Clinical Profile of Obstructive Hypertrophic Cardiomyopathy in a Nationwide Patient Cohort

Michael Butzner,[1] Phil Sarocco,[1*] Martin Maron,[2] Ethan Rowin,[2] Hiangkiat Tan,[3] Chia-Chen Teng,[3] Eric Stanek,[3] Laura Robertson[1]

[1]Cytokinetics, Incorporated, South San Francisco, CA, USA; [2]Hypertrophic Cardiomyopathy Center and Research Institute, Tufts Medical Center, Boston, MA, USA;
[3]HealthCore, Wilmington, DE, USA; [*]An employee of Cytokinetics, Incorporated, at the time the research was conducted

BACKGROUND

- Hypertrophic cardiomyopathy (HCM) is a common genetic heart disease worldwide[1]
- Obstructive hypertrophic cardiomyopathy (oHCM) is present in about two-thirds of patients with HCM[2]
- There are limited data on the clinical profile of a nonreferral-based patient cohort with oHCM
- Characterizing patients with oHCM in the general US population may expand our understanding of oHCM outcomes and improve clinical identification and treatment of this disease

OBJECTIVE

- We made use of a large, nationwide, managed-care database to characterize a patient cohort with oHCM

METHODS

- This was a retrospective observational study utilizing claims data from the HealthCore Integrated Research Database (HIRD®) during the study period of January 1, 2012 through to January 31, 2020
- The HIRD is a broad, clinically rich, and geographically diverse spectrum of longitudinal medical and pharmacy claims data, representing over 50 million lives of commercially insured and Medicare Advantage members across the United States
- We identified adult patients (≥ 18 years) with ≥ 2 claims of any oHCM *International Classification of Diseases, Ninth Revision/Tenth Revision, Clinical Modification (ICD-9/10-CM)* diagnosis codes: HCM: 425.1x, I42.1, I42.2; oHCM: 425.11, I42.1
- The index date was the earliest diagnosis of oHCM
- Patient characteristics and outcomes were reported for the 12-month period before the index date and at the 2-year follow-up

RESULTS

- A total of 1841 patients with oHCM were identified (52% male; 63.2 ± 15 years) (**Table 1**)
- Most patients with oHCM received care within a community-based cardiovascular practice; only 7% had encounters at referral HCM centers
- Sudden cardiac death had occurred in 0.7% of patients, atrial fibrillation in 26%, and hypertension in 64% by the 2-year follow-up, and heart failure had increased by 8% (16% vs 24%) (**Figure 2**)
- The percentages of patients with diagnostic procedures and myocardial imaging had increased by the 2-year follow-up (**Figures 3** and **4**)
- Drug therapy significantly increased ($p < 0.01$) from baseline to 2-year follow-up for all HCM-related medications: β-blockers (59% vs 70%), calcium channel blockers (29% vs 33%), anticoagulants (14% vs 22%), antiarrhythmics (all: 6% vs 10%; disopyramide: 1% vs 3%) (**Figure 5**)
- At the 2-year follow-up, 144 patients (8%) had received an implantable cardioverter-defibrillator for sudden death prevention, and there had been 123 (6%) septal reduction therapies (5% myectomy; 1% alcohol septal ablation) (**Figure 6**), with the mean time from initial evaluation to procedure being 218 days and 97 days, respectively

Table 1. Patient demographics

	All patients (N = 1841)
Age (years), mean (SD)	63.2 (15.0)
Male, n (%)	958 (52.0)
Index year, n (%)	
2013	536 (29.1)
2014	401 (21.8)
2015	374 (20.3)
2016	276 (15.0)
2017	238 (12.9)
2018	16 (0.9)
Health plan type, n (%)	
Health maintenance organization	492 (26.7)
Preferred provider organization	1218 (66.2)
Consumer-driven health plan	131 (7.1)
Medicare Advantage/Medicare Supplement and Part D plans, n (%)	771 (41.9)

SD, standard deviation

Figure 1. Percentage of patients: (A) age categories (years), (B) geographic region

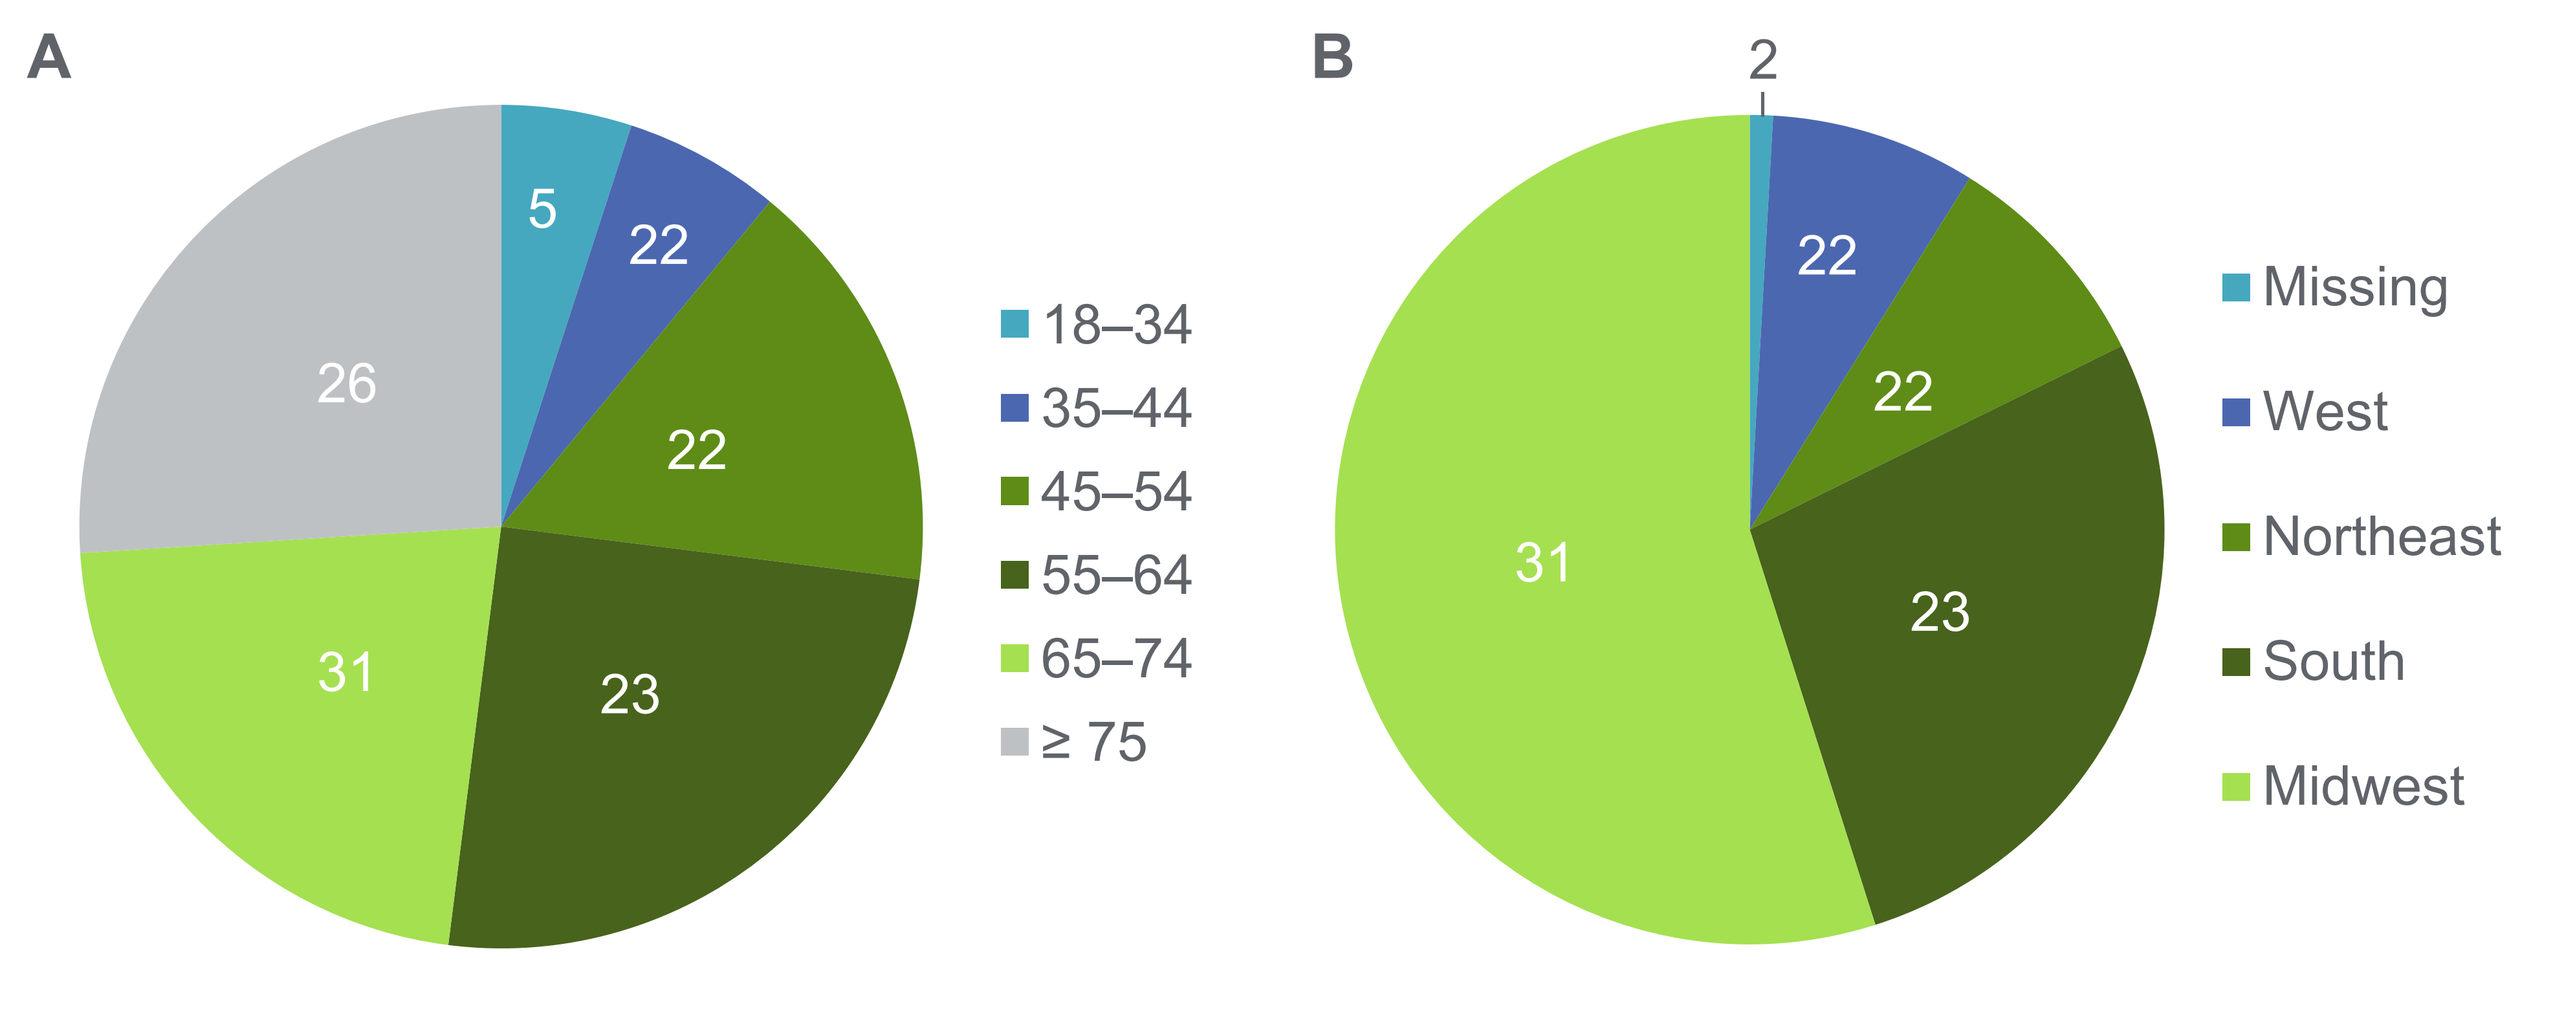
A
5
22
22
23
31
26
18–34
35–44
45–54
55–64
65–74
≥ 75
B
2
22
22
23
31
Missing
West
Northeast
South
Midwest

Figure 2. Cardiovascular comorbidities

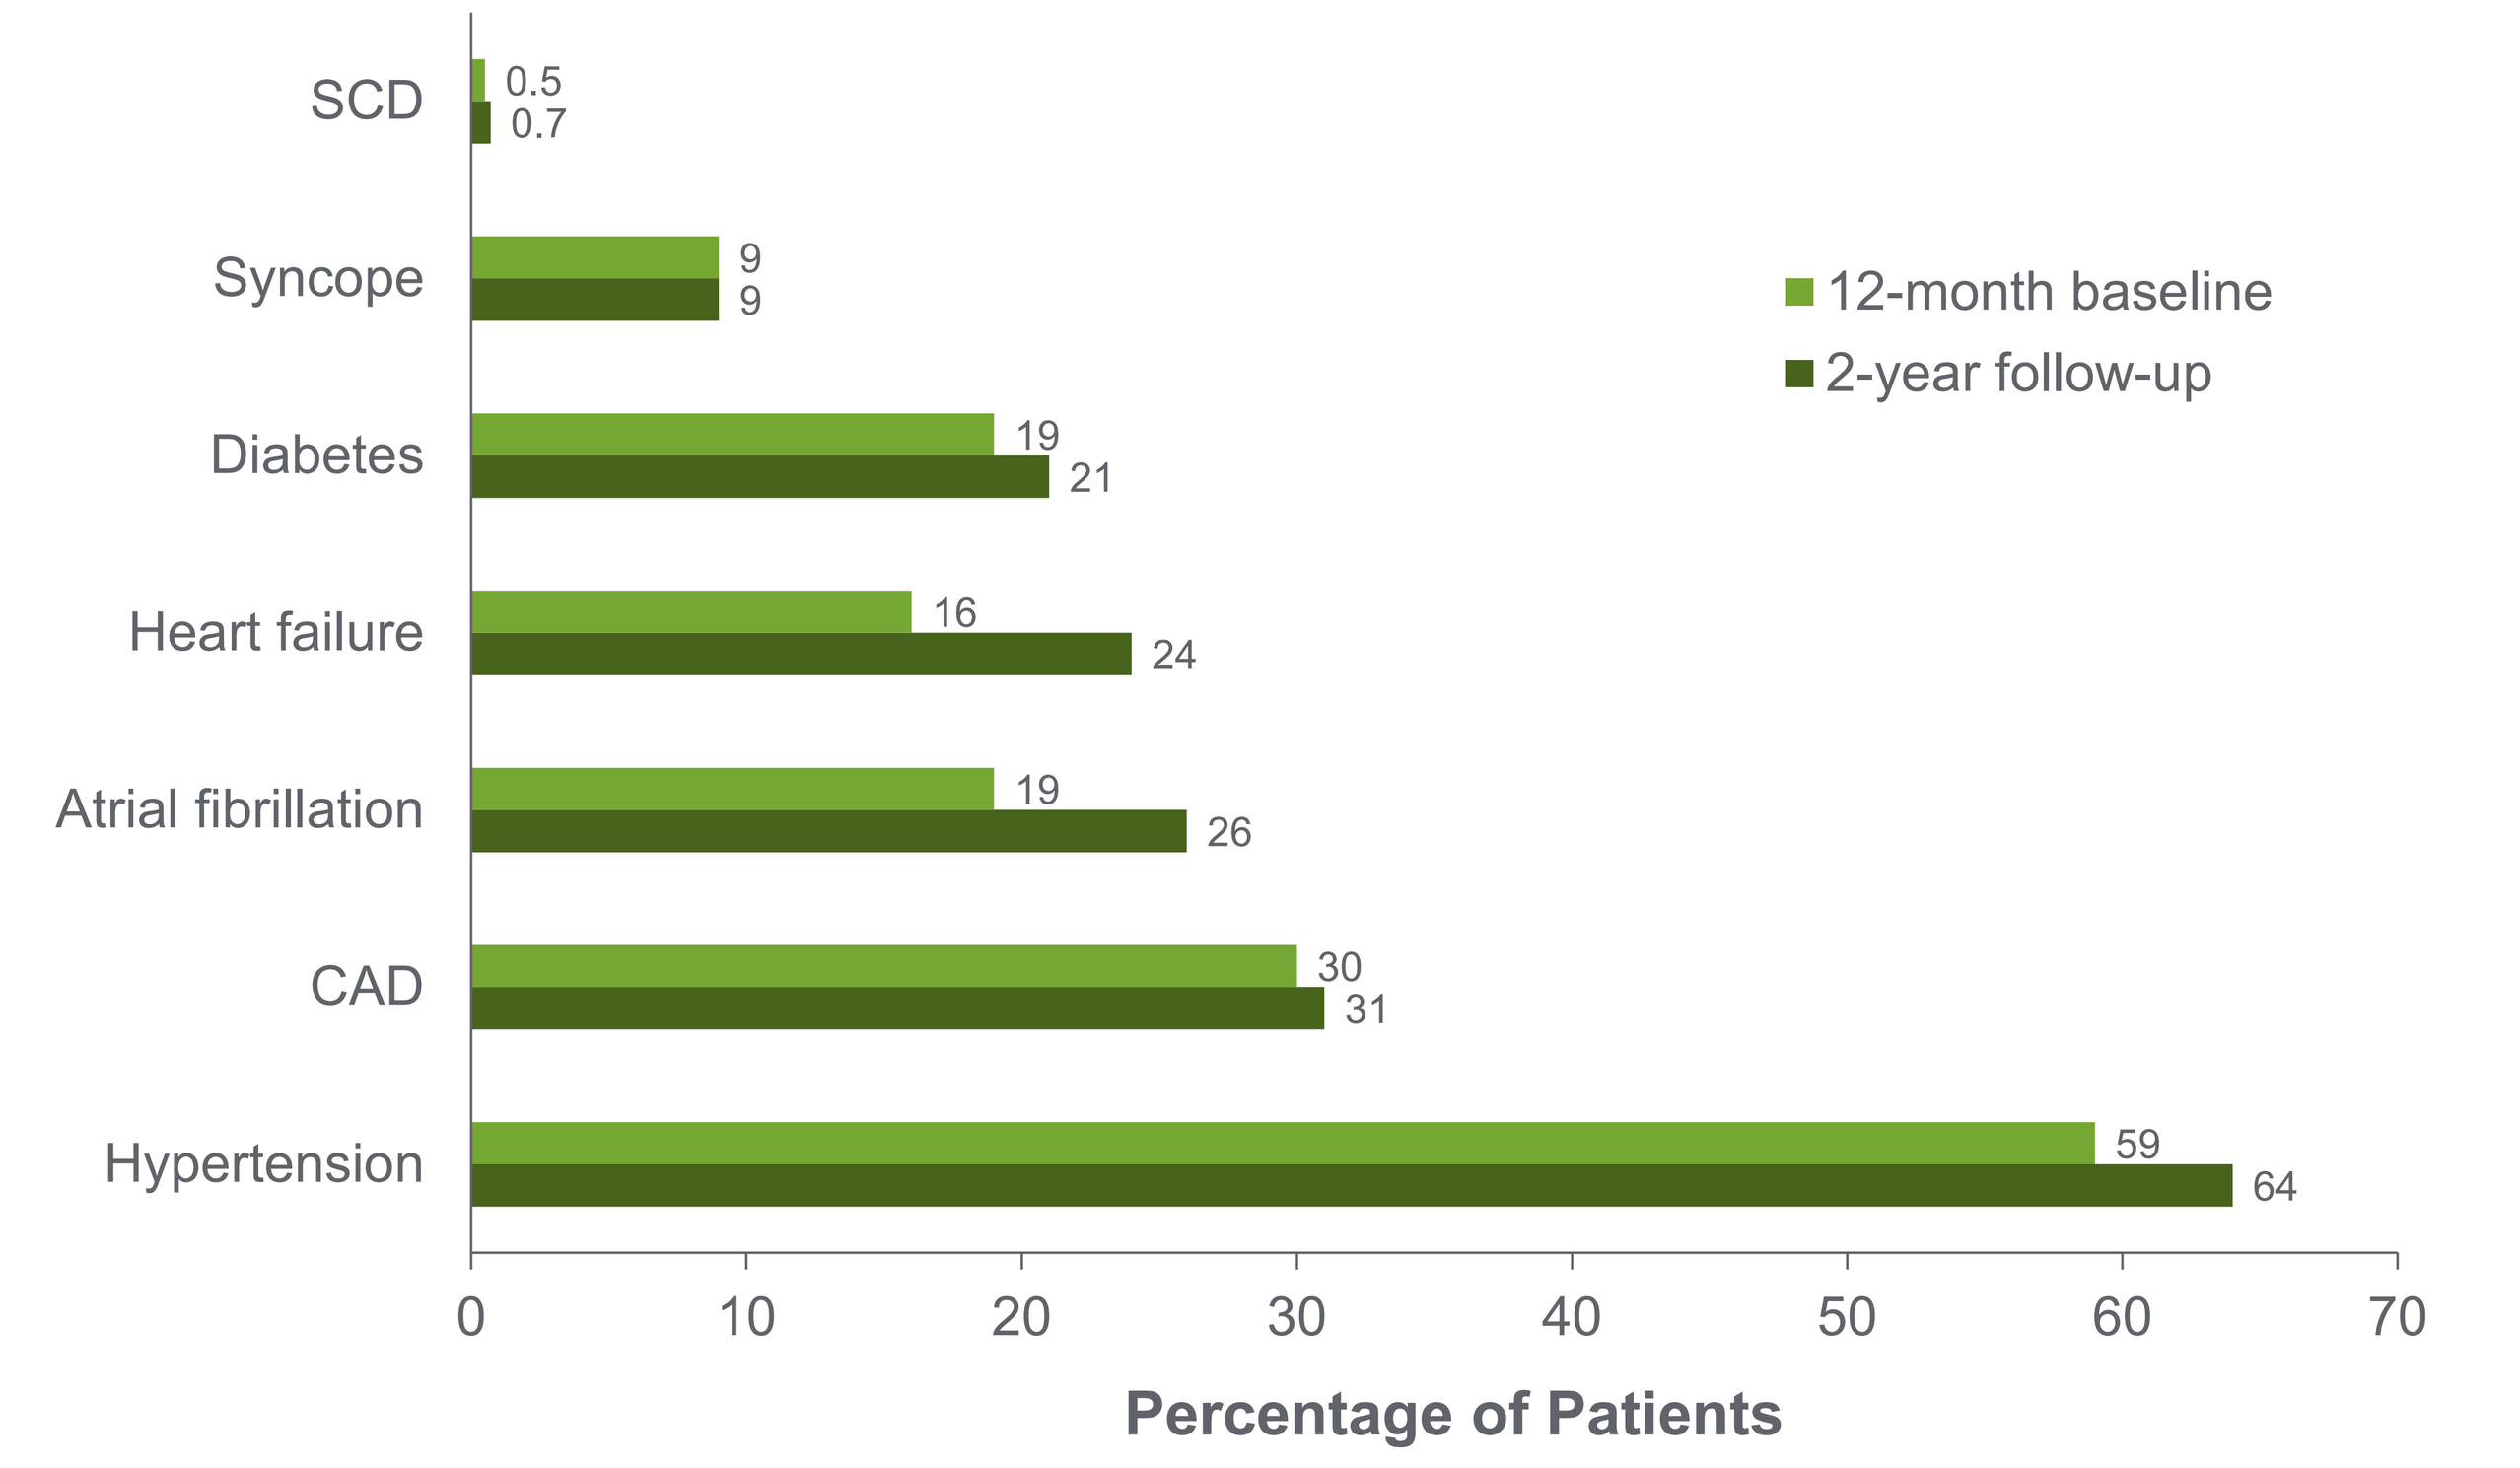
SCD 0.5 0.7
Syncope 9 9
Diabetes 19 21
Heart failure 16 24
Atrial fibrillation 19 26
CAD 30 31
Hypertension 59 64
0 10 20 30 40 50 60 70
Percentage of Patients
12-month baseline
2-year follow-up

CAD, coronary artery disease; SCD, sudden cardiac death

Figure 3. Patients with ≥ 1 claim for diagnostic procedures

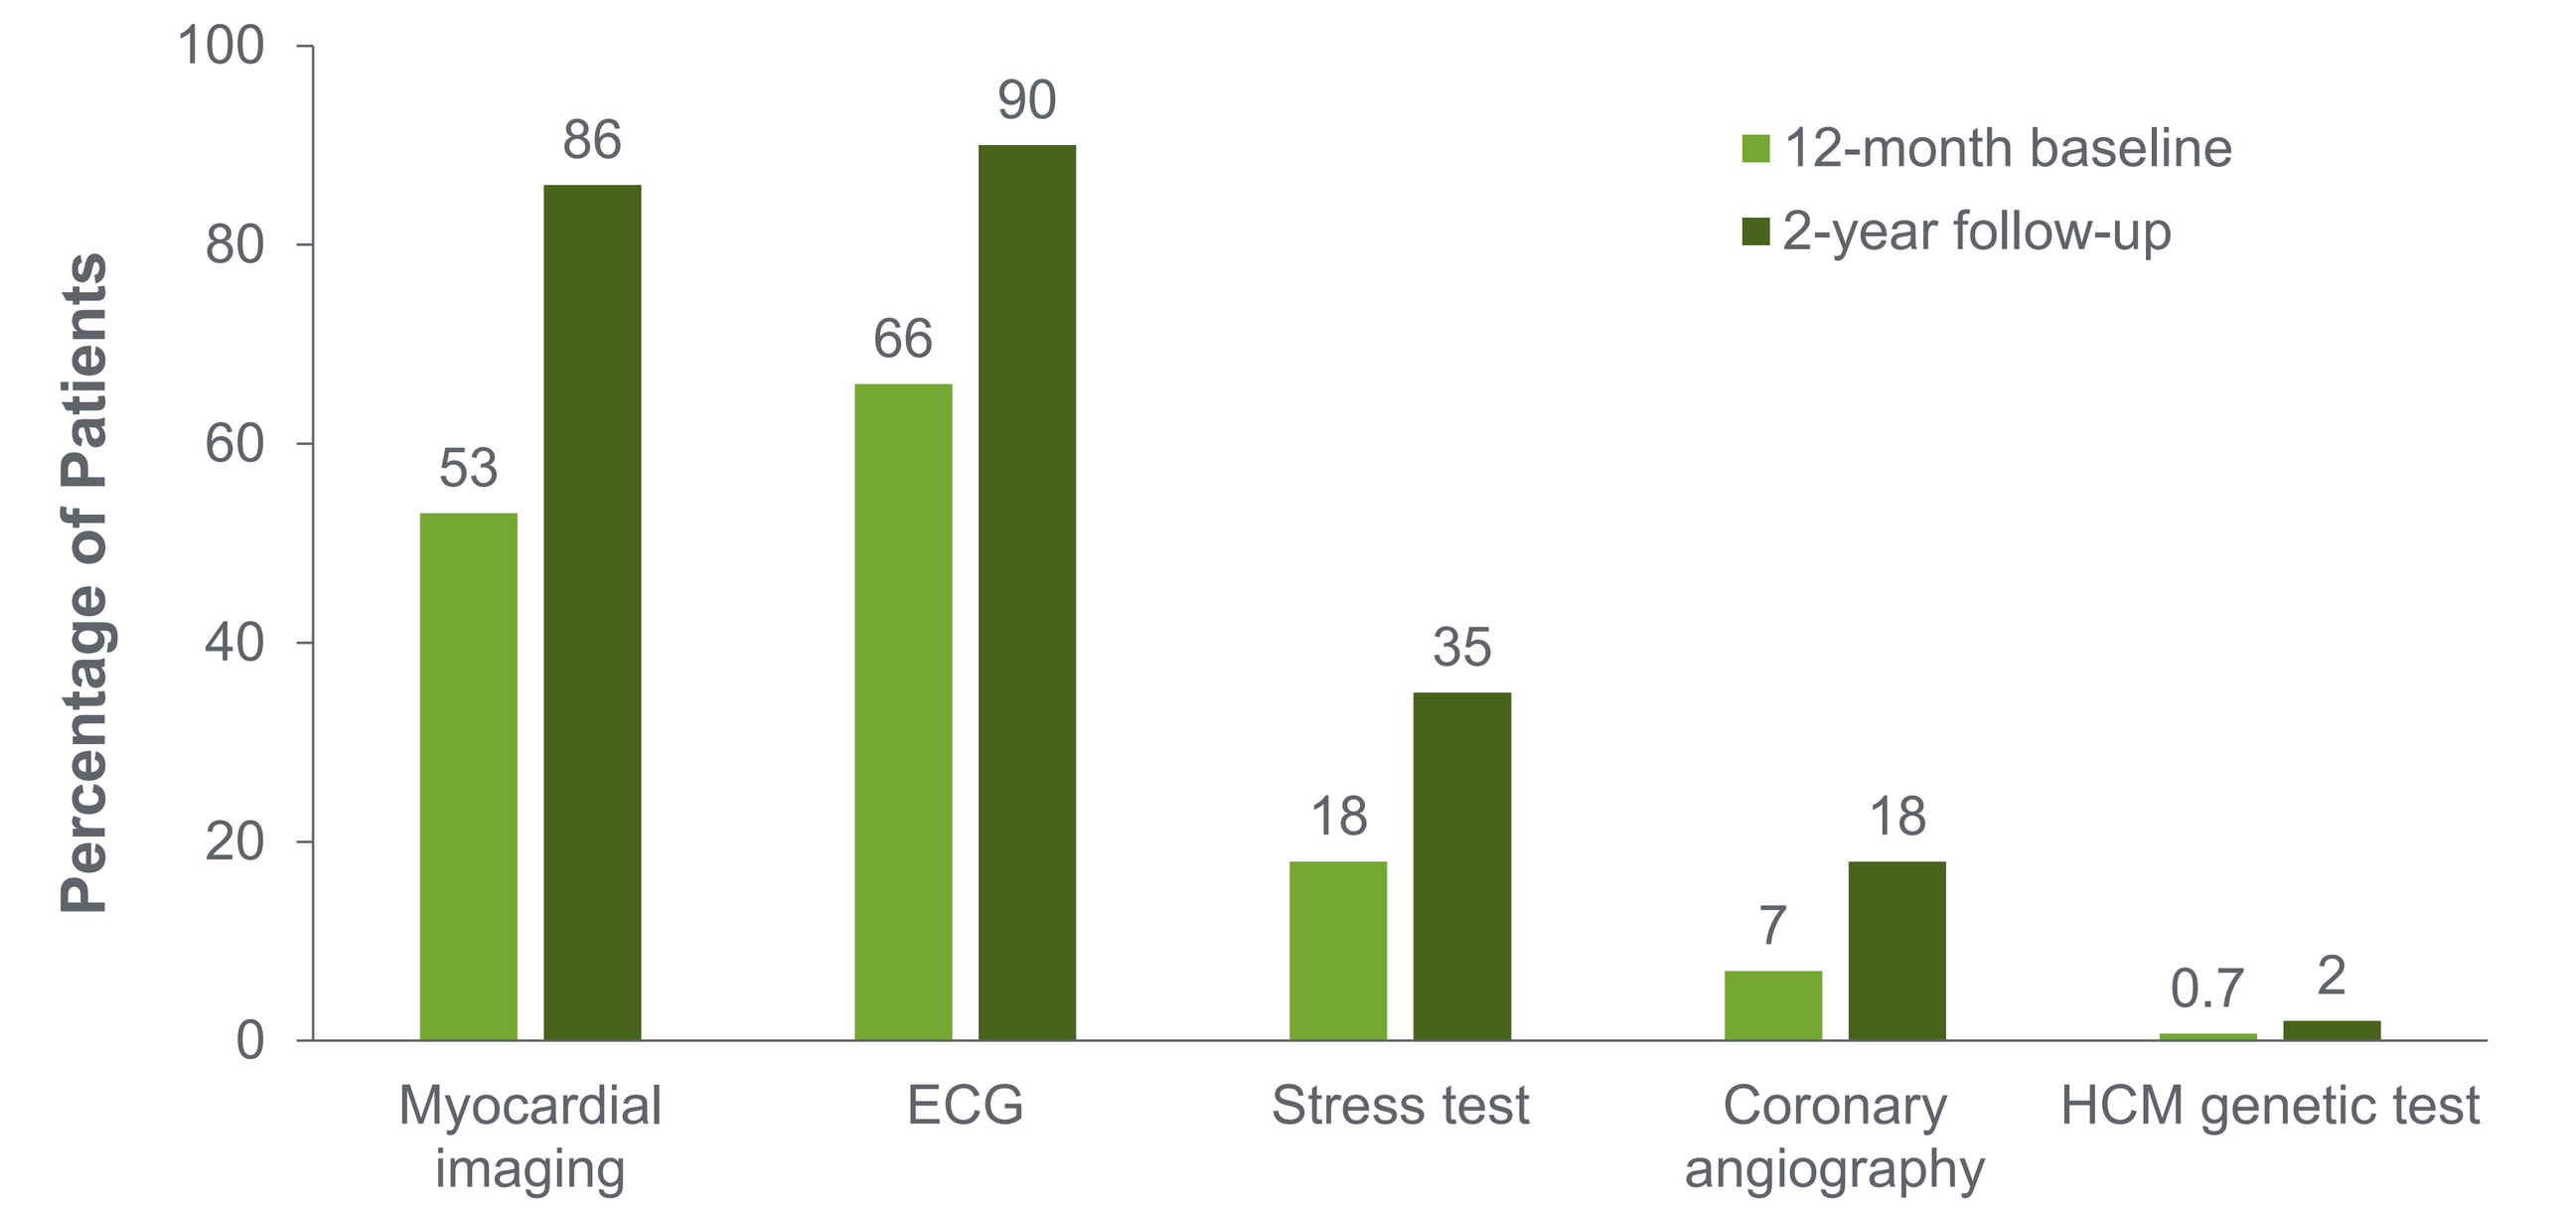
Percentage of Patients
0 20 40 60 80 100
Myocardial imaging 53 86
ECG 66 90
Stress test 18 35
Coronary angiography 7 18
HCM genetic test 0.7 2
12-month baseline
2-year follow-up

ECG, electrocardiogram; HCM, hypertrophic cardiomyopathy

Figure 4. Myocardial imaging

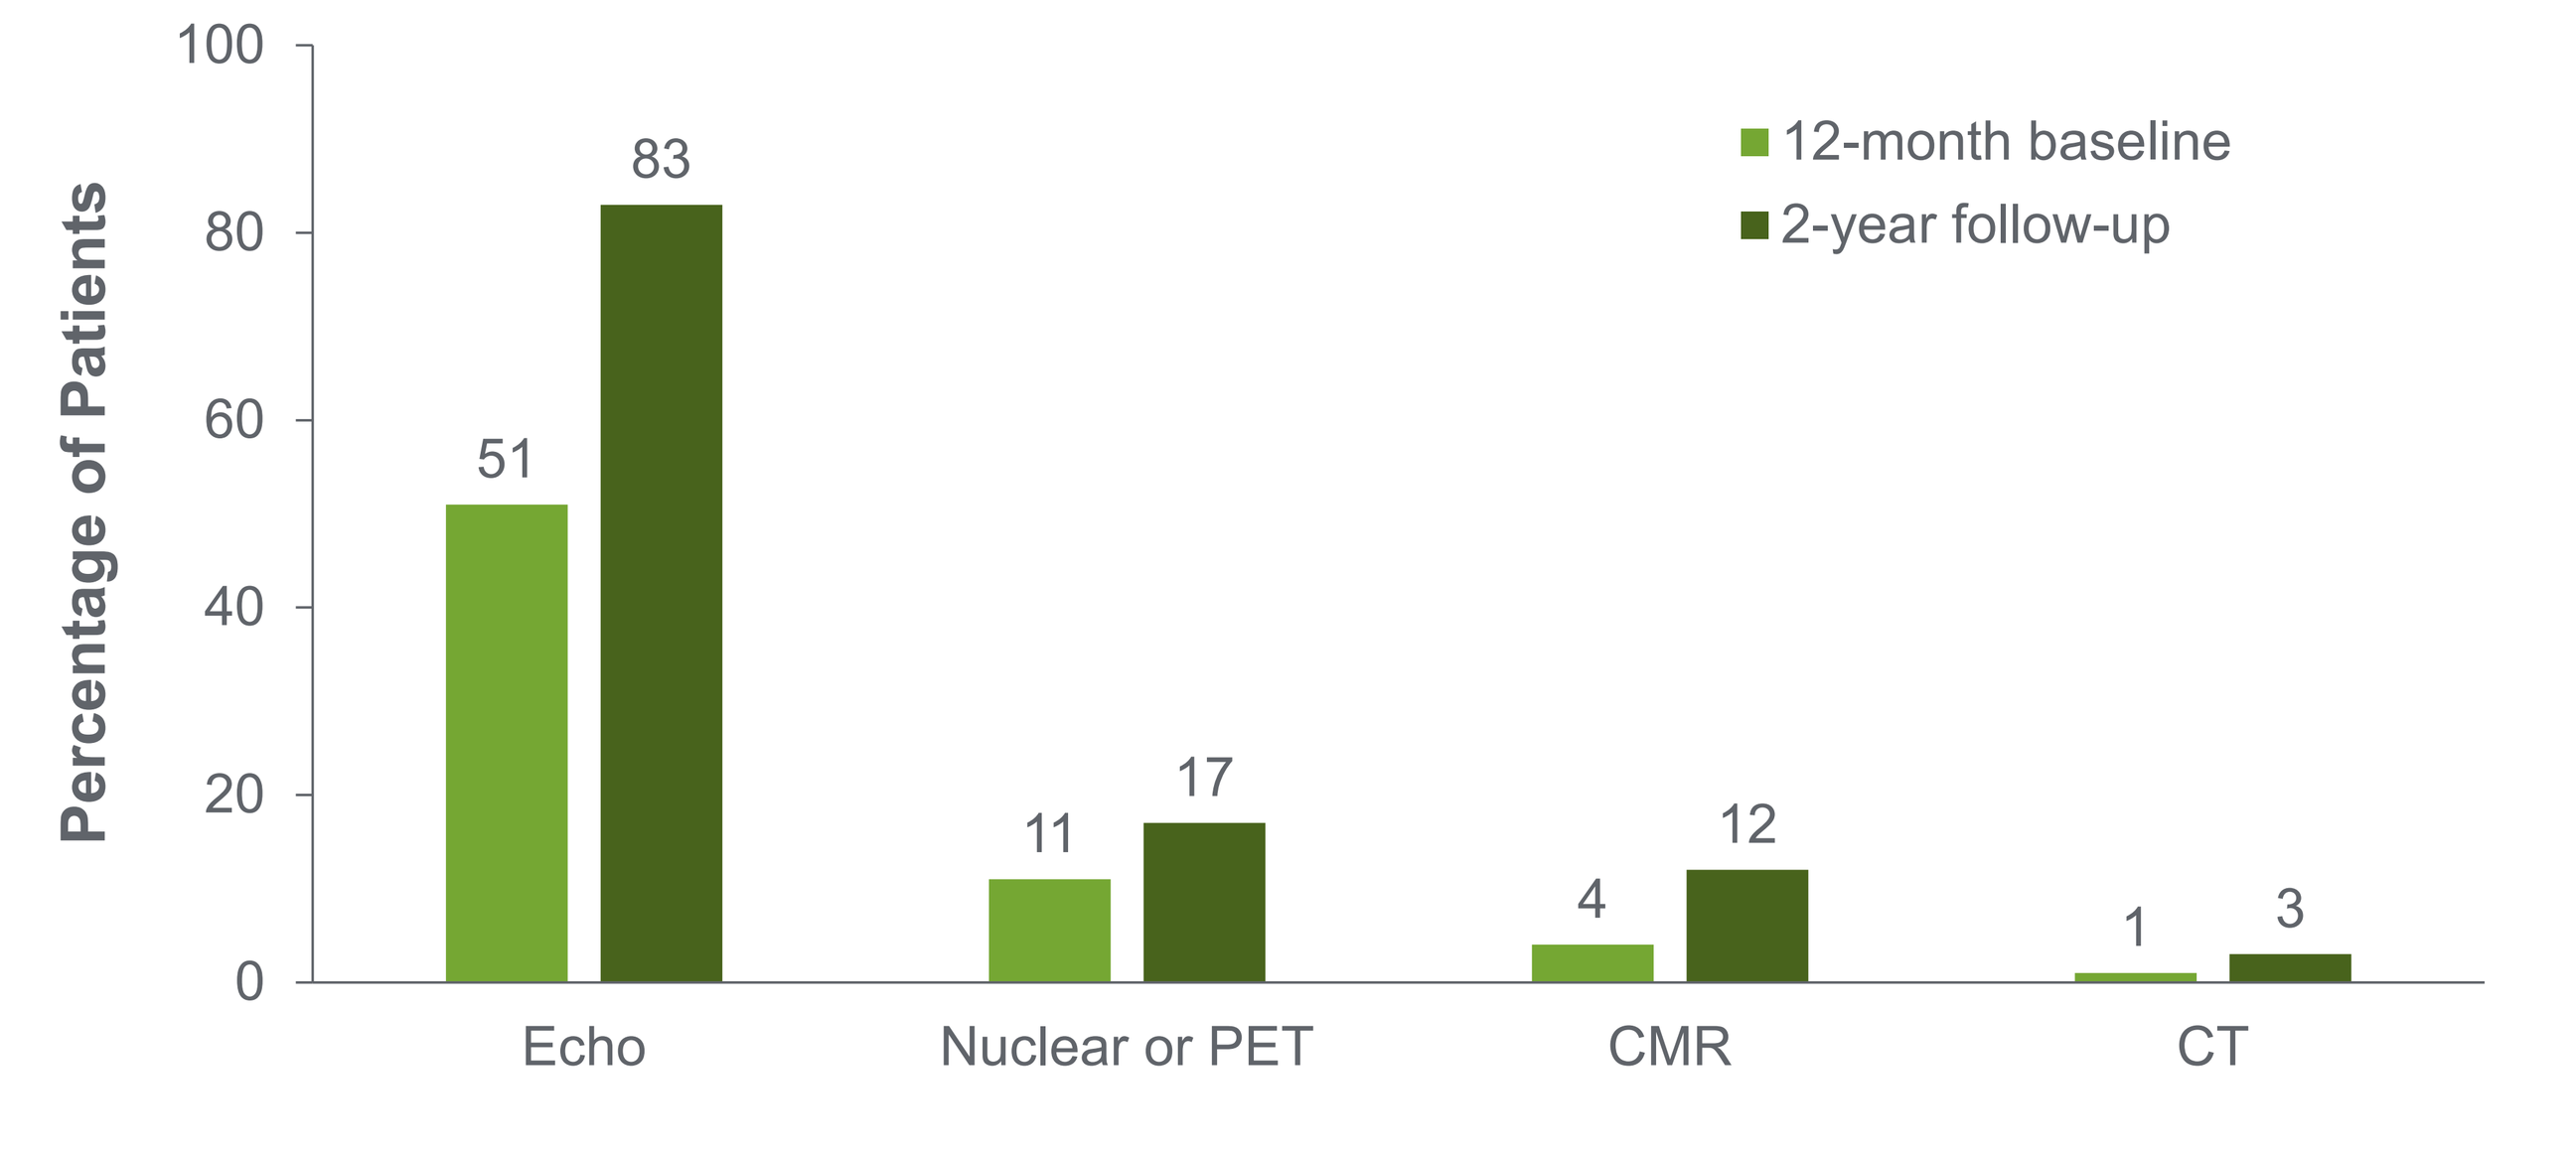
Percentage of Patients
0 20 40 60 80 100
Echo 51 83
Nuclear or PET 11 17
CMR 4 12
CT 1 3
12-month baseline
2-year follow-up

CMR, cardiac magnetic resonance; CT, computed tomography; echo, echocardiogram; PET, positron emission tomography

Figure 5. Patients with ≥ 1 claim for HCM-related prescription medication use

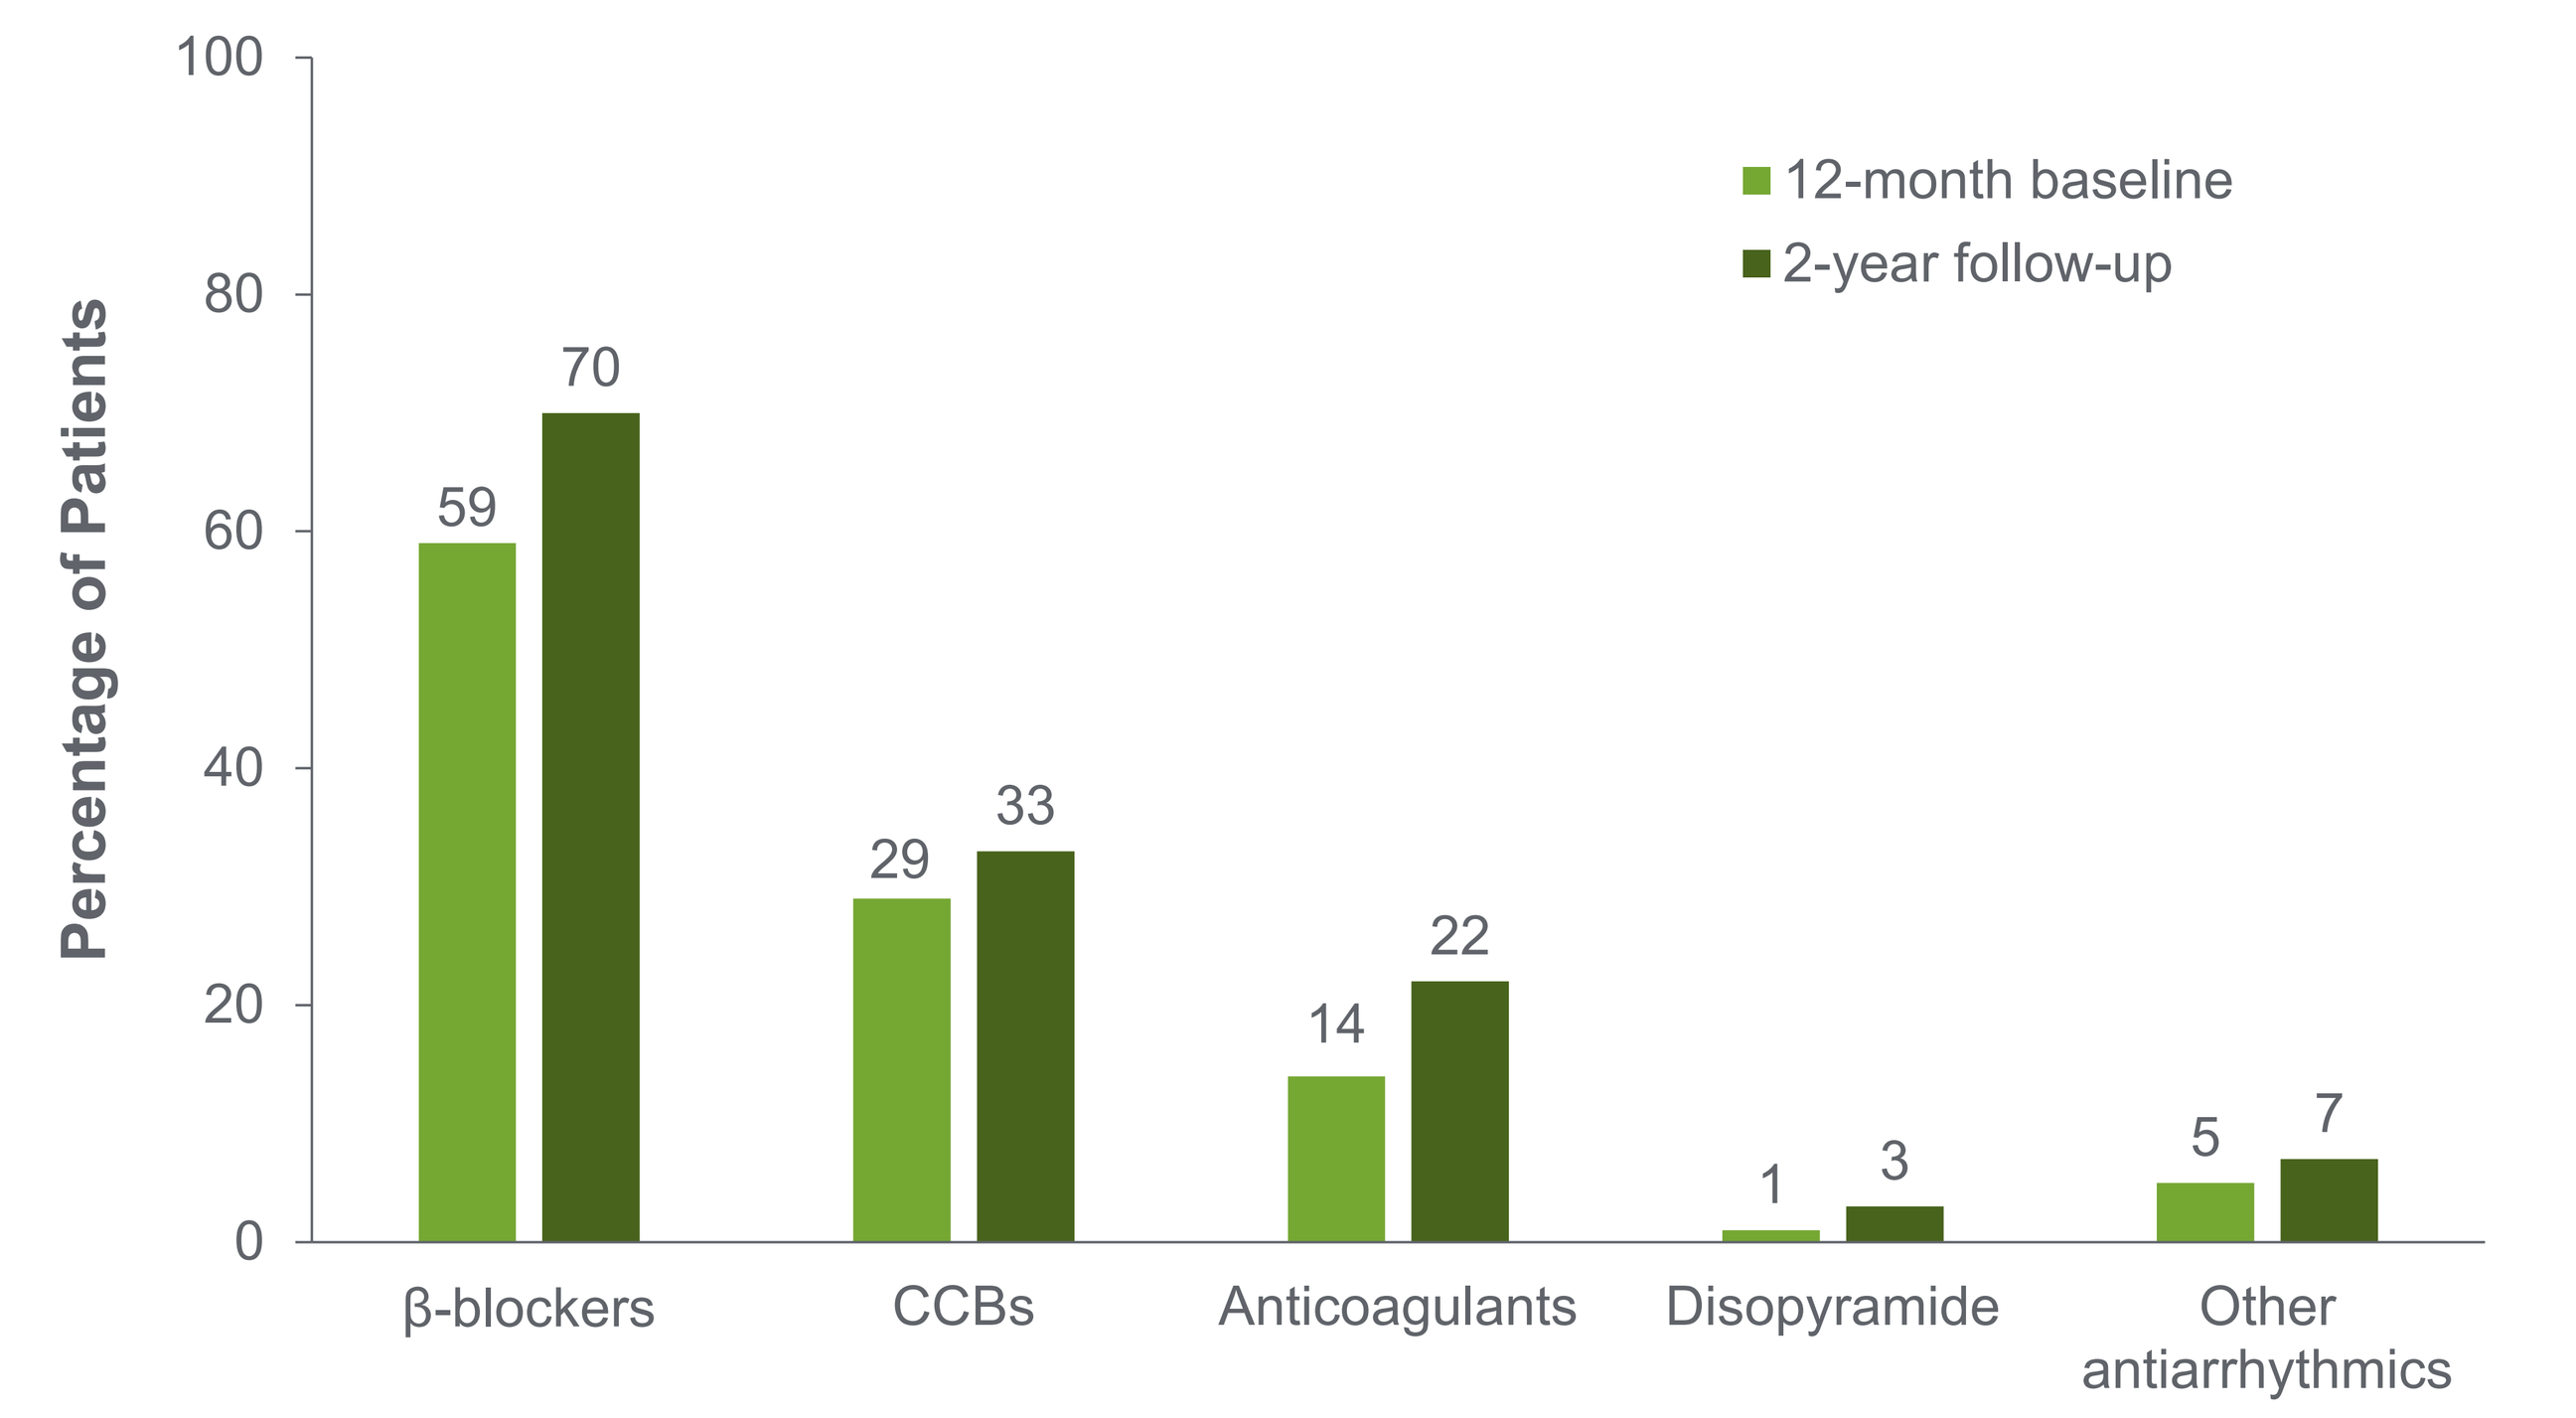
Percentage of Patients
0 20 40 60 80 100
β-blockers 59 70
CCBs 29 33
Anticoagulants 14 22
Disopyramide 1 3
Other antiarrhythmics 5 7
12-month baseline
2-year follow-up

CCBs, calcium channel blockers; HCM, hypertrophic cardiomyopathy

Figure 6. Surgical procedures at 2-year follow-up

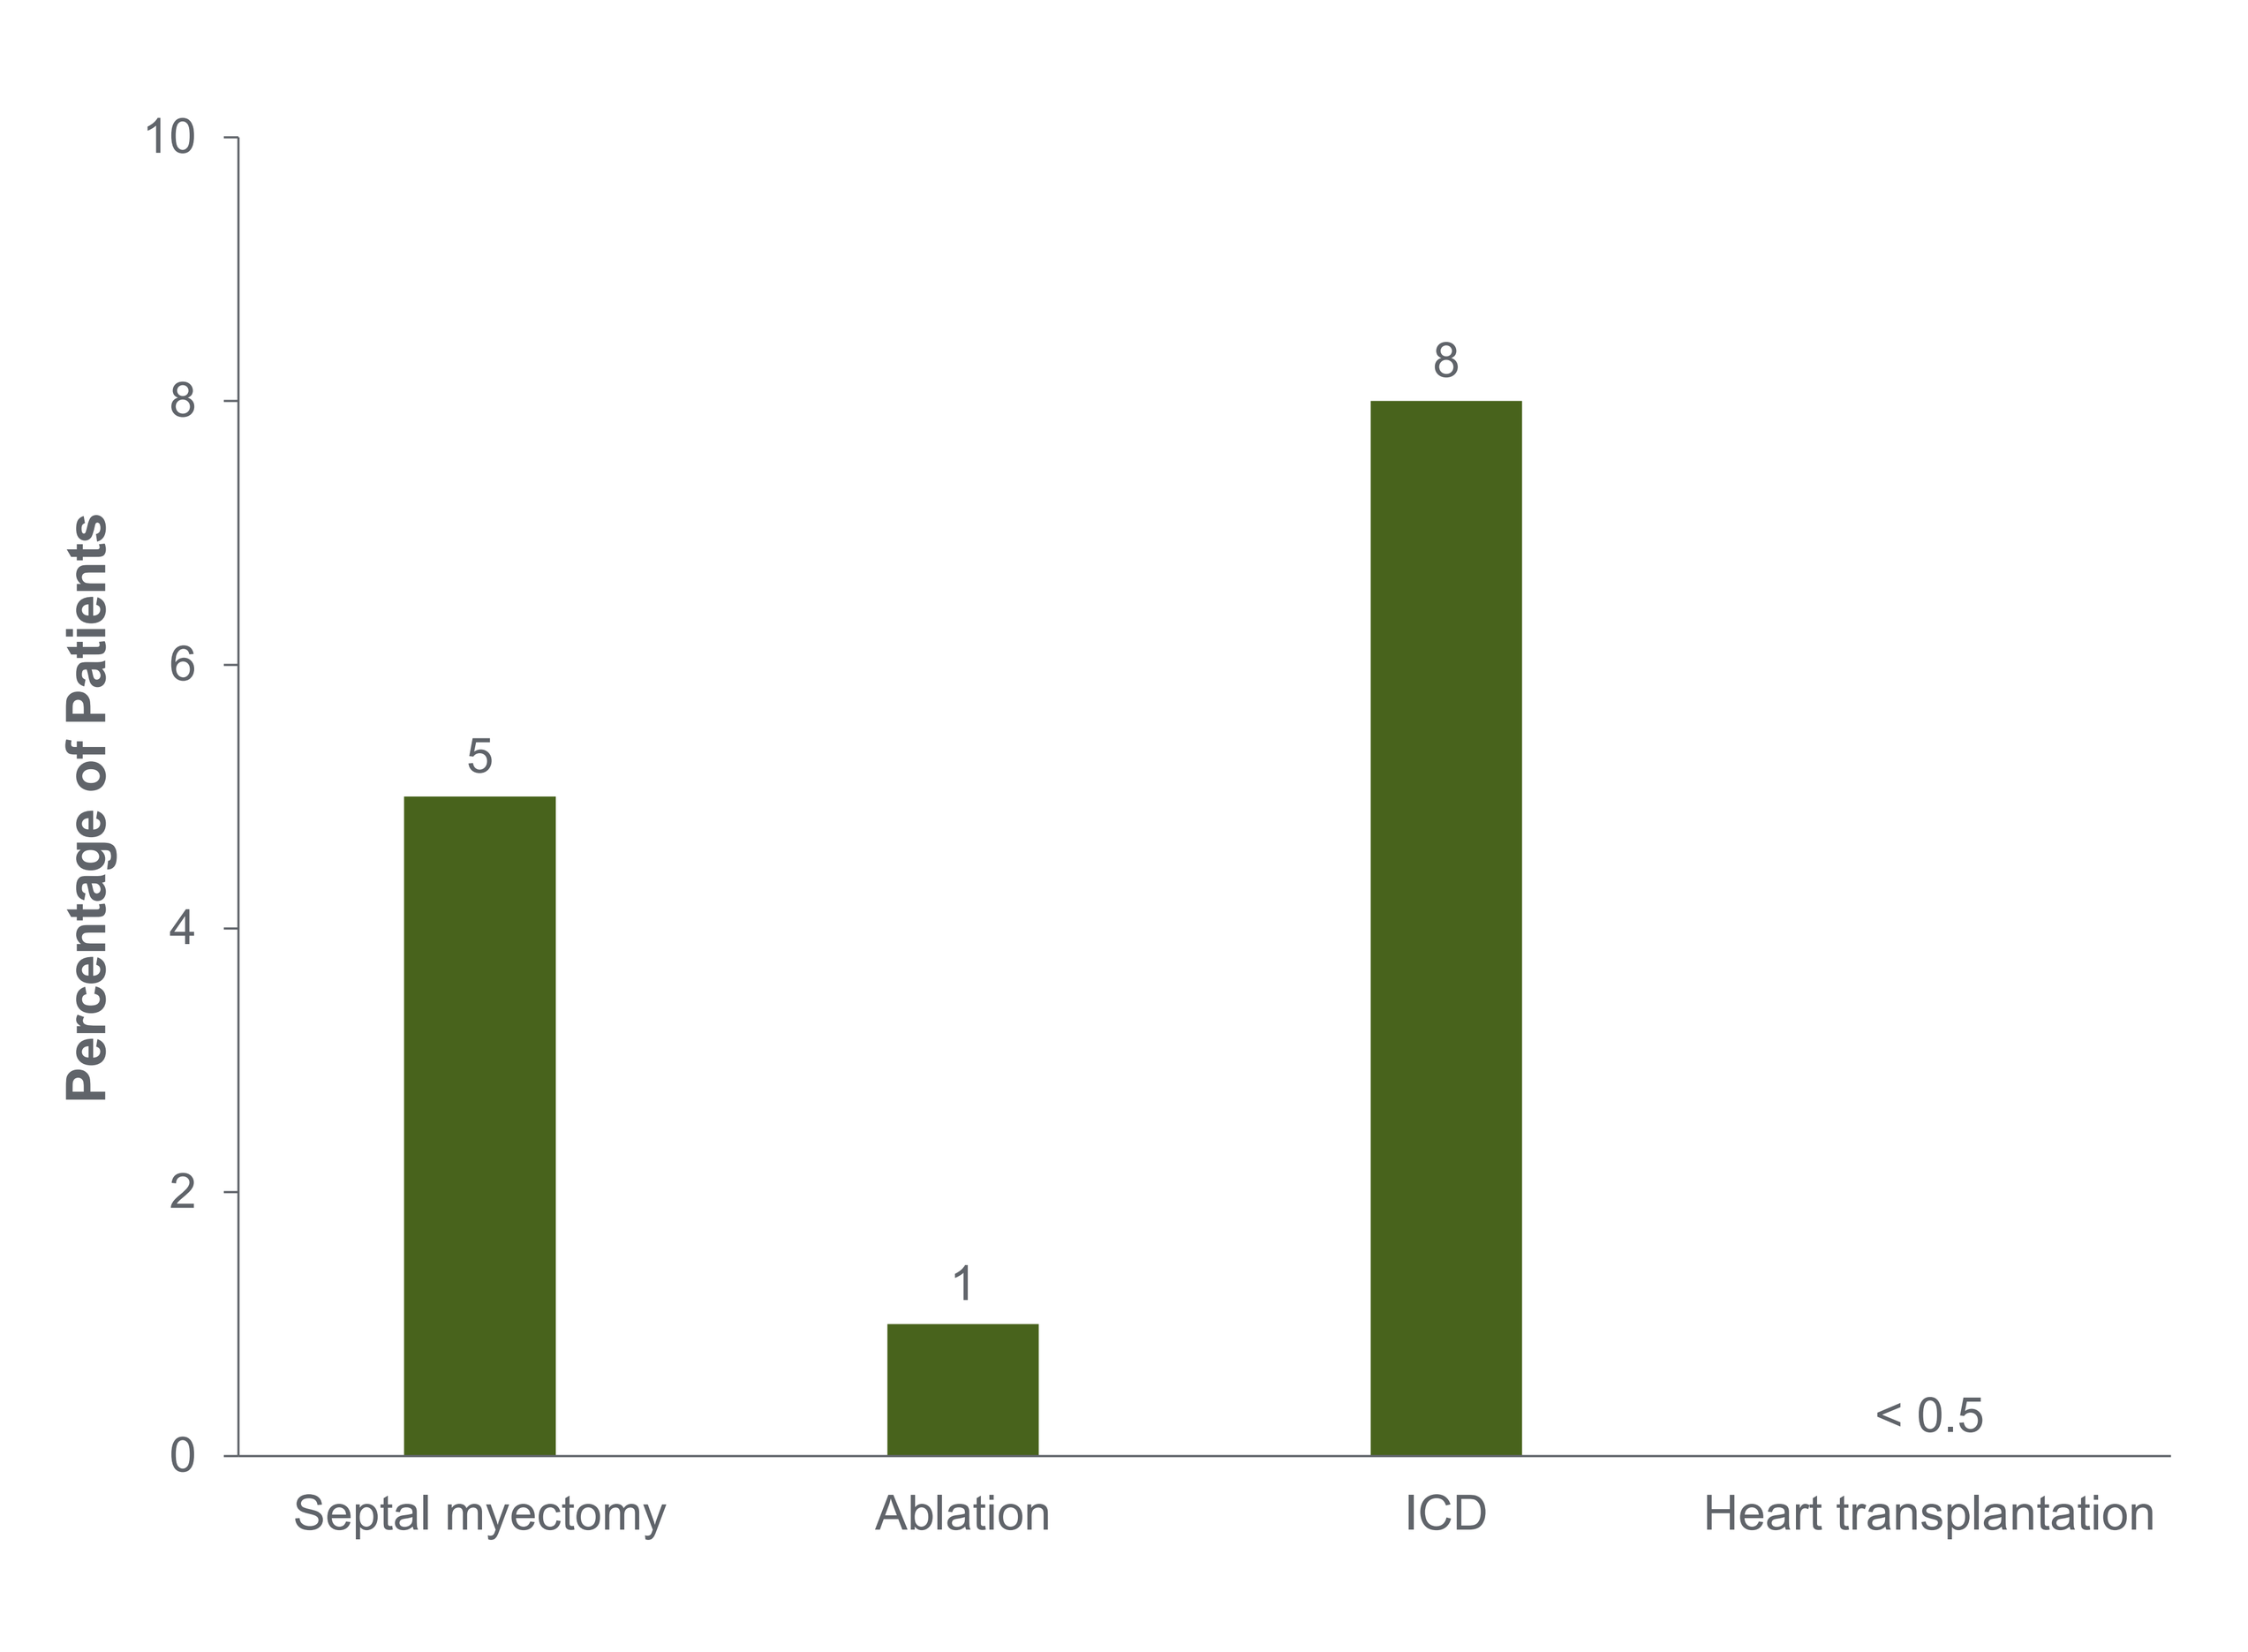
Percentage of Patients
0 2 4 6 8 10
Septal myectomy 5
Ablation 1
ICD 8
Heart transplantation < 0.5

ICD, implantable cardioverter-defibrillator

CONCLUSIONS

- This is the first study to examine a national sample of patients with oHCM using longitudinal medical and pharmacy claims data
- In a community-based population, patients with oHCM at diagnosis were, on average, in their early 60s, and they had a significant burden of cardiovascular comorbidities
- Cardiovascular medication usage was high in this oHCM population in real-world settings

References

1. Marsiglia JDC, Pereira AC. *Arq Bras Cardiol*. 2014;102(3):295–304.
2. Maron MS, et al. *Am J Cardiol*. 2016;117(10):1651–1654.

Acknowledgments

This study was funded by Cytokinetics, Incorporated.

Editorial support was provided by Charlene Rivera, PhD, on behalf of Engage Medical Affairs, Fairfield, CT, and was funded by Cytokinetics, Incorporated.

Disclosures

E Rowin and M Maron have no relevant disclosures to report. M Butzner and L Robertson are employees of Cytokinetics, Incorporated. P Sarocco was an employee of Cytokinetics, Incorporated, at the time the research was conducted. H Tan, CC Teng, and E Stanek are employees of HealthCore.

To obtain a PDF of this poster:
Scan the QR code
OR
Visit https://bit.ly/2QoIRnr
No personal information is stored.

Presented at the American College of Cardiology Scientific Sessions | Virtual Congress | May 15–17, 2021