CLT-20

Utilization of Durable Medical Equipment in FORTITUDE-ALS

Stacy A. Rudnicki,[1] Jinsy A. Andrews,[2] Angela Genge,[3] Carlayne Jackson,[4] Noah Lechtzin,[5] Timothy M. Miller,[6] Bettina M. Cockroft,[1] Fady I. Malik,[1] Lisa Meng,[1] Jenny Wei,[1] Andrew A. Wolff,[1] Jeremy M. Shefner[7]

[1]Cytokinetics, Inc., South San Francisco, CA, USA; [2]The Neurological Institute, Columbia University, New York, NY, USA; [3]Montreal Neurological Institute, Montreal, QC, Canada; [4]University of Texas Health Science Center, San Antonio, TX, USA; [5]Johns Hopkins School of Medicine, Baltimore, MD, USA; [6]Washington University School of Medicine, St. Louis, MO, USA; [7]Barrow Neurological Institute, Phoenix, AZ, USA

BACKGROUND

- The economic impact of ALS includes indirect and direct costs that include lost income, home or vehicle modifications, cost of medications, interactions with health care providers, hospitalizations, and durable medical equipment (DME)
- FORTITUDE-ALS was a double-blind, placebo-controlled, dose-ranging clinical trial of *reldesemtiv* in patients with ALS with an active treatment period of 12 weeks and a 4-week follow-up period
 - Dose-response analyses of the primary endpoint (change from baseline in slow vital capacity [SVC] at 12 weeks) and secondary endpoints (change in ALS Functional Rating Scale-Revised [ALSFRS-R] and slope of the muscle mega-score from baseline to 12 weeks) were not statistically significant
 - However, a prespecified analysis of the 2 highest doses combined and a post hoc analysis of all doses combined compared with placebo revealed statistically significant differences in change from baseline to Week 12 of the ALSFRS-R (p = 0.04 and 0.01, respectively) favoring *reldesemtiv*
- The possible impact of treatment with *reldesemtiv* on receiving DME in FORTITUDE-ALS was evaluated

OBJECTIVES

- To determine the number of patients in each treatment group prescribed and agreeing to obtain at least 1 new DME item while participating in FORTITUDE-ALS, defined as:
 - Manual wheelchair, power wheelchair, gastrostomy tube, noninvasive ventilator, or augmentative communication device
- Time to have the DME prescribed and agreed to by the patient (DME-PAP) was used as the endpoint, given the variability across insurance coverage and countries in time to receive the DME and the relatively short study duration

METHODS

FORTITUDE-ALS study

- Key inclusion/exclusion criteria:
 - Males or females between 18 and 80 years of age
 - Diagnosis of ALS for ≤ 24 months
 - Upright SVC ≥ 60% predicted for age, height, and sex at screening
 - Either not taking or on stable doses of riluzole and/or edaravone for ≥ 30 days
- Patients (N = 457) were randomized (1:1:1:1) and treated with *reldesemtiv* 150, 300, or 450 mg twice daily (bid) or placebo **(Figure 1)**

Figure 1. FORTITUDE-ALS study design

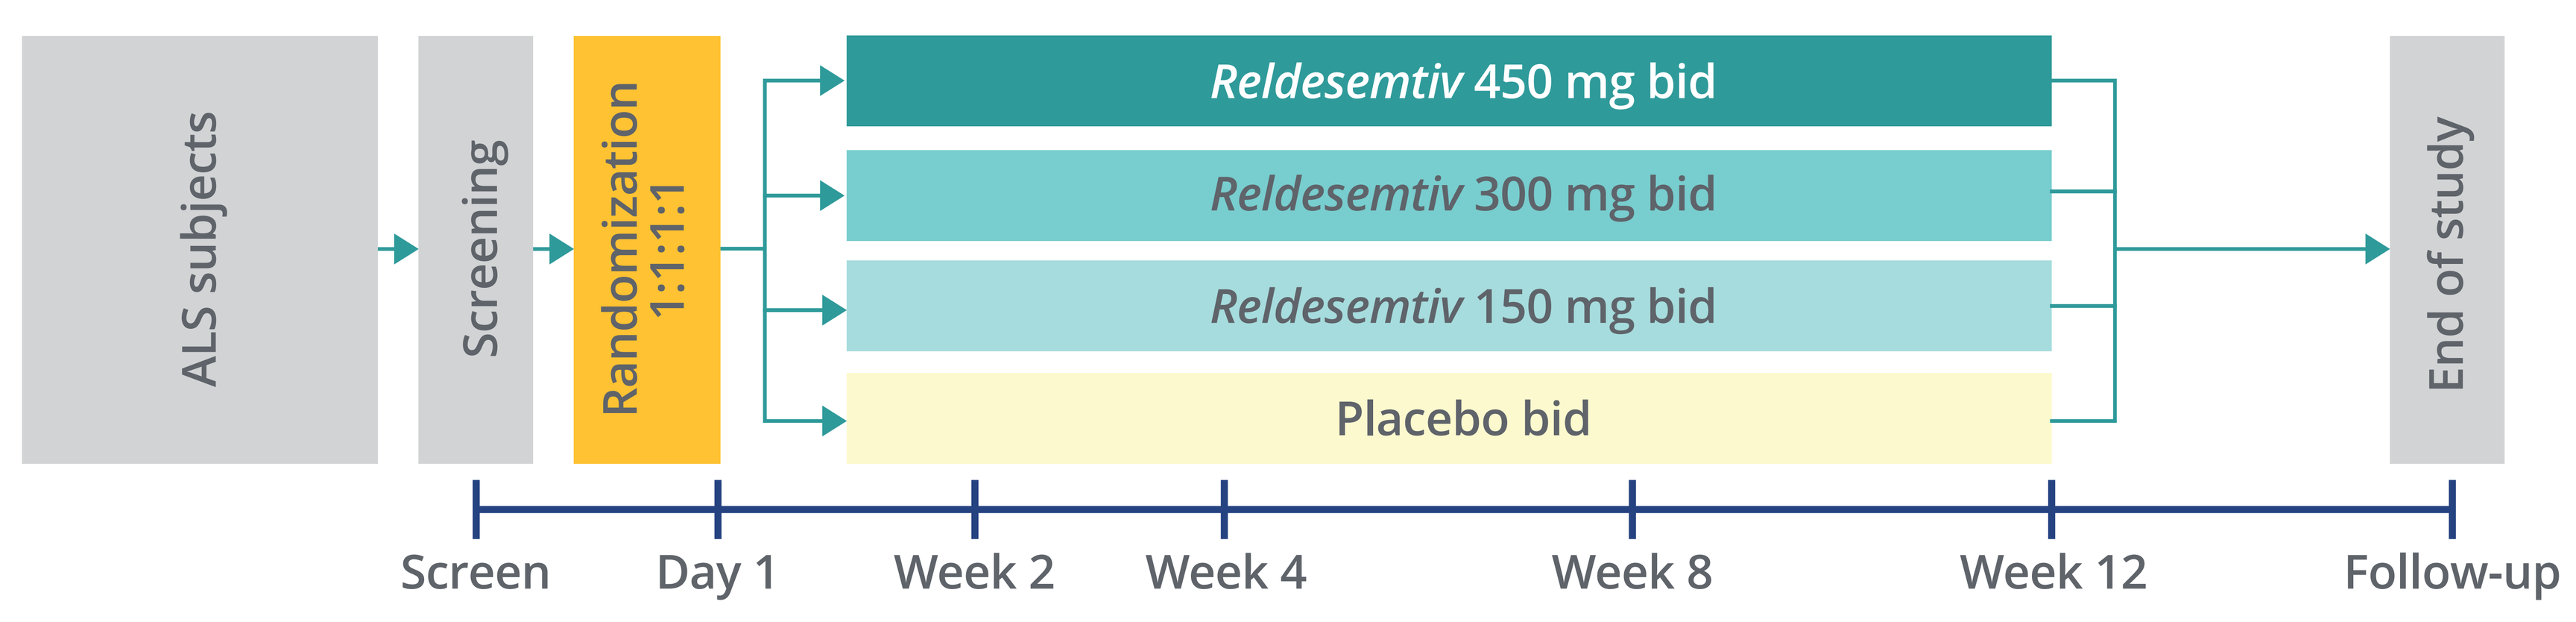
ALS subjects
Screening
Randomization 1:1:1:1
Reldesemtiv 450 mg bid
Reldesemtiv 300 mg bid
Reldesemtiv 150 mg bid
Placebo bid
End of study
Screen
Day 1
Week 2
Week 4
Week 8
Week 12
Follow-up

bid, twice daily.

DME-PAP assessment

- Information on health economic outcomes was collected for all patients in FORTITUDE-ALS, asking if and when any DMEs were prescribed and agreed to by the patient during the trial
- A prespecified Cox regression was used to compare DME-PAP on each dose level versus placebo, stratifying for riluzole/edaravone use
- A post hoc analysis compared DME-PAP for all *reldesemtiv* doses combined with placebo

RESULTS

Table 1. Baseline patient demographic and disease characteristics

Characteristic		Placebo (n = 115)	*Reldesemtiv* 150 mg (n = 112)	*Reldesemtiv* 300 mg (n = 113)	*Reldesemtiv* 450 mg (n = 117)	Overall (N = 457)
Age (years), mean (SD)		59.6 (10.6)	57.1 (10.9)	57.8 (10.2)	60.1 (11.0)	58.7 (10.7)
Male, n (%)		68 (59.1)	71 (63.4)	71 (62.8)	67 (57.3)	277 (60.6)
BMI (kg/m^2), mean (SD)		26.1 (4.4)	26.9 (5.1)	26.2 (4.4)	27.1 (4.6)	26.6 (4.6)
ALSFRS-R total score, mean (SD)		37.0 (5.6)	37.1 (5.5)	37.6 (5.6)	37.8 (5.5)	37.4 (5.5)
SVC (% predicted), mean (SD)		85.0 (14.8)	85.7 (14.8)	83.7 (14.5)	84.5 (17.1)	84.7 (15.3)
Months since diagnosis, mean (SD)		8.8 (6.3)	8.6 (6.4)	8.7 (6.1)	8.2 (5.6)	8.6 (6.1)
Months since first symptom, mean (SD)		22.1 (12.4)	23.9 (27.5)	22.5 (14.6)	22.7 (18.7)	22.8 (19.1)
ALS site of onset, n (%)	Bulbar	22 (19.1)	18 (16.1)	17 (15.0)	30 (25.6)	87 (19.0)
	Lower limb	44 (38.3)	42 (37.5)	40 (35.4)	43 (36.8)	169 (37.0)
	Upper limb	49 (42.6)	52 (46.4)	56 (49.6)	44 (37.6)	201 (44.0)
On riluzole alone, n (%)		64 (55.7)	64 (57.1)	64 (56.6)	66 (56.4)	258 (56.5)
On edaravone alone, n (%)		5 (4.3)	5 (4.5)	4 (3.5)	5 (4.3)	19 (4.2)
On riluzole and edaravone, n (%)		24 (20.9)	22 (19.6)	24 (21.2)	24 (20.5)	94 (20.6)

ALSFRS-R, ALS Functional Rating Scale-Revised; BMI, body mass index; SD, standard deviation; SVC, slow vital capacity.

DME-PAP in FORTITUDE-ALS

- New DME-PAP was significantly lower in the *reldesemtiv* 150 mg bid group and was numerically reduced in all *reldesemtiv*-treated groups versus placebo **(Figure 2)**
 - No one type of DME appeared to drive this result; with the exception of augmentative communication device, the percentage of each new DME for patients receiving placebo was higher than for all *reldesemtiv* combined
- New DME-PAP was delayed for all doses of *reldesemtiv* **(Figure 3A)**, and the probability significantly reduced for combined doses of *reldesemtiv* **(Figure 3B)** compared with placebo

Figure 2. DME-PAP by treatment group

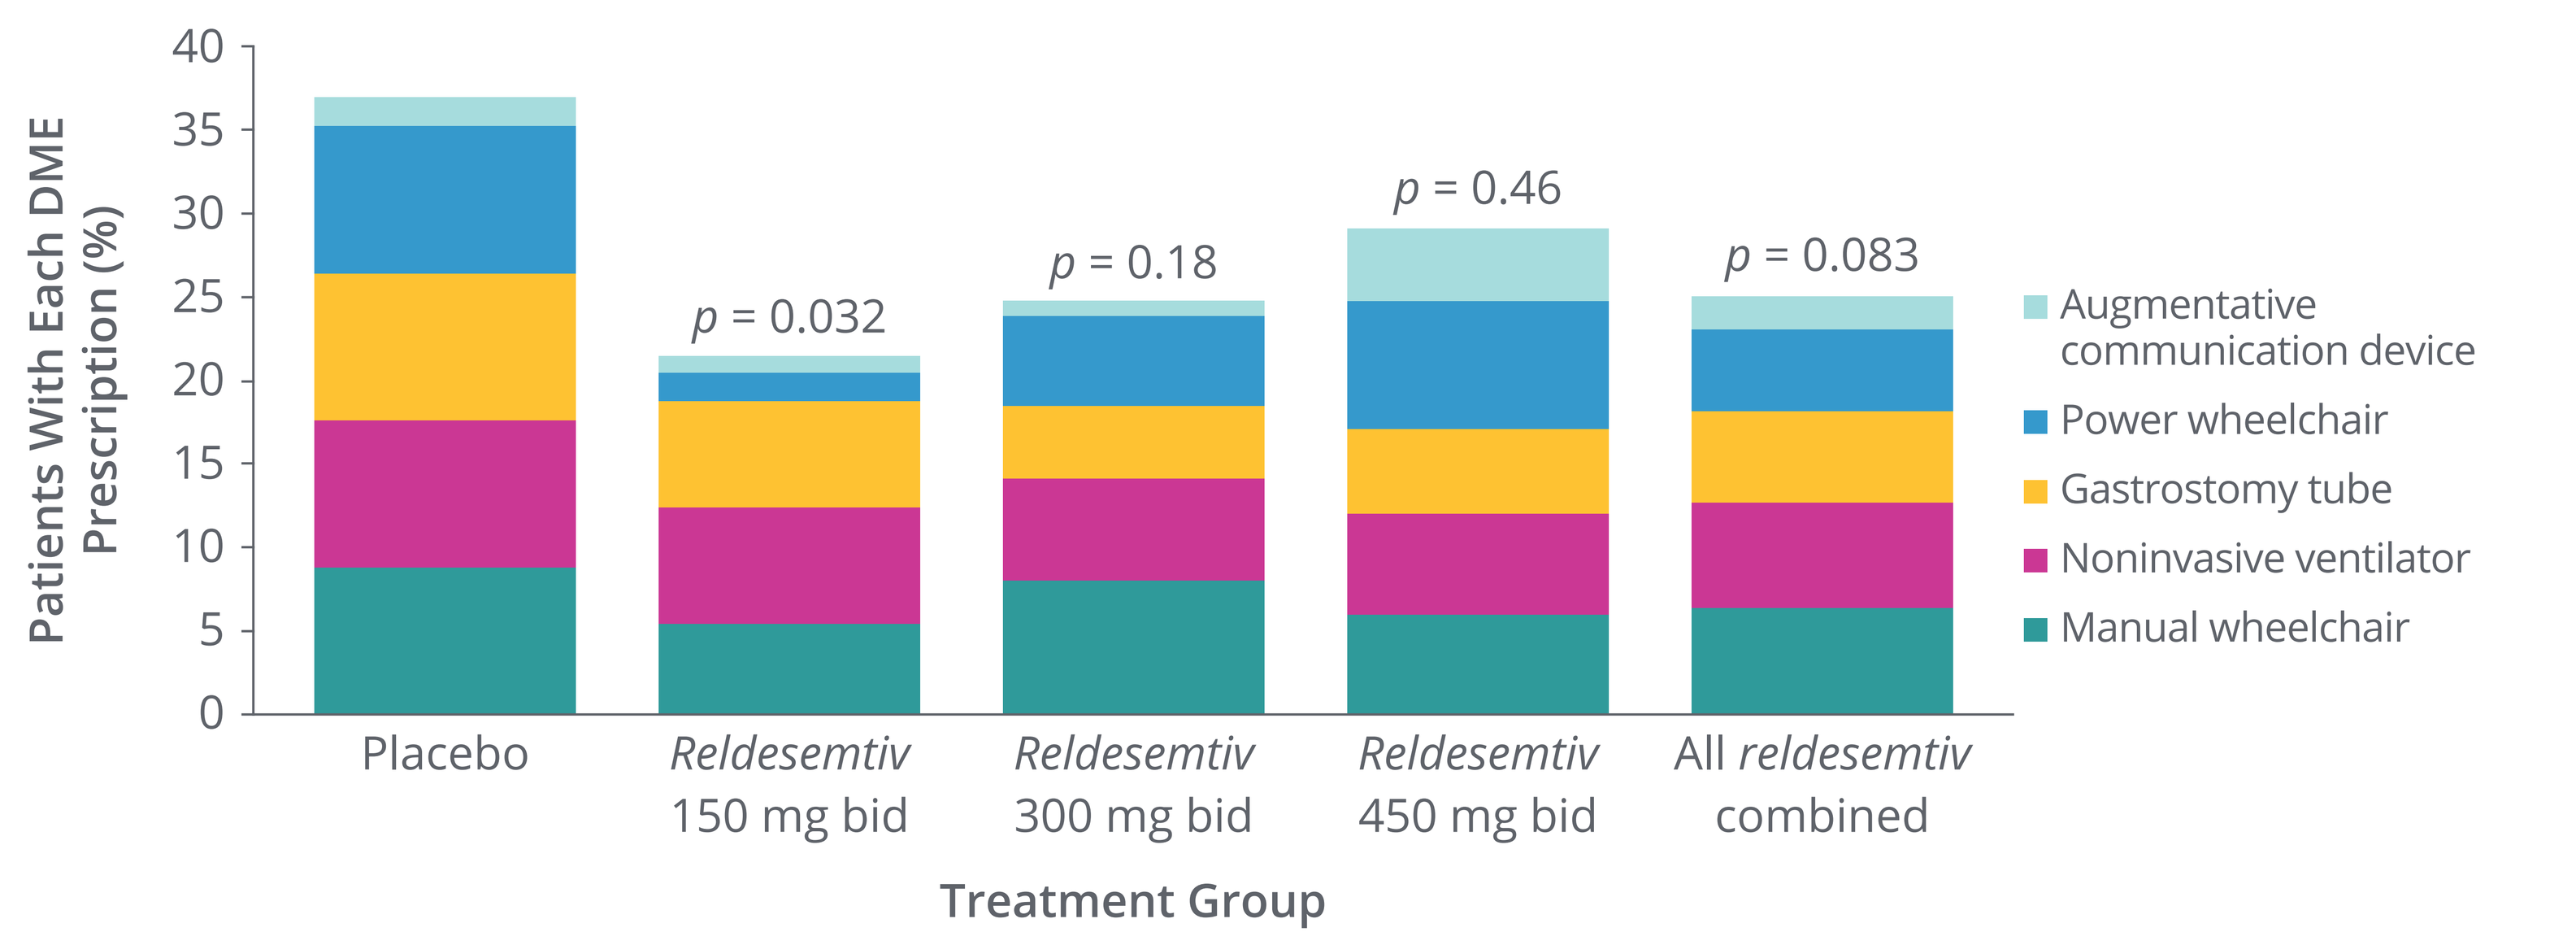
Patients With Each DME Prescription (%)
40
35
30
25
20
15
10
5
0
p = 0.032
p = 0.18
p = 0.46
p = 0.083
Placebo
Reldesemtiv 150 mg bid
Reldesemtiv 300 mg bid
Reldesemtiv 450 mg bid
All reldesemtiv combined
Treatment Group
Augmentative communication device
Power wheelchair
Gastrostomy tube
Noninvasive ventilator
Manual wheelchair

bid, twice daily; DME-PAP, durable medical equipment prescribed and agreed to by the patient.

Figure 3. Probability of no new DME-PAP over time with *reldesemtiv* treatment compared with placebo: (A) each dose; (B) all doses combined

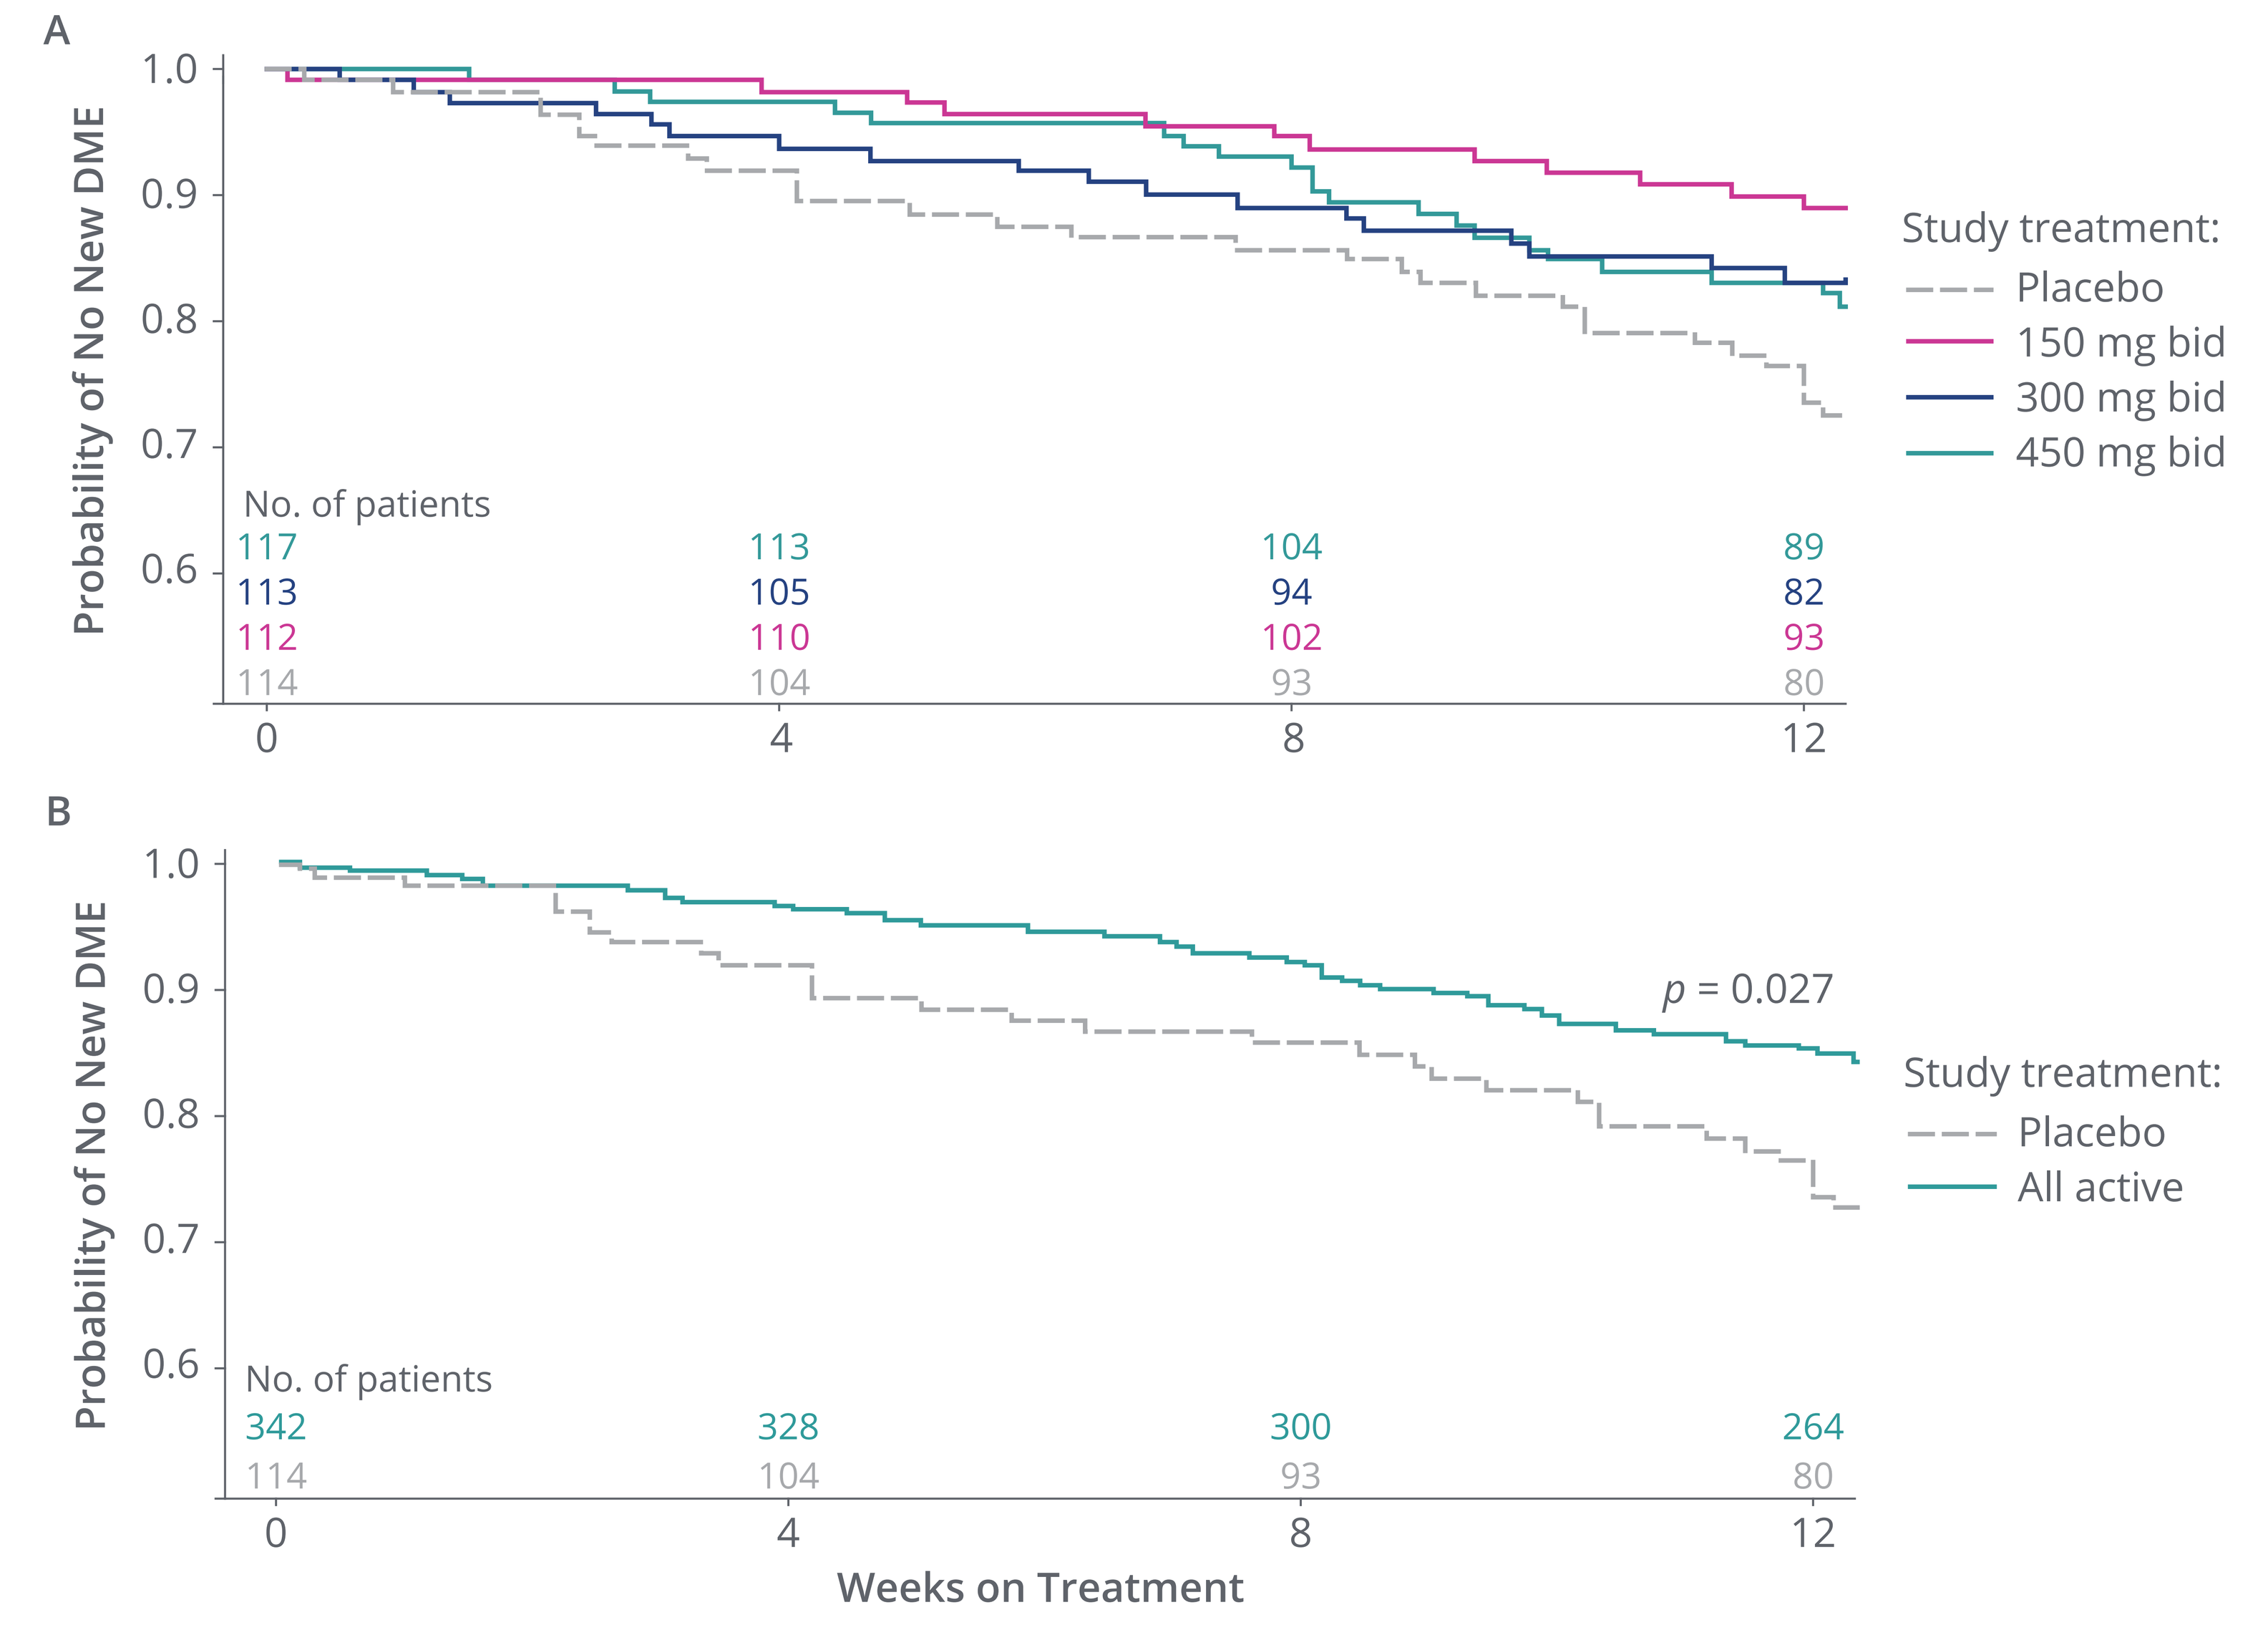
A
Probability of No New DME
1.0
0.9
0.8
0.7
0.6
No. of patients
117 113 104 89
113 105 94 82
112 110 102 93
114 104 93 80
0
4
8
12
Study treatment:
Placebo
150 mg bid
300 mg bid
450 mg bid
B
Probability of No New DME
1.0
0.9
0.8
0.7
0.6
p = 0.027
No. of patients
342 328 300 264
114 104 93 80
0
4
8
12
Weeks on Treatment
Study treatment:
Placebo
All active

bid, twice daily; DME-PAP, durable medical equipment prescribed and agreed to by the patient.

- The hazard ratio compared with placebo for accepting at least 1 DME was significantly reduced for those taking *reldesemtiv* 150 mg bid and trended toward reduction in the other dose groups **(Table 2)**
 - For all *reldesemtiv* doses combined, the hazard ratio compared with placebo was significantly lower
- *Reldesemtiv* treatment delayed 25th percentile time to agreeing to the first DME from 84 days for placebo to 120 days

Table 2. Hazard ratios compared with placebo for ≥ 1 DME-PAP

Reldesemtiv Group	Hazard Ratio Versus Placebo	95% CI	*p* Value
150 mg bid	0.45	0.25, 0.81	0.01
300 mg bid	0.72	0.42, 1.25	0.25
450 mg bid	0.70	0.42, 1.19	0.19
All doses combined	0.62	0.40, 0.95	0.03

bid, twice daily; CI, confidence interval; DME-PAP, durable medical equipment prescribed and agreed to by the patient.

CONCLUSIONS

- Patients on *reldesemtiv* in FORTITUDE-ALS, a 12-week dosing study with 4-week follow-up:
 - Had 38% lower risk of accepting DME related to impaired mobility, breathing, swallowing, or speaking compared with those receiving placebo
 - *Reldesemtiv* treatment delayed time to 25% of the population accepting DME by 36 days compared to placebo
- Using time to a patient agreeing to receive prescribed DME items as an outcome measure in ALS trials may provide additional insights into the potential impact investigational drugs may have on disease progression

Acknowledgments

We wish to thank the participants of FORTITUDE-ALS and their families for their contributions to this clinical trial, the investigators of FORTITUDE-ALS, and members of the Data Monitoring Committee and Steering Committee.

The study was funded by Cytokinetics, Inc.

Editorial support was provided by Jennifer Giel on behalf of Evidence Scientific Solutions, Inc, Philadelphia, PA, and was funded by Cytokinetics, Inc.

Disclosures

Andrews has served as a consultant for Avexis, Biohaven, and Clene Nanomedicine; has served as a consultant for Cytokinetics and is a former employee of Cytokinetics; and has received research support from Biogen, Neuraltus, Orion, and Roche. **Genge** has served as a consultant for AB Sciences, AL-S Pharma, Avexis, Biogen, Cytokinetics, MTPA, and Roche. **Jackson** has served as a consultant for Argenex, Cytokinetics, ITF Pharma, MTPA, and Strongbridge Pharmaceuticals; served on Speaker's Bureau for Avanir, CSL Behring, MTPA, and Strongbridge Pharmaceuticals; has received research support from Amylyx, Cytokinetics, and NIH; and is currently serving as a member of a Data Safety Monitoring Board for Anelixis, Brainstorm, and Mallinckrodt. **Lechtzin** has served as a consultant/advisor for Cytokinetics, Hill-Rom, and Vertex; and has received research support from AstraZeneca and Vertex. **Miller** has served as a consultant/advisor for Biogen and Cytokinetics; has received research support from Biogen and Ionis; and receives licensing fees from C2N. **Shefner** has served as a consultant for Biogen, Biohaven, Cytokinetics, MT Pharma America, and Novartis; has received research support from Amylyx, Biogen, Biohaven, Biotie, Cytokinetics, MT Pharma America, Neuraltus, and Orphazyme; and has received compensation from UpToDate for serving as neuromuscular section editor. **Cockroft** was an employee of Cytokinetics at the time of the study and owns stock in Cytokinetics. **Rudnicki, Malik, Meng, Wei,** and **Wolff** are employees of and own stock in Cytokinetics.

In collaboration with Astellas Pharma, Inc., Cytokinetics is developing *reldesemtiv* as a potential treatment for people living with ALS and certain other debilitating diseases and conditions associated with skeletal muscle weakness and/or fatigue.

To obtain a PDF of this poster:
scan the QR code
OR
visit http://bit.ly/2qwAh9c.
No personal information is stored.

astellas Cytokinetics

Presented at the 30th Symposium on ALS/MND | December 4–6, 2019 | Perth, Australia