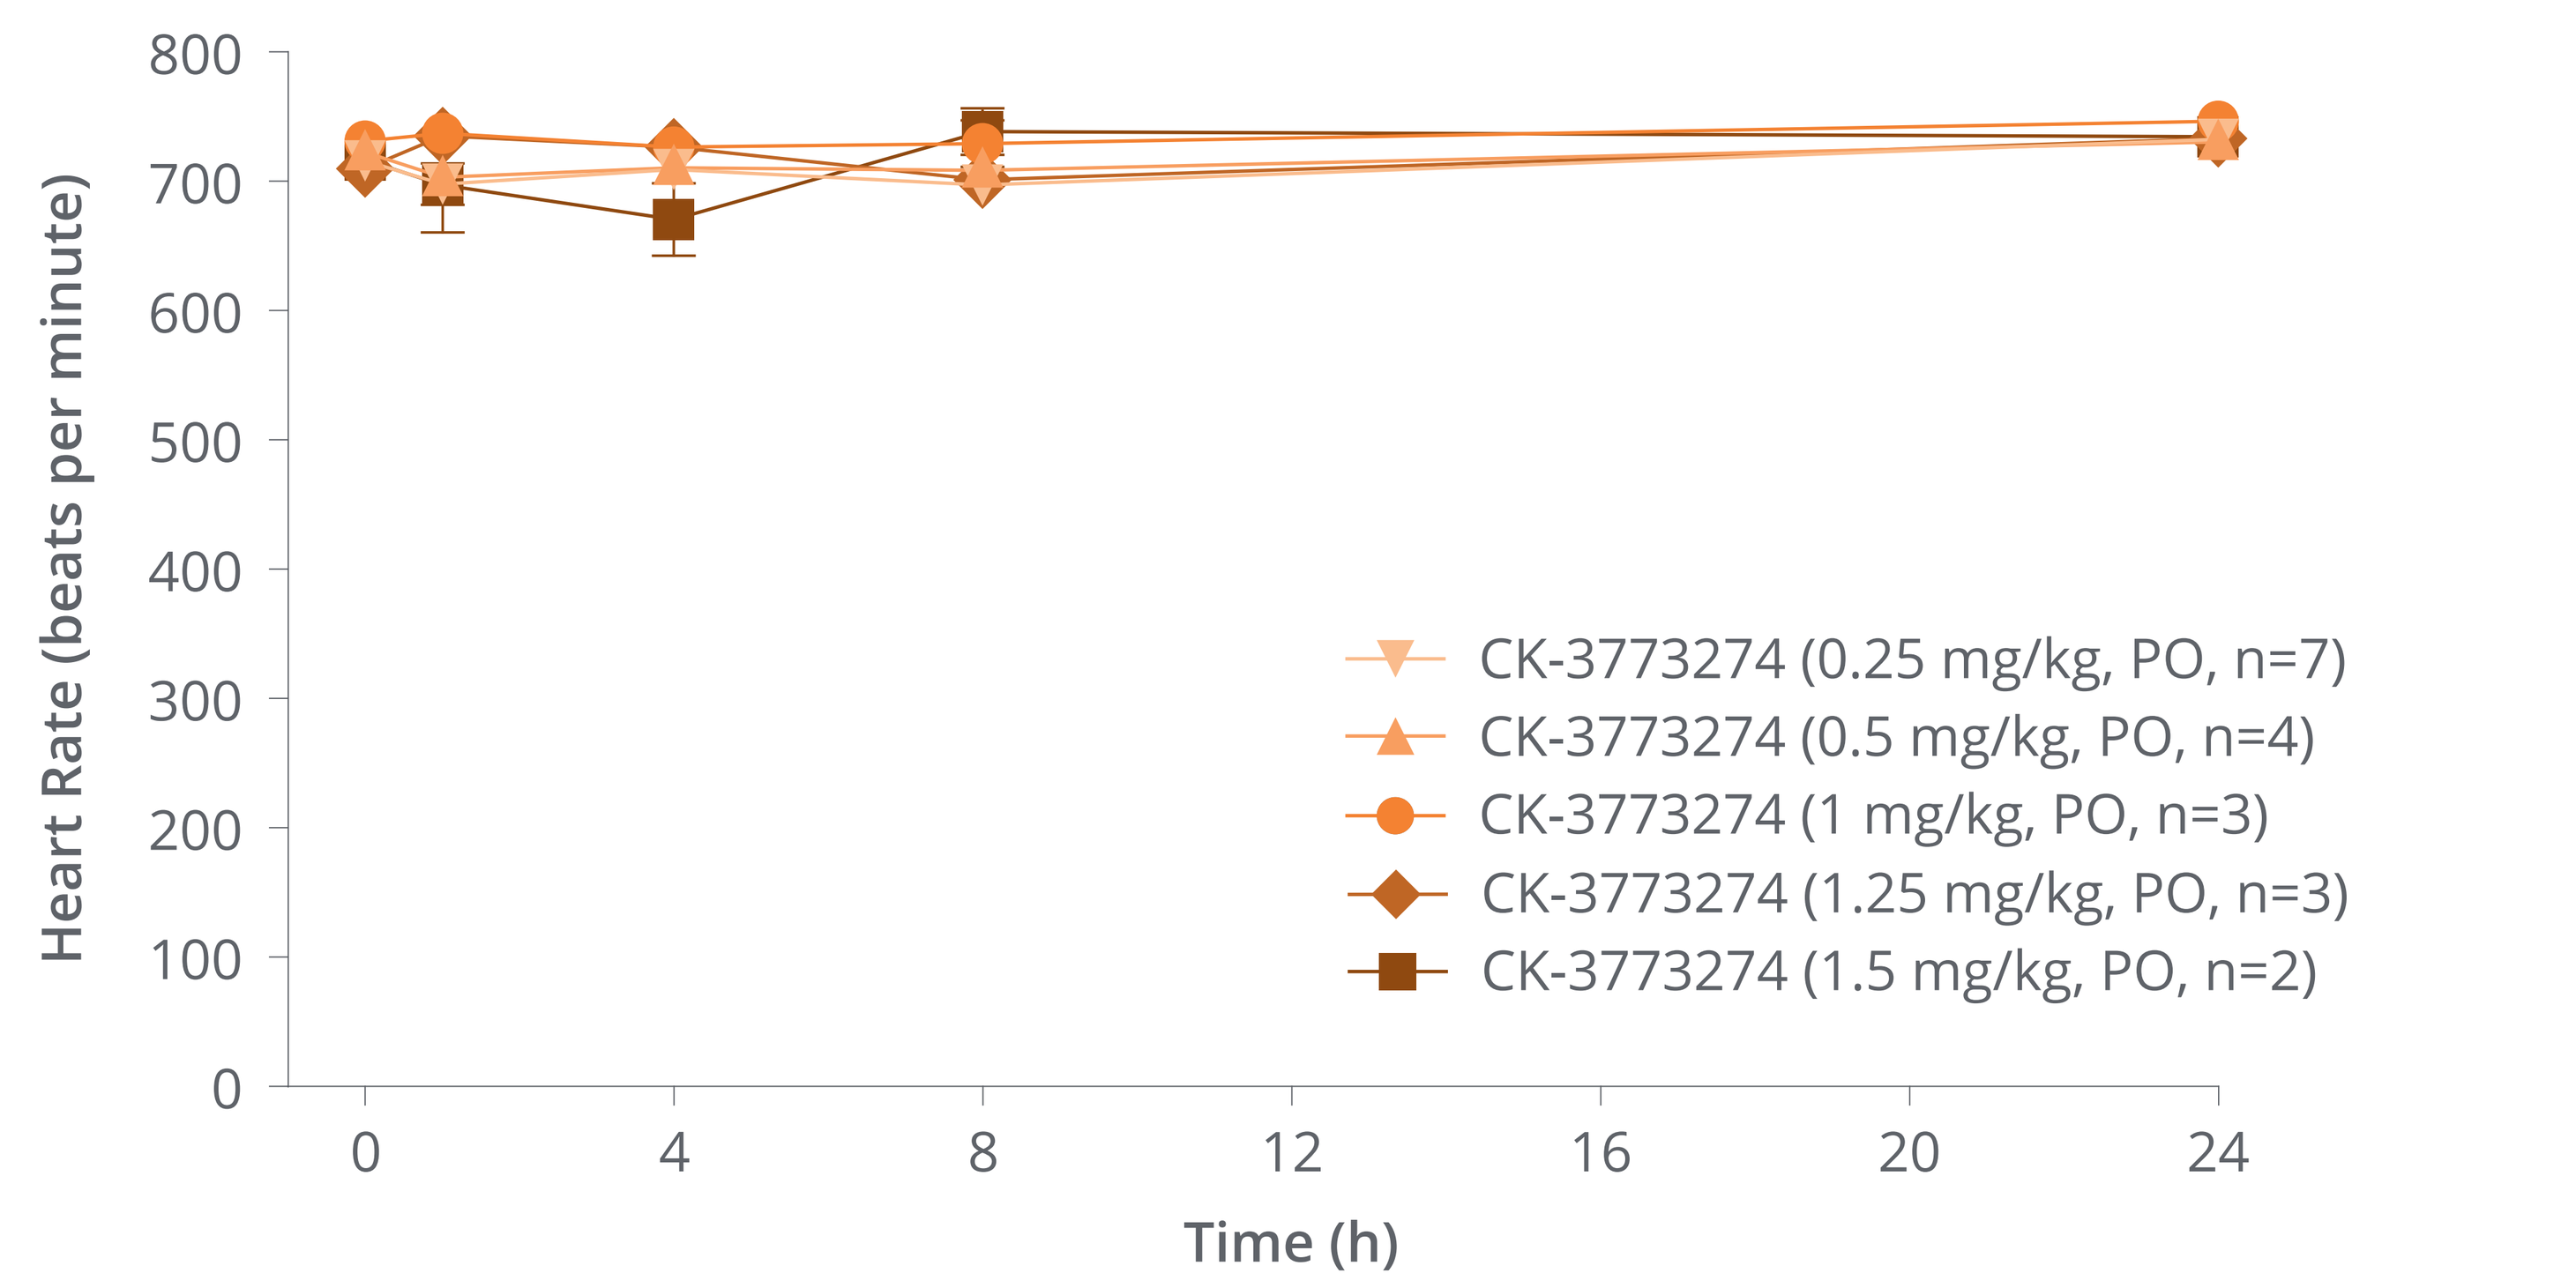
WT Mice – Heart Rate
Heart Rate (beats per minute)
Time (h)
CK-3773274 (0.25 mg/kg, PO, n=7)
CK-3773274 (0.5 mg/kg, PO, n=4)
CK-3773274 (1 mg/kg, PO, n=3)
CK-3773274 (1.25 mg/kg, PO, n=3)
CK-3773274 (1.5 mg/kg, PO, n=2)

B

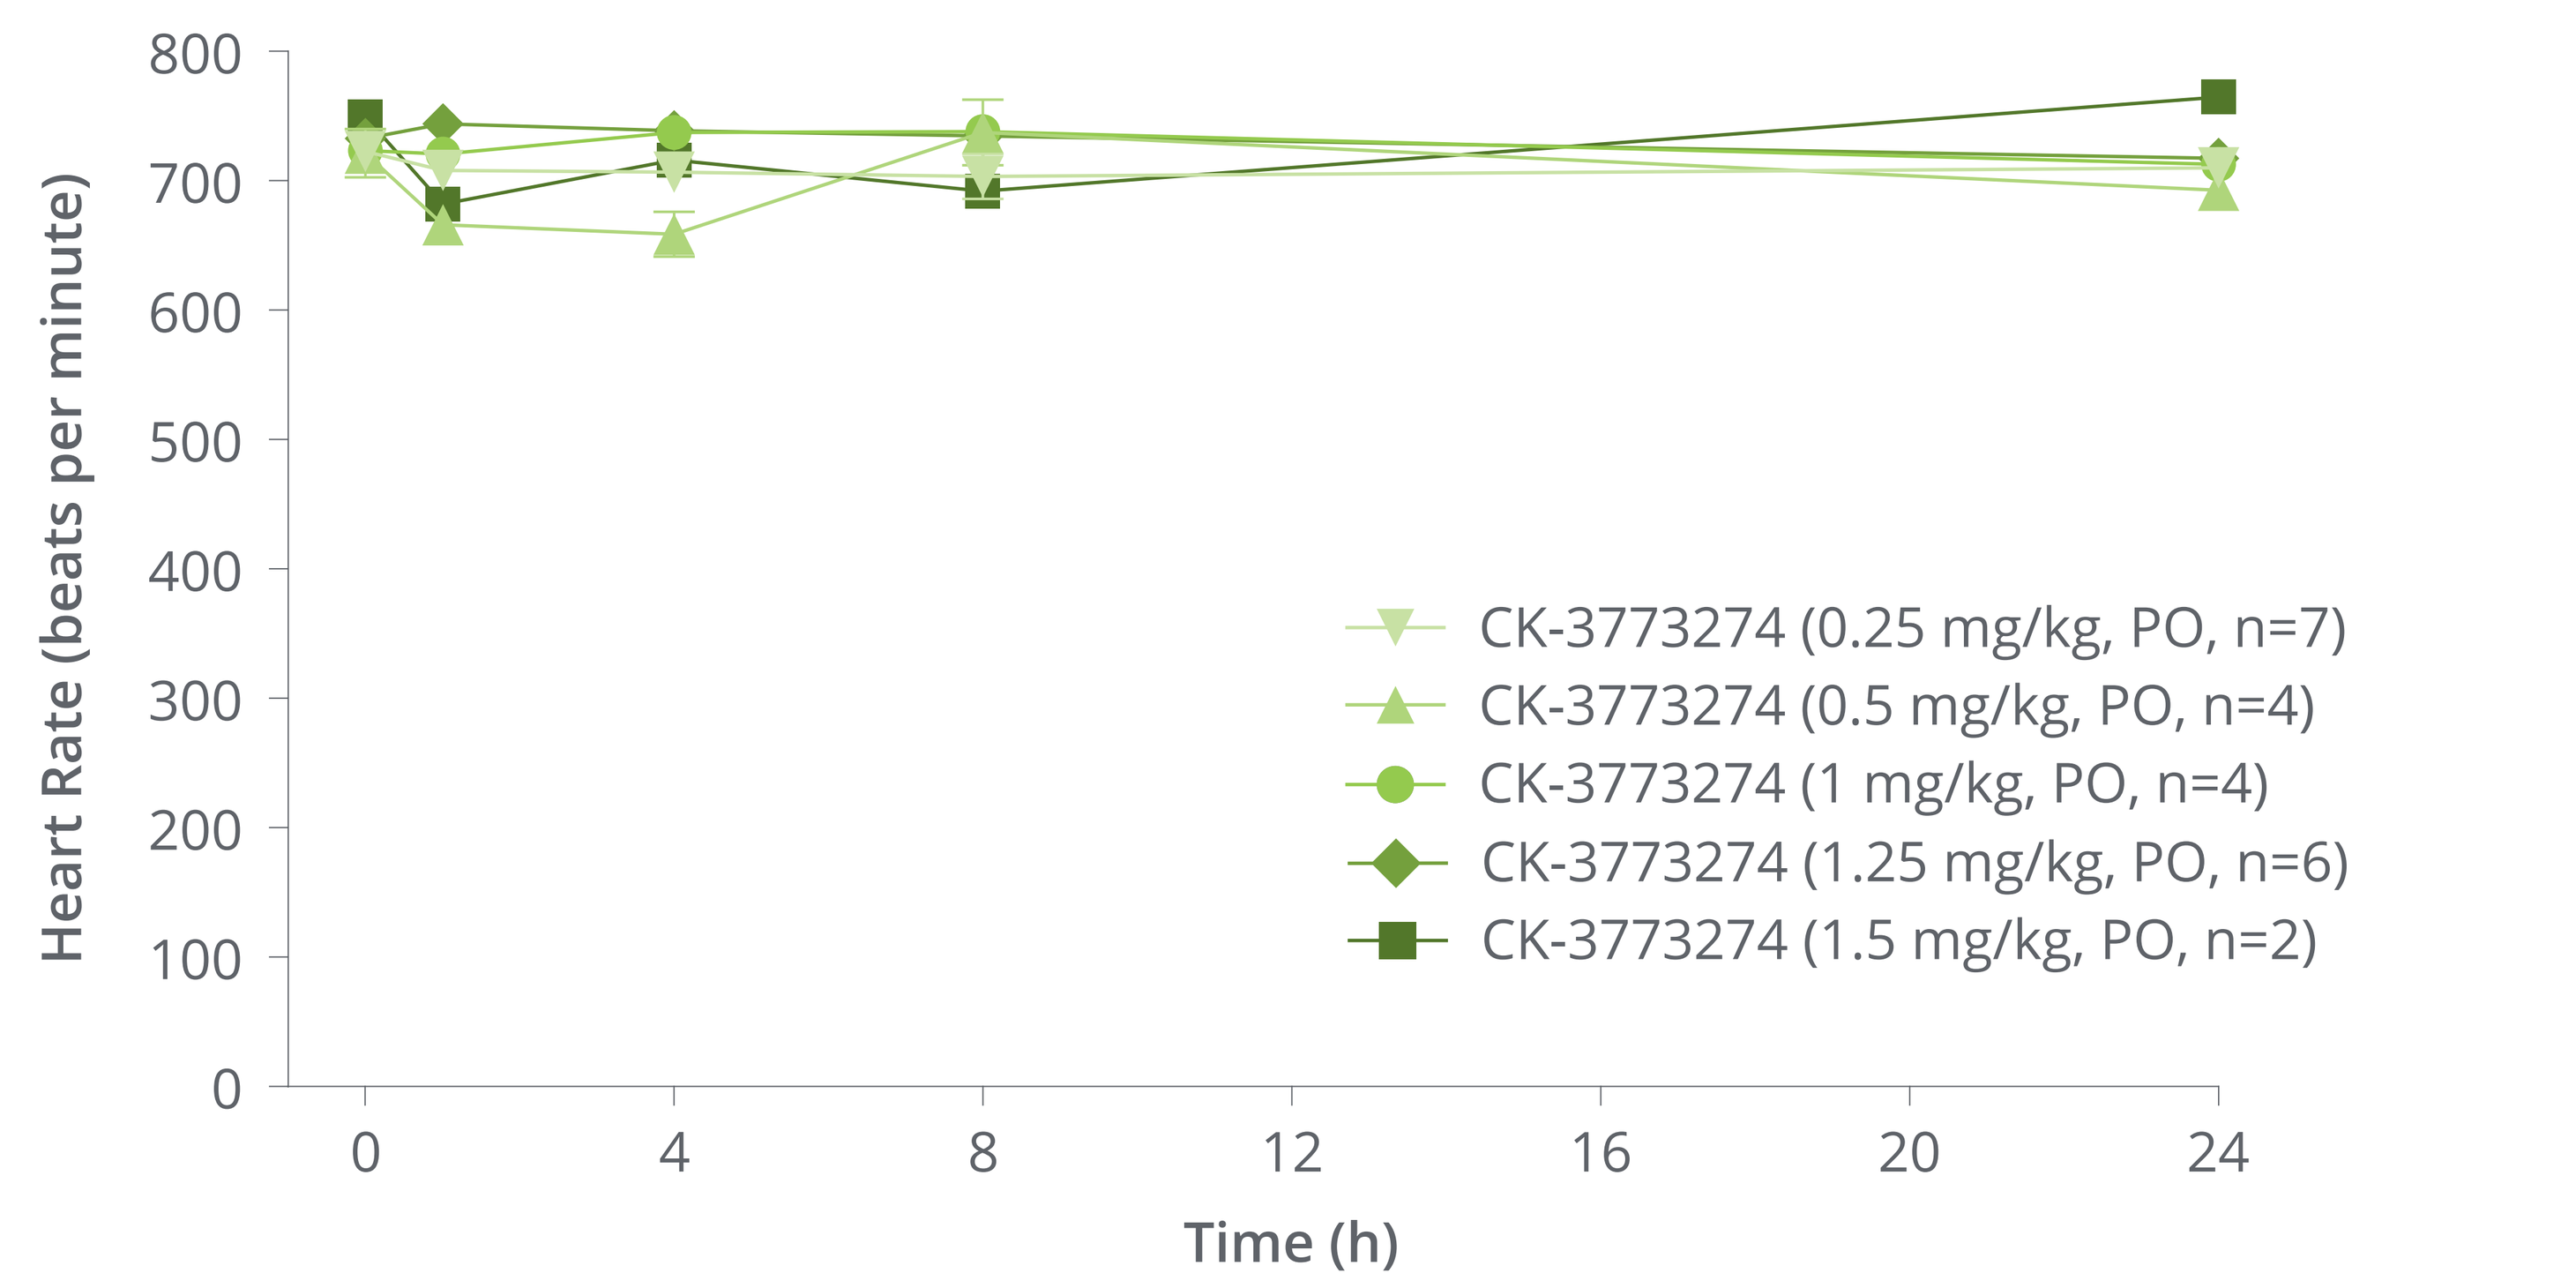
R403Q Mice – Heart Rate
Heart Rate (beats per minute)
Time (h)
CK-3773274 (0.25 mg/kg, PO, n=7)
CK-3773274 (0.5 mg/kg, PO, n=4)
CK-3773274 (1 mg/kg, PO, n=4)
CK-3773274 (1.25 mg/kg, PO, n=6)
CK-3773274 (1.5 mg/kg, PO, n=2)

Values are expressed as mean ± standard error of the mean.

Figure 2. CK-3773274 decreased fractional shortening in a dose-related manner in WT (A) and R403Q (B) mice

A

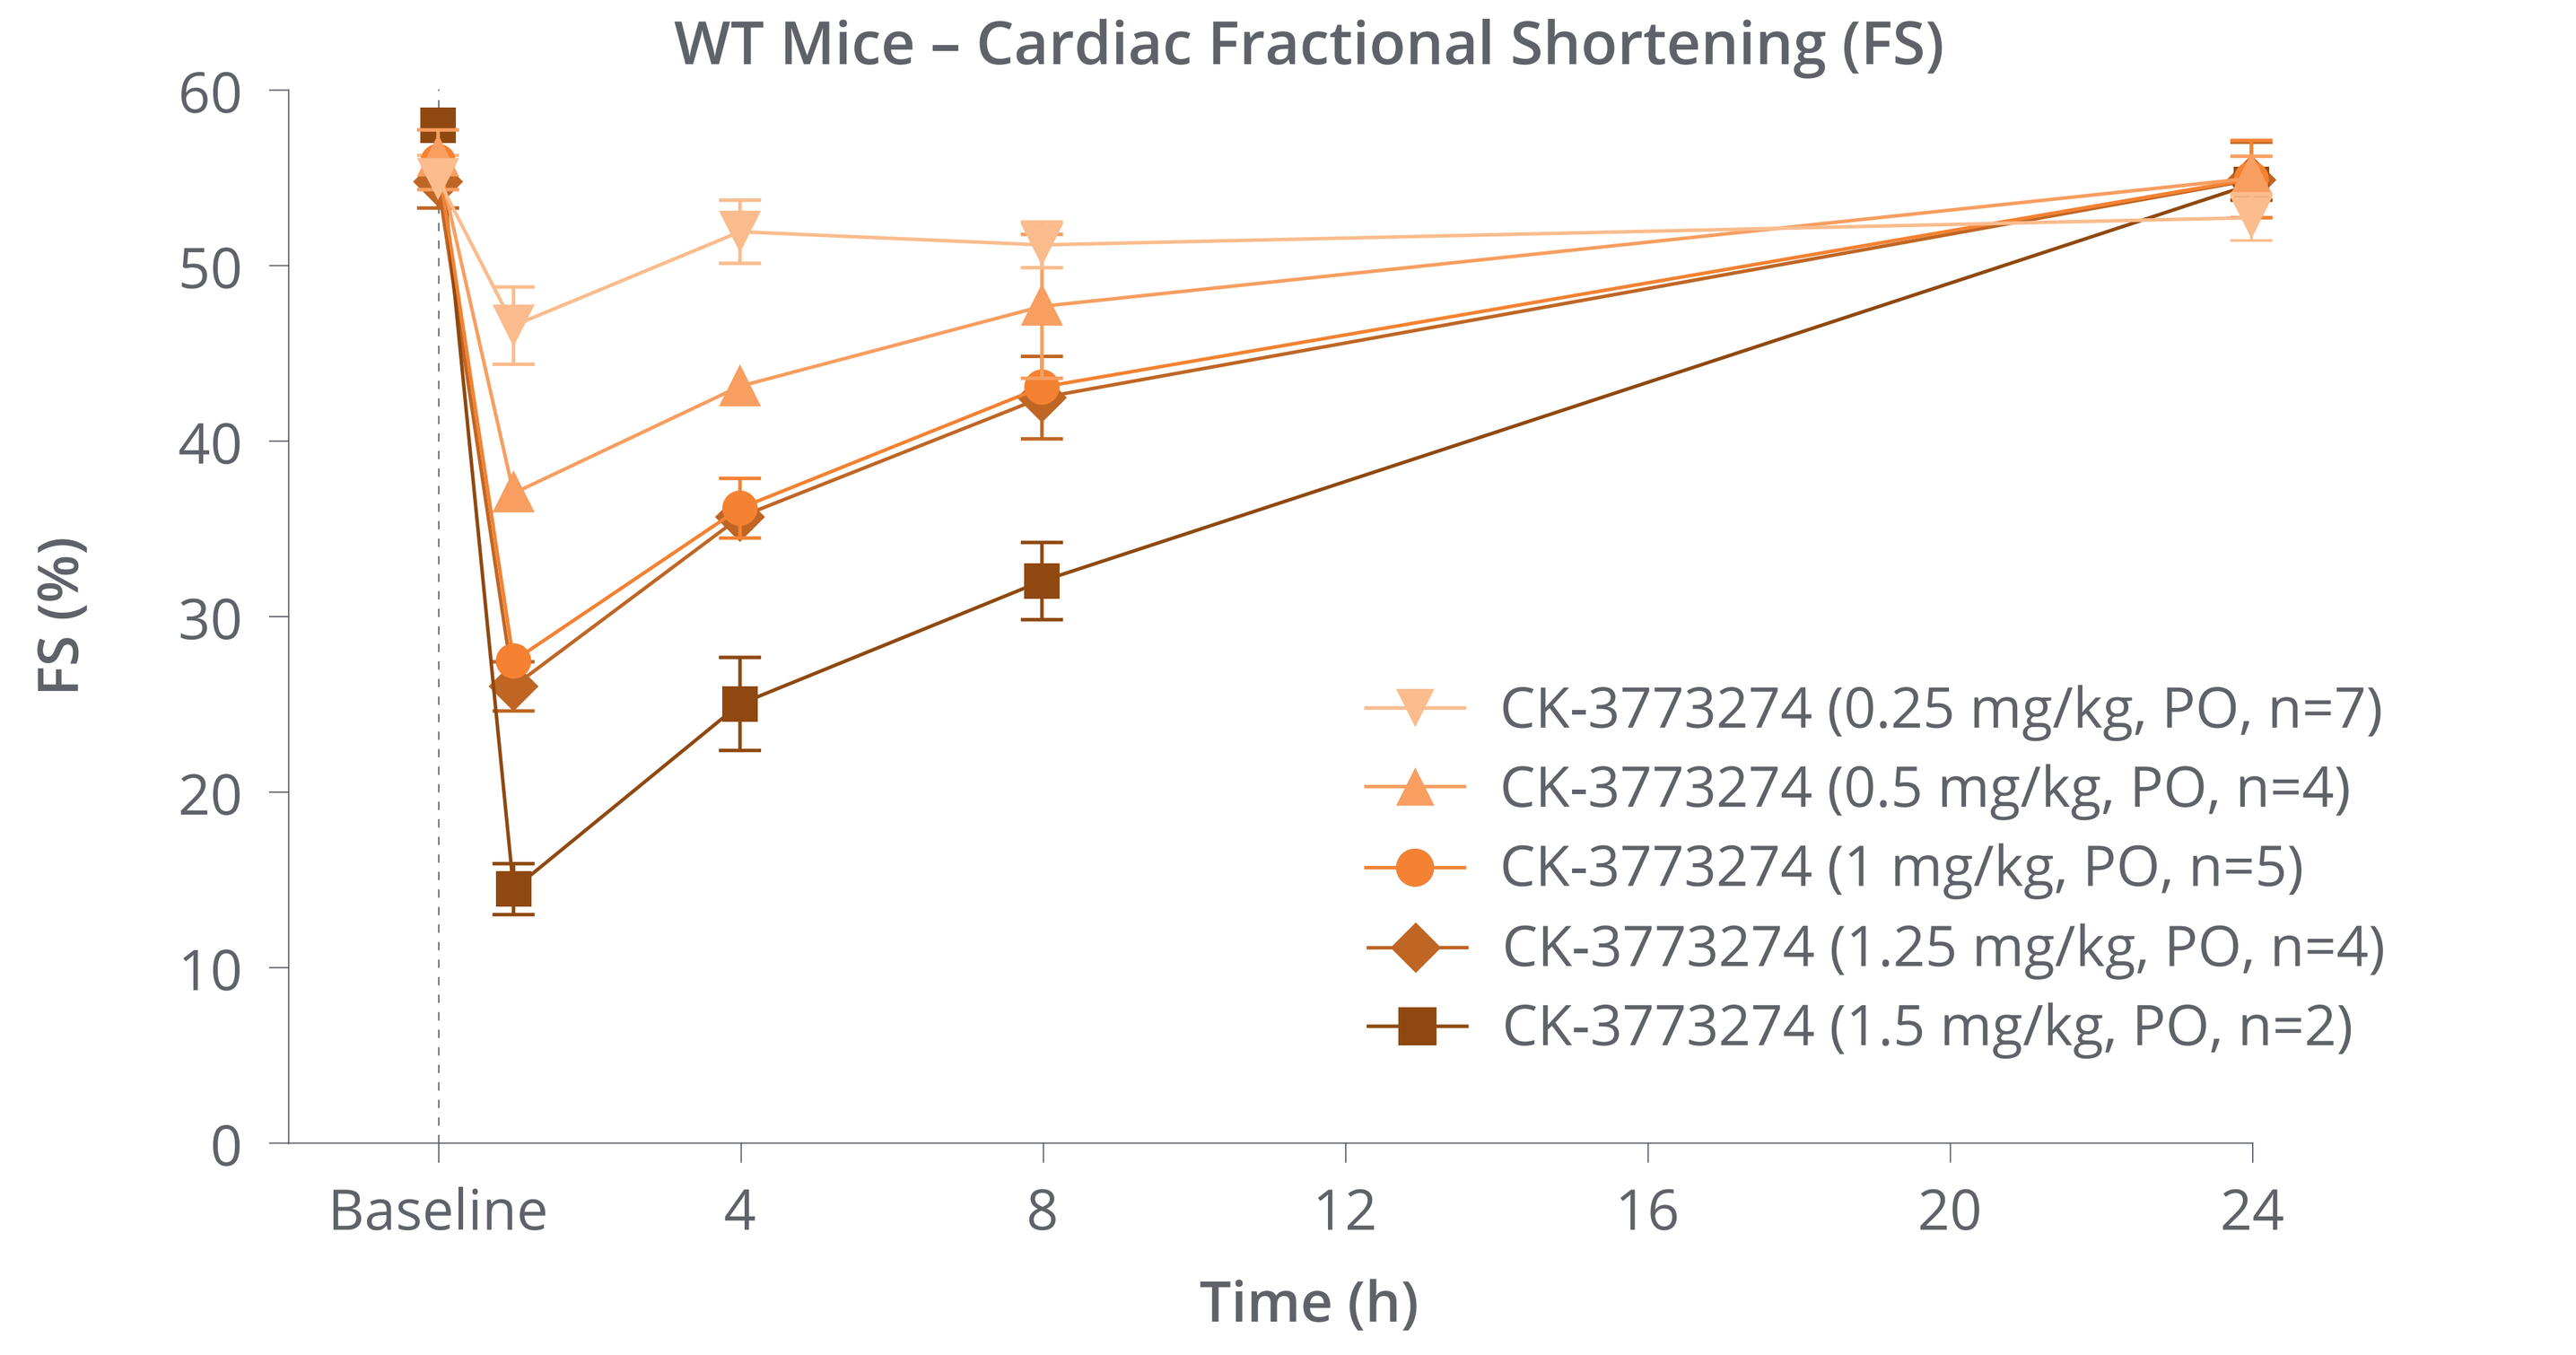
WT Mice – Cardiac Fractional Shortening (FS)
FS (%)
Time (h)
CK-3773274 (0.25 mg/kg, PO, n=7)
CK-3773274 (0.5 mg/kg, PO, n=4)
CK-3773274 (1 mg/kg, PO, n=5)
CK-3773274 (1.25 mg/kg, PO, n=4)
CK-3773274 (1.5 mg/kg, PO, n=2)

Baseline FS vs. CK-3773274 by dose and time

Dose (mg/kg)	Baseline	1 h	4 h	8 h	24 h
0.25	54.9 ± 0.9 % –	46.6 ± 2.2 % p < 0.01	51.9 ± 1.8 % NS	51.2 ± 1.3 % NS	52.7 ± 1.3 % NS
0.5	56.0 ± 1.7 % –	37.0 ± 1.0 % p <0.0001	43.1 ± 0.8 % p < 0.001	47.7 ± 4.1 % p < 0.05	54.9 ± 1.2 % NS
1	55.9 ± 1.1 % –	27.5 ± 0.8 % p < 0.0001	36.2 ± 1.7 % p < 0.0001	43.1 ± 1.1 % p < 0.0001	54.9 ± 2.2 % NS
1.25	54.8 ± 1.5 % –	26.0 ± 1.4 % p < 0.0001	35.7 ± 0.7 % p < 0.0001	42.5 ± 2.3 % p < 0.001	54.9 ± 2.2 % NS
1.5	57.9 ± 0.7 % –	14.5 ± 1.5 % p < 0.0001	25.0 ± 2.7 % p < 0.0001	32.0 ± 2.2 % p < 0.0001	54.6 ± 0 % NS

B

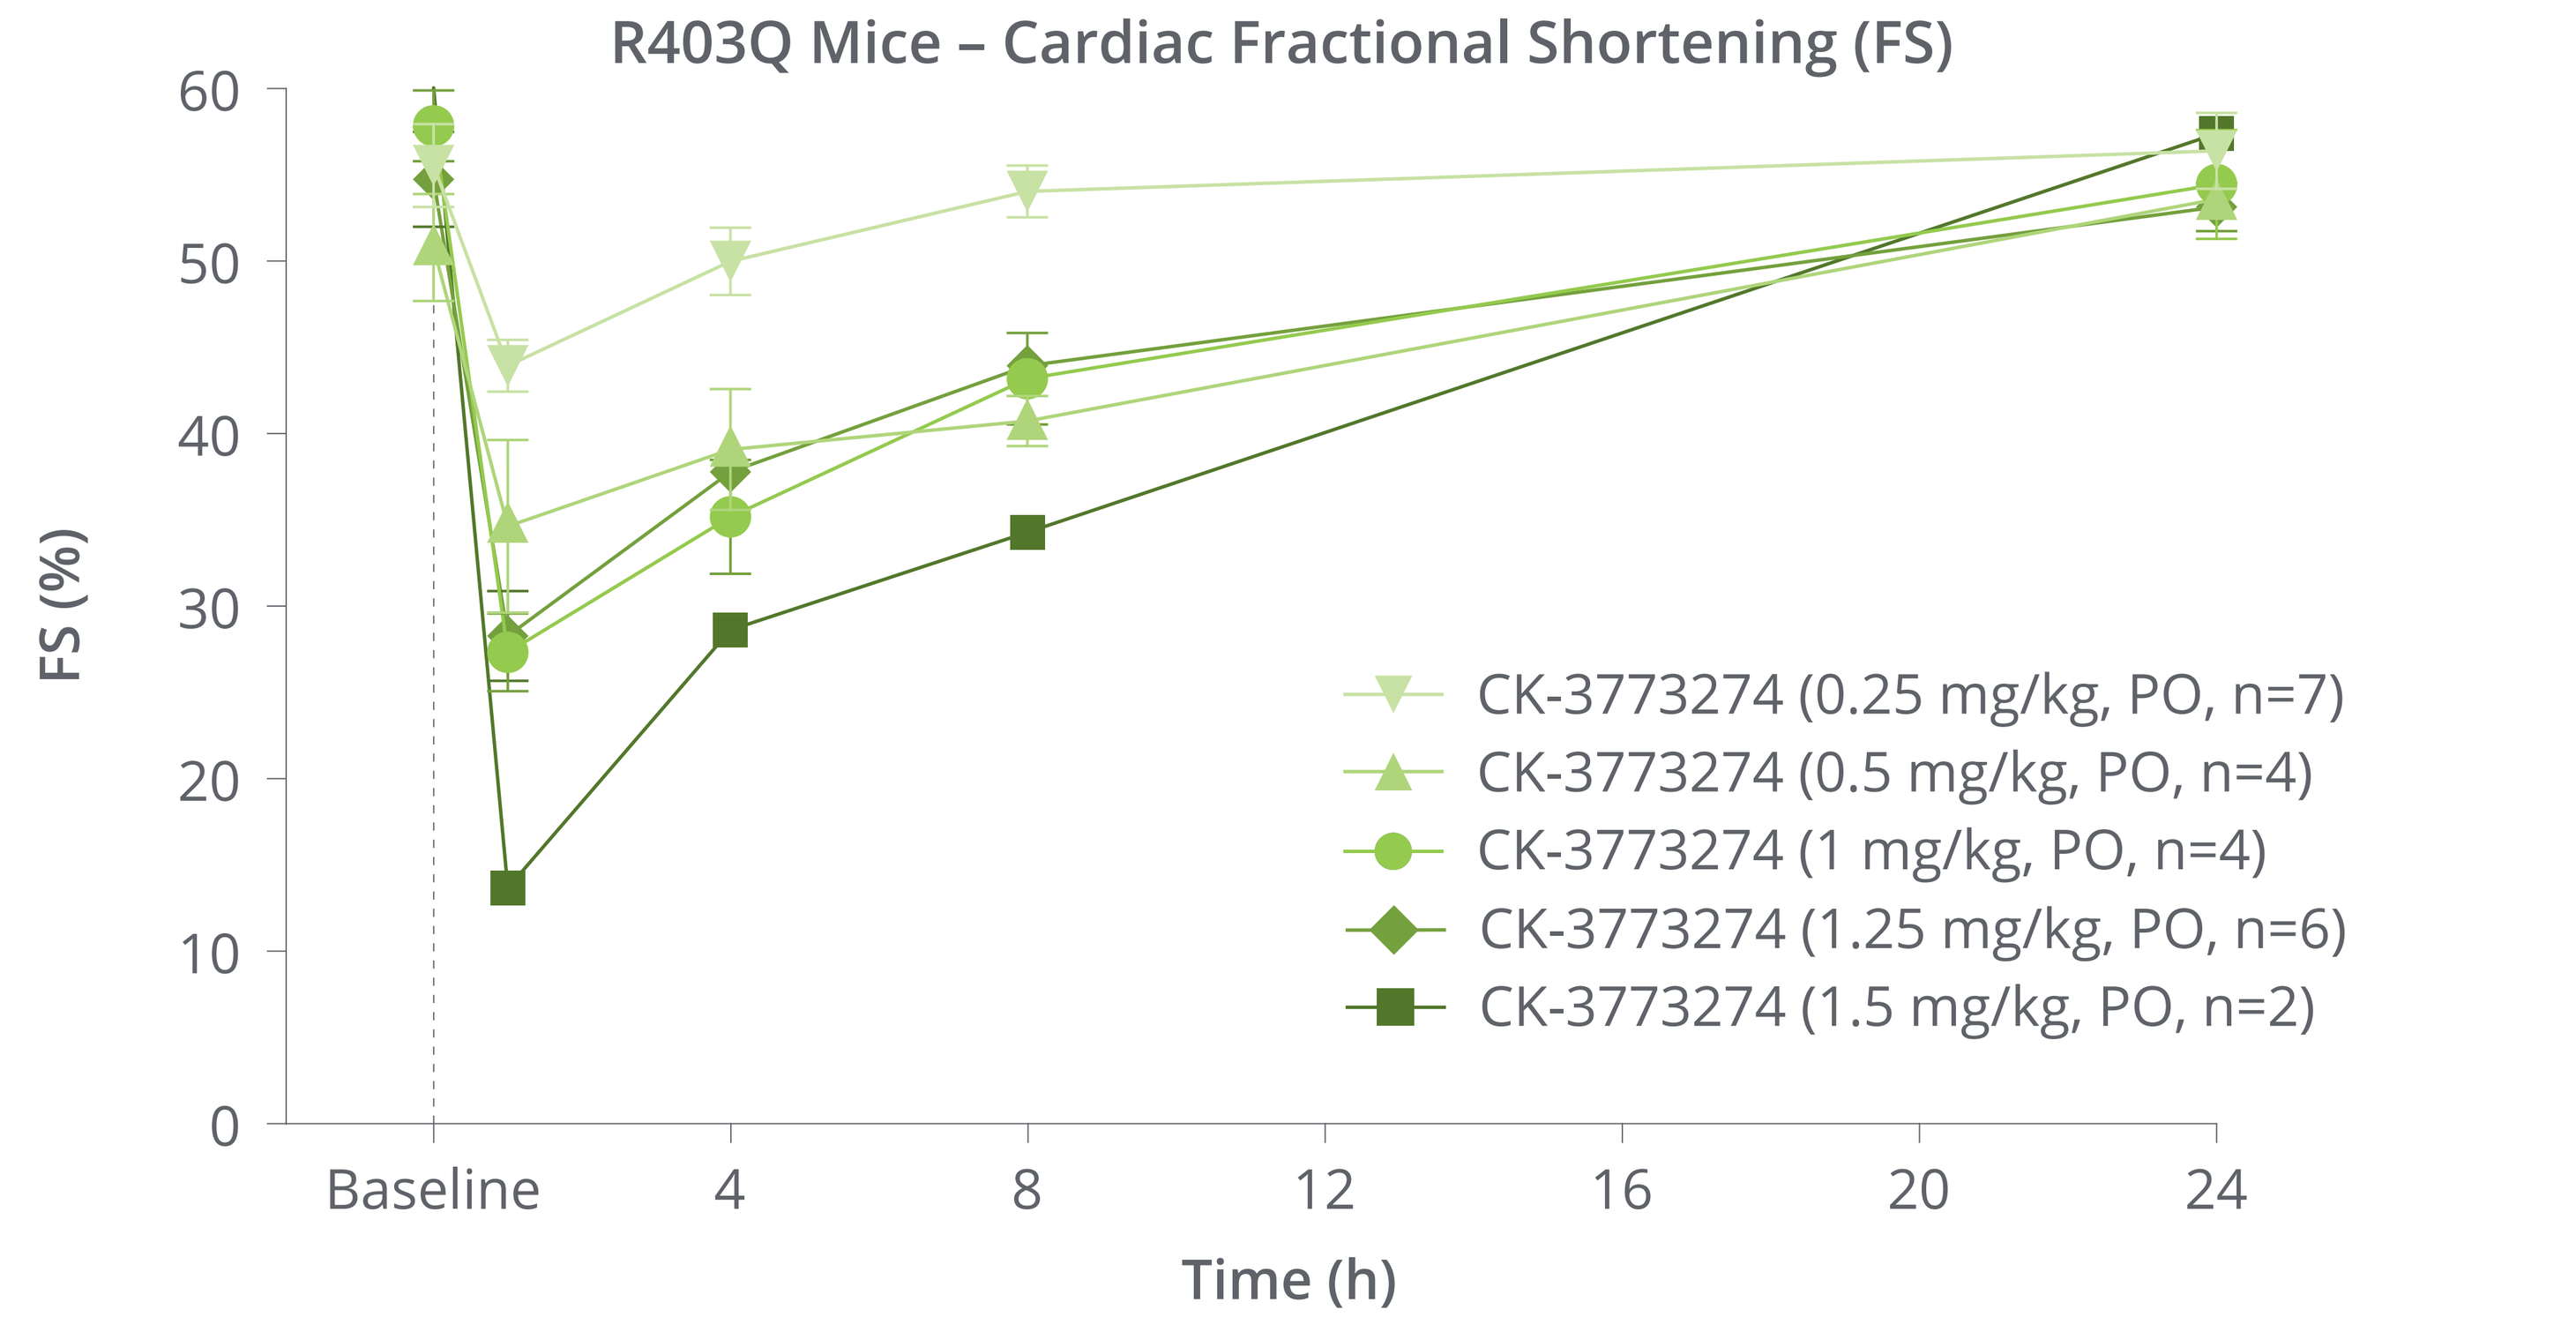
R403Q Mice – Cardiac Fractional Shortening (FS)
FS (%)
Time (h)
CK-3773274 (0.25 mg/kg, PO, n=7)
CK-3773274 (0.5 mg/kg, PO, n=4)
CK-3773274 (1 mg/kg, PO, n=4)
CK-3773274 (1.25 mg/kg, PO, n=6)
CK-3773274 (1.5 mg/kg, PO, n=2)

Baseline FS vs. CK-3773274 by dose and time

Dose (mg/kg)	Baseline	1 h	4 h	8 h	24 h
0.25	55.5 ± 2.4 % –	43.9 ± 1.5 % p < 0.0001	49.9 ± 2.0 % NS	54.0 ± 1.5 % NS	56.4 ± 2.2 % NS
0.5	50.8 ± 3.1 % –	34.6 ± 5.0 % p < 0.0001	39.1 ± 3.5 % p < 0.001	40.7 ± 1.5 % p < 0.01	53.6 ± 0.7 % NS
1	57.8 ± 2.1 % –	27.3 ± 2.2 % p < 0.0001	35.2 ± 3.3 % p < 0.0001	43.2 ± 2.7 % p < 0.001	54.4 ± 3.1 % NS
1.25	54.7 ± 2.8 % –	28.3 ± 2.6 % p < 0.0001	37.8 ± 1.1 % p < 0.0001	43.9 ± 1.2 % p < 0.01	53.1 ± 1.4 % NS
1.5	60.1 ± 1.5 % –	13.7 ± 0.6 % p < 0.0001	28.6 ± 0.9 % p < 0.0001	34.3 ± 0.5 % p < 0.0001	57.4 ± 0 % NS

At all dose levels, FS returned to baseline levels 24 hours after dosing. Statistical comparisons by %FS baseline levels vs. CK-3773274 dose level and time are presented in the accompanying table. Values are expressed as mean ± standard error of the mean.
NS, not significant.

Figure 3. CK-3773274 decreased fractional shortening to a similar extent in WT and R403Q mice in a concentration-related manner

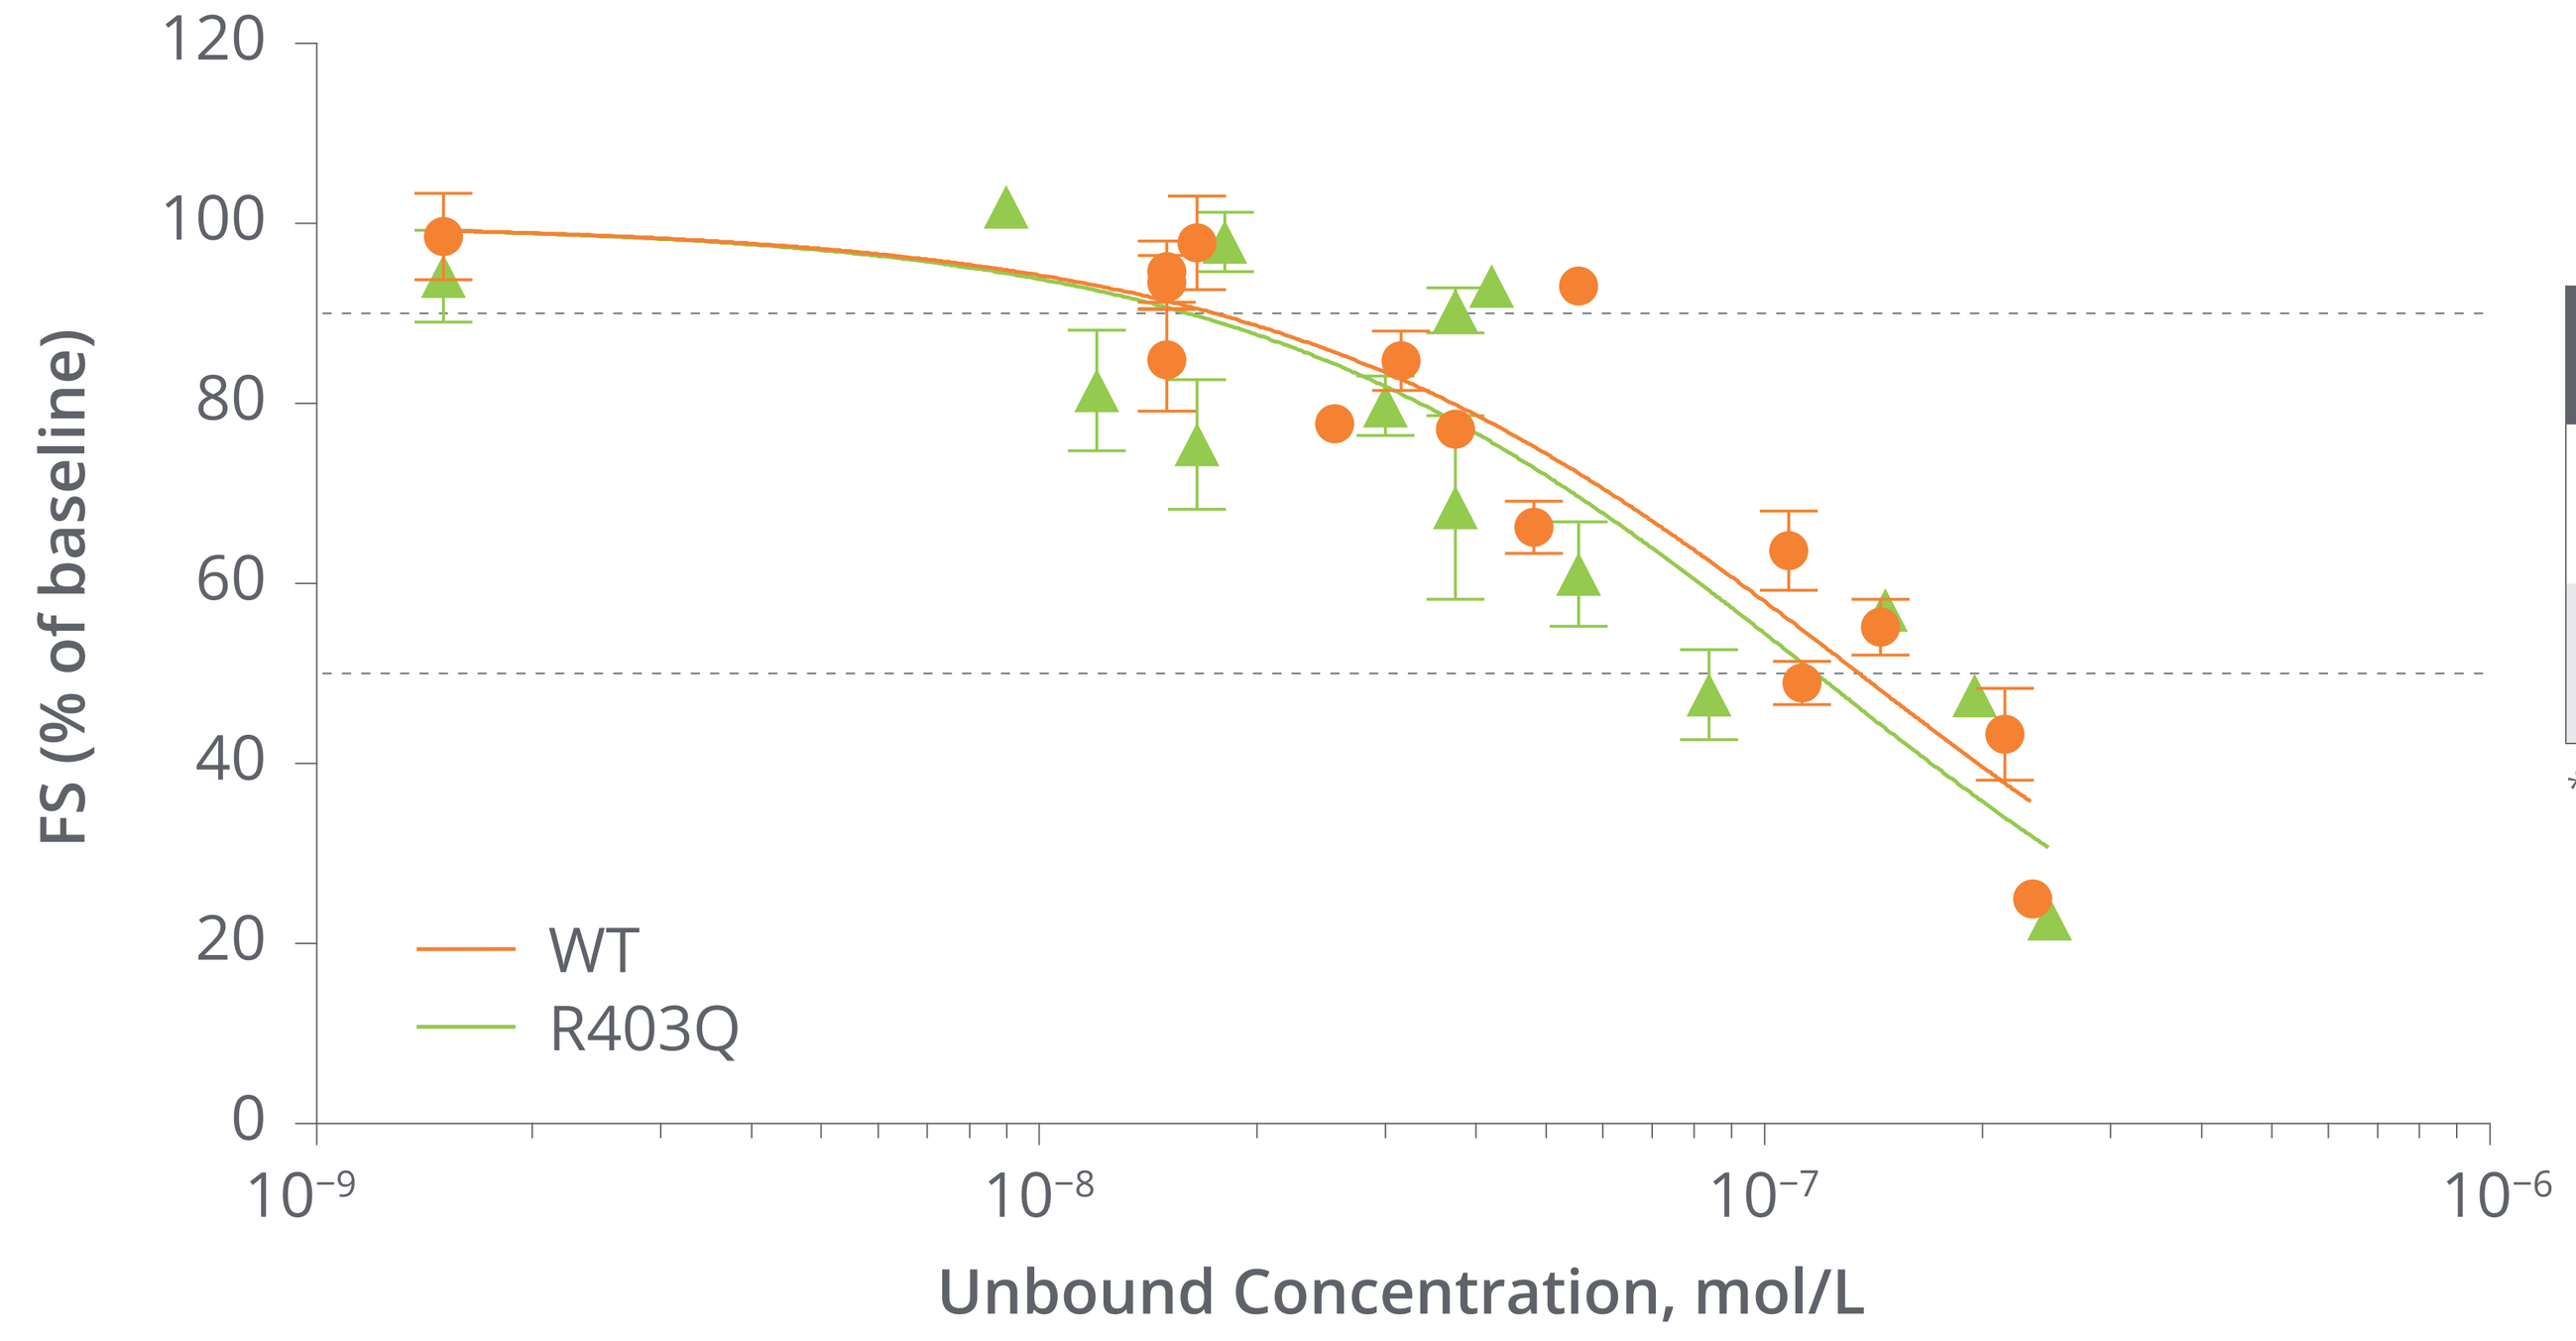
FS (% of baseline)
Unbound Concentration, mol/L
WT
R403Q

	WT	R403Q
$IC_{10,\ unbound*}$ (nM)	18	16.5
$IC_{50,\ unbound*}$ (nM)	135	117

* Mouse Plasma Protein Binding: 85%

Values are expressed as mean ± standard error of the mean.

SUMMARY

- **Single oral dose administration of the cardiac myosin inhibitor CK-3773274 reduced fractional shortening in a dose- and concentration-related manner in the genetic R403Q mouse model of hypertrophic cardiomyopathy**
 - **CK-3773274 achieved a 10% reduction (IC_{10}) in contractility at unbound nanomolar concentrations**
 - **A >7× difference between the IC_{50} and IC_{10} in both wild type and R403Q mice may translate into a measured, gradual on-target effect**
- **Cardiac myosin inhibition may be a viable approach to reduce underlying hypercontractility of the cardiac sarcomere in hypertrophic cardiomyopathies**

References

1. Green, et al. *Science*. 2016;351:617–621.
2. Malik, et al. *Science*. 2011;331:1439–1443.

Disclosures

All authors were employees of Cytokinetics at the time of the study and were compensated financially for their work
This study was funded by Cytokinetics, Inc.

Presented at the Basic Cardiovascular Sciences 2019 Scientific Sessions; July 29–August 1, 2019; Boston, MA, USA