ASE2022 HYBRID
June 10-13, 2022 • Seattle, WA

Early Cardiac Structural and Functional Reverse Remodeling in Obstructive Hypertrophic Cardiomyopathy after 10 Weeks of *Aficamten* Therapy: Analyses from REDWOOD-HCM

T. ABRAHAM[1], A. MASRI[2], M. MARON[3], L. CHOUDHURY[4], I. OLIVOTTO[5], S. SABERI[6], A. WANG[7], P. GARCIA-PAVIA[8], N. LAKDAWALA[9], S. NAGUEH[10], F. RADER[11], A. TOWER-RADER[12], A. TURER[13], C. COATS[14], M. FIFER[12], A. OWENS[15], H. WATKINS[16], R. BARRIALES-VILLA[17], C. KRAMER[18], T. WONG[19], S.B. HEITNER[20], D.L. JACOBY[20], S. KUPFER[20], F.I. MALIK[20], L. MENG[20], S.L. PAIGE[20], A. TUGCU[20], A. WOHLTMAN[20], and S. SOLOMON[9]

1 University of California, San Francisco, CA; 2 Oregon Health Science University, Portland, OR; 3 Tufts Medical Center, Boston, MA; 4 Northwestern University, Chicago, IL; 5 Azienda Ospedaliera Universitaria Careggi, Florence, Italy; 6 University of Michigan Medical Center, Ann Arbor, MI; 7 Duke University, Durham, NC; 8 Hospital Universitario Puerta de Hierro de Majadahonda, Madrid, Spain; 9 Brigham and Women's Hospital, Boston, MA; 10 Methodist DeBakey Heart and Vascular Center, Houston, TX; 11 Cedars Sinai Medical Center, Los Angeles, CA; 12 Massachusetts General Hospital, Boston, MA; 13 UT Southwestern Medical Center, Dallas, TX; 14 University of Glasgow, Glasgow, United Kingdom; 15 University of Pennsylvania Perelman School of Medicine, Philadelphia, PA; 16 University of Oxford, Oxford, United Kingdom; 17 Complexo Hospitalario Universitario de A Coruña, A Coruña, Spain; 18 University of Virginia Health System, Charlottesville, VA; 19 University of Pittsburgh Medical Center, Pittsburgh, PA; 20 Cytokinetics Incorporated, South San Francisco, CA

INTRODUCTION

Hypertrophic cardiomyopathy (HCM) is a common genetic disease that affects roughly 20 million people worldwide. Patients with HCM may experience debilitating symptoms and are at an increased risk of sudden cardiac death. In many cases, the underlying cause of HCM is due to mutations in genes encoding proteins of the sarcomere. The resulting cardiac **hypercontractility** and hypertrophy, combined with systolic anterior motion (SAM) of the mitral valve, can lead to left ventricular outflow tract (LVOT) obstruction. **Despite standard-of-care medication therapy,** including beta-blockers, calcium channel blockers, and disopyramide, as well as septal reduction therapy (SRT), **many patients remain symptomatic**.

Aficamten is a next-in-class cardiac myosin inhibitor that **counteracts the hypercontractility** underlying the pathophysiology of HCM. *Aficamten* has been engineered to leverage 3 key pharmacological attributes:

- **Half life of approximately 3.4 days**
 - → once daily dosing
 - → steady state by 2 weeks
 - → rapid reversibility
- **Shallow exposure-response relationship**
 - → wide therapeutic window
 - → echocardiographically guided dose titration
- **No significant CYP inhibition or induction**
 - → minimal drug-drug interactions

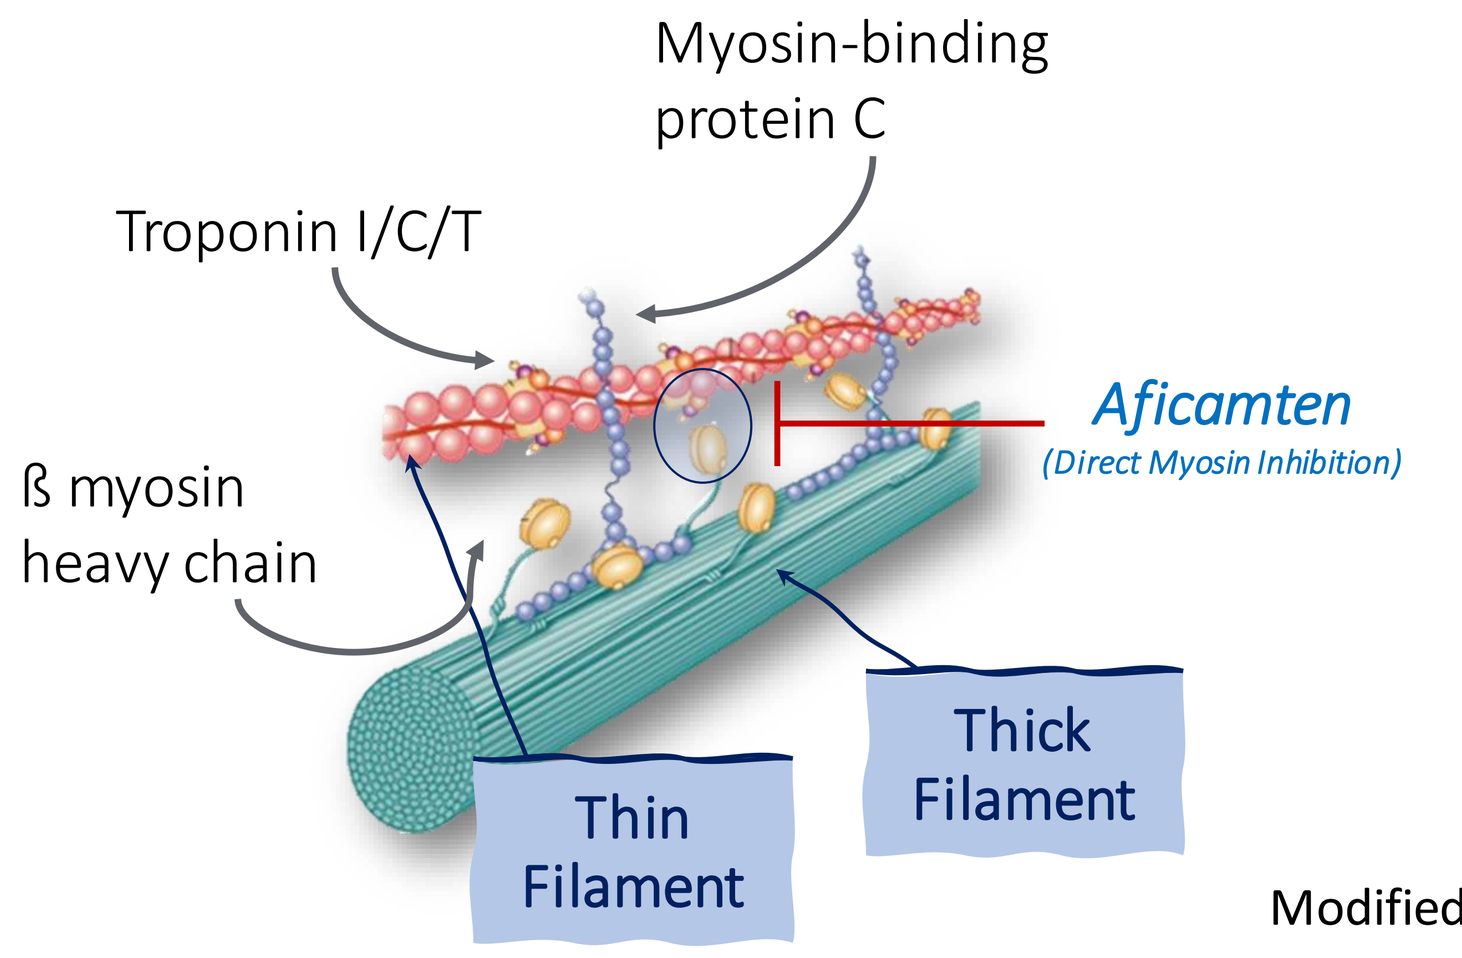
Myosin-binding protein C
Troponin I/C/T
ß myosin heavy chain
Aficamten
(Direct Myosin Inhibition)
Thick Filament
Thin Filament

Modified from Semsarian et al. (2015)

AIM

Randomized Evaluation of Dosing With CK-274 in Obstructive Outflow Disease in HCM (REDWOOD-HCM:NCT04219826) is a multicenter, randomized, controlled trial of *aficamten* in patients with symptomatic obstructive HCM. We sought to **evaluate echocardiographic metrics of cardiac structure and function** beyond the previously reported reductions in resting and provoked LVOT gradients.

METHODS

Key Inclusion Criteria

- 18-85 years old
- Unexplained LV wall thickness ≥ 15 mm
- Symptomatic (NYHA class II or III)
- Obstructive HCM
 - Resting LVOT-G ≥ 50 mmHg
 -OR-
 - Resting LVOT-G ≥ 30 mmHg AND Valsalva LVOT-G ≥ 50 mmHg
- Baseline LVEF ≥ 60%
- Stable doses of background medical therapy (beta-blockers, calcium channel blockers)

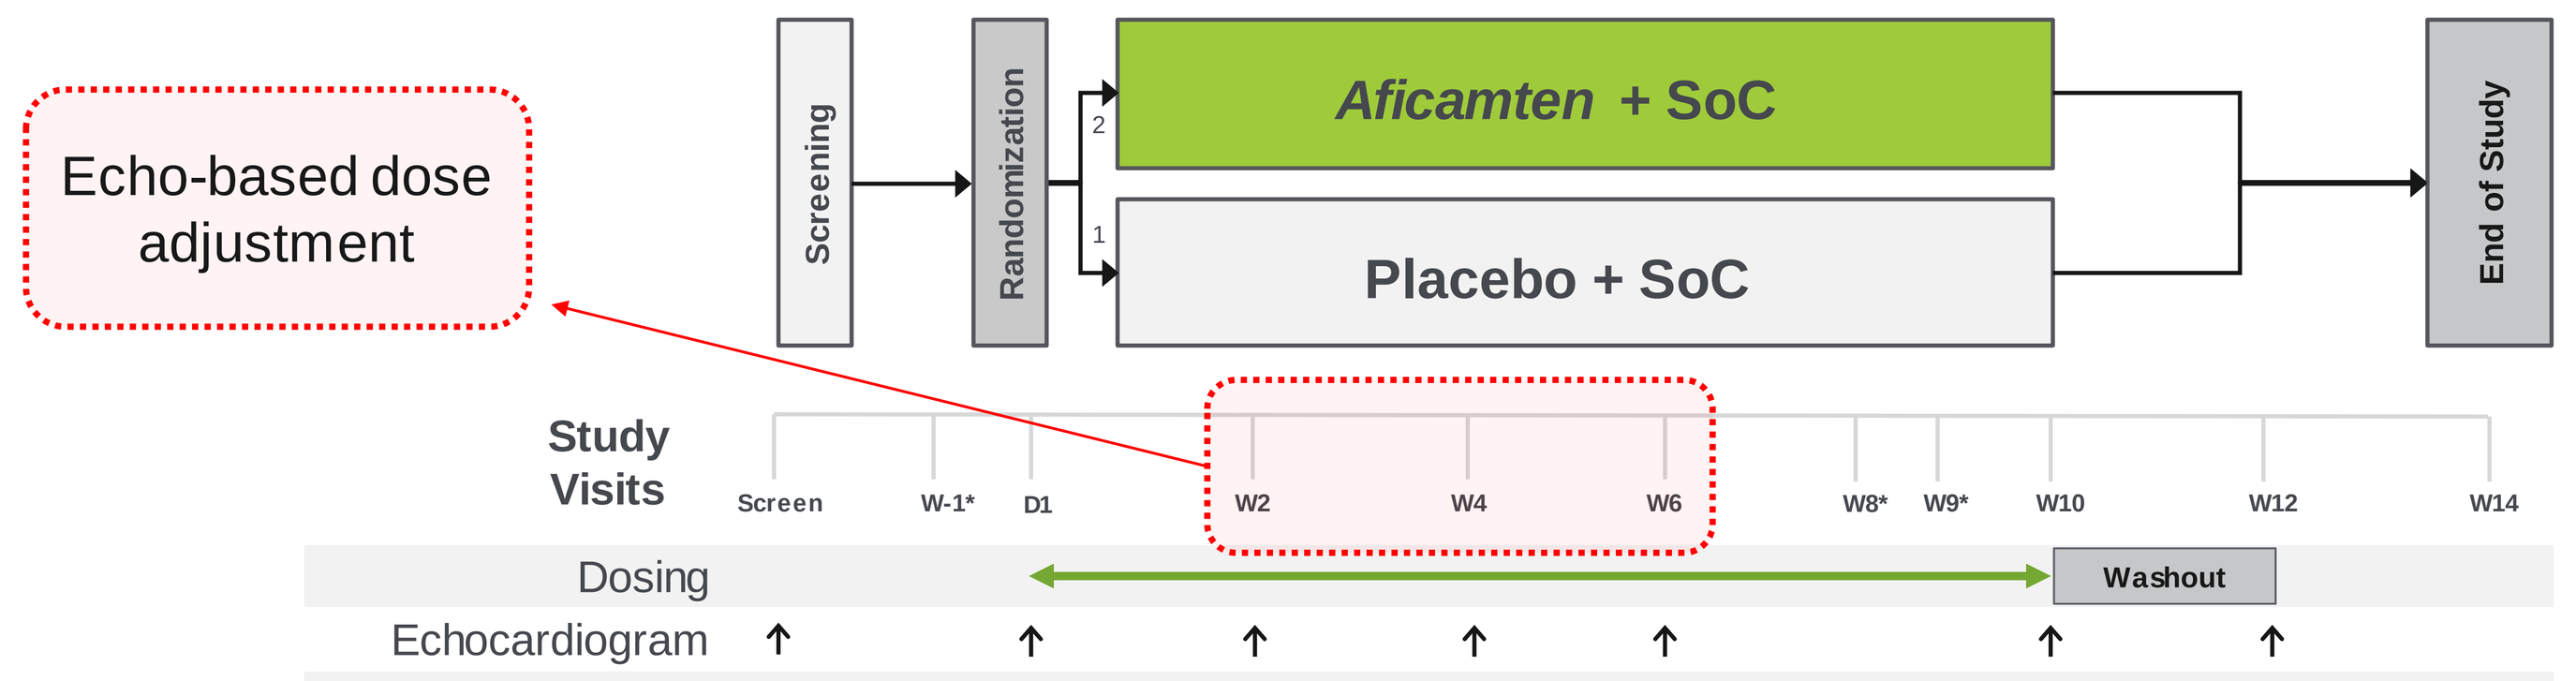
Echo-based dose adjustment
Screening
Randomization
Aficamten + SoC
Placebo + SoC
End of Study
Study Visits
Screen W-1* D1 W2 W4 W6 W8* W9* W10 W12 W14
Dosing
Washout
Echocardiogram

	Dose 1	Dose 2	Dose 3
Cohort 1 (N = 21)	5 mg	10 mg	15 mg
Cohort 2 (N = 22)	10 mg	20 mg	30 mg

RESULTS

Baseline Characteristics

Characteristic	Placebo (n = 13)	*Aficamten* (n = 28)
Age (Years), Mean (SD) [Range]	57.2 (9.6) [36-69]	56.6 (13.6) [33-78]
Female, n (%)	8 (62%)	15 (54%)
Race = white, n (%)	12 (92%)	28 (100%)
NYHA Class, n (%)		
Class II	11 (85%)	17 (61%)
Class III	2 (15%)	11 (39%)
NT-proBNP (pg/mL), Median (IQR)	532 (129-958)	388 (202-1261)
Echocardiographic Characteristics		
LVEF (%), Mean (SD)*	73.6 (5.9)	71.7 (8.0)
LVOT-G at Rest (mmHg), Mean (SD)*	70.0 (28.0)	61.1 (29.8)
LVOT-G with Valsalva (mmHg), Mean (SD)*	93.3 (27.2)	89.3 (31.5)
Lateral E/e' ratio, Mean (SD)	17.4 (10.0)	13.8 (6.3)
Lateral e' (cm/s), Mean (SD)	5.8 (2.1)	6.7 (2.3)
LAVI (mL/m^2), Mean (SD)	31.4 (7.5)	32.5 (8.1)
LVMI (g/m^2), Mean (SD)	103.3 (25.1)	109.8 (28.6)
Presence of MR, n (%)	11 (84.6%)	26 (92.9%)
Presence of Eccentric MR, n (%)	3 (25.0%)	12 (42.9%)
Presence of SAM, n (%)	12 (92.3%)	24 (85.7%)

*Site reads from screening echocardiograms

RESULTS

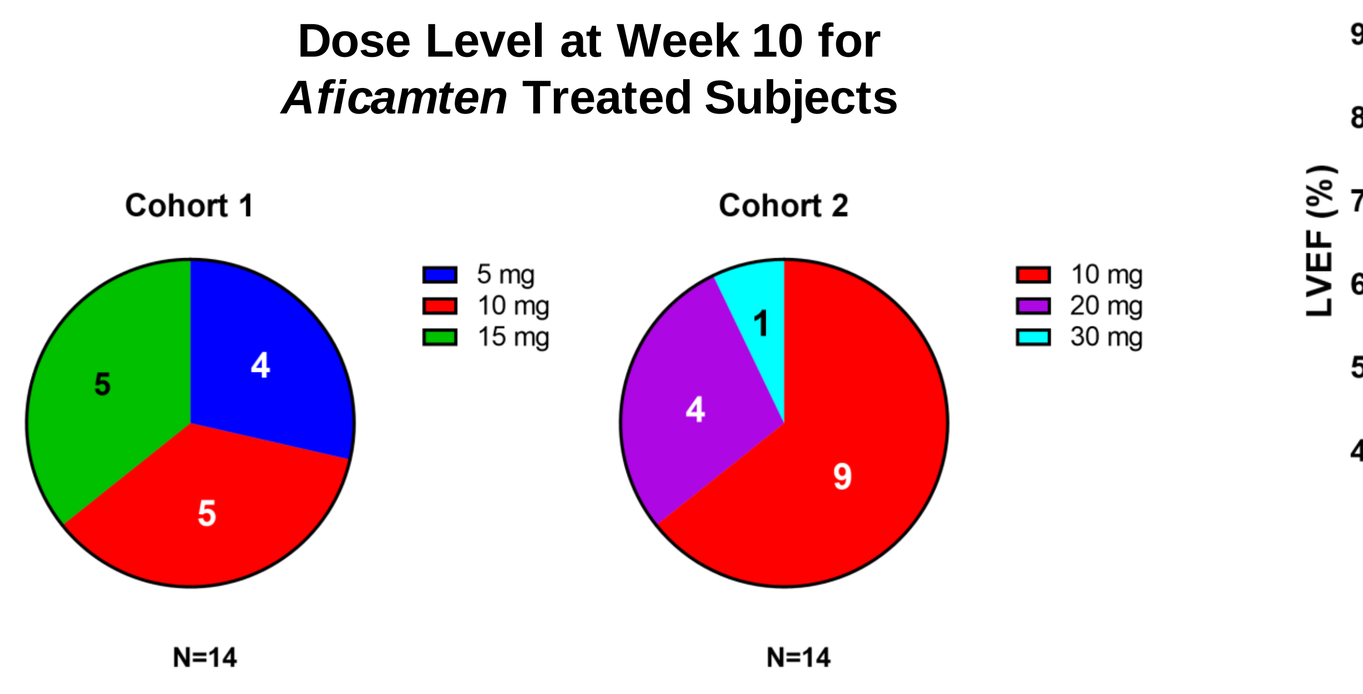
Dose Level at Week 10 for Aficamten Treated Subjects
Cohort 1
Cohort 2
5 mg
10 mg
15 mg
10 mg
20 mg
30 mg
N=14
N=14

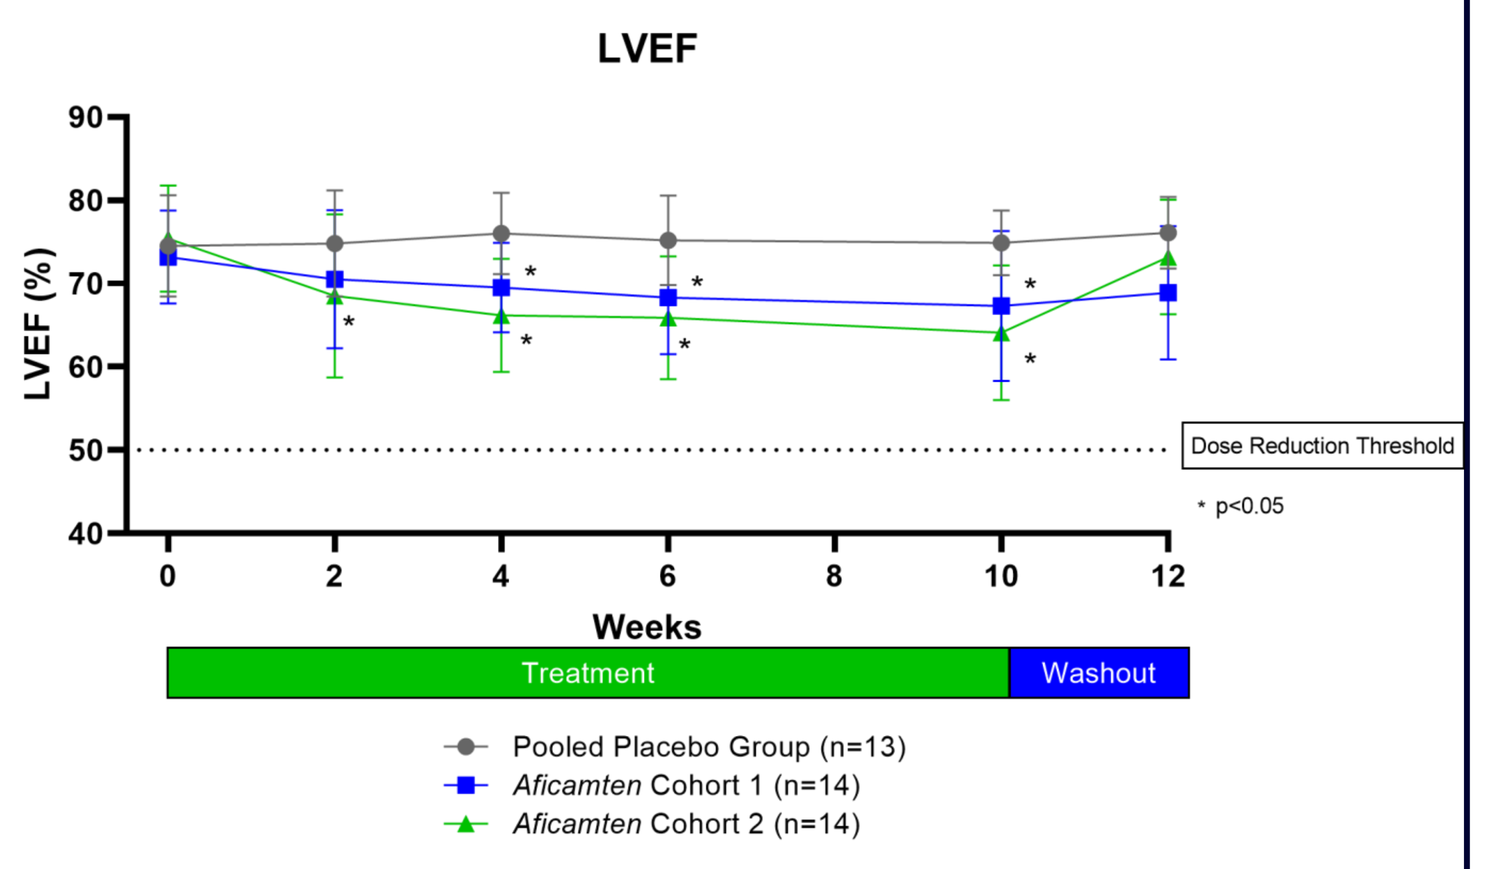
LVEF
LVEF (%)
Dose Reduction Threshold
* p<0.05
Weeks
Treatment
Washout
Pooled Placebo Group (n=13)
Aficamten Cohort 1 (n=14)
Aficamten Cohort 2 (n=14)

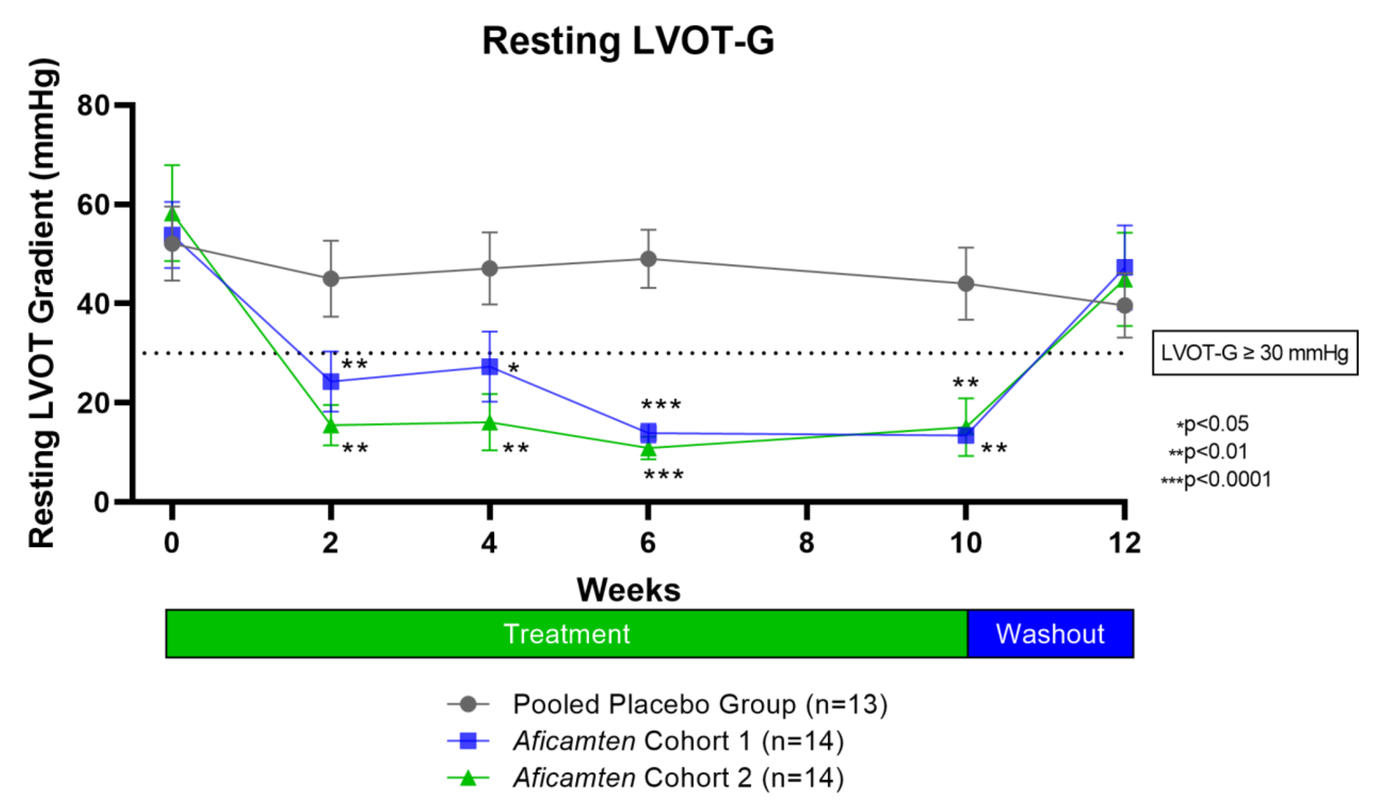
Resting LVOT-G
Resting LVOT Gradient (mmHg)
LVOT-G ≥ 30 mmHg
*p<0.05
**p<0.01
***p<0.0001
Weeks
Treatment
Washout
Pooled Placebo Group (n=13)
Aficamten Cohort 1 (n=14)
Aficamten Cohort 2 (n=14)

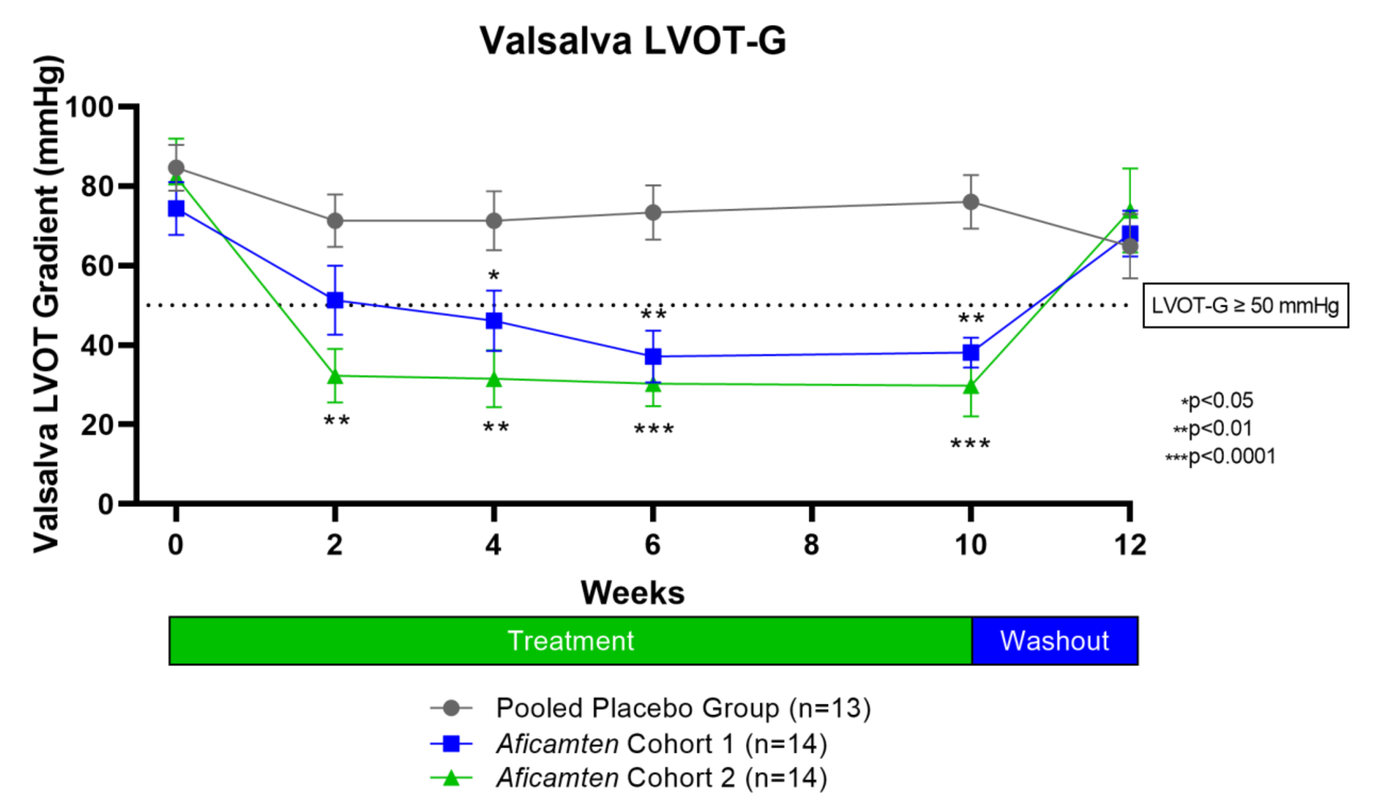
Valsalva LVOT-G
Valsalva LVOT Gradient (mmHg)
LVOT-G ≥ 50 mmHg
*p<0.05
**p<0.01
***p<0.0001
Weeks
Treatment
Washout
Pooled Placebo Group (n=13)
Aficamten Cohort 1 (n=14)
Aficamten Cohort 2 (n=14)

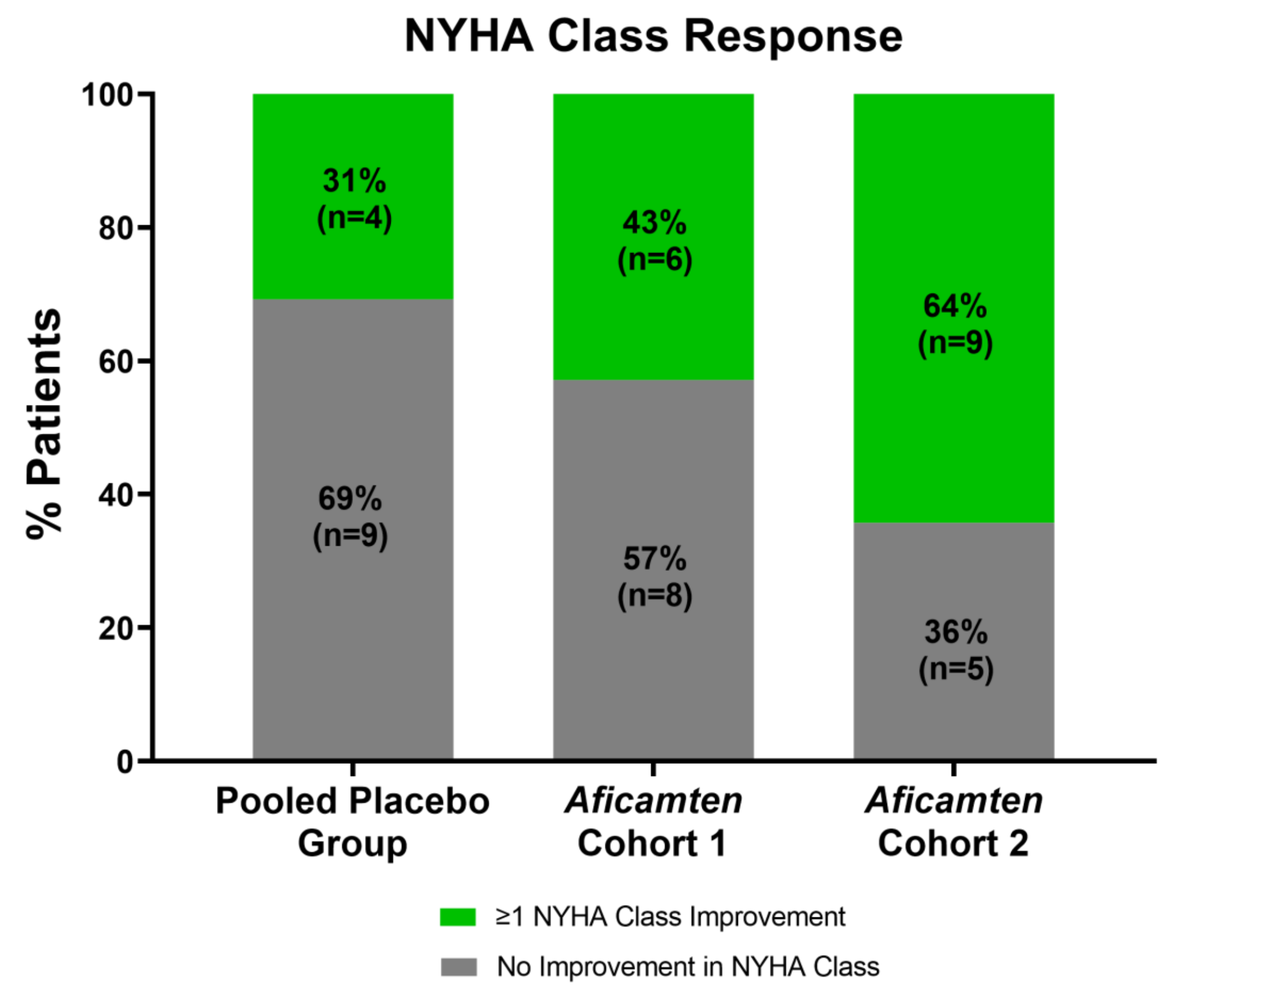
NYHA Class Response
% Patients
31% (n=4)
69% (n=9)
43% (n=6)
57% (n=8)
64% (n=9)
36% (n=5)
Pooled Placebo Group
Aficamten Cohort 1
Aficamten Cohort 2
≥1 NYHA Class Improvement
No Improvement in NYHA Class

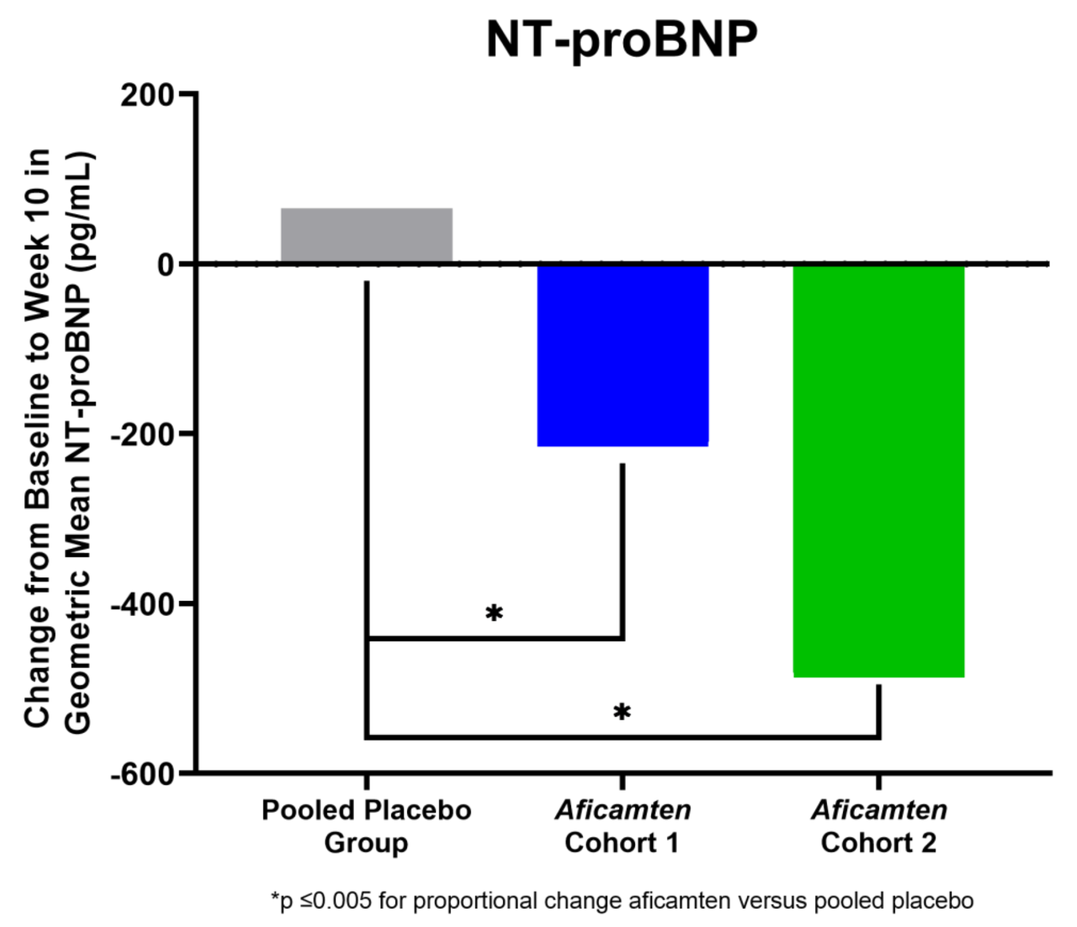
NT-proBNP
Change from Baseline to Week 10 in Geometric Mean NT-proBNP (pg/mL)
Pooled Placebo Group
Aficamten Cohort 1
Aficamten Cohort 2
*p ≤0.005 for proportional change aficamten versus pooled placebo

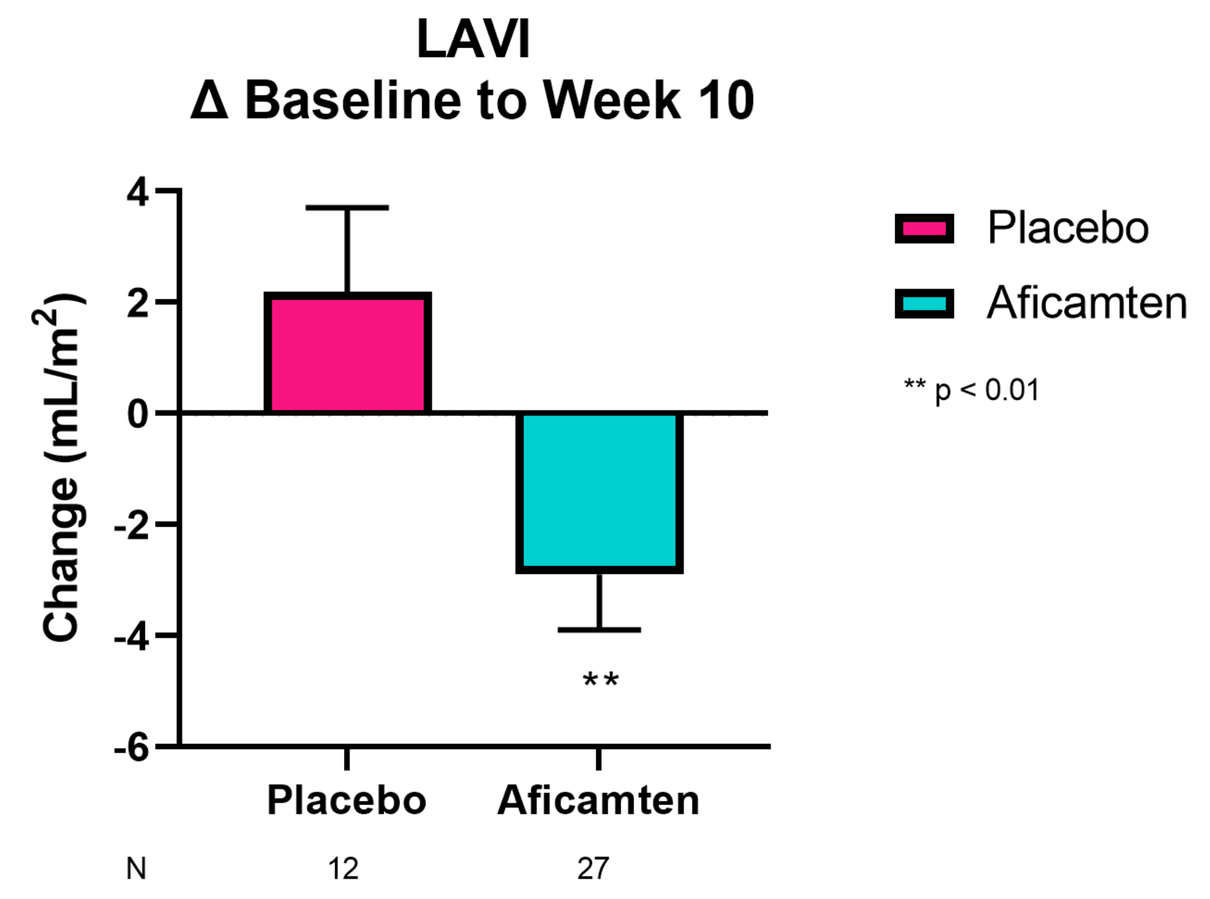
LAVI
Δ Baseline to Week 10
Change (mL/m2)
Placebo
Aficamten
** p < 0.01
N 12 27

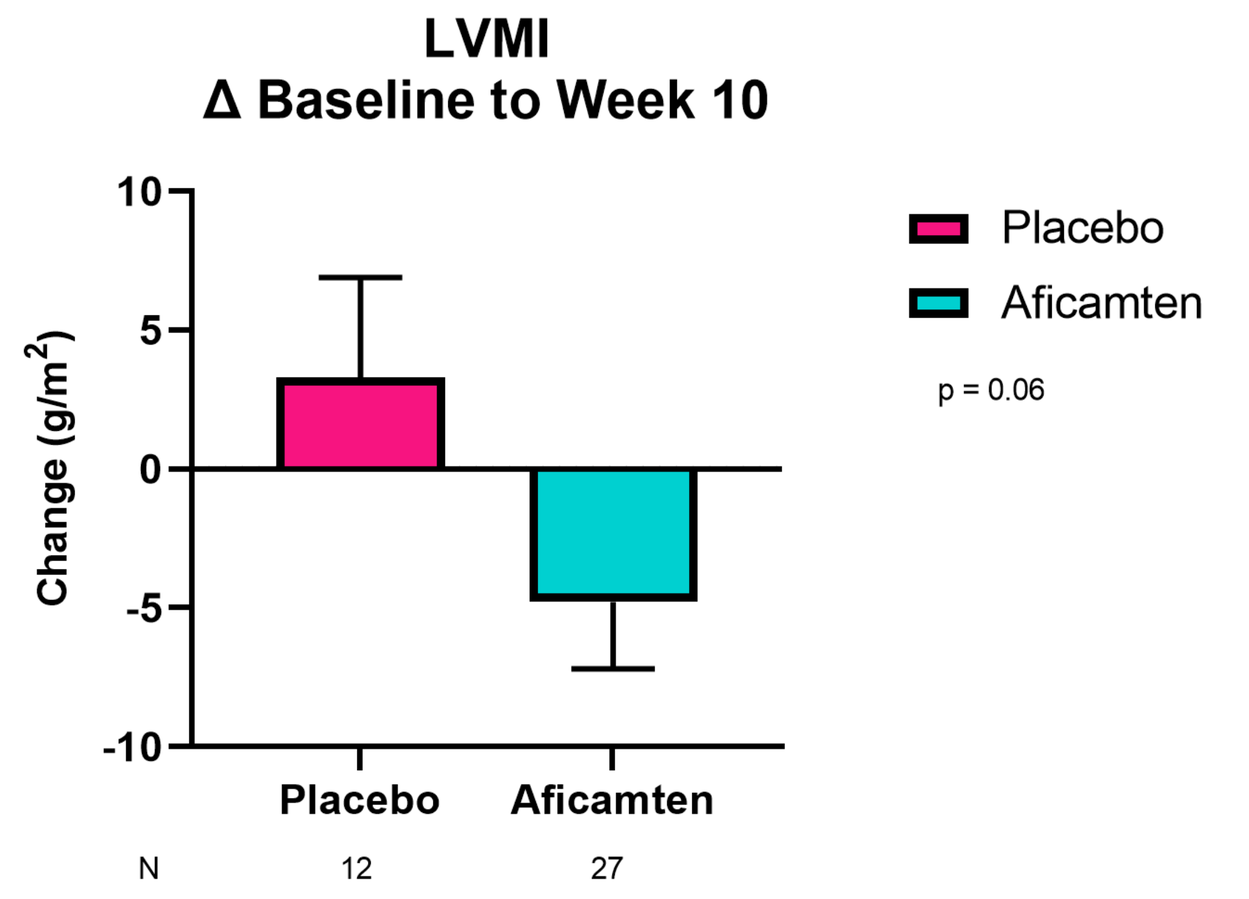
LVMI
Δ Baseline to Week 10
Change (g/m2)
Placebo
Aficamten
p = 0.06
N 12 27

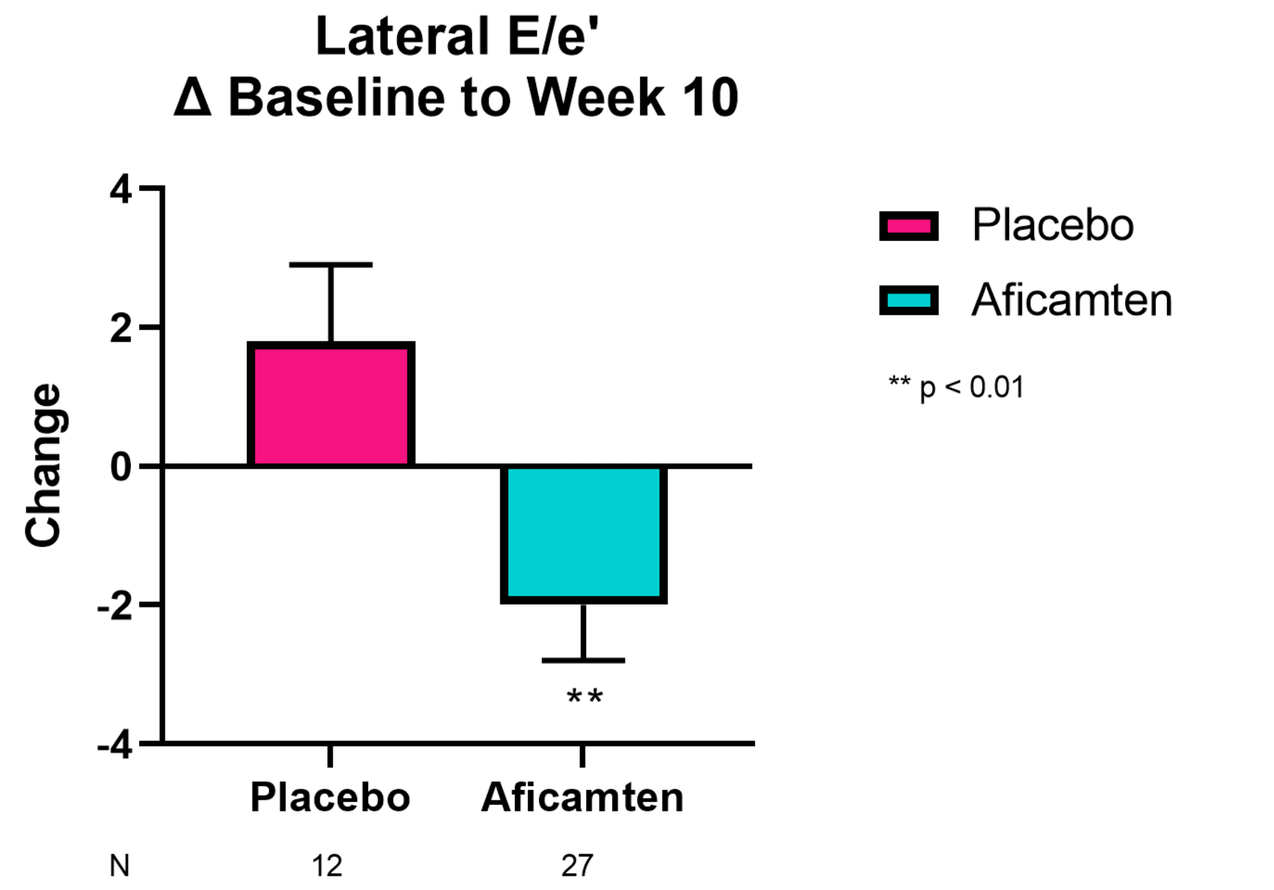
Lateral E/e'
Δ Baseline to Week 10
Change
Placebo
Aficamten
** p < 0.01
N 12 27

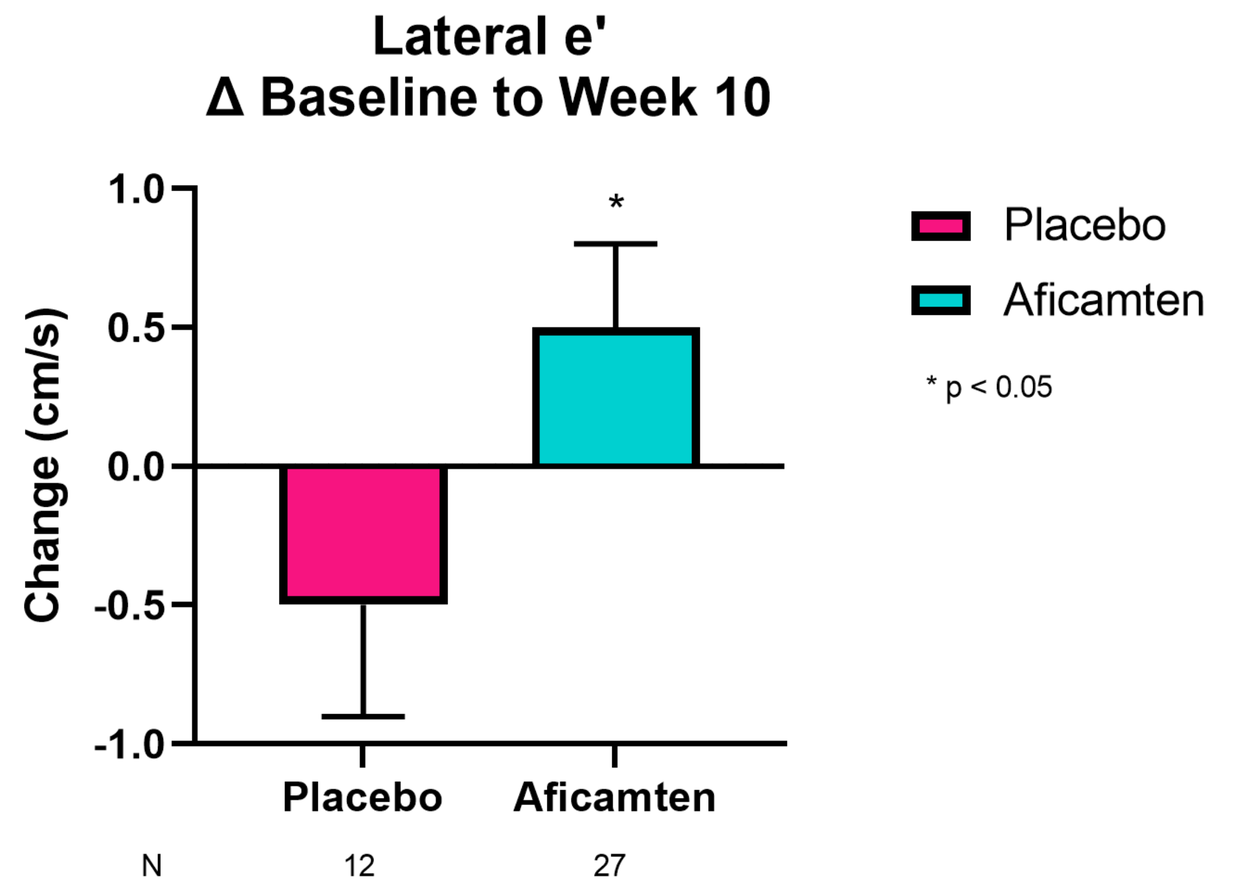
Lateral e'
Δ Baseline to Week 10
Change (cm/s)
Placebo
Aficamten
* p < 0.05
N 12 27

RESULTS

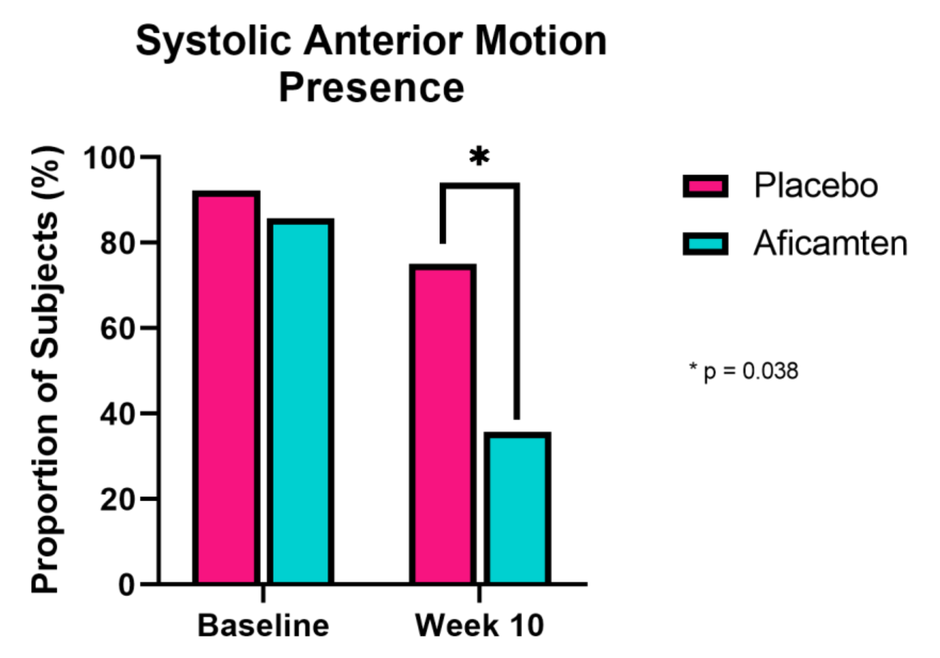
Systolic Anterior Motion Presence
Proportion of Subjects (%)
Baseline
Week 10
Placebo
Aficamten
* p = 0.038

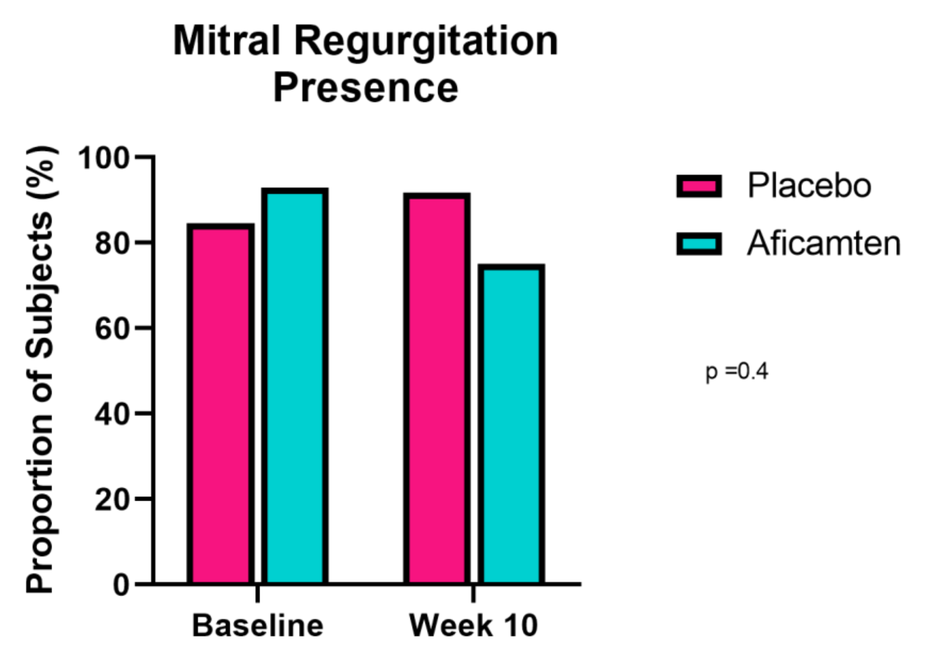
Mitral Regurgitation Presence
Proportion of Subjects (%)
Baseline
Week 10
Placebo
Aficamten
p =0.4

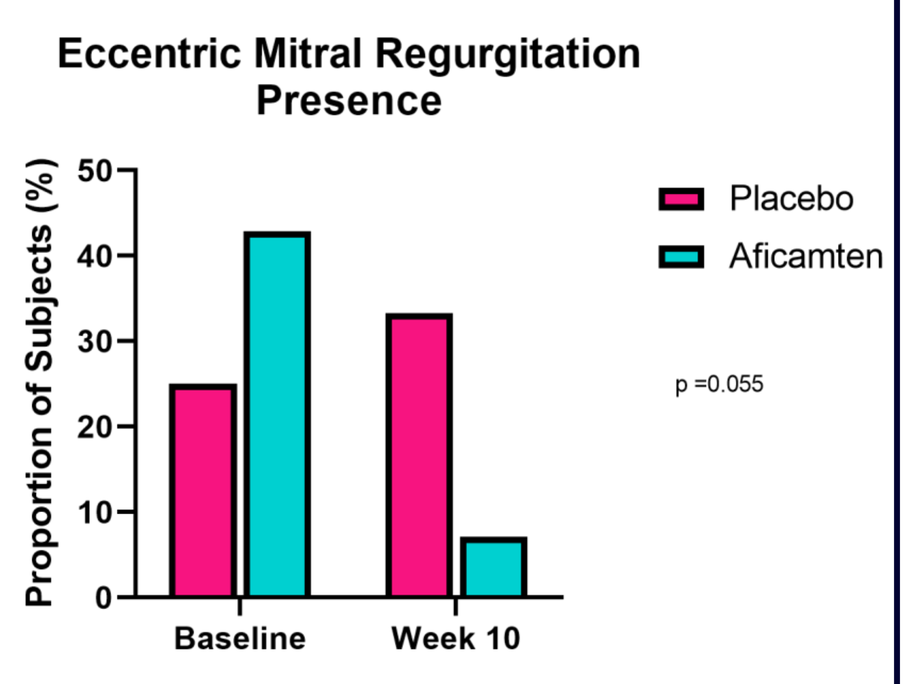
Eccentric Mitral Regurgitation Presence
Proportion of Subjects (%)
Baseline
Week 10
Placebo
Aficamten
p =0.055

CONCLUSIONS

- After only 10 weeks of treatment with *aficamten*:
 - **Improved Cardiac Structure**
 - Reduction in LAVI
 - **Improved Myocardial Relaxation**
 - Increased Lateral e'
 - Reduction in Lateral E/e'
 - **Improved Mitral Valve Mechanics**
 - Less SAM
 - Less eccentric MR
- Echocardiographic improvements paralleled by:
 - **Reduction in resting and provoked LVOT obstruction**
 - **Reduction in plasma NT-proBNP**
- *Aficamten* was well tolerated
 - **No dose interruptions or discontinuations**
 - **1 case whereby dose was successfully down-titrated in response to a LVEF <50% (site read 48.2%) and subsequent increase of LVEF**
- Longer-term echocardiographic and cardiac MRI data from REDWOOD-HCM OLE and the Phase 3 study SEQUOIA-HCM will provide further insights into the longer-term effects of *aficamten* on cardiac structure and function

REFERENCES

- Maron et al., "REDWOOD-HCM: A randomized, double-blind, placebo-controlled, dose-finding trial of the cardiac myosin inhibitor, aficamten, in obstructive hypertrophic cardiomyopathy" (*Heart Failure Society of America* Annual Scientific Meeting, September 2021).
- Semsarian et al., "New perspectives on the prevalence of hypertrophic cardiomyopathy." *J Am Coll Cardiol.* 2015;65(12):1249-1254.