77

FORTITUDE-ALS: Who Received the Most Benefit, and Translation to Real-world Events

Jeremy M. Shefner,[1] Jinsy A. Andrews,[2] Angela Genge,[3] Carlayne Jackson,[4] Noah Lechtzin,[5] Timothy M. Miller,[6] Bettina M. Cockroft,[7] Fady I. Malik,[7] Lisa Meng,[7] Jenny Wei,[7] Andrew A. Wolff,[7] Stacy A. Rudnicki,[7] and the FORTITUDE-ALS Study Group

[1]Barrow Neurological Institute, Phoenix, AZ, USA; [2]The Neurological Institute, Columbia University, New York, NY, USA; [3]Montreal Neurological Institute, Montreal, QC, Canada; [4]University of Texas Health Science Center, San Antonio, TX, USA; [5]Johns Hopkins School of Medicine, Baltimore, MD, USA; [6]Washington University School of Medicine, St. Louis, MO, USA; [7]Cytokinetics, Inc., South San Francisco, CA, USA

BACKGROUND

- The rate of progression of amyotrophic lateral sclerosis (ALS) varies among patients and may influence the ability to detect the response to therapeutic interventions
- Translation of the rate of progression during a clinical trial to real-world events of importance to patients, such as use of durable medical equipment (DME), is critical
- FORTITUDE-ALS was a large, Phase 2, double-blind, placebo-controlled, dose ranging clinical trial of *reldesemtiv*, a selective fast skeletal muscle troponin activator, in ALS patients that suggested a clinical benefit in patients treated for 3 months

HYPOTHESIS

- Prestudy progression rates from patients in the FORTITUDE-ALS trial were used to evaluate change in the ALS Functional Rating Scale-Revised (ALSFRS-R), and the effect of treatment with *reldesemtiv* on the use of DME
- Here we present data showing that benefit was more evident if slower progressing patients were excluded, and that DME use was altered by active treatment

METHODS

- Key inclusion/exclusion criteria for FORTITUDE-ALS:
 - Males or females between 18 and 80 years of age
 - Diagnosis of ALS for ≤ 24 months
 - Upright slow vital capacity (SVC) ≥ 60% predicted for age, height, and sex at screening
 - Either not taking or on stable doses of riluzole and/or edaravone
- Patients (N=458) were randomized to *reldesemtiv* 150, 300, or 450 mg twice daily (bid) or placebo **(Figure 1)**
- Disease progression was estimated using onset date of ALS symptoms and baseline ALSFRS-R score (defined as the latest ALSFRS-R before the first dose of study treatment)
- Patients were divided into slow, medium, and fast progressing tertiles (SP, MP, FP) for prestudy ALSFRS-R total score reduction per month
 - SP: ≤ 0.37
 - MP: > 0.37 ≤ 0.67
 - FP: > 0.67
- Timing and use of a new DME (ie, manual wheelchair, power wheelchair, gastrostomy tube, noninvasive ventilator, or augmentative communication device) for all patients were analyzed using Cox regression to compare utilization of DME for each dose with placebo

Figure 1. FORTITUDE-ALS study design

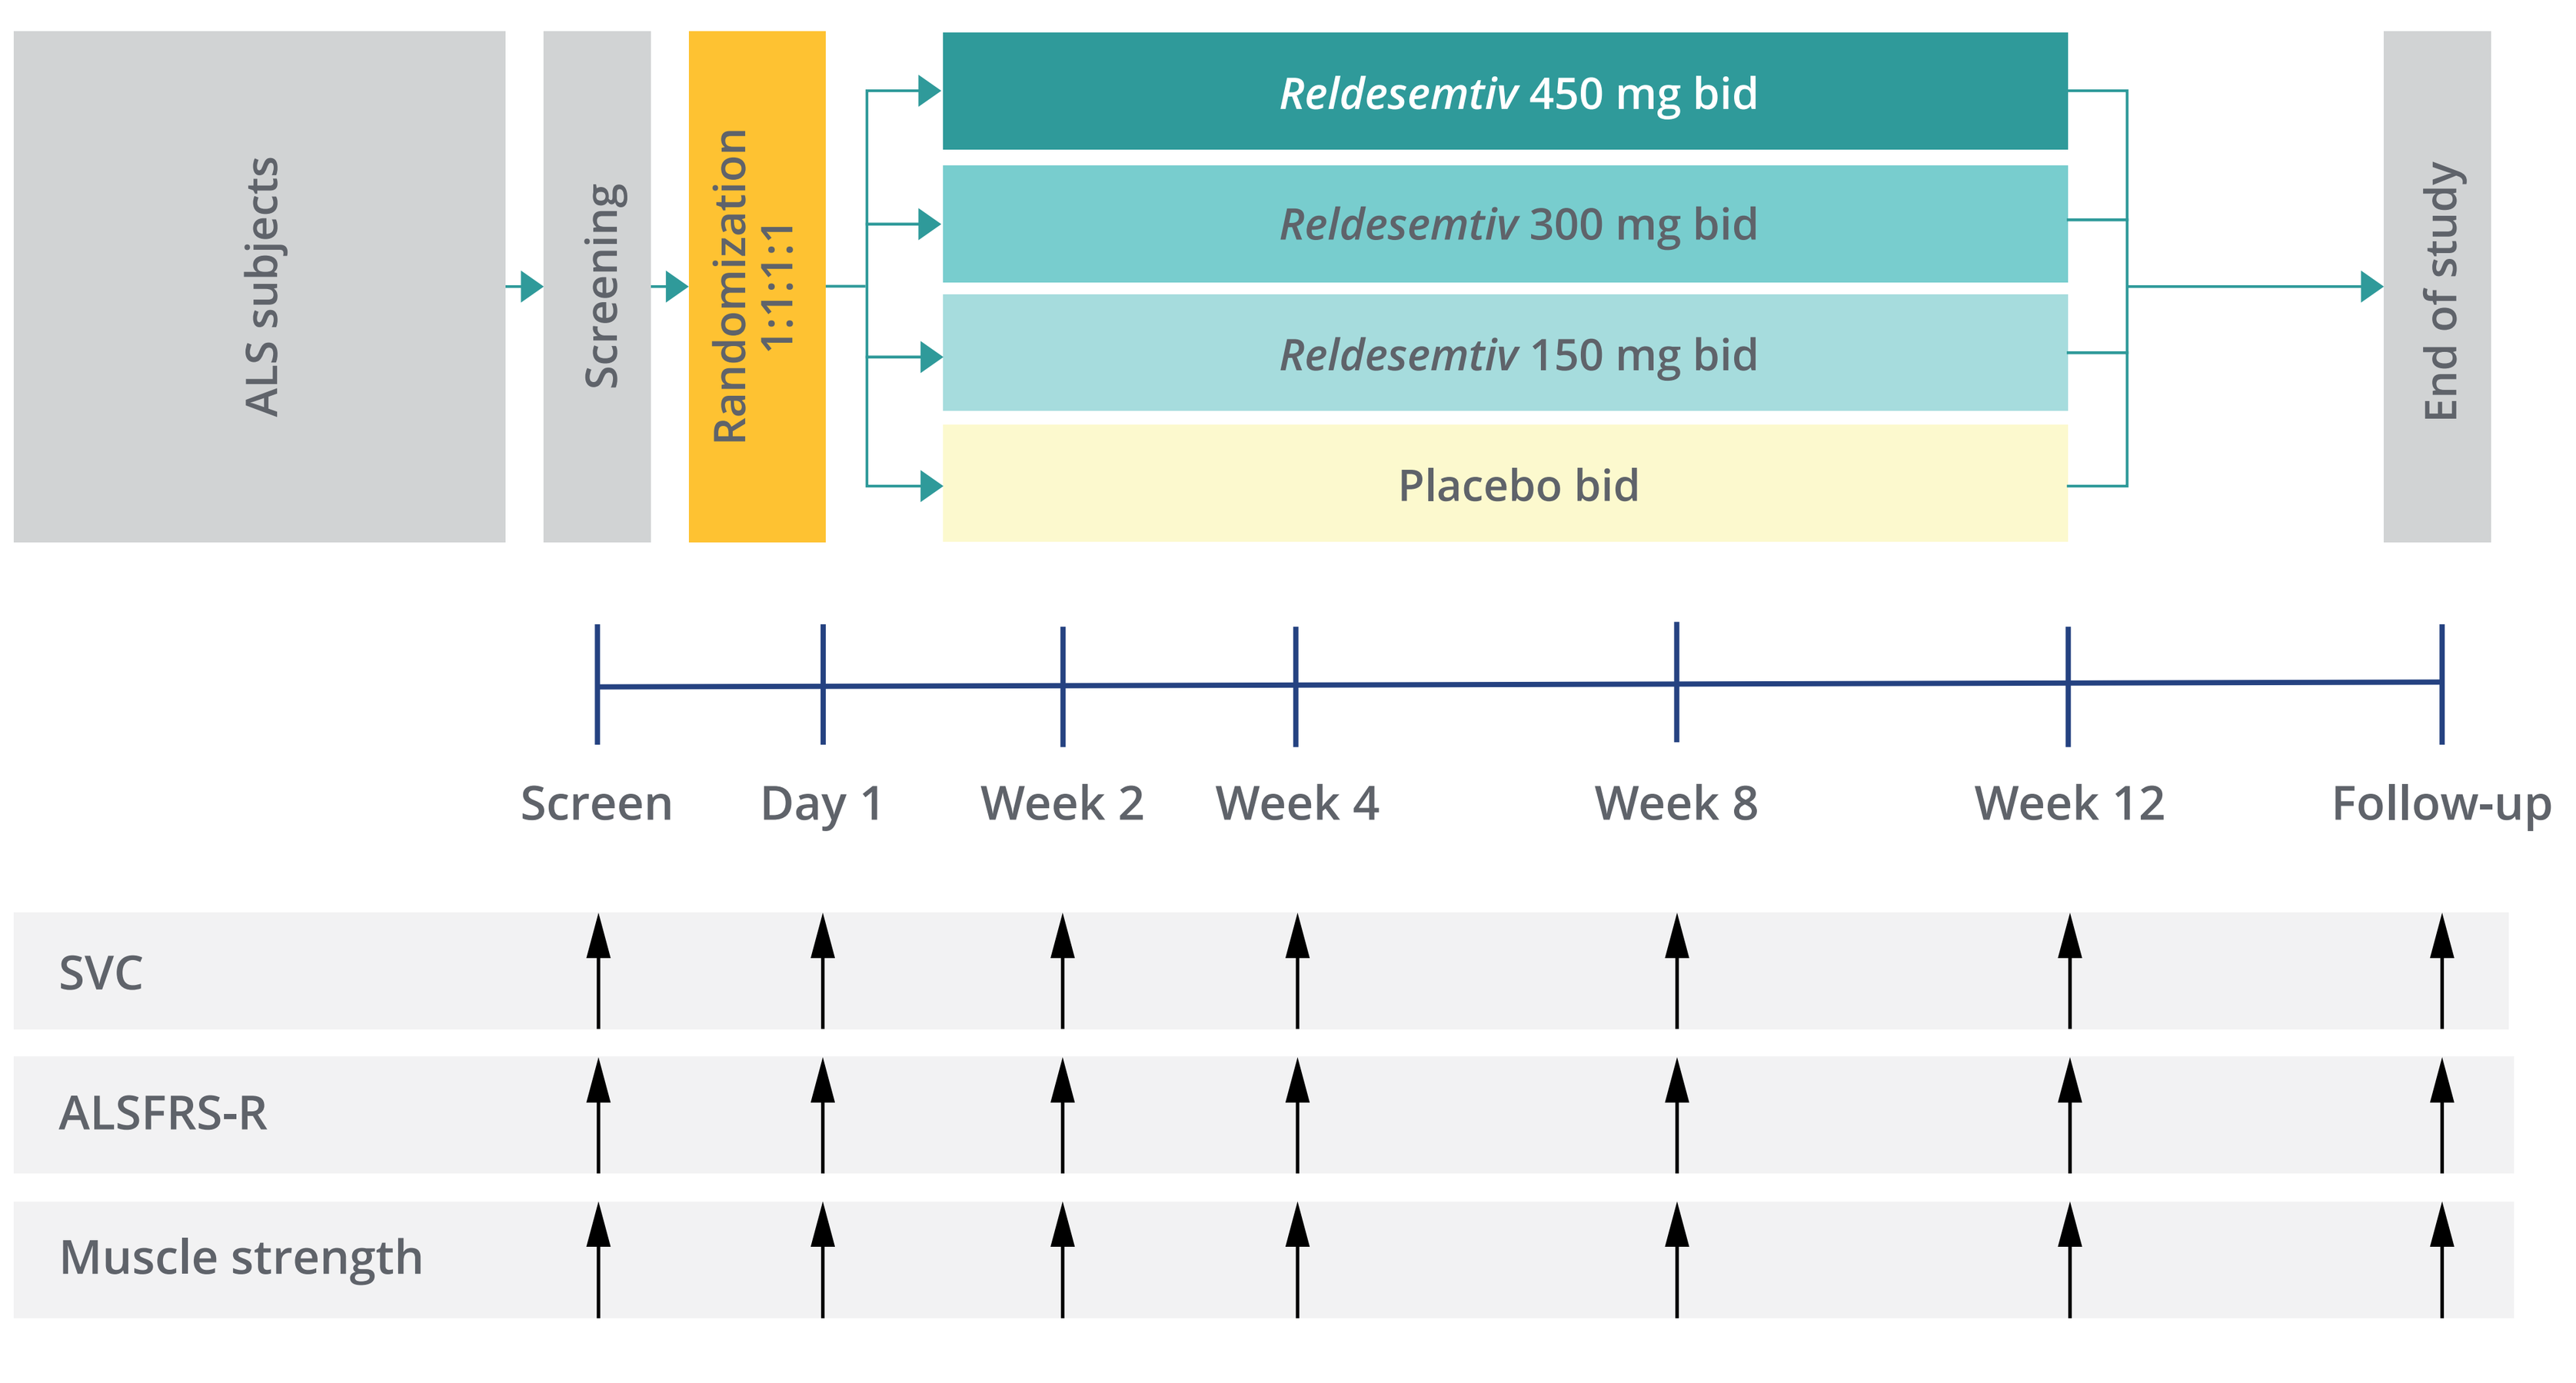
ALS subjects
Screening
Randomization 1:1:1:1
Reldesemtiv 450 mg bid
Reldesemtiv 300 mg bid
Reldesemtiv 150 mg bid
Placebo bid
End of study
Screen
Day 1
Week 2
Week 4
Week 8
Week 12
Follow-up
SVC
ALSFRS-R
Muscle strength

ALS, amyotrophic lateral sclerosis; ALSFRS-R, ALS Functional Rating Scale-Revised; bid, twice daily; SVC, slow vital capacity.

RESULTS

Table 1. Baseline demographic and disease characteristics by prestudy rate of ALS progression

Characteristic	SP tertile (≤ 0.37) Placebo (n = 35)	SP tertile (≤ 0.37) *Reldesemtiv* (n = 117)	MP tertile (> 0.37 ≤ 0.67) Placebo (n = 40)	MP tertile (> 0.37 ≤ 0.67) *Reldesemtiv* (n = 112)	FP tertile (> 0.67) Placebo (n = 40)	FP tertile (> 0.67) *Reldesemtiv* (n = 113)	Overall (n = 457)
Age (years), mean (SD)	60.6 (11.1)	58.4 (10.9)	58.8 (9.1)	58.2 (10.9)	59.6 (11.7)	58.4 (10.6)	58.7 (10.7)
Male, n (%)	21 (60.0)	76 (65.0)	23 (57.5)	63 (56.3)	24 (60.0)	70 (61.9)	277 (60.6)
BMI (kg/m²), mean (SD)	25.8 (4.2)	27.2 (4.9)	26.3 (4.6)	26.9 (4.4)	26.2 (4.4)	26.1 (4.7)	26.6 (4.6)
ALSFRS-R total score, mean (SD)	41.3 (4.2)	41.8 (3.4)	36.3 (4.4)	36.9 (4.4)	33.9 (5.5)	33.7 (5.2)	37.4 (5.5)
SVC (% predicted), mean (SD)	88.2 (15.7)	89.6 (15.2)	85.4 (12.3)	85.1 (16.0)	81.7 (15.9)	79.0 (13.4)	84.7 (15.3)
Months since diagnosis, mean (SD)	9.6 (7.0)	9.7 (6.7)	10.0 (6.9)	9.6 (6.2)	6.8 (4.6)	6.2 (4.1)	8.6 (6.1)
Months since first symptom, mean (SD)	29.4 (16.3)	33.2 (31.5)	23.3 (8.0)	21.7 (8.2)	14.6 (6.6)	13.9 (5.9)	22.8 (19.1)
ALS site of onset: bulbar, n (%)	7 (20.0)	25 (21.4)	8 (20.0)	15 (13.4)	7 (17.5)	25 (22.1)	87 (19.0)
On riluzole alone, n (%)	21 (60.0)	69 (59.0)	19 (47.5)	59 (52.7)	24 (60.0)	66 (58.4)	258 (56.5)
On edaravone alone, n (%)	1 (2.9)	3 (2.6)	2 (5.0)	5 (4.5)	2 (5.0)	6 (5.3)	19 (4.2)
On riluzole and edaravone, n (%)	6 (17.1)	21 (17.9)	11 (27.5)	32 (28.6)	7 (17.5)	17 (15.0)	94 (20.6)

ALS, amyotrophic lateral sclerosis; ALSFRS-R, ALS Functional Rating Scale-Revised; FP, fast progressing; MP, medium progressing; SD, standard deviation; SP, slow progressing.

Rate of ALSFRS-R progression

- In the combined MP and FP tertiles, change from baseline in ALSFRS-R total score at Week 12 was significantly smaller in patients who received any dose of *reldesemtiv* versus placebo **(Figure 2)**

Figure 2. Change from baseline in ALSFRS-R score by prestudy progressor tertiles

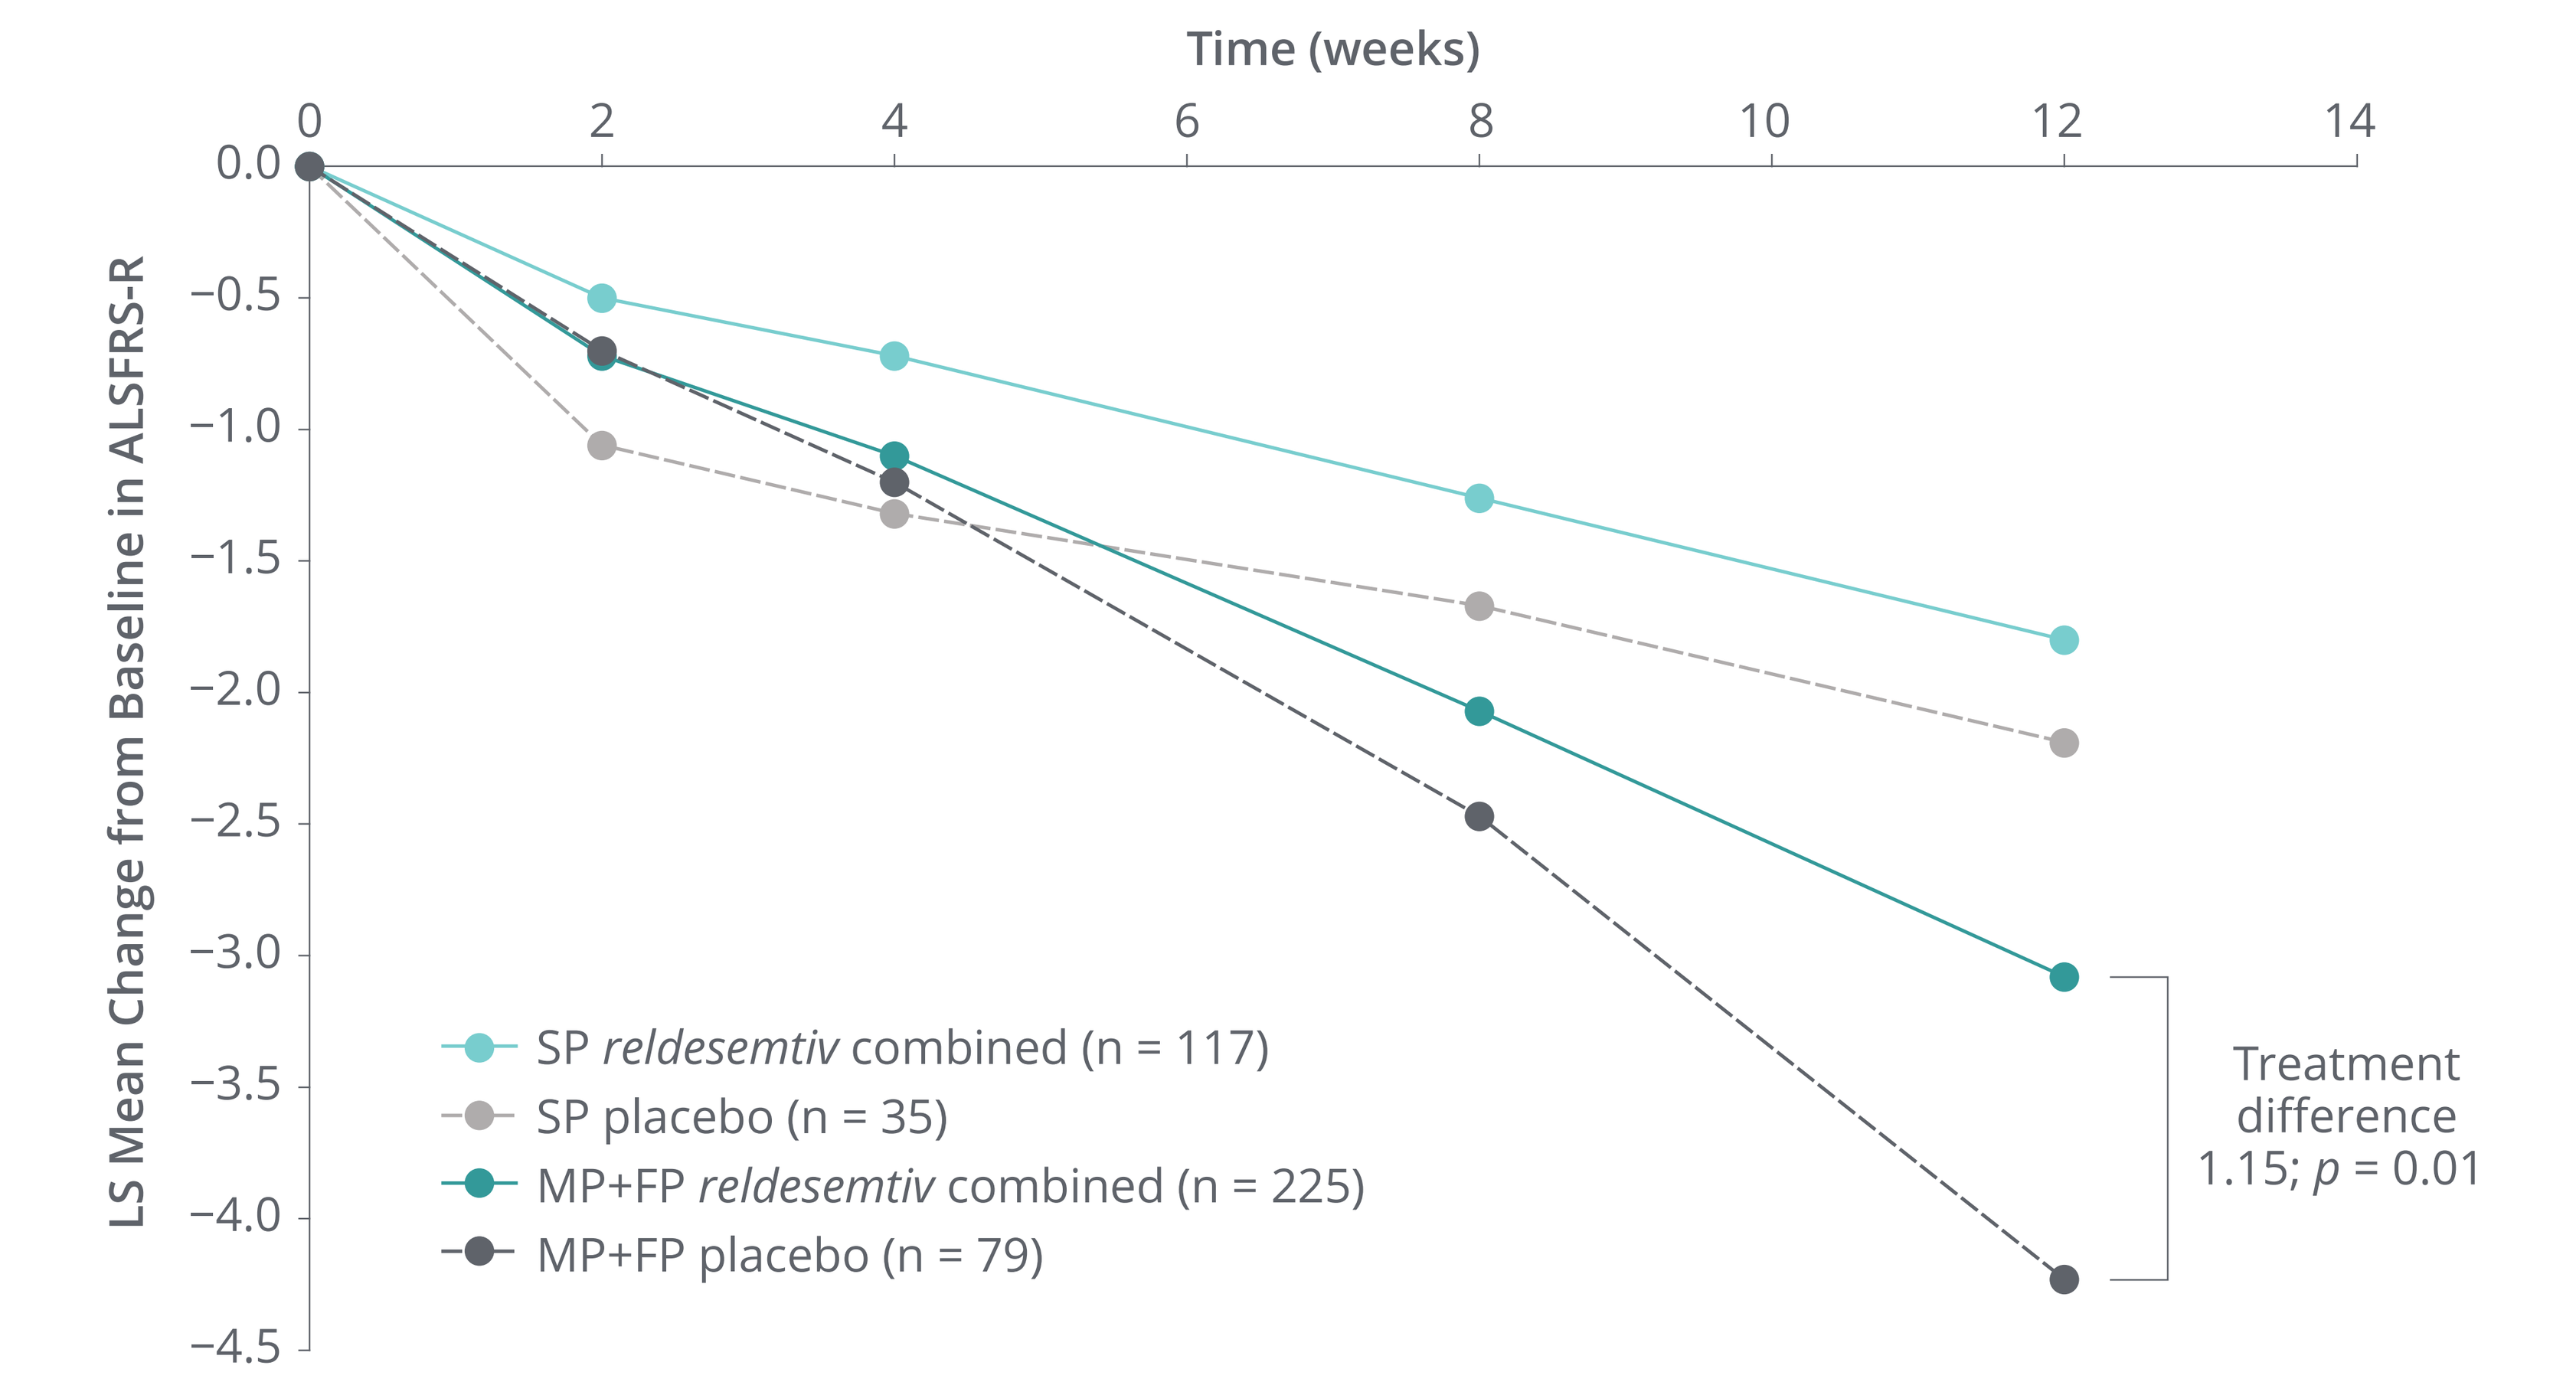
Time (weeks)
0
2
4
6
8
10
12
14
0.0
−0.5
−1.0
−1.5
−2.0
−2.5
−3.0
−3.5
−4.0
−4.5
LS Mean Change from Baseline in ALSFRS-R
SP *reldesemtiv* combined (n = 117)
SP placebo (n = 35)
MP+FP *reldesemtiv* combined (n = 225)
MP+FP placebo (n = 79)
Treatment difference 1.15; *p* = 0.01

ALSFRS-R, ALS Functional Rating Scale-Revised; FP, fast progressing; MP, medium progressing; SP, slow progressing.

DME use with *reldesemtiv*

- A smaller percentage of patients on any *reldesemtiv* dose accepted new DME, and the acceptance of a new DME was significantly later as compared with patients on placebo **(Figure 3)**
 - The hazard ratio compared with placebo for accepting at least one DME was significantly reduced for those taking *reldesemtiv* 150 mg bid (0.45 [95% confidence interval (CI): 0.25, 0.81]; $p = 0.0079$), and trended toward reduction in the other groups
 - For all *reldesemtiv* doses combined, the hazard ratio compared with placebo was 0.62 (CI: 0.40, 0.95; $p = 0.027$): a statistically significantly lower risk (reduced by 38%) of needing a new piece of DME compared with placebo

Figure 3. Probability of no new DME use over time with *reldesemtiv* treatment: (A) each dose; (B) all doses combined compared with placebo

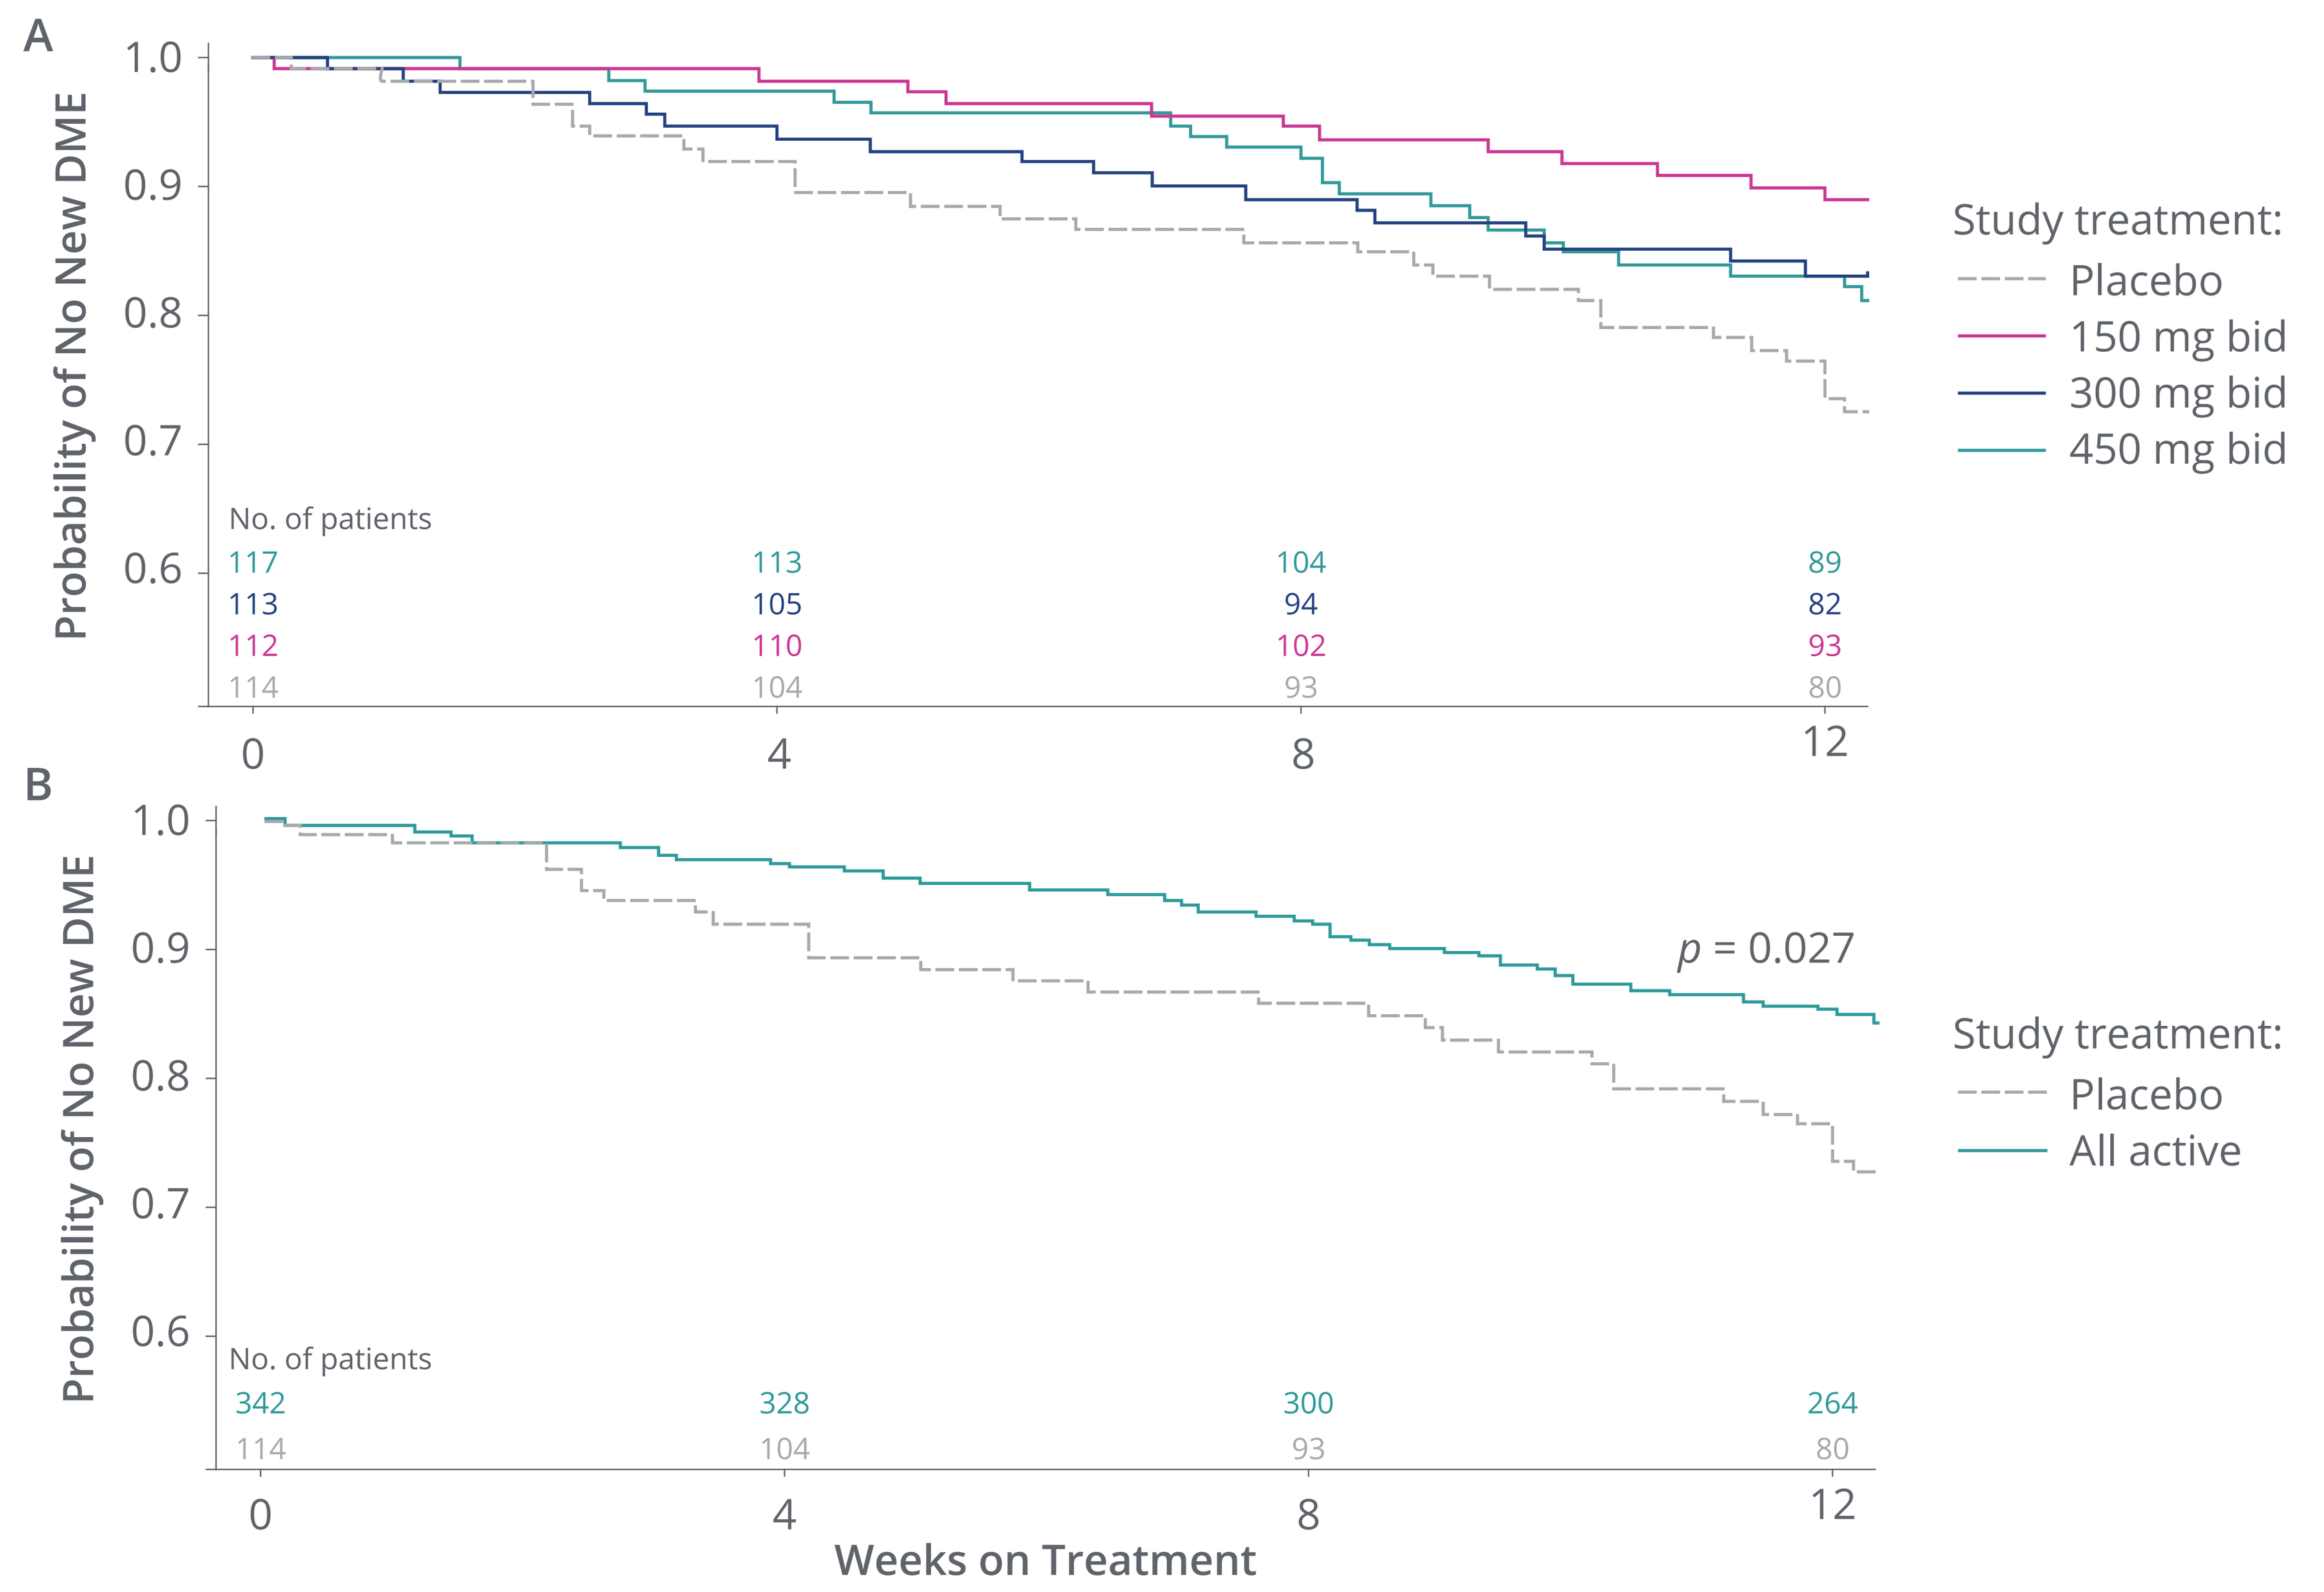
A
1.0
0.9
0.8
0.7
0.6
Probability of No New DME
No. of patients
117
113
104
89
113
105
94
82
112
110
102
93
114
104
93
80
0
4
8
12
Study treatment:
Placebo
150 mg bid
300 mg bid
450 mg bid
B
1.0
0.9
0.8
0.7
0.6
Probability of No New DME
p = 0.027
No. of patients
342
328
300
264
114
104
93
80
0
4
8
12
Weeks on Treatment
Study treatment:
Placebo
All active

bid, twice daily; DME, durable medical equipment.

- For each DME type, the percentage of new DME in the placebo group was higher than the all active group suggesting the result is not driven by any one type

CONCLUSIONS

- **Statistically significant effects of *reldesemtiv* treatment were seen in MP and FP tertiles (based on estimated prestudy disease progression rate using ALSFRS-R) in the ALSFRS-R total score at Week 12**
- **The timing and use of DME appear to be influenced by *reldesemtiv* treatment, suggesting a real-world impact of *reldesemtiv***
- **Findings from FORTITUDE-ALS indicate that patients with more rapid disease progression may enable detection of a treatment benefit in ALS trials of shorter duration, and treatment with *reldesemtiv* may have beneficial effects on real-world life experiences of importance to patients with ALS**

Acknowledgments

We wish to thank the participants of FORTITUDE-ALS and their families for their contributions to this clinical trial, the investigators of FORTITUDE-ALS, and members of the Data Monitoring Committee and Steering Committee.

The study was funded by Cytokinetics, Inc.

Editorial support was provided by Jennifer Geil on behalf of Evidence Scientific Solutions, Inc, Philadelphia, PA, and was funded by Cytokinetics, Inc.

In collaboration with Astellas Pharma, Inc., Cytokinetics is developing *reldesemtiv* as a potential treatment for people living with ALS and certain other debilitating diseases and conditions associated with skeletal muscle weakness and/or fatigue.

Disclosures

Shefner has served as a consultant for Biogen, Biohaven, Cytokinetics, Neuraltus, and Pharnext; has received research support from Amylyx, Biogen, Biohaven, Blotie, Cytokinetics, Neuraltus, Orion, and Orphazyme; and has received compensation from UpToDate for serving as neuromuscular section editor. **Andrews** has served as a consultant for Avexis, Biohaven, and Clene Nanomedicine; has served as a consultant for Cytokinetics; is a former employee of Cytokinetics; and has received research support from Biogen, Neuraltus, Orion, and Roche. **Genge** has served as a consultant for AB Sciences, AL-S Pharma, Avexis, Biogen, Cytokinetics, MTPA, and Roche. **Jackson** has served as a consultant for Argenex, Cytokinetics, ITF Pharma, MTPA, and Strongbridge Pharmaceuticals; served on Speaker's Bureau for Avanir, CSL Behring, MTPA, and Strongbridge Pharmaceuticals; has received research support from Amylyx, Cytokinetics, and NIH; and is currently serving as a member of a Data Safety Monitoring Board for Anelixis, Brainstorm, and Mallinckrodt. **Lechtzin** has served as a consultant/advisor for Cytokinetics, Hill-Rom, and Vertex; and has received research support from AstraZeneca and Vertex. **Miller** has served as a consultant/advisor for Biogen and Cytokinetics; has received research support from Biogen and Ionis; and receives licensing fees from C2N. **Cockroft, Malik, Meng, Wei, Wolff,** and **Rudnicki** are employees of and own stock in Cytokinetics.

To obtain a PDF of this poster:
scan the QR code
OR
visit http://bit.ly/2n1rWZt.
No personal information is stored.

Presented at the 18th Annual Northeast Amyotrophic Lateral Sclerosis Consortium (NEALS) Meeting | Clearwater Beach, FL | October 2–4, 2019