i186

Sudden Changes in the ALSFRS-R in Three ALS Clinical Trials

Jeremy M. Shefner,[1] Stuart Kupfer,[2] Fady I. Malik,[2] Lisa Meng,[2] Stacy A. Rudnicki,[2] Andrew A. Wolff,[2] Jenny Wei[2]

[1]Barrow Neurological Institute, University of Arizona, and Creighton University, Phoenix, AZ, USA; [2]Cytokinetics Incorporated, South San Francisco, CA, USA

BACKGROUND

ALSFRS-R as an outcome measure

- The ALSFRS-R (Amyotrophic Lateral Sclerosis Functional Rating Scale Revised) is widely used to assess disease severity and progression in patients with ALS
- It is also the most commonly used primary outcome measure in late-phase clinical trials for potential ALS therapies

ALSFRS-R variability

- The ALSFRS-R has been shown to be sensitive in detecting modest clinical benefits, although there is concern that the effectiveness of detecting change can be reduced by inconsistency in measurement or by variability in patient function
 - It is well known that patients have “good days” and “bad days”, and a measurement on these days may appear to indicate a sudden change

OBJECTIVE

- To determine the frequency of sudden visit-to-visit variations in the ALSFRS-R score in three large clinical trials in patients with ALS, as well as the impact on results

METHODS

Patients and assessments

- We evaluated ALSFRS-R total scores from the placebo arms of phase 2 and 3 studies of *tirasemtiv* in ALS (BENEFIT-ALS, VITALITY-ALS),[1,2] and the phase 2 study of *reldesemtiv* in ALS (FORTITUDE-ALS)[3]
 - Across the three clinical trials, 512 patients were randomized to placebo
 - Participants were followed for 16 weeks (in BENEFIT-ALS and FORTITUDE-ALS) or 24 weeks (in VITALITY-ALS)
 - ALSFRS-R was assessed regularly throughout the clinical trials

ALSFRS-R variability analysis

- We identified visit-to-visit pairs in which the score of a given visit was ≥5 points more than the preceding visit
 - An identical analysis was applied for increases of ≥4 points
- To determine the extent to which these anomalous score changes impacted group data, ALSFRS-R slopes from the entire group of patients were compared with subgroups excluding:
 - All data from the participants who had large increases
 - Only the anomalous points (retaining that participant’s scores at other weeks)
- We also evaluated whether patient characteristics predicted the ≥4- or ≥5-point increases

RESULTS

- Overall, 1.4% of patients had visit-to-visit pairs with ≥5-point increases, and 6.3% of patients had pairs with ≥4-point increases
 - The increases tended to occur at earlier visits (**Table 1**)
 - No clinical trial site had more than one participant with anomalous pairs
- The slopes of change in the ALSFRS-R score over time for participants with and without anomalous increases were very similar, as were slopes calculated with and without the anomalous data point included (**Figure 1**)
 - To illustrate this, **Table 2** shows the slope of decline for VITALITY-ALS, when sudden changes were included or when sudden changes were removed from the analysis

Table 1. Anomalous data points tended to occur early – frequency of sudden changes by study week

Second visit, weeks from baseline	Number of participants with sudden increase in ALSFRS-R total score	
	≥5-point increase	≥4-point increase
4	2	8
8	2	9
12	1	6
16	1	7
20	0	2
24	1	3

Second visit is the second of consecutive visits showing a sudden increase.

RESULTS continued

Figure 1. ALSFRS-R total scores over time for all participants with ≥4-point increase, with and without anomalous points

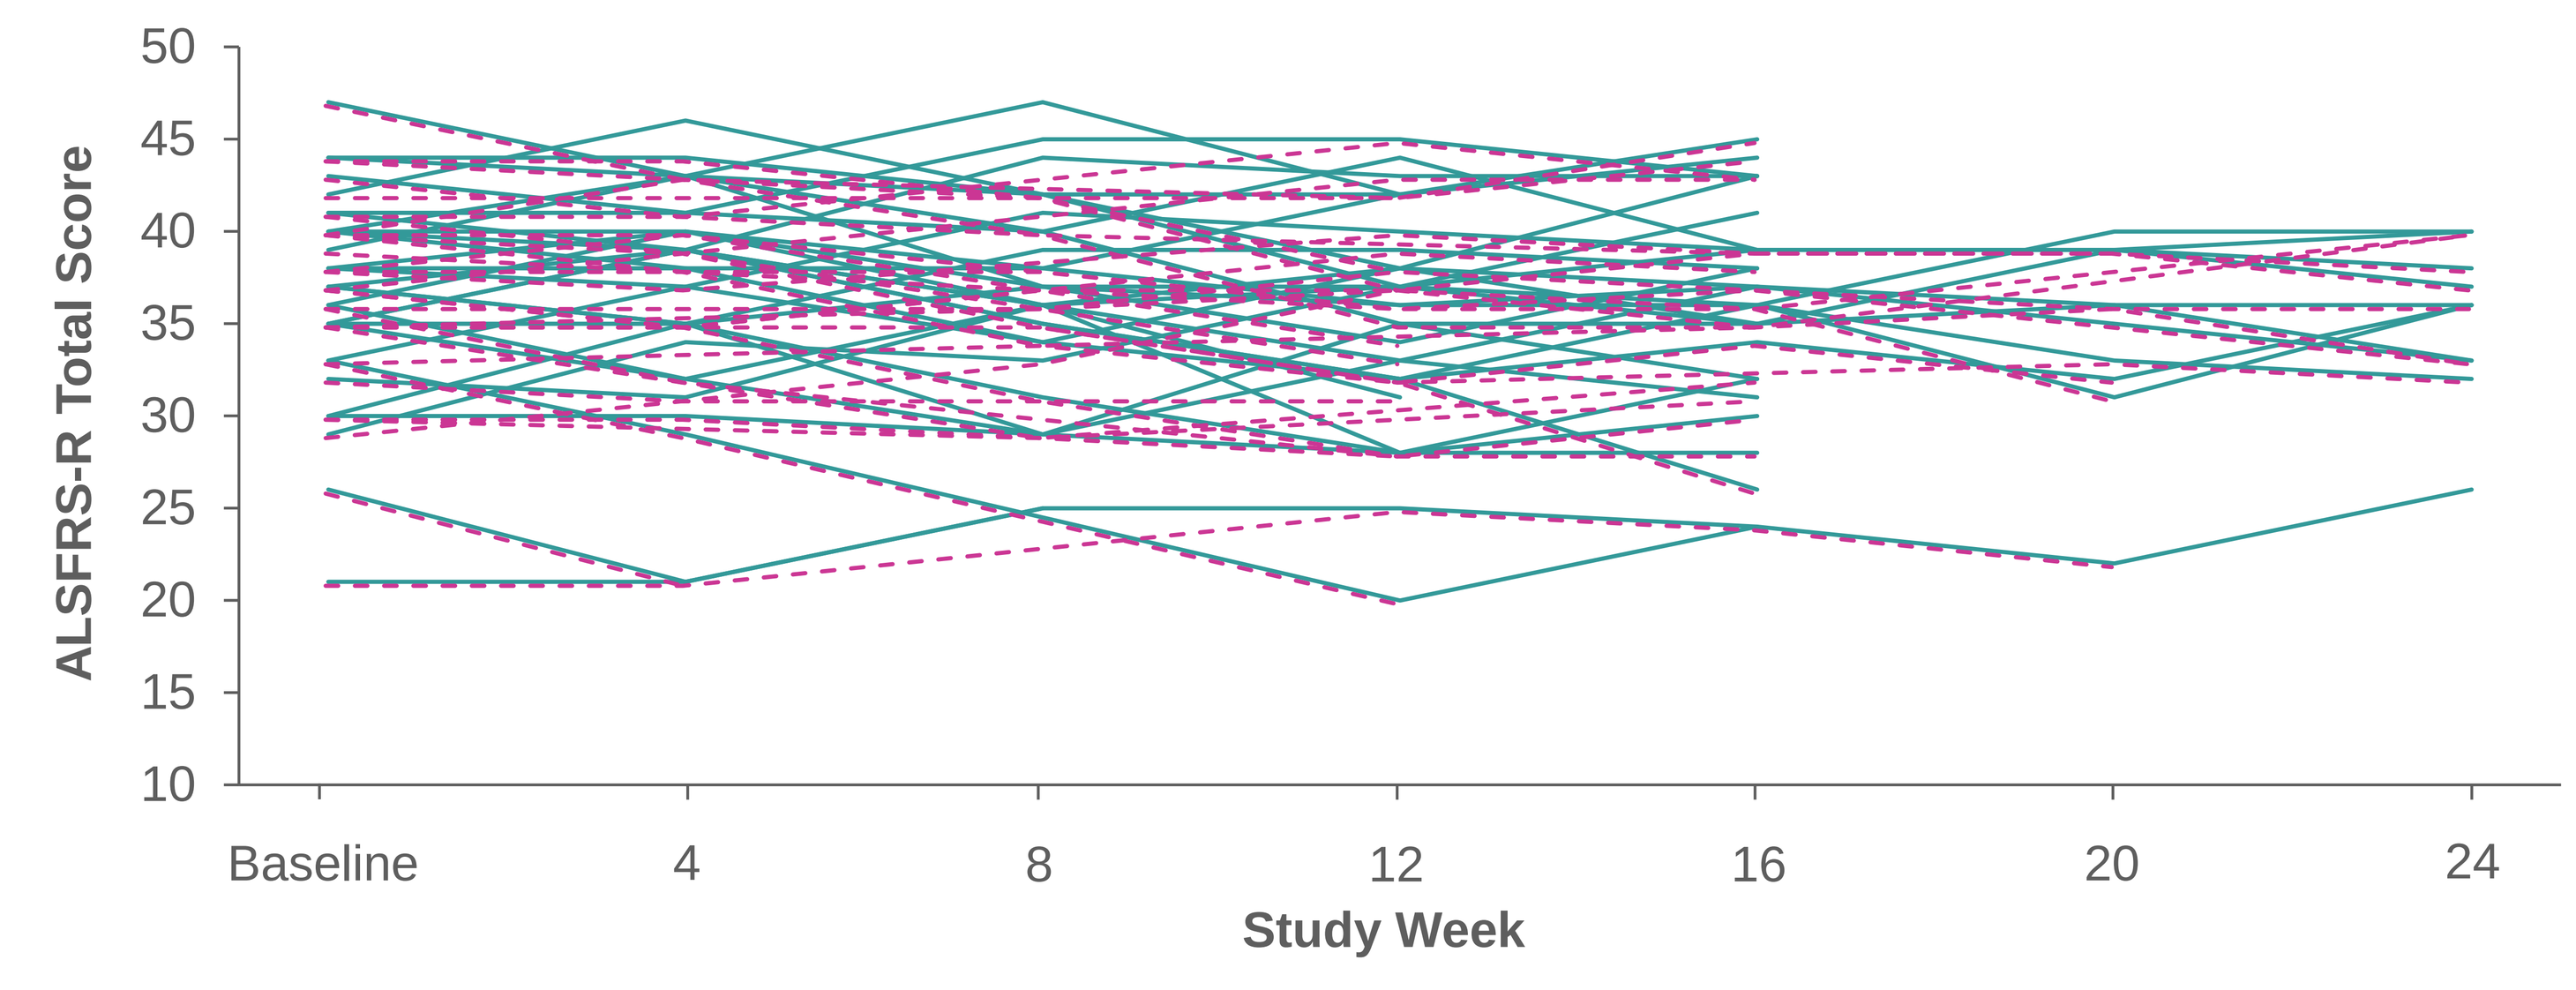
ALSFRS-R Total Score
50
45
40
35
30
25
20
15
10
Baseline
4
8
12
16
20
24
Study Week

Data are shown from all three clinical trials. Lines indicate ALSFRS-R total scores for individual patients with an increase of ≥4 points at some timepoint during the clinical trials. Pink dotted lines show data without anomalous data points; teal lines show data with anomalous points.

Table 2. Slopes of decline in ALSFRS-R total score over 0–24 weeks in VITALITY-ALS

	Slope of the change from baseline in ALSFRS-R total score (points per day)	
	Mean	95% confidence interval
All placebo-group participants	–0.032	–0.037, –0.026
Subgroups excluding sudden increases:		
Excluding patients with ≥4-point increase	–0.033	–0.038, –0.027
Excluding ≥4-point increases	–0.032	–0.037, –0.026
Excluding patients with ≥5-point increase	–0.032	–0.037, –0.026
Excluding ≥5-point increases	–0.032	–0.037, –0.026

Results are illustrated for VITALITY-ALS (study duration 24 weeks). Similar results were seen for the two 16-week studies. Subgroups excluding patients with increases exclude all data for that patient; subgroups excluding increases removed only the data point with the anomalous increase.

Analysis of association with patient characteristics

- For both ≥4 and ≥5-point increases, no difference in occurrences was seen as a function of:
 - Site of ALS onset
 - Time from symptom onset
 - Baseline ALSFRS-R total score

CONCLUSIONS

- Overall, in the three studies reviewed here, a very small number of participants had sudden changes in ALSFRS-R scores
 - These contributed very little to the overall estimate of slope in the study population because these data points were randomly scattered among participants
- Sudden ≥5-point increases occurred in <2% of participants, who were distributed across study sites, with no site having more than one participant with anomalous data points
 - This is in contrast to a previous analysis of two multicenter studies in which ≥5-point increases were noted in 12% of patients and events were clustered in particular study sites[4]
- Although we found no predictors of anomalous evaluations, other data suggest this occurs most often when different evaluators are testing the same participant

References

1. Shefner JM et al. *Amyotroph Lateral Scler Frontotemporal Degener* 2016;17:426-35.
2. Shefner JM et al. *Amyotroph Lateral Scler Frontotemporal Degener* 2019;20:584-94.
3. Shefner JM et al. *Amyotroph Lateral Scler Frontotemporal Degener* 2021;22:287-99.
4. Bakers JNE et al. *Amyotroph Lateral Scler Frontotemporal Degener* 2021;epub ahead of print.

Acknowledgments

BENEFIT-ALS and **VITALITY-ALS** were conducted by Cytokinetics, Incorporated.

FORTITUDE-ALS was conducted by Cytokinetics, Incorporated in collaboration with Astellas Pharma Inc.

We wish to thank the participants and their families for their contributions to these clinical trials, the investigators and study site staff, and members of the Data Monitoring Committees and Steering Committees.

Editorial support for the preparation of this poster was provided by Geraldine Thompson on behalf of Engage Scientific Solutions, Horsham, UK, and was funded by Cytokinetics, Incorporated.

©2022 CYTOKINETICS, All Rights Reserved.
CYTOKINETICS® and the CYTOKINETICS and C-shaped logo are registered trademarks of Cytokinetics in the U.S. and certain other countries.

Scan the QR code to obtain a PDF of this poster.
No personal information is stored.

Presented at the European Network for the Cure of ALS (ENCALS)
Edinburgh, UK | June 1–3, 2022

Barrow Neurological Institute | Gregory W. Fulton ALS Center

VITALITY ALS FORTITUDE ALS Cytokinetics