Characterization of the Cardiac Myosin Inhibitor CK-3773274: a Potential Therapeutic Approach for Hypertrophic Cardiomyopathy

2912-Pos

James J. Hartman, Darren T. Hwee, Jingying Wang, Yangsong Wu, Julia Schaletzky, Preeti Paliwal, Ken Lee, Khanha D. Taheri, Eddie Wehri, Todd J.A. Ewing, Joseph P. Michel, Chihyuan Chuang, Eva R. Chin, Bradley P. Morgan, Fady I. Malik

Cytokinetics Inc, South San Francisco, CA, USA

ABSTRACT

Hypercontractility of the cardiac sarcomere may be essential for the underlying pathological hypertrophy and fibrosis in genetic hypertrophic cardiomyopathies. Here, we characterize the small molecule, CK-3773274, as a novel cardiac myosin inhibitor that decreases contractility in vitro and in vivo. In bovine cardiac myofibrils, CK-3773274 decreased myosin ATPase activity in a concentration-dependent fashion (IC_{50}:1.26 μM). CK-3773274 selectively targets cardiac myosin, as it reduced myosin ATPase activity in the absence of other sarcomere proteins, including actin, troponin, and tropomyosin. Importantly, CK-3773274 did not inhibit the ATPase activity of smooth muscle myosin. In transient kinetic studies, CK-3773274 substantially slowed the rate of actin-activated phosphate release, likely stabilizing myosin conformations that bind weakly to actin. Binding of CK-3773274 to cardiac myosin is mutually exclusive with the nonspecific myosin II inhibitor blebbistatin, suggesting they bind to the same or overlapping sites. Consistent with its biochemical phenotype, CK-3773274 (10 μM) reduced fractional shortening (FS) by 84% in electrically paced, isolated adult rat cardiomyocytes relative to control without any effect on the calcium (Ca^{2+}) transient. The effect of CK-3773274 on cardiac contractility in vivo was assessed in healthy male Sprague Dawley (SD) rats using single oral doses ranging from 0.5 to 4 mg/kg. Left ventricular dimensions and FS were determined by echocardiography at select time points over a 24-hour period. One hour after dose administration, CK-3773274 significantly reduced FS in a dose-related fashion by 20% to 70% relative to vehicle treatment without any changes to heart rate. In conclusion, CK-3773274 is a novel, small molecule, cardiac myosin inhibitor that reduces cardiac contractility in vitro and in vivo. Cardiac myosin inhibition may be a viable approach to treat the underlying hypercontractility of the cardiac sarcomere in hypertrophic cardiomyopathies.

INTRODUCTION

- Hypercontractility of the cardiac sarcomere appears to underlie pathological hypertrophy and fibrosis in select genetic hypertrophic cardiomyopathies.
- Direct modulation of the sarcomere is a novel approach to potentially treat conditions with maladaptive changes in cardiac contractility (Malik et al. 2011, Green et al. 2016).
- The objective of this study was to characterize the biochemical mechanism of action of the small molecule CK-3773274, and the ability to modulate cardiac contractility in vitro and in vivo.

METHODS

Preparation of Reagents

- Myofibrils were prepared from flash-frozen bovine cardiac, bovine masseter, and rabbit psoas tissue as described in Hwee et al. (2015). Bovine cardiac myosin subfragment-1 (S1) was prepared as described in Malik et al. (2011).

ATPase Assays

- Steady-state ATPase activity was measured using a pyruvate kinase and lactate dehydrogenase-coupled enzyme system as described in Hwee et al. (2015) and Malik et al. (2011). Non-myosin ATPase activity was subtracted from cardiac and slow skeletal myofibril assays (where indicated) by subtracting the ATPase activity in the presence of a saturating concentration of the non-selective myosin II inhibitor (-)-blebbistatin.

Blebbistatin Binding Assays

- Binding of (-)-blebbistatin to bovine cardiac myosin subfragment-1 was measured in a buffer consisting of 12 mM K-PIPES pH 6.8, 2 mM $MgCl_2$, 1 mM DTT, and 2 mM ADP-vanadate. Fluorescence emission spectra were recorded using a PTI QM-6 (λ_{ex} 426 nm). Compound titrations were performed using a SpectraMax Gemini XS spectrofluorimeter (λ_{ex} 426 nm, λ_{em} 575 nm).

Transient Kinetics

- Transient kinetics were measured at 25°C using a SF-61DX stopped-flow instrument (TgK Scientific) in a buffer consisting of 12 mM K-PIPES pH 6.8, 2 mM $MgCl_2$, and 1 mM DTT using the methods of De La Cruz and Ostap (2009) and Malik et al. (2011). ATP binding and hydrolysis were monitored by intrinsic tryptophan fluorescence (λ_{ex} 295 nm) using a 320 nm longpass filter. Actin-activated phosphate release was monitored using MDCC-labeled phosphate binding protein (λ_{ex} 434 nm) using a 455 nm longpass filter.

Measurement of Cardiomyocyte Contractility and Calcium Transients

- Adult rat ventricular cardiomyocytes were isolated and loaded with Fura-2 as described in Malik et al. (2011). Cardiomyocyte contractility and calcium transients were measured by edge-detection video microscopy and fluorescence photometery (IonOptix, Milton, MA) as described in Malik et al. (2011).

Rat Echocardiography Assessment

- Adult male Sprague Dawley rats were anesthetized with inhaled isoflurane (1-5%) throughout the echocardiography procedure. Baseline contractility was assessed 1 day prior to CK-3773274 treatment. Animals were orally dosed with vehicle (0.5% hydroxypropylmethylcellulose (HMPC)/ 0.1% Tween-80) or CK-3773274 (0.5, 1, 2, or 4 mg/kg) and measures of left ventricular contractility were assessed 1, 4, 8, and 24 hours post-dose. Using a GE Vivid7 machine, a 10 MHz probe was placed at the level of the papillary muscles and 2D M-mode images of the left ventricle were captured. Images and measurements were obtained in parasternal long axis view. In vivo percent fractional shortening was determined by analysis of the M-mode images using the GE Vivid7 ultrasound software.

RESULTS

Figure 1. CK-3773274 inhibits the ATPase activity of bovine cardiac myofibrils

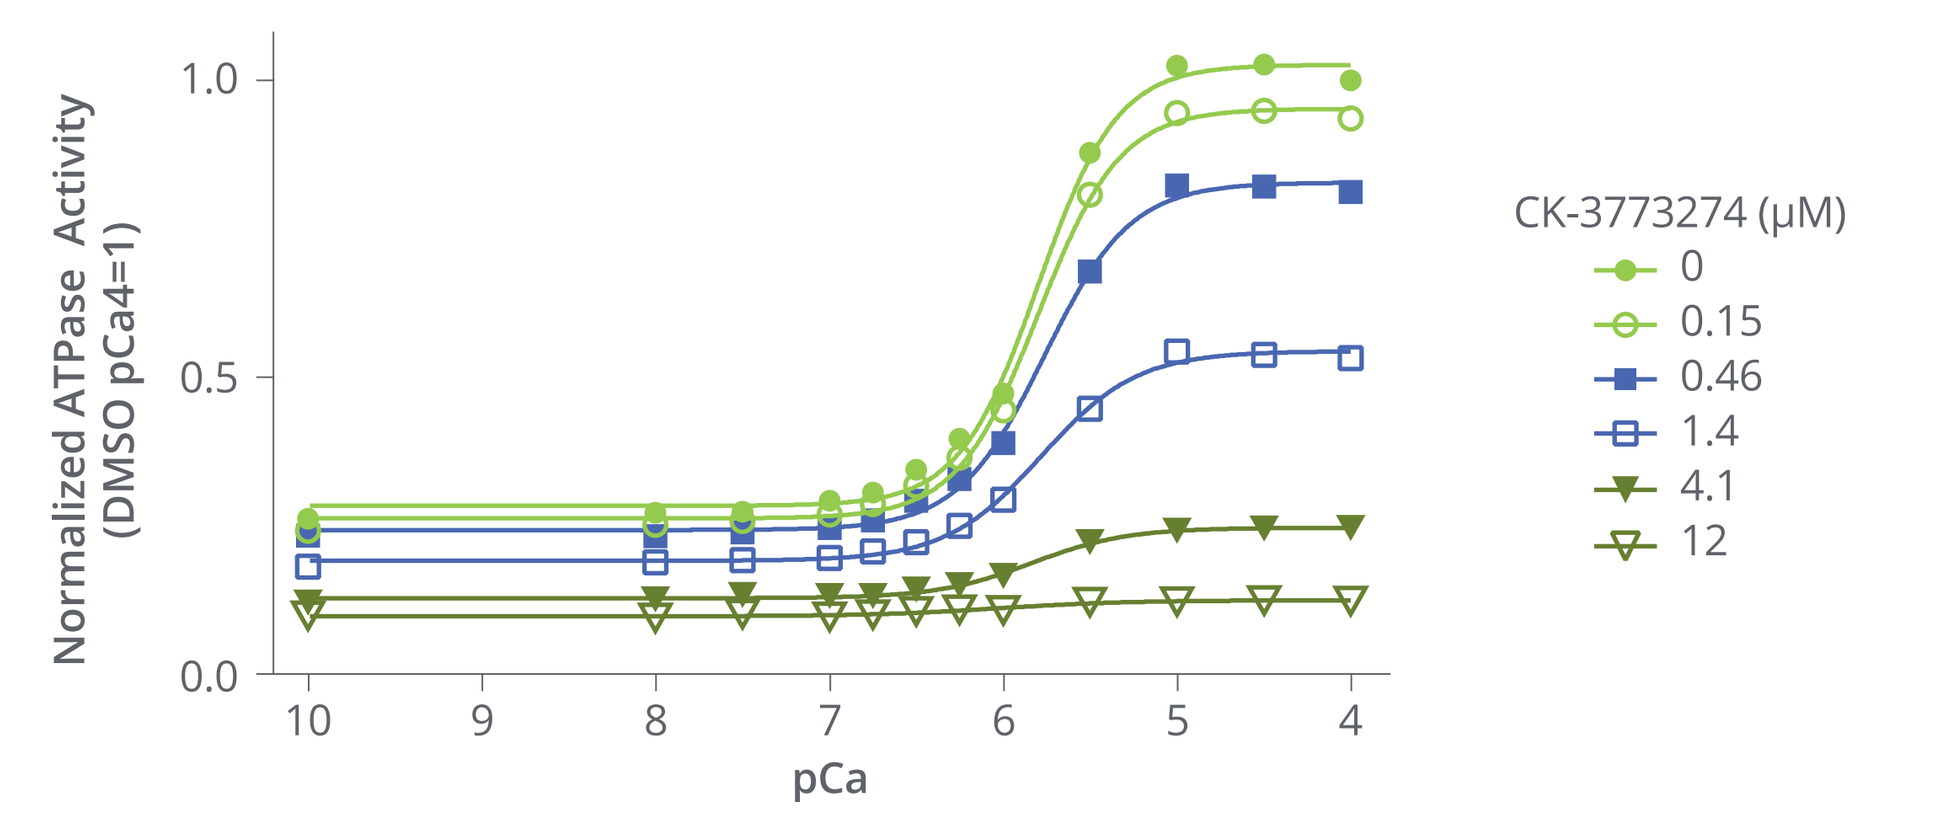
Normalized ATPase Activity (DMSO pCa4=1)
pCa
CK-3773274 (μM)
0
0.15
0.46
1.4
4.1
12

Best-Fit Values	DMSO	CK-3773274 0.15 μM	0.46 μM	1.4 μM	4.1 μM	12 μM
pCa_{50}	5.80 (5.77 to 5.83)	5.80 (5.77 to 5.82)	5.76 (0.573 to 5.79)	5.78 (5.75 to 5.81)	5.83 (5.77 to 5.90)	6.03 (5.72 to 6.33)
Hill slope	−1.91 (−2.09 to −1.73)	−1.85 (−2.00 to −1.71)	−1.75 (−1.90 to −1.59)	−1.58 (−1.72 to −1.45)	−1.59 (−1.90 to −1.28)	−1.16 (−2.02 to −0.30)
Minimum	0.28 (0.27 to 0.29)	0.26 (0.25 to 0.27)	0.24 (0.24 to 0.25)	0.19 (0.19 to 0.20)	0.13 (0.12 to 0.13)	0.10 (0.09 to 0.10)
Maximum	1.03 (1.01 to 1.04)	0.95 (0.94 to 0.96)	0.83 (0.82 to 0.84)	0.54 (0.54 to 0.55)	0.25 (0.24 to 0.25)	0.12 (0.12 to 0.13)

- The ATPase activity of Triton X-100-skinned bovine cardiac myofibrils was measured using a pyruvate kinase/ lactate dehydrogenase-coupled assay as described in Hwee et al. (2015). The table shows the results of fitting to a four-parameter dose-response equation along with the 95% confidence interval of the fitted parameters.

Data shown are mean values ± standard deviation (SD) (n=4).

Figure 2. CK-3773274 selectively inhibits the ATPase activity of cardiac and slow skeletal myofibrils compared with fast skeletal myofibrils

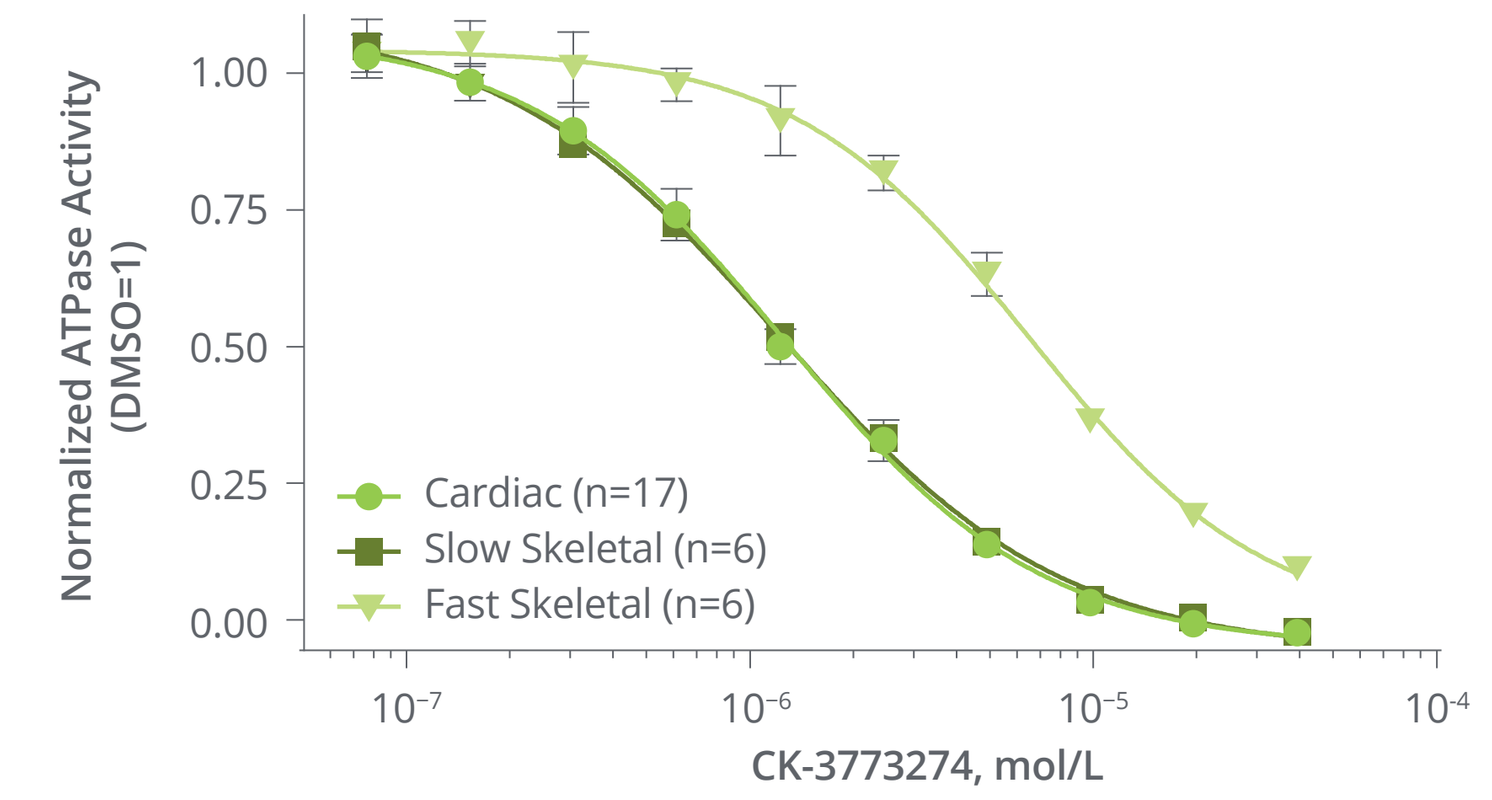
Normalized ATPase Activity (DMSO=1)
CK-3773274, mol/L
Cardiac (n=17)
Slow Skeletal (n=6)
Fast Skeletal (n=6)

	Cardiac	Slow Skeletal	Fast Skeletal
IC_{50} (μM)	1.26 (1.20 to 1.33)	1.23 (1.17 to 1.29)	6.52 (5.72 to 7.71)
Hill slope	−1.16 (−1.24 to −1.10)	−1.06 (−1.12 to −1.00)	−1.27 (−1.49 to −1.08)
Top	1.07 (1.05 to 1.09)	1.10 (1.08 to 1.12)	1.04 (1.02 to 1.07)
Bottom	−0.04 (−0.06 to −0.03)	−0.05 (−0.07 to −0.04)	−0.005 (−0.09 to 0.06)

- Dose-response analysis was performed with cardiac (bovine, n=17), slow skeletal (bovine, n=6), and fast skeletal (rabbit, n=6) detergent-skinned myofibrils as described in Hwee et al. (2015). Free Ca^{2+} concentrations were fixed at approximately the pCa_{75} for each myofibril type. Raw ATPase rates were normalized to reactions containing an equivalent concentration of DMSO, and for cardiac and slow skeletal reactions, ATPase rates in the presence of the nonselective myosin inhibitor blebbistatin were subtracted to eliminate the effects of non-myosin ATPases. The table shows the results of fitting to a four-parameter dose-response equation along with the 95% confidence interval.

Data shown are mean values ± SD.

Figure 3. CK-3773274 inhibits the ATPase activity of bovine cardiac myosin subfragment-1

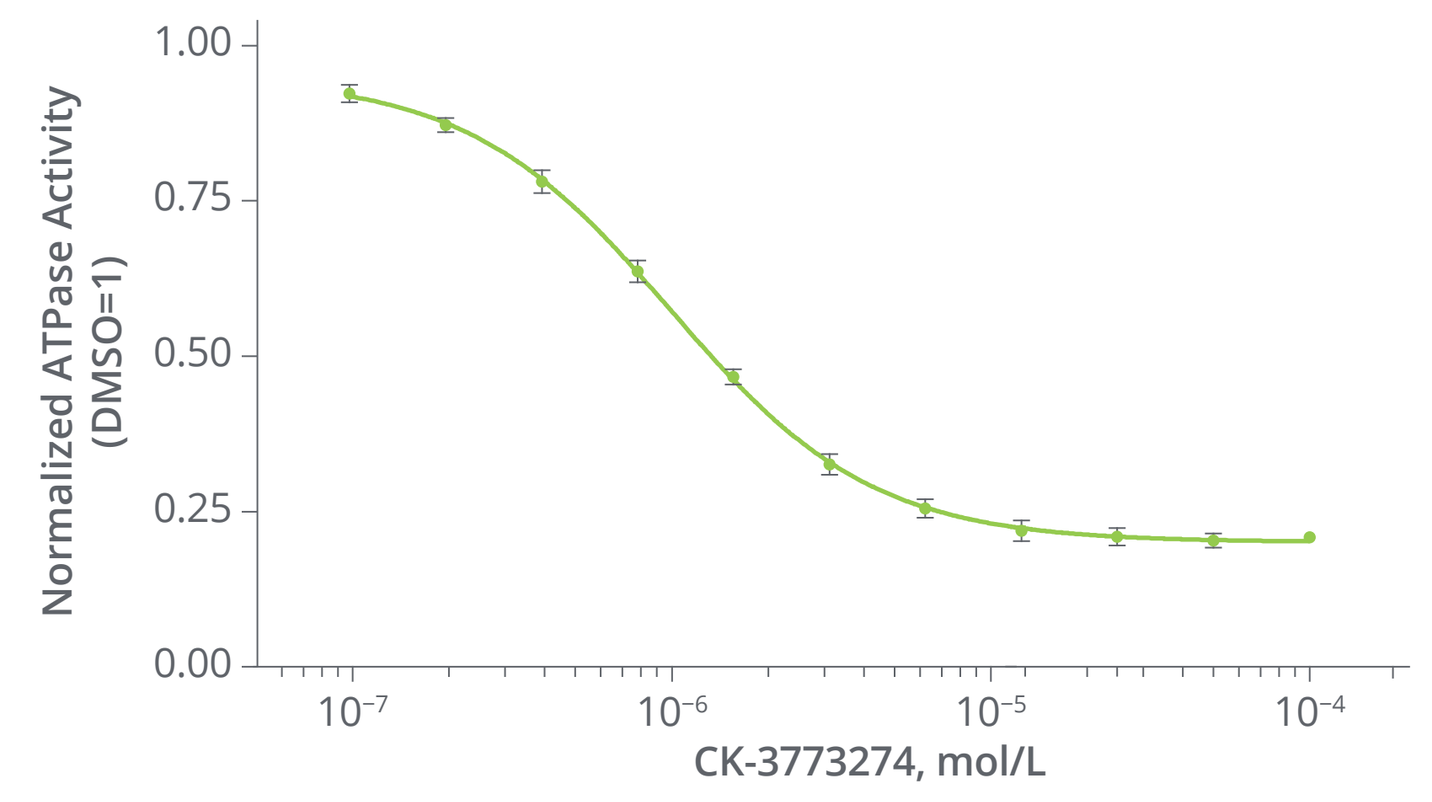
Normalized ATPase Activity (DMSO=1)
CK-3773274, mol/L

- The basal ATPase activity of bovine cardiac myosin subfragment-1 (1 μM) was measured using a pyruvate kinase/lactate dehydrogenase-coupled assay in a buffer consisting of 12 mM K-PIPES pH 6.8, 2 mM $MgCl_2$, 1 mM DTT, and 1 mM ATP. ATPase activity was normalized to control reactions containing 2% DMSO. Data were fitted using a four-parameter dose response equation (95% CI): IC_{50} = 0.99 μM (0.95 to 1.03), Hill slope = −1.37 (−1.44 to −1.30). Top = 0.95 (0.94 to 0.96), Bottom = 0.20 (0.19 to 0.21).

Data shown are mean values ± SD (n=8 reactions).

Figure 4. CK-3773274 does not appreciably inhibit the actin-activated ATPase activity of smooth muscle myosin subfragment-1

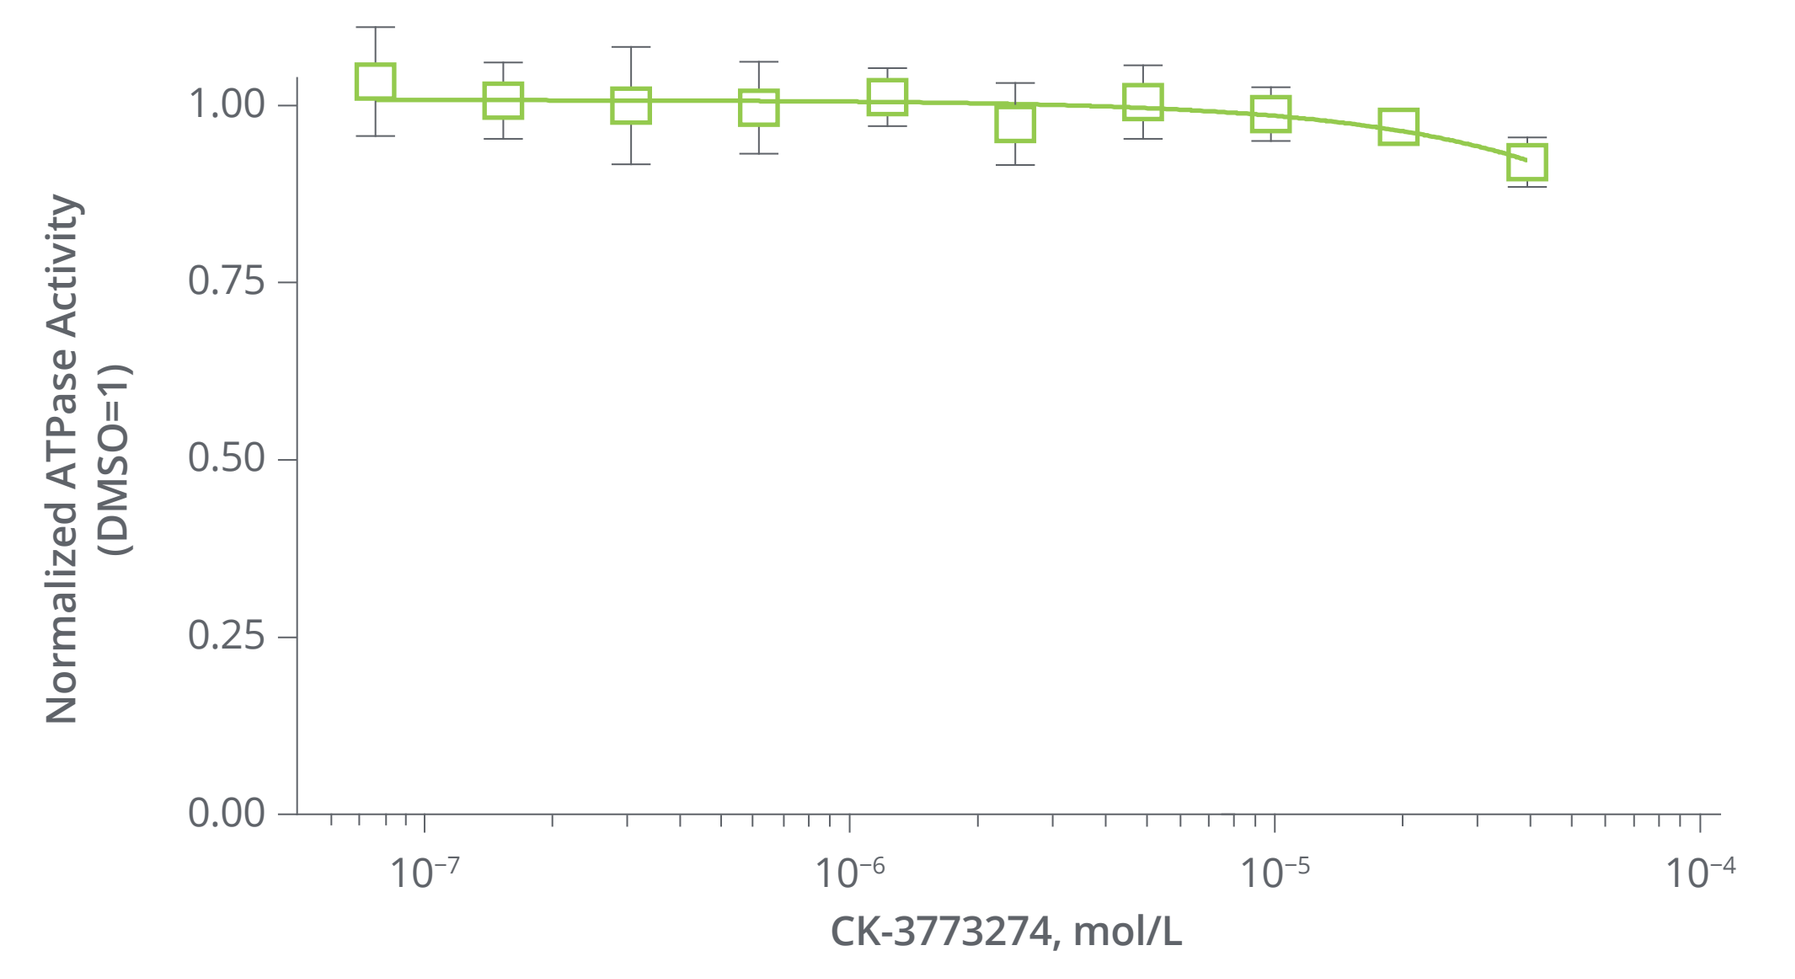
Normalized ATPase Activity (DMSO=1)
CK-3773274, mol/L

- The ATPase activity of chicken gizzard smooth muscle myosin subfragment-1 was measured after crosslinking to bovine cardiac actin using EDC/NHS to maximize the actin-activated ATPase activity of the enzyme. ATPase activity was measured using a pyruvate kinase/lactate dehydrogenase-coupled assay in a buffer consisting of 12 mM K-PIPES pH 6.8, 2 mM $MgCl_2$, 1 mM DTT, and 1 mM ATP. ATPase activity was normalized to control reactions containing 2% DMSO.

Data shown are mean values ± SD (n=6 reactions).

Figure 5. CK-3773274 and blebbistatin binding to cardiac myosin is mutually exclusive

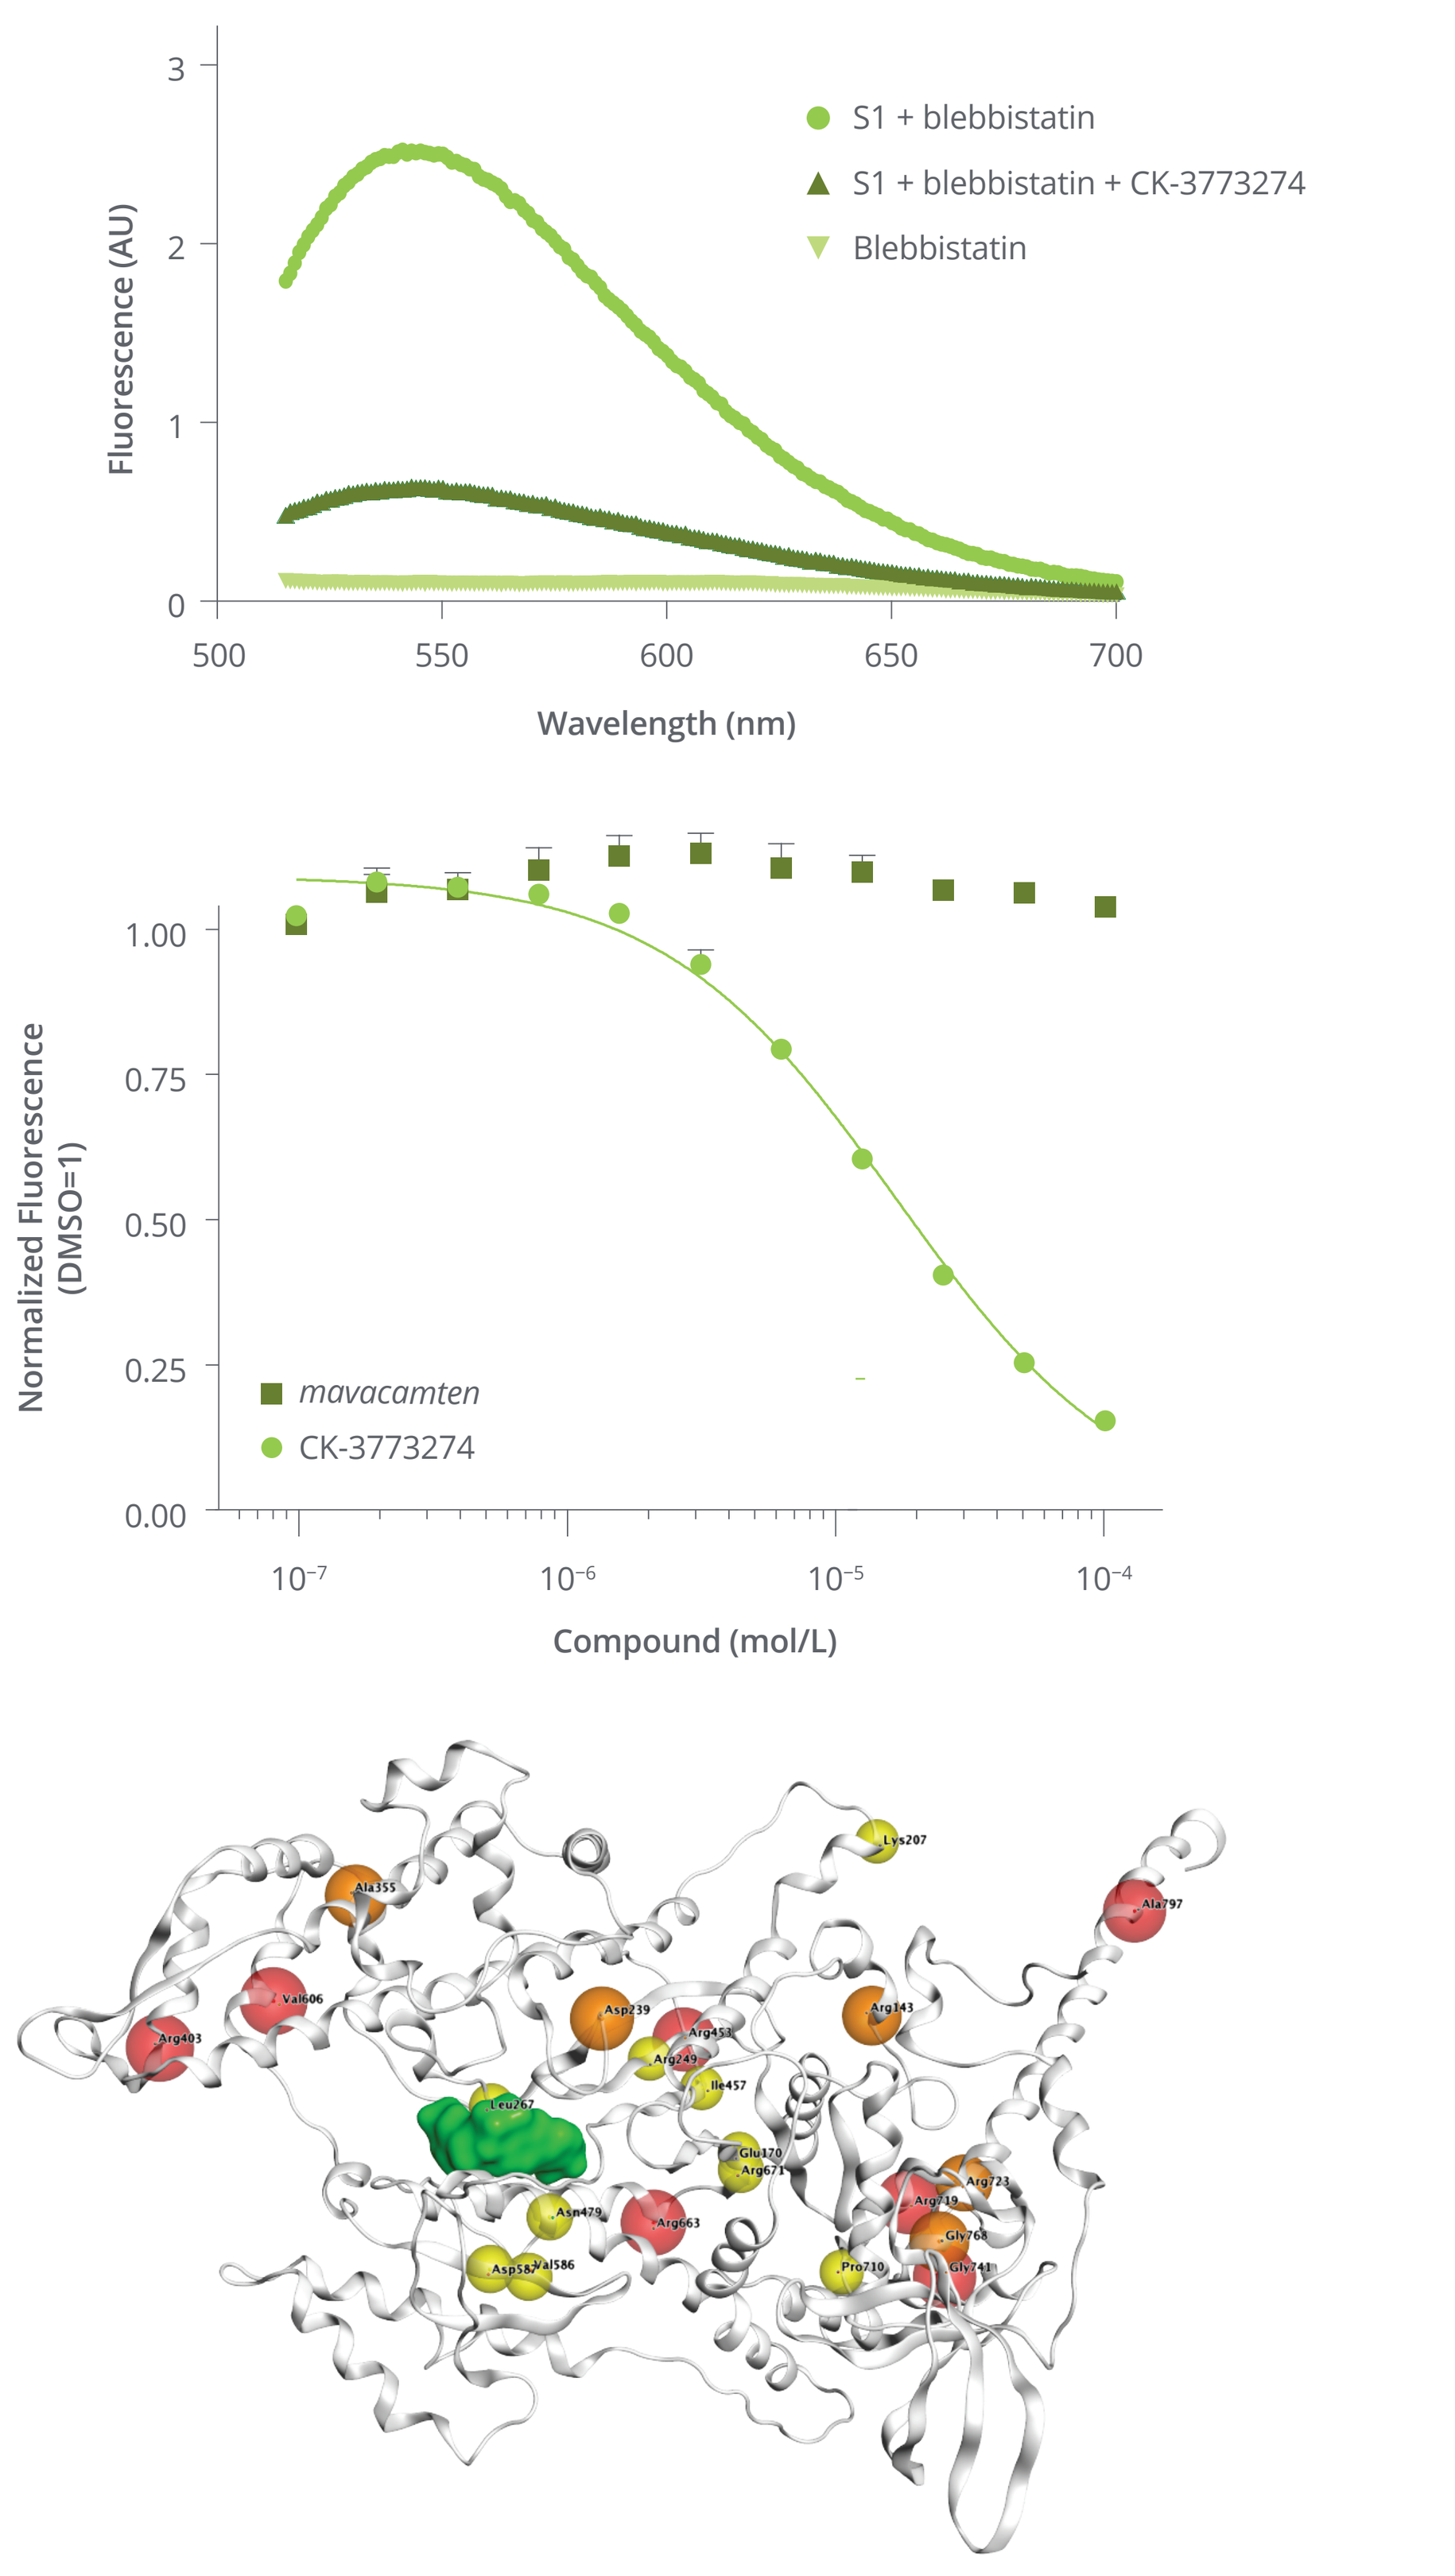
(A)
Fluorescence (AU)
S1 + blebbistatin
S1 + blebbistatin + CK-3773274
Blebbistatin
Wavelength (nm)
(B)
Normalized Fluorescence (DMSO=1)
mavacamten
CK-3773274
Compound (mol/L)
(C)

A. Fluorescence emission of (-)-blebbistatin (5 μM) increases upon binding to bovine cardiac myosin subfragment-1 (1 μM) in the ADP-VO_4 state. This increase is reduced when a high concentration of CK-3773274 (50 μM) is present. **B.** CK-3773274 (but not *mavacamten*) reduces myosin-enhanced blebbistatin fluorescence in a concentration-dependent manner, consistent with mutually exclusive binding. **C.** Structure of bovine cardiac myosin (PDB 5N69, Planelles-Herrero et al. [2017] with the blebbistatin binding site (PDB 1YV3) identified by Allingham et al. (2005) indicated in green. Also highlighted are select HCM-associated MYH7 mutations with high (red, >0.003%), medium (orange, 0.001-0.003%), and low (yellow, <0.001%) prevalence based on Walsh et al. (2017).

Figure 6. CK-3773274 slows actin-activated phosphate release without affecting ATP binding or hydrolysis

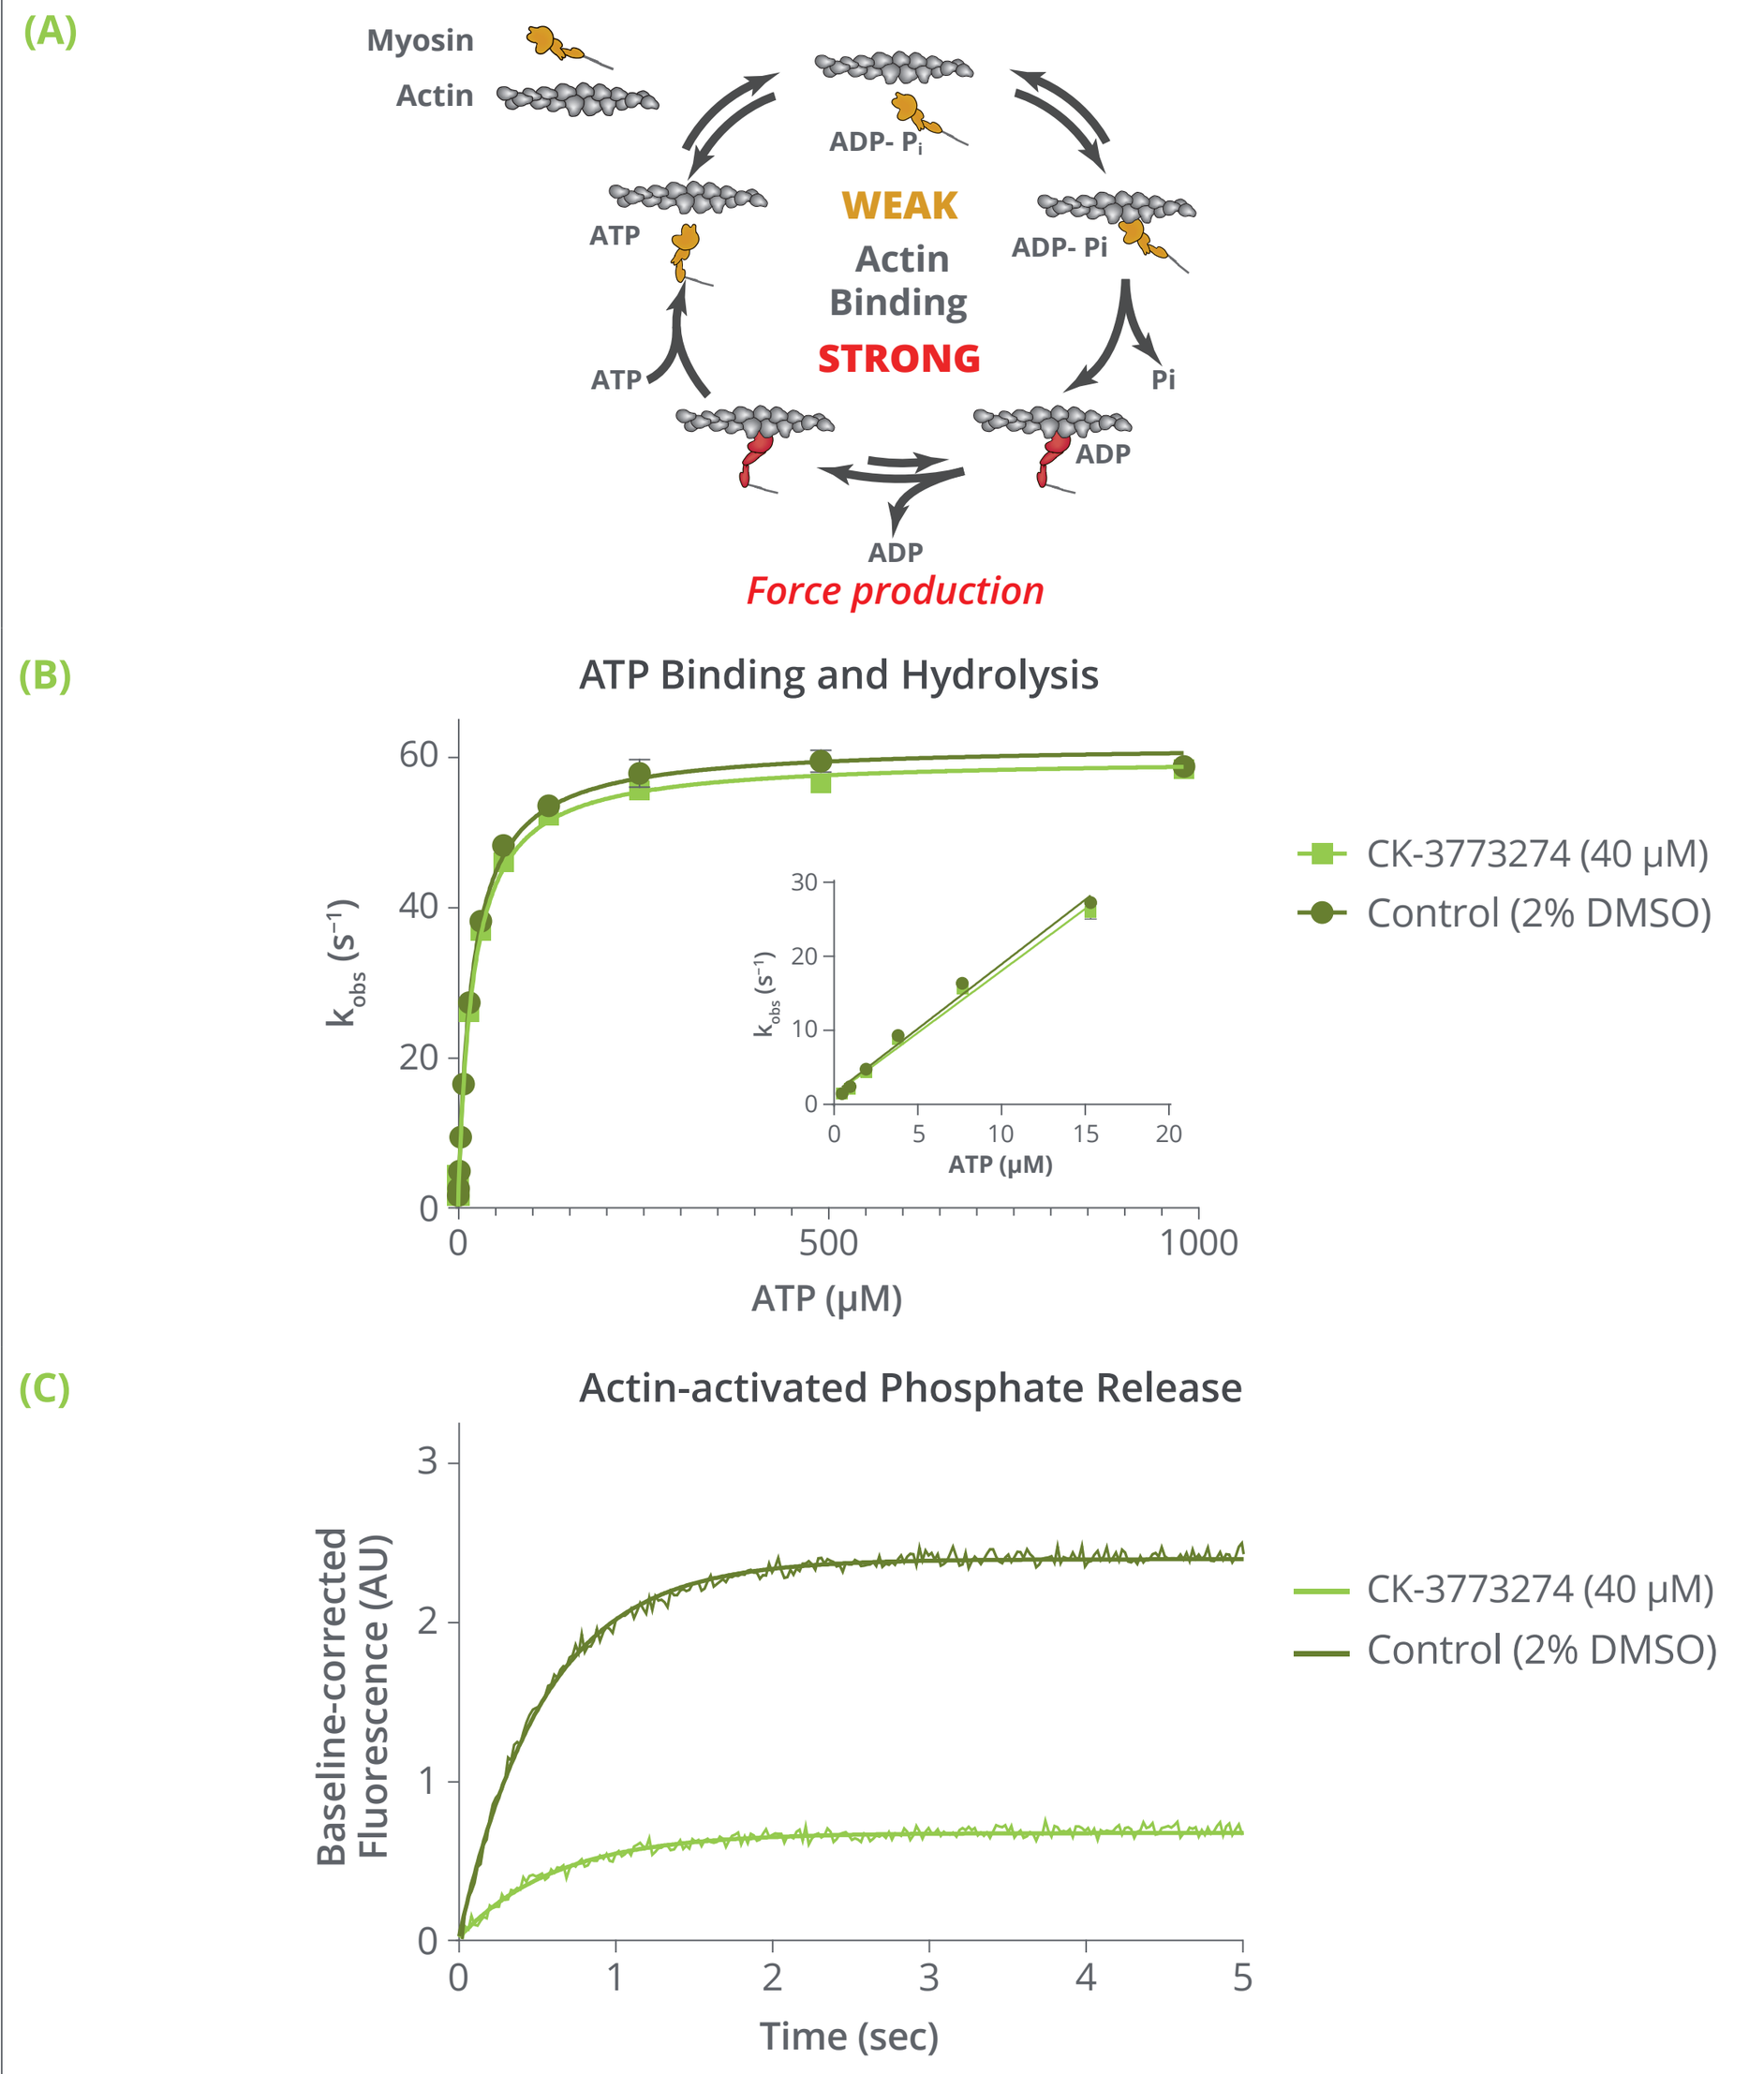
(A)
Myosin
Actin
WEAK
Actin Binding
STRONG
Force production
(B)
ATP Binding and Hydrolysis
ATP (μM)
CK-3773274 (40 μM)
Control (2% DMSO)
(C)
Actin-activated Phosphate Release
Baseline-corrected Fluorescence (AU)
Time (sec)
CK-3773274 (40 μM)
Control (2% DMSO)

A. Myosin chemomechanical cycle. **B.** ATP binding and hydrolysis were measured by following the intrinsic tryptophan fluorescence of bovine cardiac myosin subfragment-1 (1 μM final concentration) after rapid mixing with varying concentrations of ATP. Three to five fluorescence transients were averaged and fit to a single exponential. Data shown are mean ± SD (n=3 separate experiments). DMSO: $k_{+H} + k_{-H}$ = 62 s^{-1}, ATP binding rate = 1.7E6 M^{-1} s^{-1}. CK-3773274: $k_{+H} + k_{-H}$ = 60 s^{-1}, ATP binding rate 1.7E6 M^{-1} s^{-1}. **C.** Actin-activated phosphate release was measured by rapidly mixing bovine cardiac myosin subfragment-1 (2 μM) with ATP (1 μM), aging the reaction for 30 seconds, followed by mixing with bovine cardiac actin (28 μM) and MDCC-PBP (10 μM). The graph shows the average of 4-5 fluorescence transients, averaged and fit to a single exponential. DMSO: rate = 1.8 s^{-1}, amplitude = 2.4. CK-3773274: rate = 1.6 s^{-1}, amplitude = 0.67.

Figure 7. CK-3773274 decreases shortening of isolated adult rat ventricular myocytes without altering Ca^{2+} transients

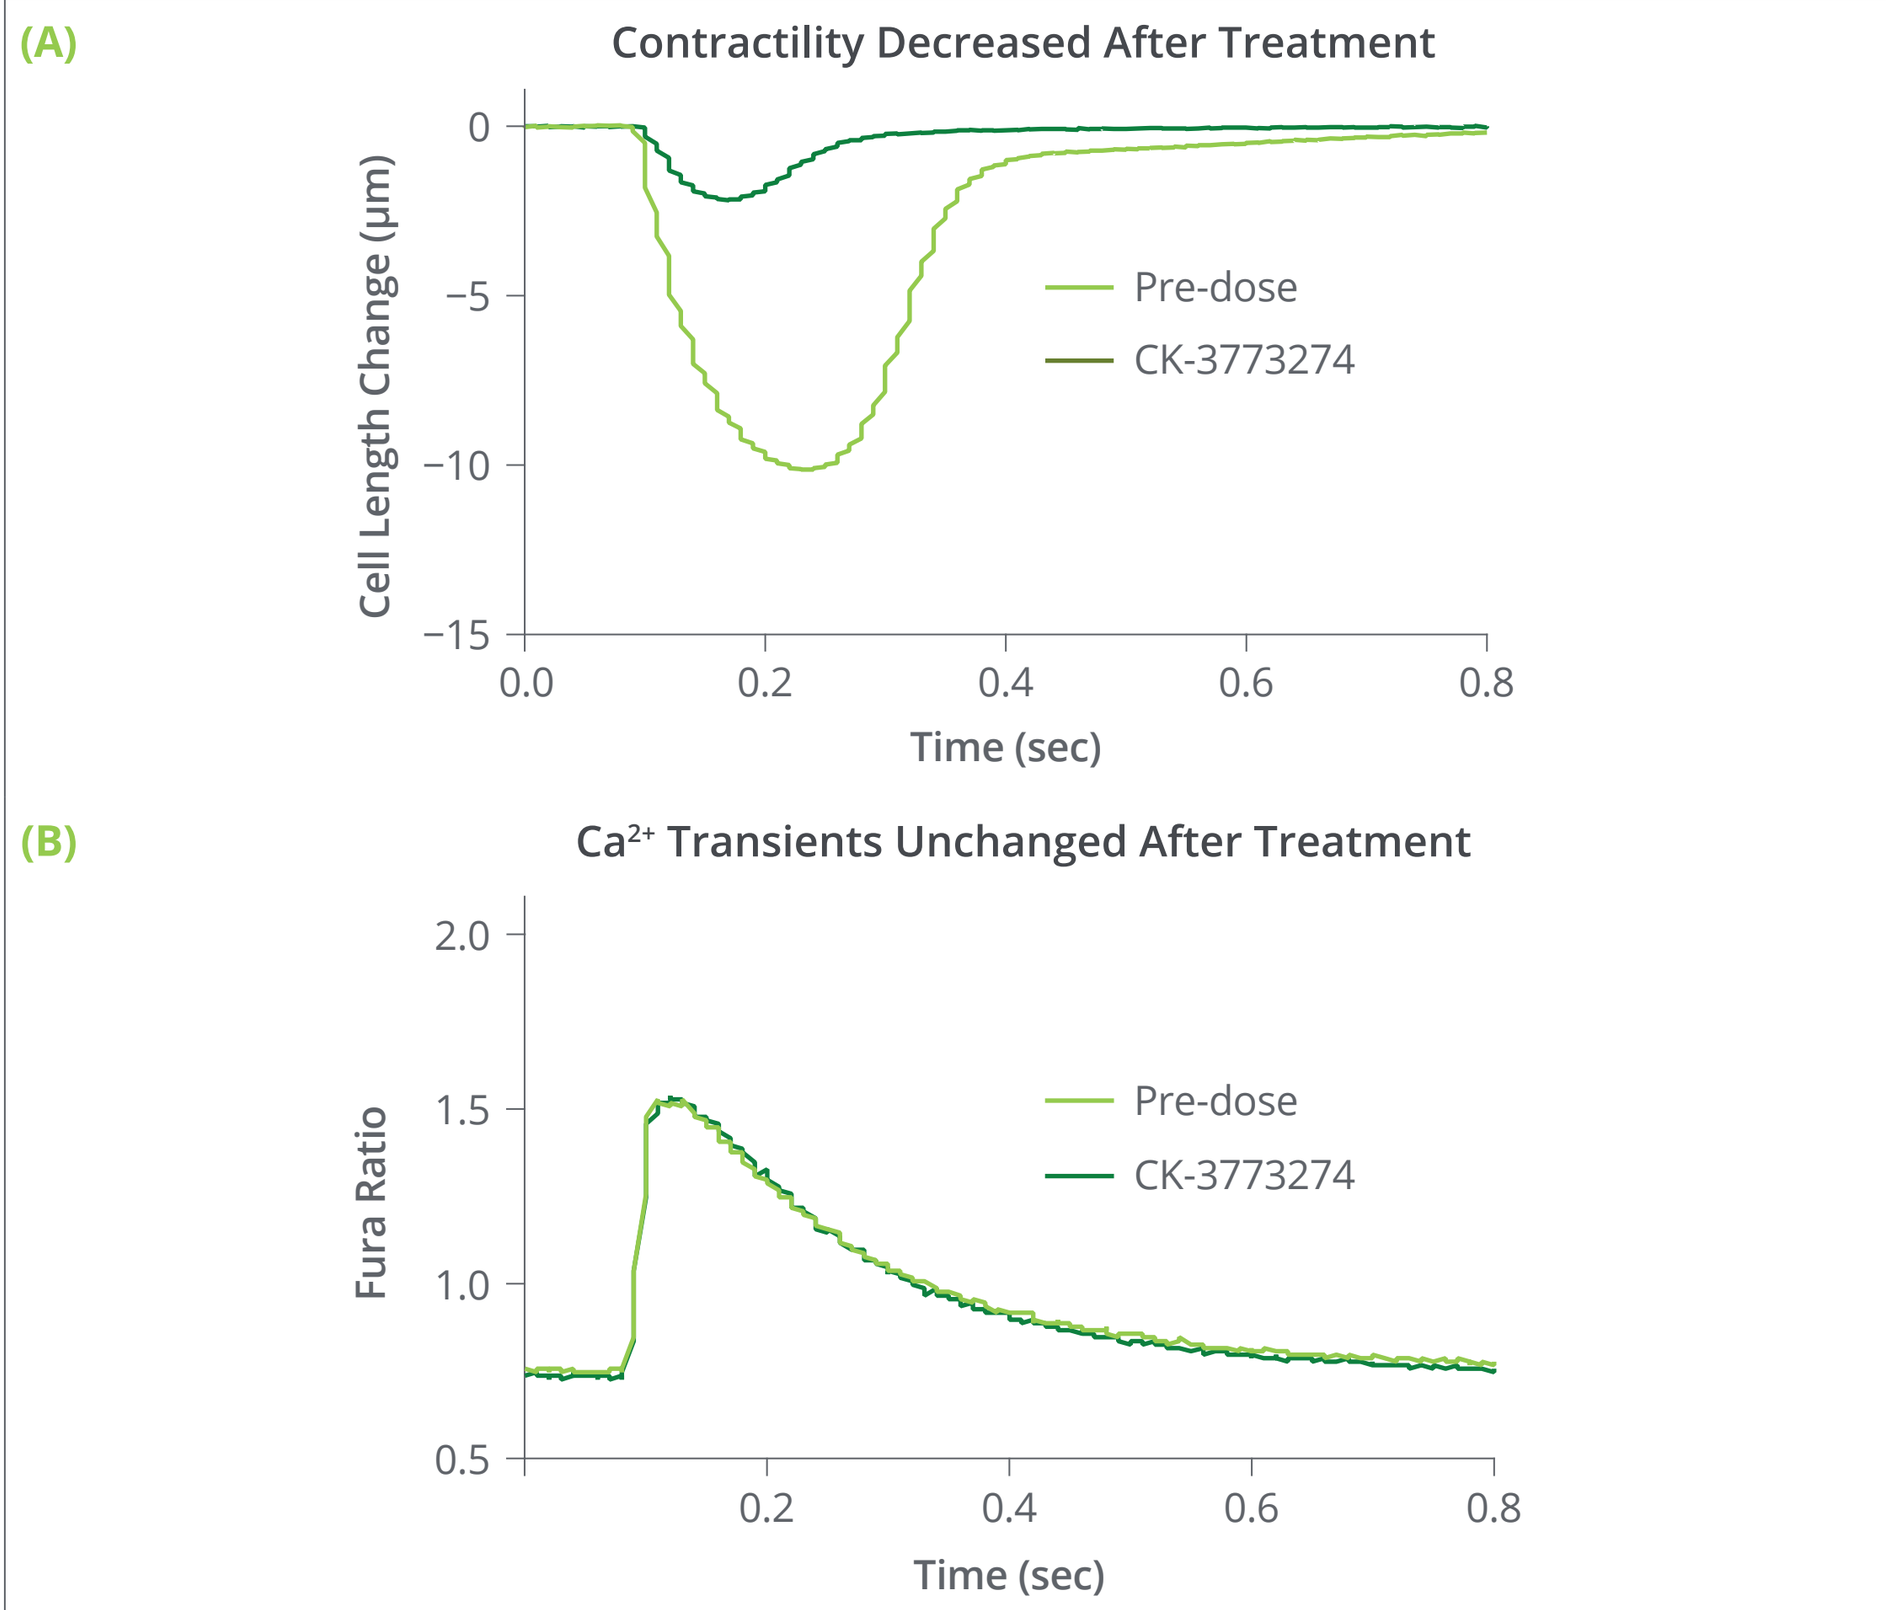
(A)
Contractility Decreased After Treatment
Cell Length Change (μm)
Pre-dose
CK-3773274
Time (sec)
(B)
Ca2+ Transients Unchanged After Treatment
Fura Ratio
Pre-dose
CK-3773274
Time (sec)

Treatment	N	FS (% of basal)	Diastolic Fura Ratio	Systolic Fura Ratio	T_{75} (seconds)
Basal	5	100	0.83 ± 0.07	1.58 ± 0.13	0.25 ± 0.03
10 μM CK-3773274	5	16.4 ± 3.4*	0.81 ± 0.09	1.56 ± 0.06	0.26 ± 0.02

- Shown are an average of 10 cell shortening **(A)** and calcium transients **(B)** for a representative adult rat cardiomyocyte before and after exposure to 10 μM CK-3773274. Pooled data are shown in table. Basal reference values are: diastolic cell length = 137.6 ± 9.7 μm. Fractional shortening (FS) = 6.79 ± 1.05 μm. Contraction velocity (CV) = 182.1 ± 31.1 μm/sec, relaxation velocity (RV) = 124.3 ± 34.3 μm/sec, time to peak = 0.116 ± 0.015 sec, and time to baseline (T_{50}) = 0.183 ± 0.015 sec.

Data presented as mean ± standard error of the mean (SEM). *p < 0.05 vs. basal FS.

Figure 8. CK-3773274 reduced fractional shortening (FS) in vivo in Sprague Dawley rats in a (A) dose- and (B) concentration-related manner

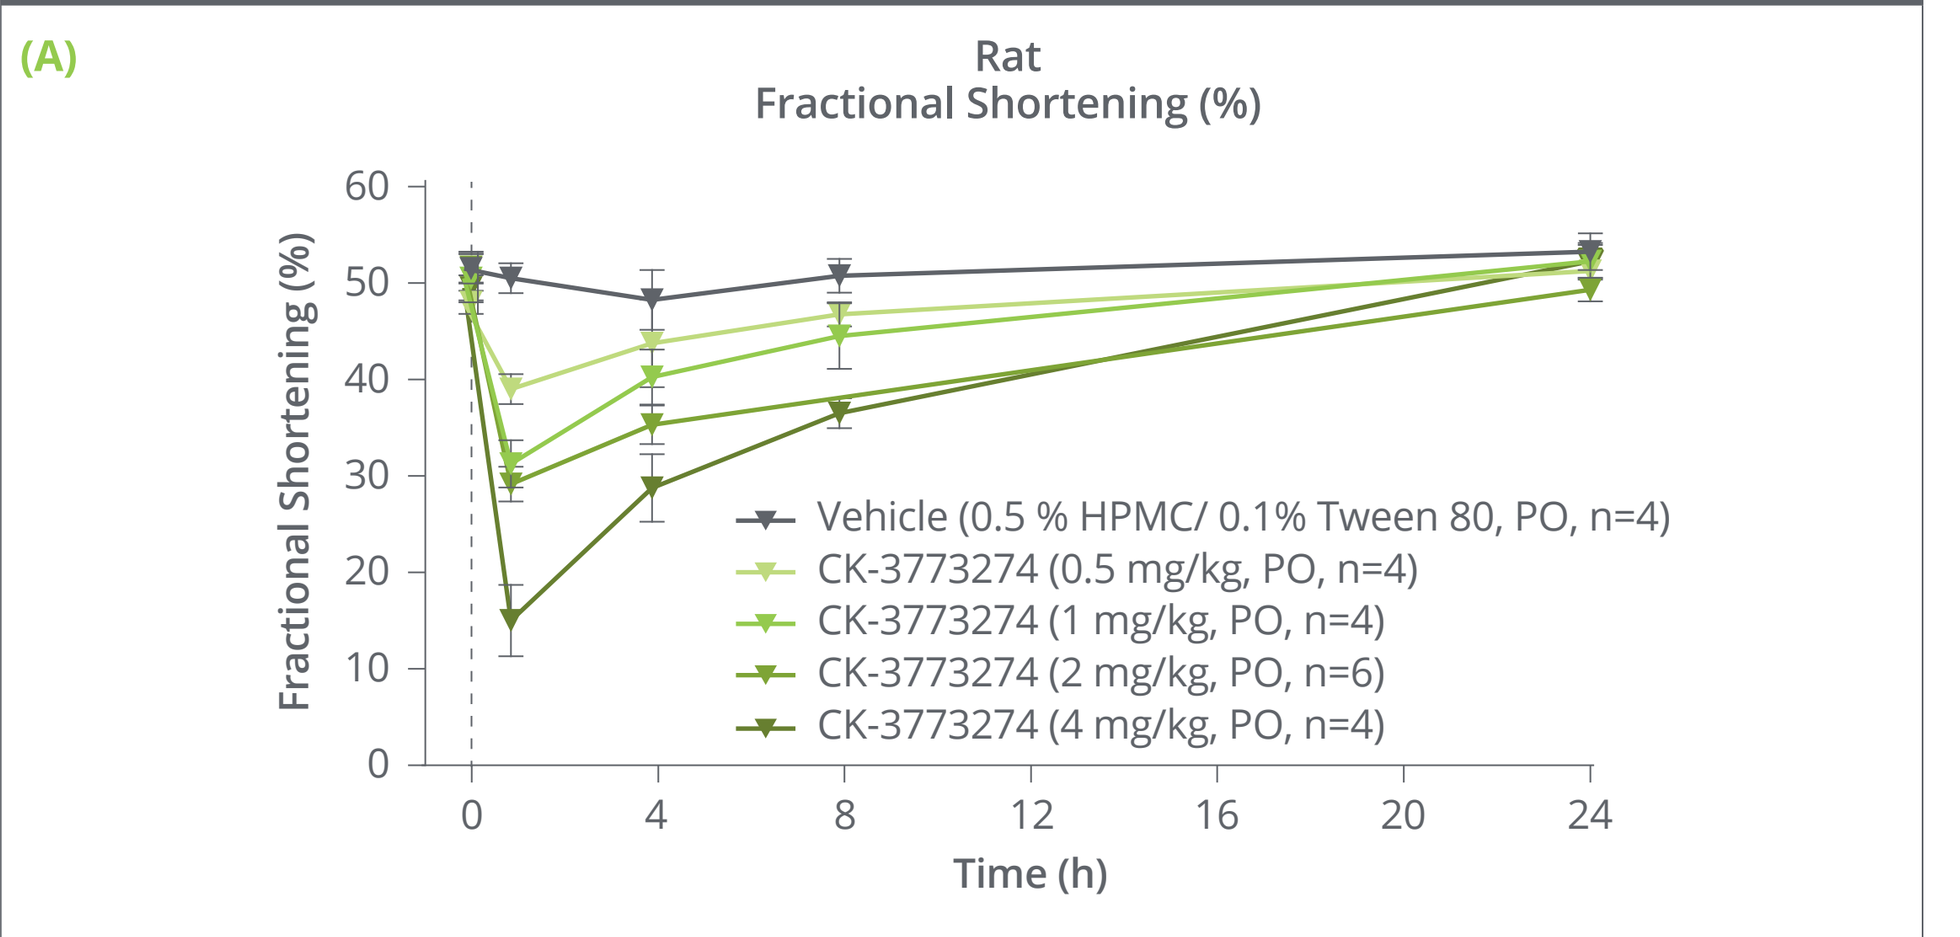
(A)
Rat
Fractional Shortening (%)
Time (h)
Vehicle (0.5 % HPMC/ 0.1% Tween 80, PO, n=4)
CK-3773274 (0.5 mg/kg, PO, n=4)
CK-3773274 (1 mg/kg, PO, n=4)
CK-3773274 (2 mg/kg, PO, n=6)
CK-3773274 (4 mg/kg, PO, n=4)

Dose (mg/kg)	Fractional Shortening (%) Pre-dose Baseline	1 h	4 h	8 h	24 h
Vehicle	50.7 ± 1.1	47.9 ± 1.3	47.5 ± 1.8	50.8 ± 1.8	50.4 ± 1.5
0.5	49.0 ± 1.1	39.0 ± 1.6*	43.8 ± 4.6	46.8 ± 1.3	51.3 ± 0.6
1	49.3 ± 1.7	31.3 ± 2.5**	40.3 ± 2.9	44.5 ± 3.4	52.3 ± 1.7
2	51.3 ± 1.6	29.2 ± 1.8**	35.3 ± 2.0	–	49.3 ± 1.2
4	49.3 ± 1.1	15.0 ± 3.7***	28.8 ± 3.5*	36.5 ± 1.6**	52.3 ± 1.9

Values are expressed as mean ± SEM. *p<0.05, **p<0.01, ***p<0.001 vs. baseline values within each timepoint by 2-way analysis of variance (ANOVA).

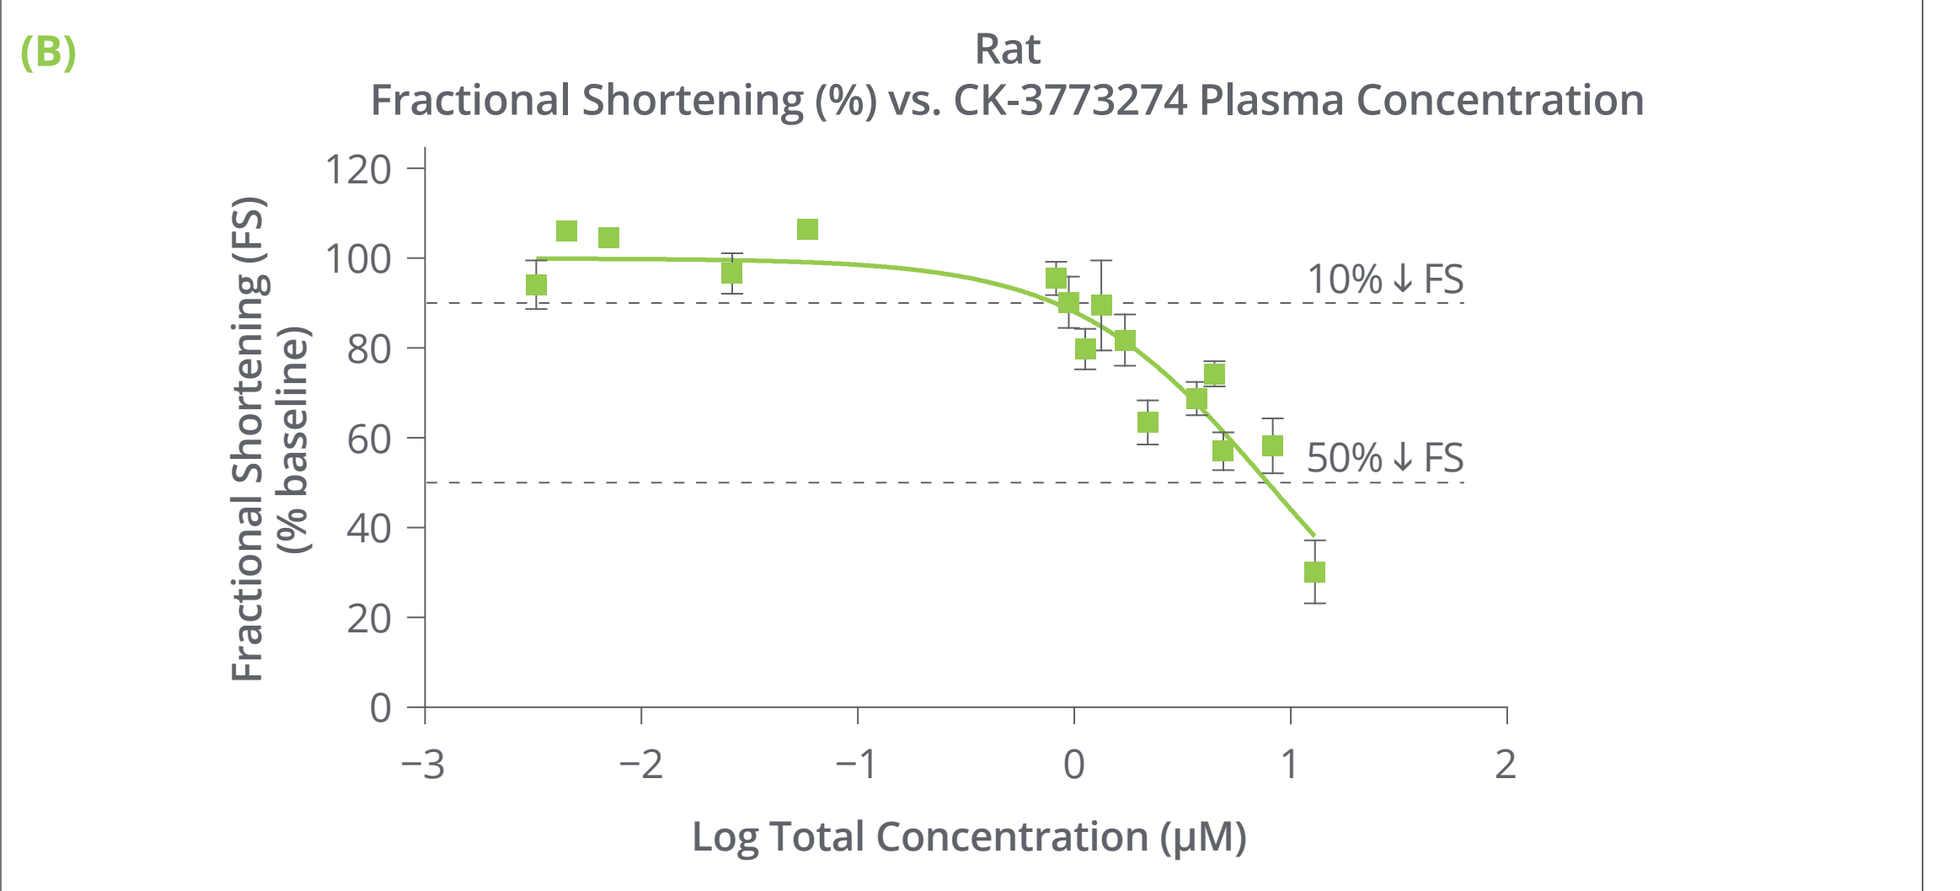
(B)
Rat
Fractional Shortening (%) vs. CK-3773274 Plasma Concentration
Fractional Shortening (FS) (% baseline)
10% ↓ FS
50% ↓ FS
Log Total Concentration (μM)

Fractional Shortening	
IC_{10} (μM)	0.8
IC_{50} (μM)	7.9

* Rat Plasma Protein Binding: 98.2%

A. Sprague Dawley rats received vehicle or CK-3773274 (0.5, 1, 2, or 4 mg/kg, PO) and echocardiography assessments were performed at select timepoints over 24 hours.
B. CK-3773274 concentration-fractional shortening response plot with the horizontal dotted lines indicating a 10% and 50% reduction of FS relative to baseline (IC_{10} and IC_{50}).

Values are expressed mean ± SEM.

SUMMARY

- **CK-3773274 is a novel small molecule that selectively inhibits cardiac myosin ATPase activity and contractility in vitro.**
- **Binding of CK-3773274 is mutually exclusive with blebbistatin, suggesting they bind to the same or overlapping sites on cardiac myosin. In contrast, *mavacamten* and blebbistatin binding are not mutually exclusive, consistent with CK-3773274 and *mavacamten* having distinct binding sites.**
- **CK-3773274 slows actin-activated phosphate release with little effect on ATP binding and hydrolysis, consistent with a mechanism that stabilizes myosin in weak actin-binding conformations.**
- **CK-3773274 reduced cardiac contractility in vivo in Sprague Dawley rats in a dose- and concentration-related manner.**
- **Cardiac myosin inhibition may be a viable approach to treat the underlying hypercontractility of the cardiac sarcomere in hypertrophic cardiomyopathies.**

References

Allingham et al. (2005). *Nature Str Mol Biol.* 12(4):378–379.
De La Cruz and Ostap (2009). *Methods Enzymol.* 455:157–192.
Green et al. (2016). *Science.* 351(6273):617–621.
Hwee et al. (2015). *J Pharmacol Exp Ther.* 353(1):159–168.
Malik et al. (2011). *Science.* 331(6023):1439–1443.
Planelles-Herrero et al. (2017). *Nat Commun.* 8(1):190
Walsh et al. (2017). *Genet Med.* 19(2):192–203.

To obtain a PDF of this poster: scan the QR code OR visit http://bit.ly/2HbxyaE. No personal information is stored.

Disclosures

All authors are current or former employees at Cytokinetics and were financially compensated for their work.

This study was funded by Cytokinetics, Inc.

Presented at the Annual Meeting of the Biophysical Society, San Diego, CA, February 15–19, 2020