210

A First-in-Human Study of the Selective Cardiac Myosin Inhibitor, CK-3773274

Laura A. Robertson,[1] Danielle R. Armas,[2] Edward Robbie,[1] Anna Osmukhina,[1] Hanbin Li,[3] Fady I. Malik,[1] Scott D. Solomon[4]

[1]Cytokinetics, Inc., South San Francisco, CA; [2]Celerion, Inc., Tempe, AZ; [3]Certara, Inc., Menlo Park, CA; [4]Brigham and Women's Hospital, Boston, MA

INTRODUCTION

- Hypercontractility of the cardiac sarcomere appears to underlie pathological hypertrophy, outflow obstruction, and fibrosis in select genetic hypertrophic cardiomyopathies (HCM)[1]
- CK-3773274 (CK-274) decreases cardiac contractility in preclinical models by directly binding to the cardiac myosin motor domain and inhibiting the myosin ATPase, stabilizing the motor in a non–force-producing state[2]
- The preclinical pharmacologic and pharmacokinetic properties of CK-274 were optimized to facilitate the potential ease of dose titration and related therapeutic window in patients[2]
- In preclinical models of cardiac function, including the R403Q mouse model of HCM, CK-274 reduced cardiac contractility in a predictable dose and exposure dependent fashion[3]
- Here we report on the first-in-human experience of single and multiple dosing with CK-274

STUDY OBJECTIVES

- **Primary**
 - Determine the safety and tolerability of single and multiple ascending doses of CK-274 administered orally to healthy adult participants
- **Secondary**
 - Evaluate the pharmacokinetics (PK) of CK-274 following single and multiple oral doses
 - Identify dose(s) of CK-274 that reduce left ventricular ejection fraction (LVEF)
 - Describe the pharmacokinetic-pharmacodynamic (PK/PD) relationship between CK-274 and cardiac function
- This study was not designed to identify a maximum tolerated dose

METHODS

Single Ascending Dose (SAD)

- The SAD cohort design incorporated a sentinel group
- PK sampling occurred on Days 1, 2, 3, 4, 5, and 10 following dosing
- Echocardiography was performed pre-dose, at 1.5 hours, 4 hours, or 6 hours depending on cohort, and at 24 hours following dosing

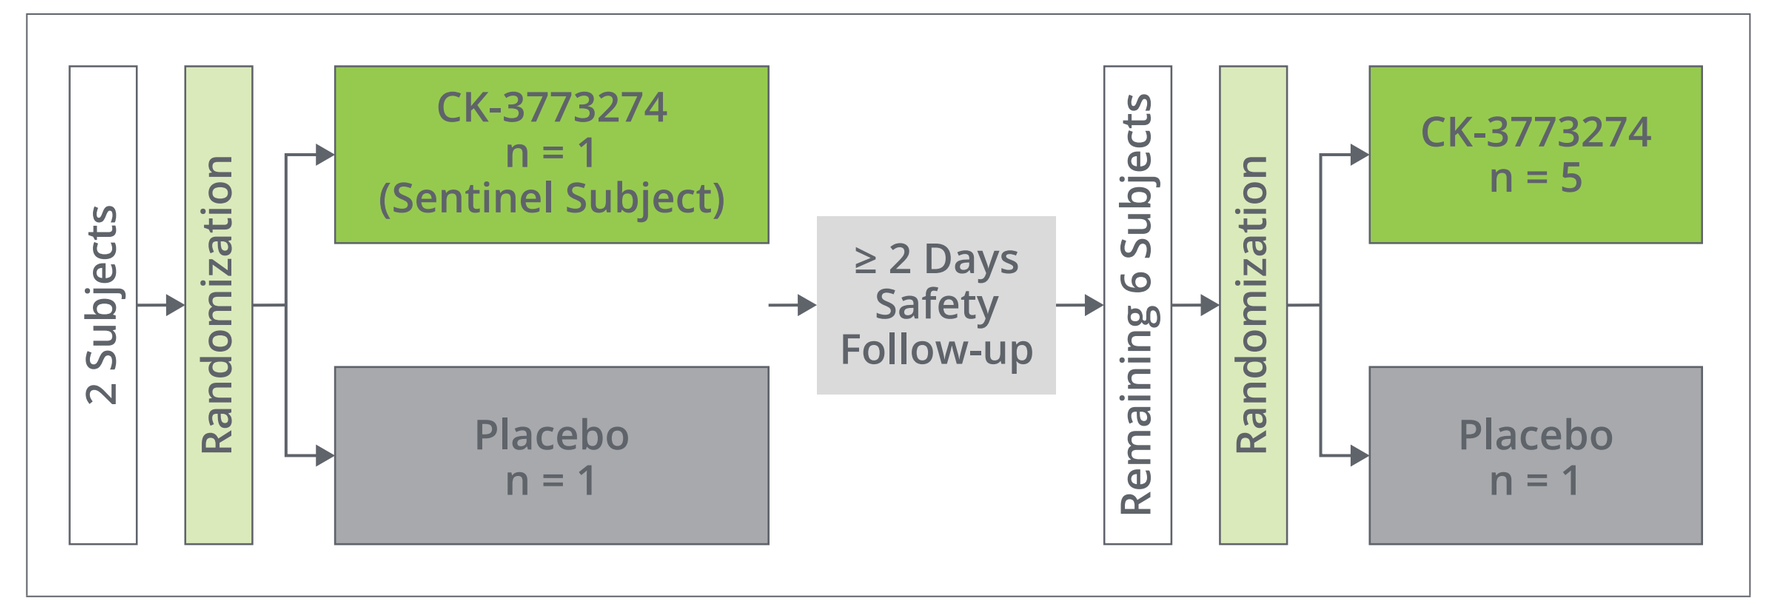
2 Subjects
Randomization
CK-3773274
n = 1
(Sentinel Subject)
Placebo
n = 1
≥ 2 Days
Safety
Follow-up
Remaining 6 Subjects
Randomization
CK-3773274
n = 5
Placebo
n = 1

Multiple Ascending Dose (MAD)

- The MAD cohorts started following completion of SAD Arm D
- Participants received study drug once daily for 14 days (5 and 10 mg) or 17 days (7.5 mg)
- PK sampling occurred daily during dosing, daily for 3 days after the end of dosing, with a final sample 7 days after the completion of dosing
- Echocardiography was performed pre-dose, at Days 2, 4, 9, 14, or 17 depending on cohort, and at 24 hours following dosing

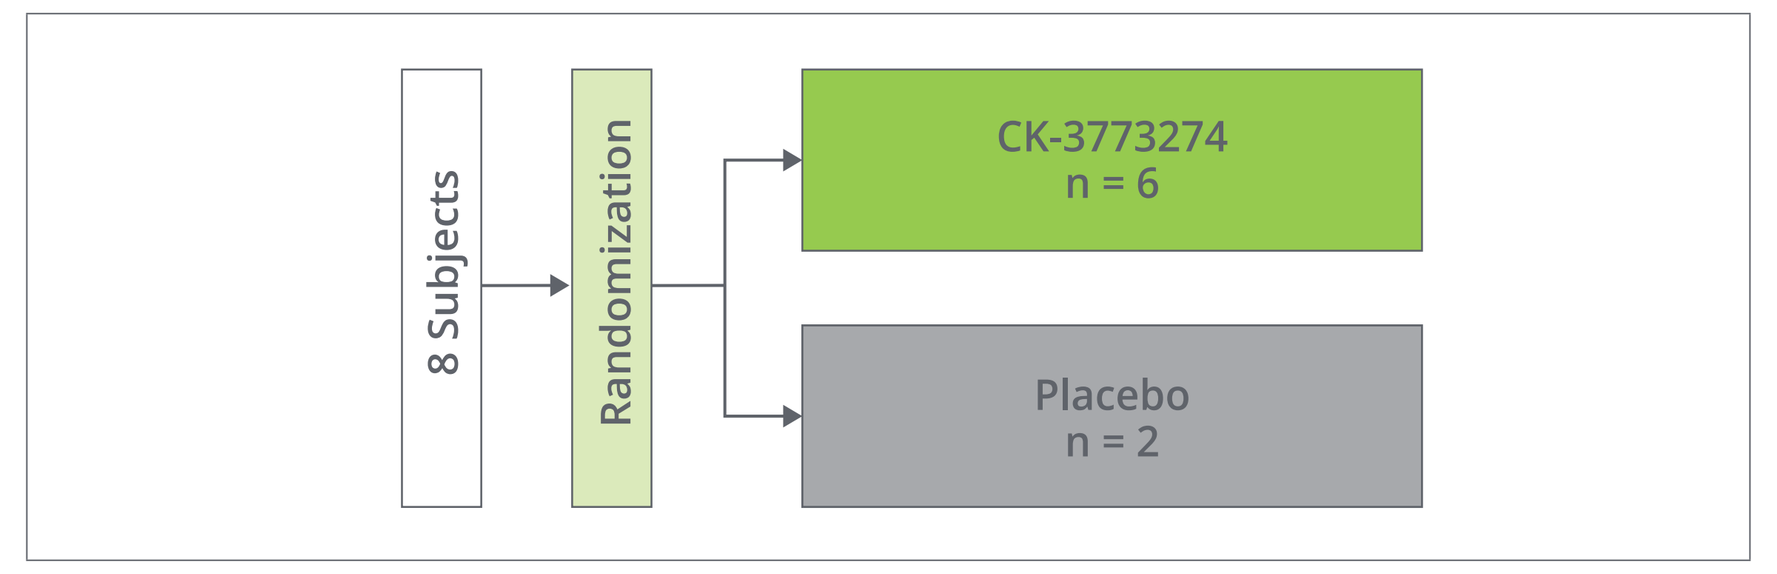
8 Subjects
Randomization
CK-3773274
n = 6
Placebo
n = 2

- Additional cohorts to assess the effect of CYP2D6 poor metabolizer genotype and food on the PK of CK-274 were also performed but are not reported here

Key Inclusion Criteria (SAD/MAD Cohorts)

- Males and females (of non-childbearing potential) between 18 and 55 years of age, inclusive
- Body weight > 55.0 kg and body mass index within 18.0 to 32.0 kg/m^2, inclusive
- Normal cardiac structure and function, or if abnormalities are present, they are deemed not clinically significant
- SAD Arms A–D: LVEF ≥ 60%
- SAD Arms E–G and MAD Arms: LVEF ≥ 65%
- Normal electrocardiogram (ECG) or, if abnormalities are present, they are deemed not clinically significant
- Clinical laboratory findings within normal range
- Does not have CYP2D6 poor metabolizer genotype

Dose Escalation Criteria

- Dose escalation recommendations were provided by the study team and reviewed by an unblinded Dose Level Review Committee (DLRC)
 - The DLRC included an external cardiologist (S. Solomon) with relevant expertise and designated staff members of the study sponsor (or designee) representing clinical science, safety science, clinical pharmacology, and biostatistics
- The following rules were applied to dose escalation decisions:
 - No participants have experienced cardiac serious adverse events (SAEs) related to the study drug
 - No two participants have experienced similar, non-cardiac SAEs in the same organ system that appear to be related to the study drug
 - No two participants treated with CK-274 have a decrease in LVEF > 15% from last pre-dose value (determined by the DLRC)
 - No more than two participants have an LVEF < 50% (unless determined not to be related to the study drug by the DLRC and investigator) and have symptoms of decreased cardiac output
 - No participant has an LVEF < 45% (unless determined not to be related to the study drug by the DLRC and investigator) and has symptoms of decreased cardiac output
 - The investigator and DLRC both approve the escalation and the next dose level based on their clinical judgment

Study Conduct

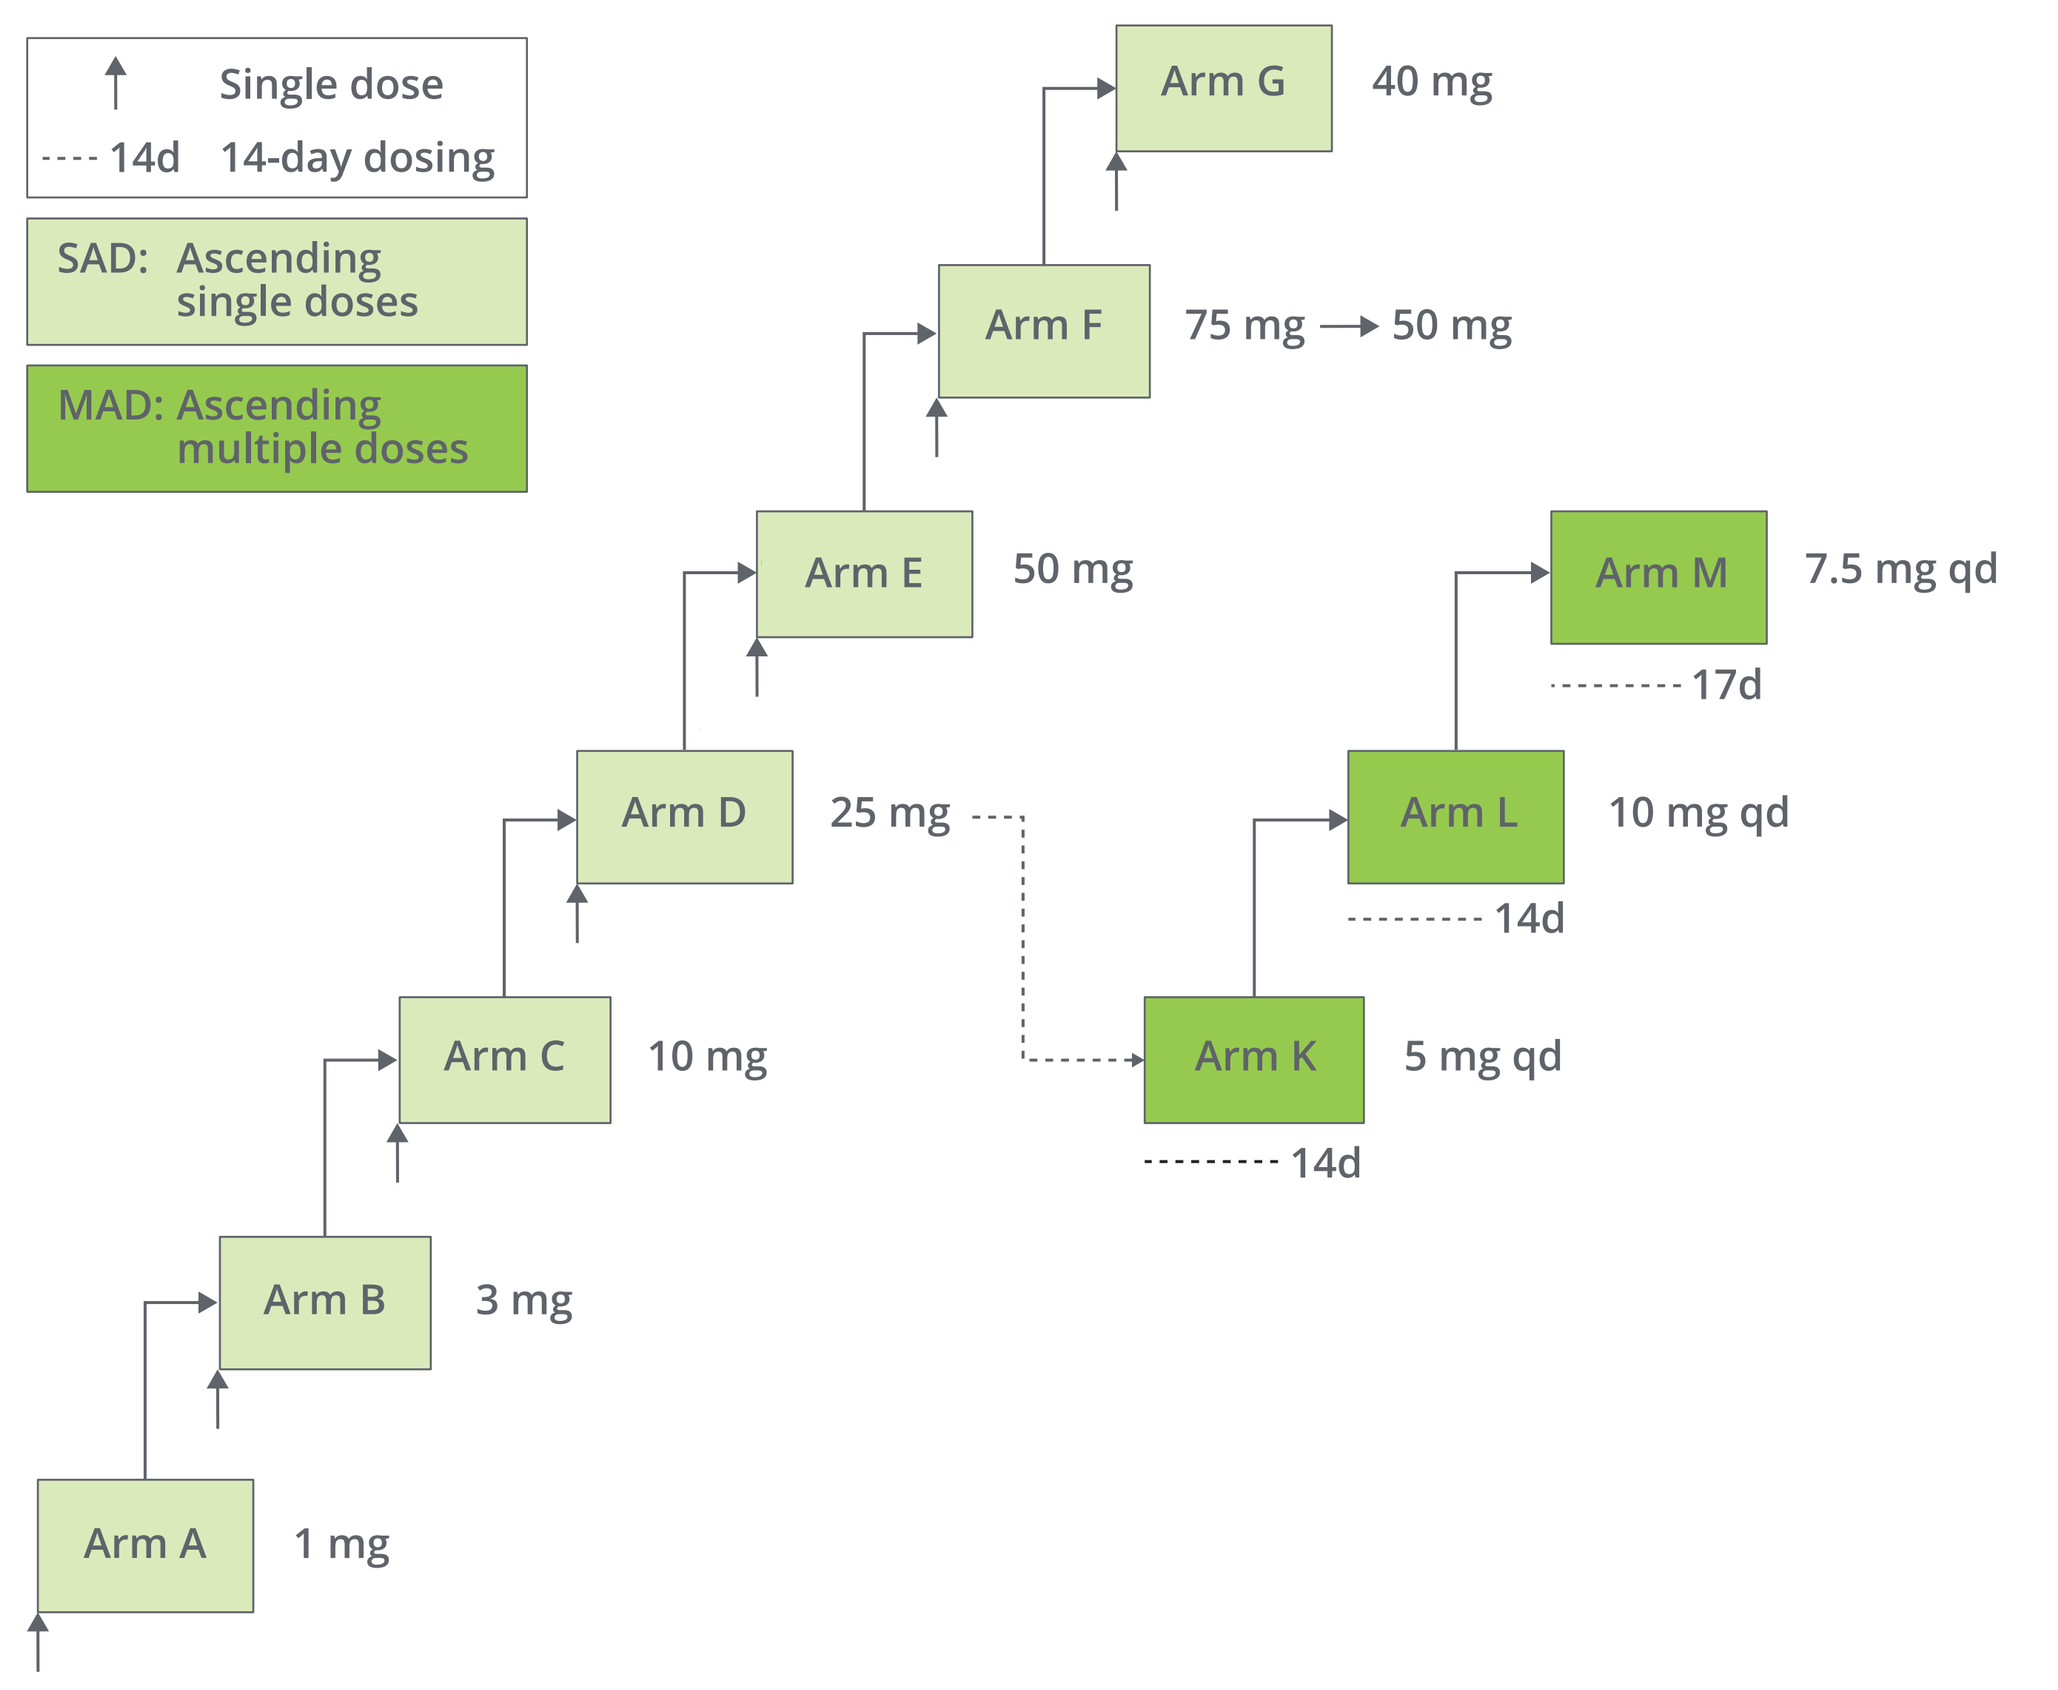
Single dose
14d 14-day dosing
SAD: Ascending single doses
MAD: Ascending multiple doses
Arm A 1 mg
Arm B 3 mg
Arm C 10 mg
Arm D 25 mg
Arm E 50 mg
Arm F 75 mg → 50 mg
Arm G 40 mg
Arm K 5 mg qd
14d
Arm L 10 mg qd
14d
Arm M 7.5 mg qd
17d

- Criteria to stop dose escalation were met in the SAD 75-mg dose cohort
 - One participant in sentinel group had LVEF < 45% at 1.5 hours, asymptomatic and recovered to normal EF at 6 hours
- Criteria to stop dose escalation were met in the MAD 10-mg dose cohort
 - Two participants had LVEF < 50% at 14 days, asymptomatic and recovered to normal EF within 24–48 hours

Stopping criteria were evaluated based on initial read of echocardiogram prior to the final core lab determination.
d, day; MAD, multiple ascending dose; qd, once daily; SAD, single ascending dose.

RESULTS

Baseline Characteristics

Characteristic Mean (SD)*	SAD (n = 57)	MAD (n = 24)
Age (years, mean [range])	39.6 (18–55)	40.4 (28–54)
Sex (male, %)	72	75
Weight (kg)	79.4 (10.3)	77.0 (9.1)
BMI (kg/m^2)	27.8 (2.7)	27.0 (2.3)
LVEF (%)	65.8 (2.44)	67.5 (1.24)
SBP (mmHg)	113.6 (9.69)	114 (7.38)
DBP (mmHg)	71.5 (6.72)	72.6 (4.94)
HR (bpm)	63.3 (8.42)	61.3 (6.80)

* Except where noted otherwise.
BMI, body mass index; DBP, diastolic blood pressure; HR, heart rate; MAD, multiple ascending dose; SAD, single ascending dose; SBP, systolic blood pressure; SD, standard deviation.

Safety

SAD Adverse Events Occurring in Two or More Participants

Preferred Term n (%)	Pooled Placebo (n = 15)	CK-274 1 mg (n = 6)	CK-274 3 mg (n = 6)	CK-274 10 mg (n = 6)	CK-274 25 mg (n = 6)	CK-274 40 mg (n = 6)	CK-274 50 mg (n = 11)	CK-274 75 mg (n = 1)	Total (n = 57)
Headache	1 (6.7)	–	–	1 (16.7)	2 (33.3)	–	1 (9.1)	–	5 (8.8)
Ejection fraction decreased	–	–	–	–	–	1 (16.7)	1 (9.1)	1 (100)	3 (5.3)
Abdominal pain upper	–	–	–	–	2 (33.3)	–	–	–	2 (3.5)
Abdominal tenderness	–	–	–	–	1 (16.7)	–	1 (9.1)	–	2 (3.5)
Chest pain	–	–	–	–	–	–	1 (9.1)*	1 (100)†	2 (3.5)
Dyspepsia	–	–	–	–	1 (16.7)	–	1 (9.1)	–	2 (3.5)
Flatulence	–	–	–	–	1 (16.7)	–	1 (9.1)	–	2 (3.5)
Nausea	–	–	–	–	2 (33.3)	–	–	–	2 (3.5)
Upper respiratory tract infection	2 (13.3)	–	–	–	–	–	–	–	2 (3.5)

- 17 participants experienced TEAEs
- There were no SAEs; all AEs were of mild or moderate severity

* "Bubbling sensation to the left chest" consistent with a gastrointestinal association rather than a cardiac association.
† Onset approximately 60 hours post dose.
AE, adverse event; SAD, single ascending dose; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

MAD Adverse Events

Preferred Term n (%)	Pooled Placebo (n = 6)	CK-274 5 mg qd x 14d (n = 6)	CK-274 7.5 mg qd x 17d (n = 6)	CK-274 10 mg qd x 14d (n = 6)	Total (n = 24)
Headache	1 (16.7)	–	–	1 (16.7)	2 (8.3)
Chapped lips	–	1 (16.7)	–	–	1 (4.2)
Cough	–	1 (16.7)	–	–	1 (4.2)
Feeling hot	1 (16.7)	–	–	–	1 (4.2)
Nausea	1 (16.7)	–	–	–	1 (4.2)
Salivary hypersecretion	–	1 (16.7)	–	–	1 (4.2)
Upper respiratory tract infection	–	–	–	1 (16.7)	1 (4.2)
Vomiting	1 (16.7)	–	–	–	1 (4.2)

- Four participants experienced TEAEs
- There were no SAEs; all AEs were of mild severity

AE, adverse event; d, day; MAD, multiple ascending dose; qd, once daily; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

- No clinically meaningful changes in ECG, laboratory test results, or vital signs in both SAD and MAD cohorts

Pharmacokinetics

SAD Pharmacokinetics Are Generally Dose Proportional

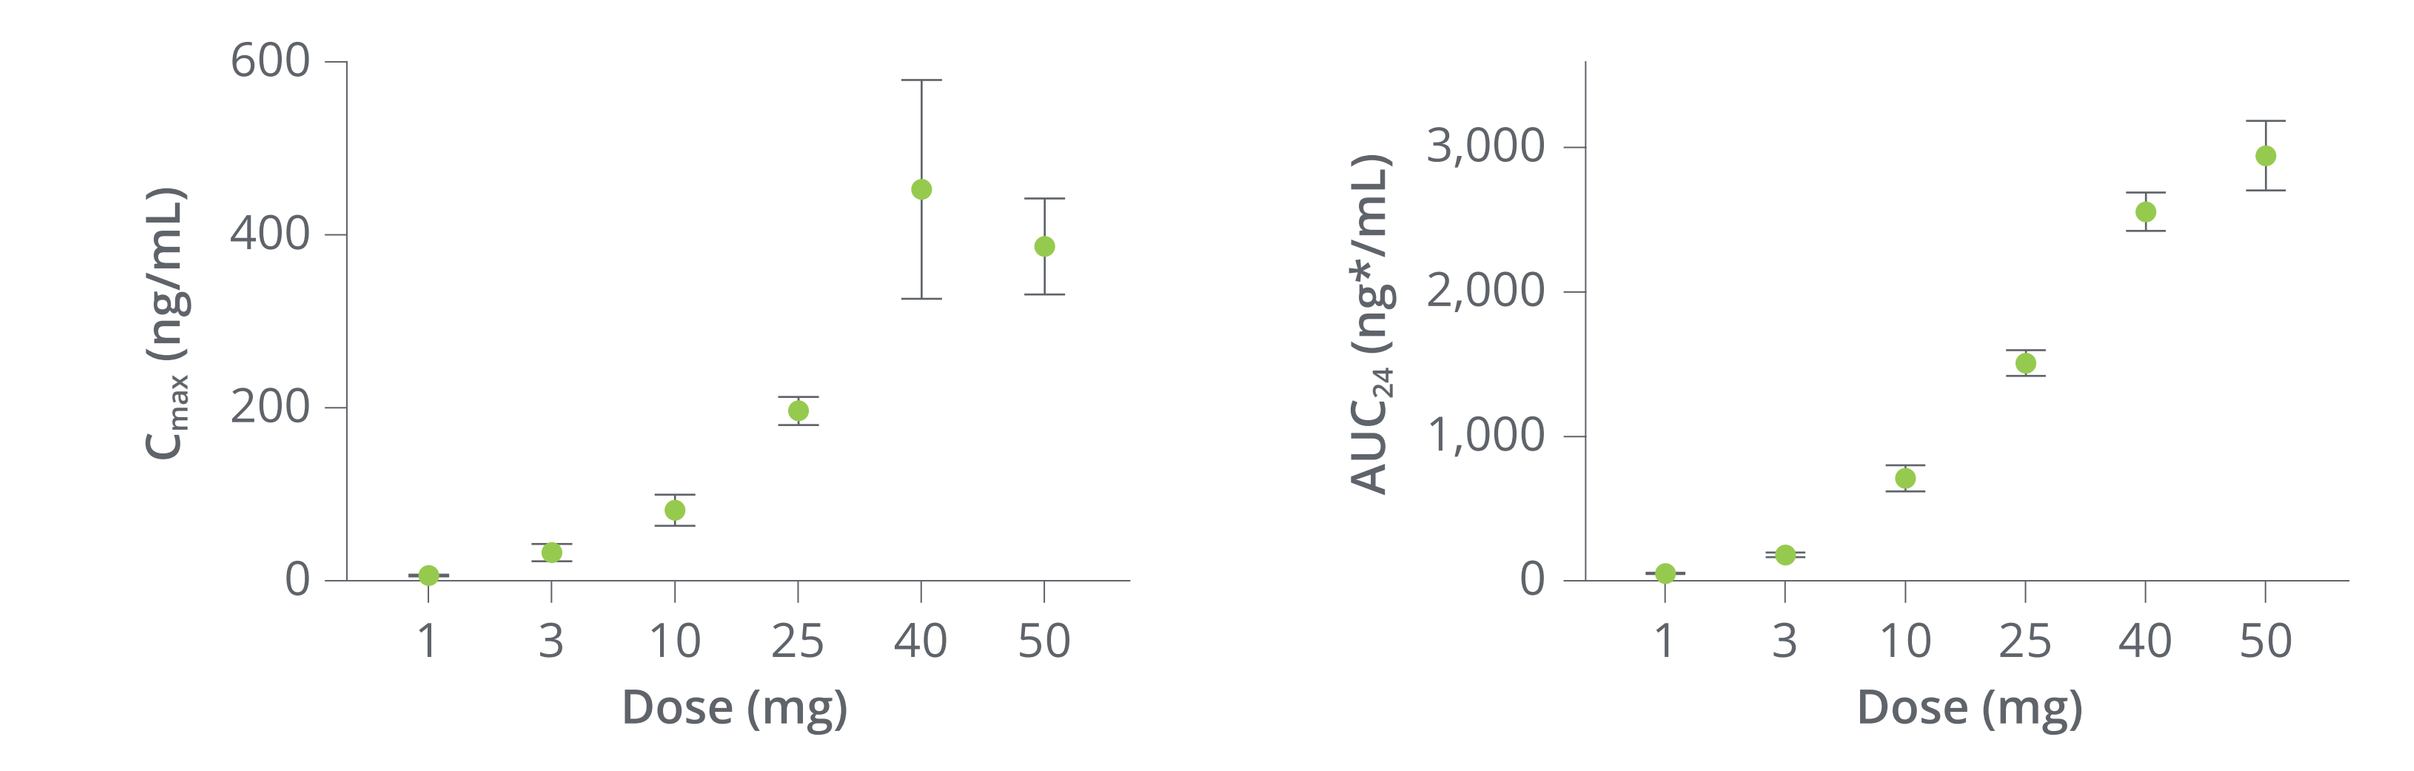
600
400
200
0
C_{max} (ng/mL)
1 3 10 25 40 50
Dose (mg)
3,000
2,000
1,000
0
AUC_{24} (ng*/mL)
1 3 10 25 40 50
Dose (mg)

Data points represent mean ± standard error of the mean.
C_{max}, maximum drug plasma concentration; AUC, area under the plasma concentration curve; SAD, single ascending dose.

Steady-state Appears Evident After 14 Days of Dosing

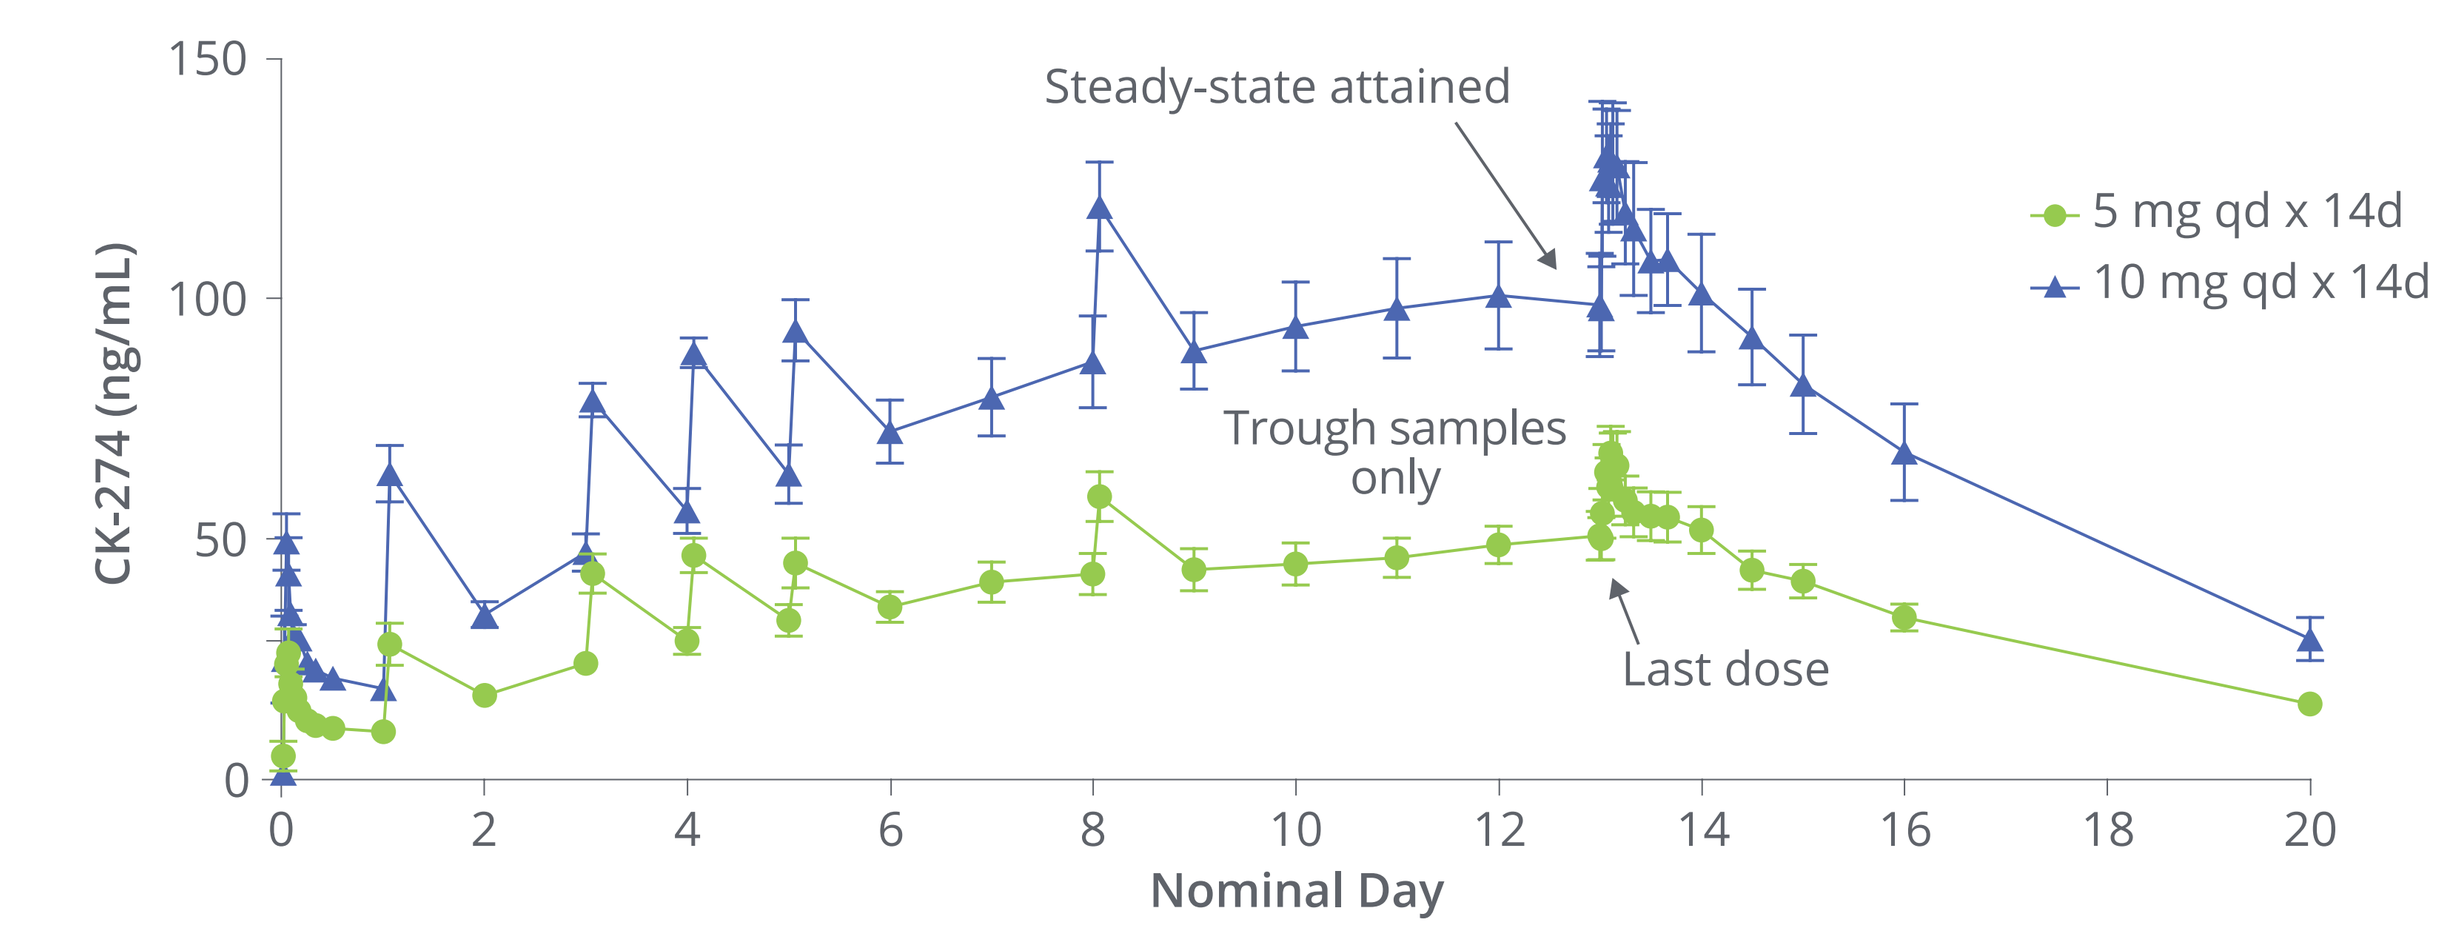
150
100
50
0
CK-274 (ng/mL)
0 2 4 6 8 10 12 14 16 18 20
Nominal Day
Steady-state attained
Trough samples only
Last dose
5 mg qd x 14d
10 mg qd x 14d

Data points represent mean ± standard error of the mean.
d, day; qd, once daily.

MAD Pharmacokinetic Parameters

Dose (n)	C_{max} (ng/mL)	t_{max} (h)	AUC_{24} (ng•h/mL)	$t_{½}$ (h)	AR
	PK Parameter, Geometric Mean (%CV)*				
5 mg (6)	69 (23.2%)	2.75 (1.5–4)	1,320 (23.0%)	86.3 (11.9)	4.71
7.5 mg (6)	148 (39.5%)	1.0 (0.5–5)	2,518 (25.8%)	76.9 (14.5)	4.50
10 mg (6)	141 (19.7%)	2.5 (0.5–3)	2,631 (22.8%)	79.7 (14.1)	4.79

- Half-life of CK-274 at steady-state is ~81 hours (3.4 days) on average

* Except data for t_{max} shown as median (minimum-maximum), and $t_{½}$ shown as the arithmetic mean (standard deviation). AR (accumulation ratio) calculated as (AUC_{24} on Day 14 or 17)/(AUC_{24} on Day 1).
%CV, percent coefficient of variation; C_{max}, maximum plasma concentration; AUC_{24}, area under the plasma concentration curve; MAD, multiple ascending dose; $t_{½}$, apparent plasma terminal elimination half-life; t_{max}, time to maximum observed plasma concentration.

PK/PD Relationship of CK-274 for Ejection Fraction (LVEF)

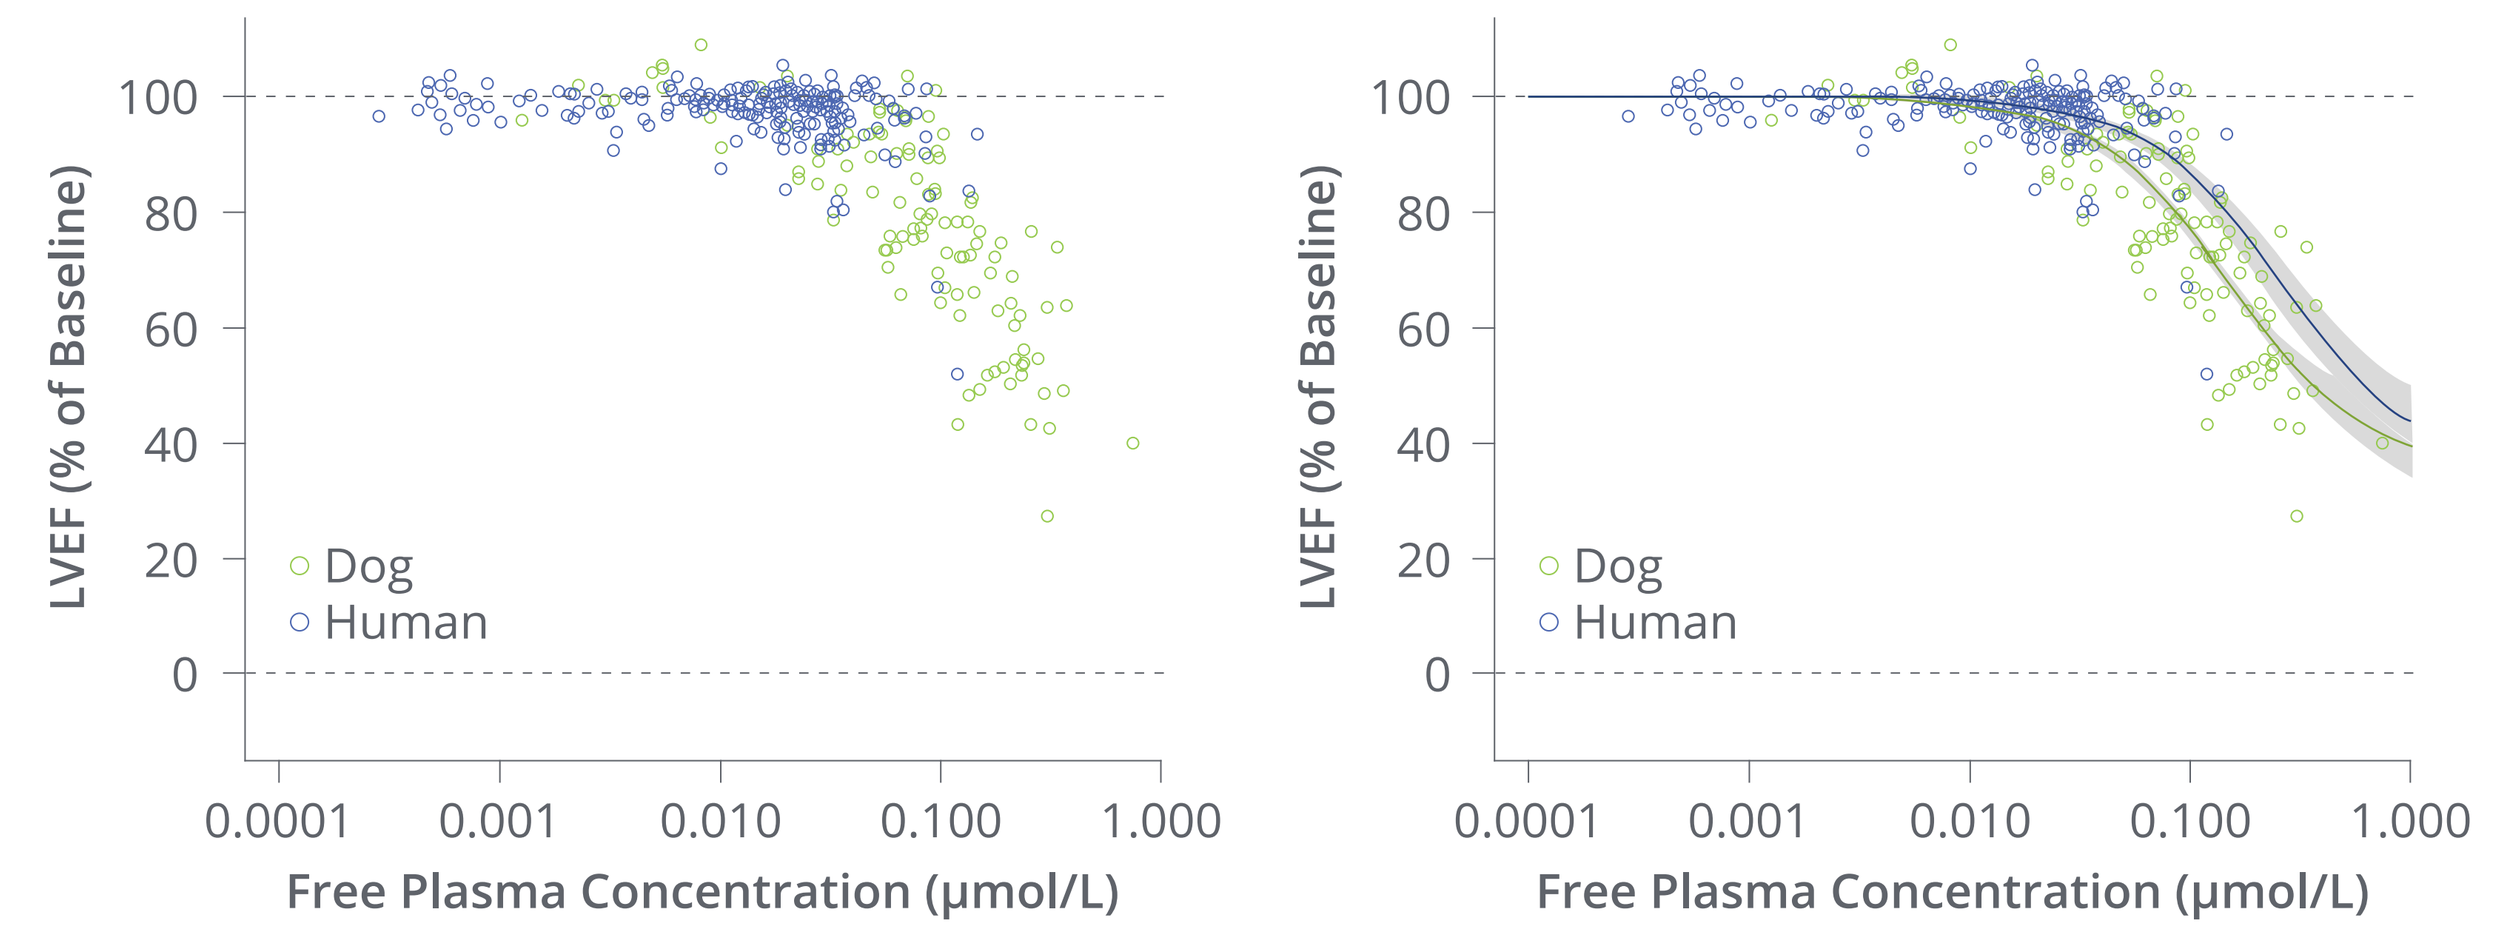
LVEF (% of Baseline)
100
80
60
40
20
0
Dog
Human
0.0001 0.001 0.010 0.100 1.000
Free Plasma Concentration (μmol/L)
LVEF (% of Baseline)
Dog
Human
0.0001 0.001 0.010 0.100 1.000
Free Plasma Concentration (μmol/L)

- Decrease in LVEF as function of exposure is similar in humans and dogs
 - Right panel shows model prediction curves with solid lines representing fitted model for dog (green curve) and human (blue curve), and shaded areas represent 90% confidence intervals of the model prediction
- The PK/PD relationship for CK-274 observed in humans was similar to that observed preclinically when adjusted for differences in protein binding

PHASE 2 OVERVIEW

Study Schema

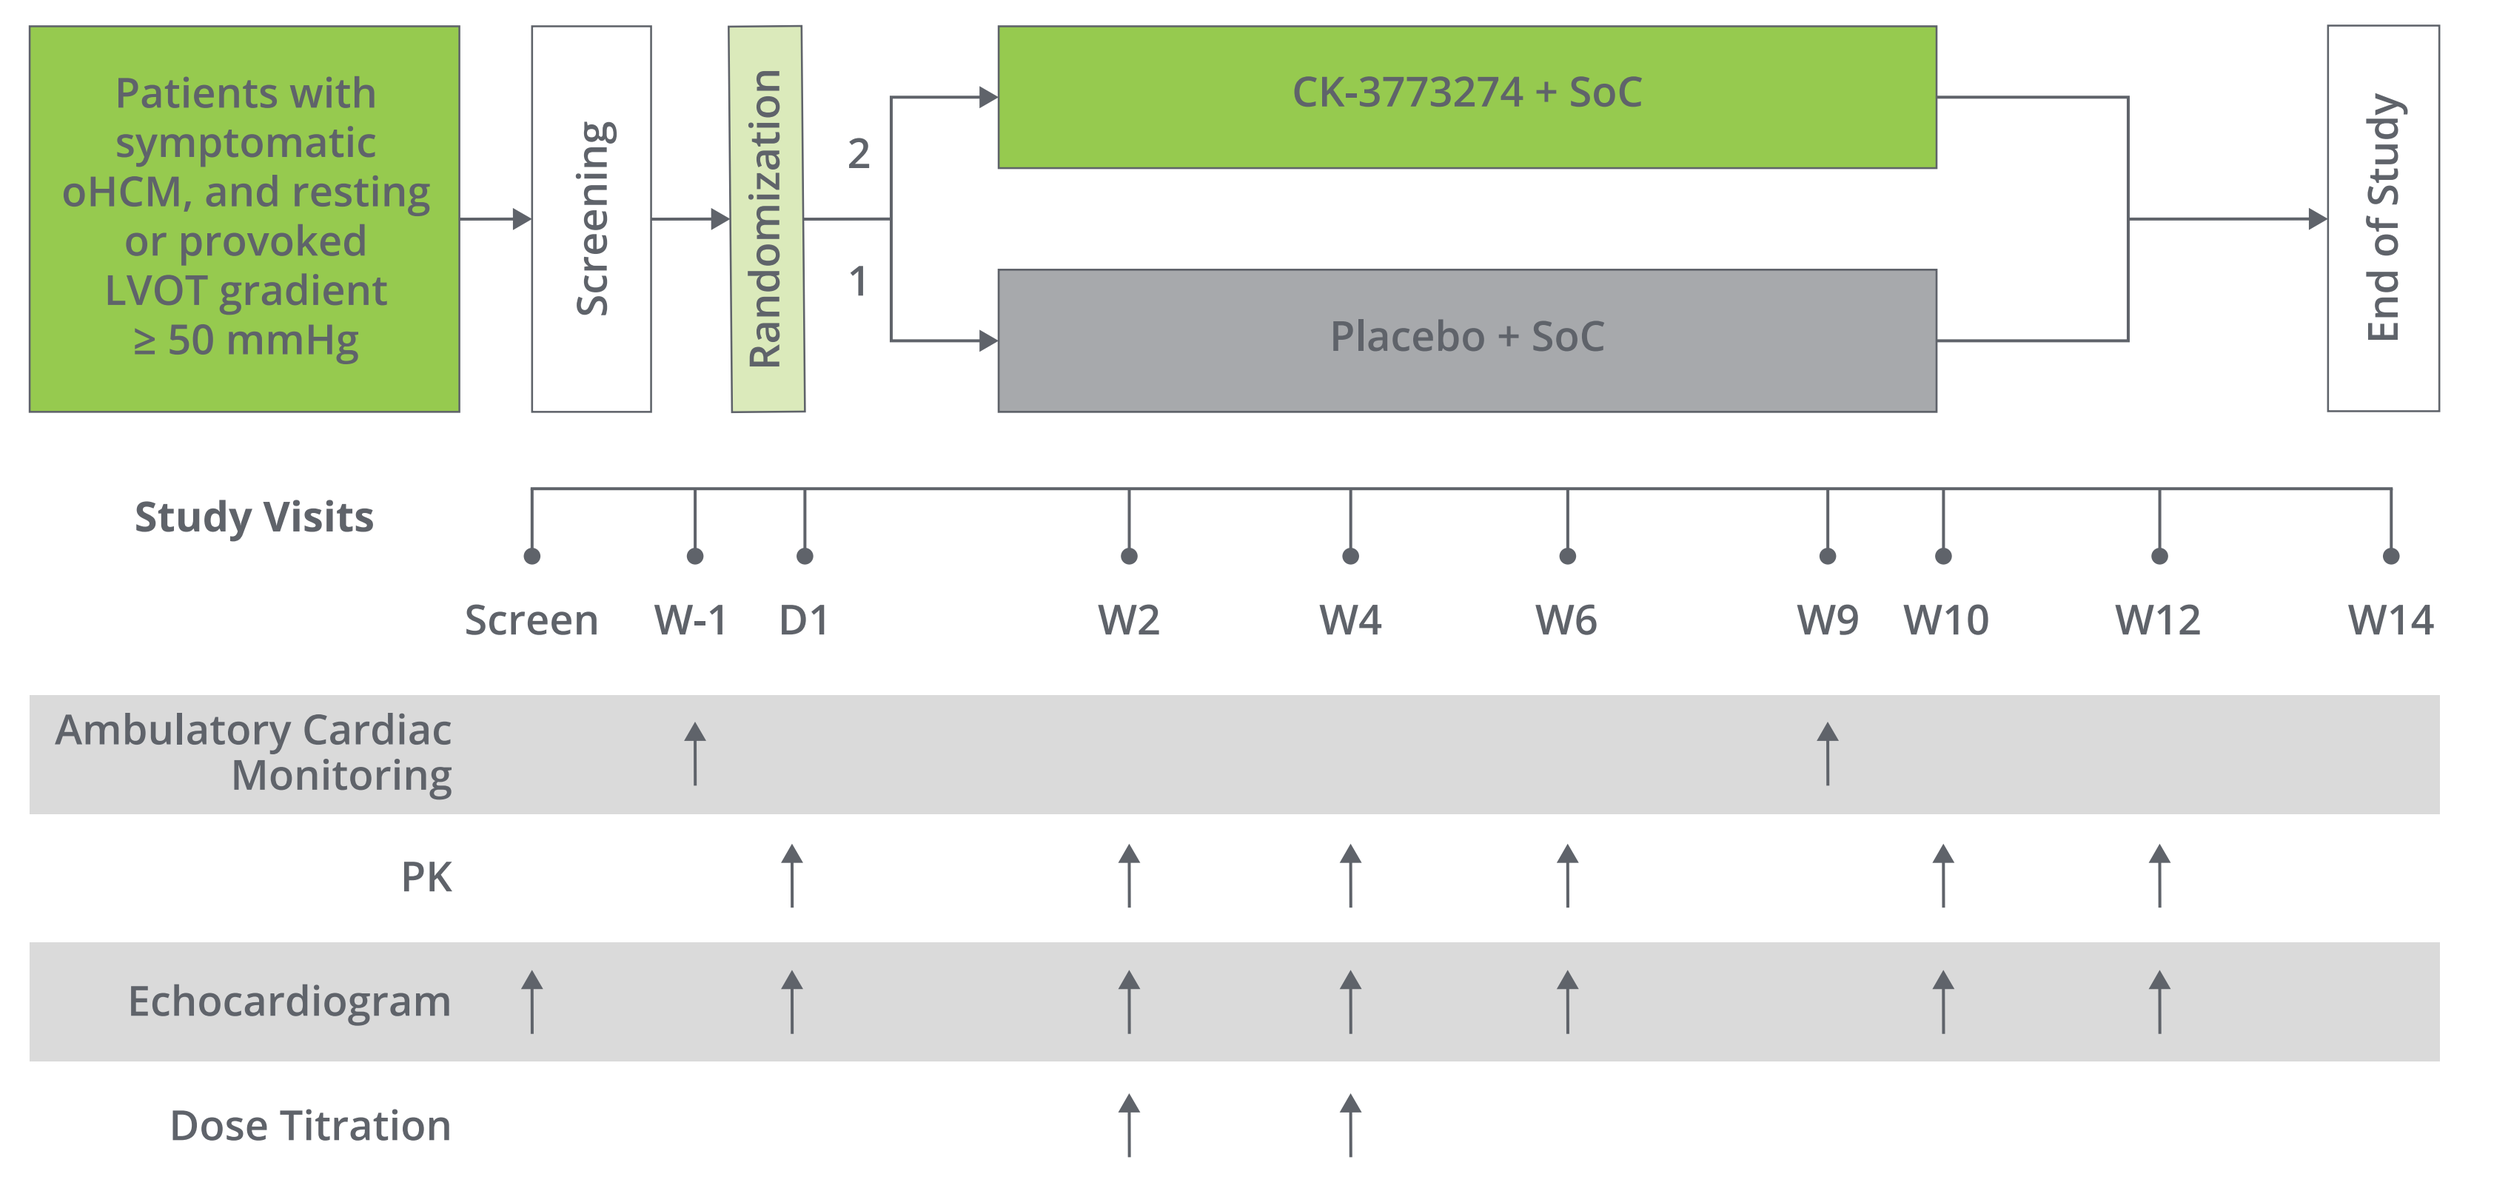
Patients with symptomatic oHCM, and resting or provoked LVOT gradient ≥ 50 mmHg
Screening
Randomization
2
1
CK-3773274 + SoC
Placebo + SoC
End of Study
Study Visits
Screen W-1 D1 W2 W4 W6 W9 W10 W12 W14
Ambulatory Cardiac Monitoring
PK
Echocardiogram
Dose Titration

- **Primary objective:** safety and tolerability
- **Secondary objectives:** PK and PD
- **Dose titration:** every 2 weeks guided by echocardiography

oHCM, obstructive hypertrophic cardiomyopathies; LVOT, left ventricular overflow tract; SoC, standard of care.

Study Population

- Men and women between 18 and 70 years of age
- Patients with obstructive HCM with:
 - Resting left ventricular overflow tract gradient (LVOT-G) ≥ 50 mmHg
 OR
 - Resting LVOT-G ≥ 30 mmHg and < 50 mmHg + post-Valsalva LVOT-G ≥ 50 mmHg
- LVEF ≥ 60%
- New York Heart Association class II or III
- Stable background medical therapy allowed

CONCLUSIONS

- **CK-274 was safe and well tolerated in healthy participants; there were no SAEs and no clinically meaningful changes in vital signs, ECGs, or laboratory tests**
- **Criteria for stopping dose escalation were reached after a single dose of 75 mg and after 14 days of a daily 10 mg dose**
- **Decreases in ejection fraction below 50% were readily reversible within 6 hours following single doses and within 24–48 hours following 14 days of dosing**
- **Pharmacokinetics (C_{max} and AUC_{24}) were generally dose linear; steady-state appeared evident after 14 days of daily dosing**
- **The shallow exposure-response relationship observed preclinically appears to translate to humans and thereby may enable flexible dose optimization in humans**
- **These Phase 1 data support progression of CK-274 into a placebo-controlled, double-blind Phase 2 study in patients with obstructive HCM who:**
 - **Remain on their background therapy for HCM**
 - **Can undergo echo-guided dose titration every 2 weeks**

Acknowledgments

We wish to thank the participants and their families for their contributions to this clinical trial.

References

1. Marian AJ, Braunwald E. *Circ Res.* 2017;121:749–770.
2. Hwee DT, et al. Poster #332 Presented at the Basic Cardiovascular Sciences 2019 Scientific Sessions, July 29–August 1, 2019; Boston, MA.
3. Hwee DT, et al. Poster #615 Presented at the Basic Cardiovascular Sciences 2019 Scientific Sessions, July 29–August 1, 2019; Boston, MA.

Disclosures

Robertson, **Robbie**, **Osmukhina**, and **Malik** are employees of and own stock in Cytokinetics, Inc. **Armas** is an employee of Celerion, Inc., where the CY 6011 study was conducted. **Li** is an employee of Certara, Inc. and a consultant to Cytokinetics, Inc. **Solomon** has received research support from and is a consultant to Cytokinetics, Inc.

The study was funded by Cytokinetics, Inc. Editorial support was provided by Kakuri Omari on behalf of Evidence Scientific Solutions, Inc, Philadelphia, PA and was funded by Cytokinetics, Inc.

To obtain a PDF of this poster: scan the QR code OR visit http://bit.ly/2KEPPQ5. No personal information is stored.

Presented at the Heart Failure Society of America 23rd Annual Scientific Meeting; Philadelphia, PA; September 13–16, 2019