Medication Adherence and Adverse Drug Effects Across Patients with Obstructive Hypertrophic Cardiomyopathy Receiving Pharmacotherapy

Ralph J. Riello III[1,2], Michael Butzner[3], Eros Papademetriou[4], Sanatan Shreay[3], Nihar R. Desai[1]

[1]Department of Internal Medicine, Yale School of Medicine, New Haven, CT, USA; [2]Clinical and Translational Research Accelerator, Yale School of Medicine, New Haven, CT, USA; [3]Cytokinetics, Inc., South San Francisco, CA, USA; [4]Putnam Associates, HEOR & RWE, New York, NY, USA

INTRODUCTION

- Historically, standard of care (SoC) pharmacologic management of symptomatic obstructive hypertrophic cardiomyopathy (oHCM) includes beta-blockers (BB), calcium channel blockers (CCB), or combination therapy with disopyramide.
- These agents may provide symptomatic relief due to left ventricular outflow tract obstruction, but use is generally limited by tolerability.
- While disease-modifying therapies for oHCM are emerging, rates of adverse drug effects (ADEs) and adherence for generic pharmacotherapy are not well described.

OBJECTIVES

- To evaluate the incidence of ADEs and medication adherence in patients with oHCM using the Symphony real-world claims database.

METHODS

Study Design

- Retrospective study of adults diagnosed with oHCM in the US from 2016 to 2024, using administrative medical and pharmacy claims data from the Symphony Integrated Dataverse (IDV).
- The IDV contains longitudinal data that capture adjudicated prescription, medical, and hospital claims across the US for all insurance types: claims from over 65,000 pharmacies, 1500 hospitals, 800 outpatient facilities, and 80,000 physician practices across the US for nearly 180 million patients.

Simplified eligibility criteria

- Patients had ≥2 claims for oHCM (ICD-10 I42.1) >30 days apart (index date first oHCM claim; see full selection criteria on **Table 1**).
- Treatment with a BB, CCB, and/or disopyramide after index date.
- At least 18 years of age as of the index date.

Baseline enrollment

- Continuous enrollment with medical and pharmacy benefits for 1 year prior to the index date.

Exclusion criteria

- Patients with evidence of Fabry disease or amyloidosis during the study period were excluded.
- Patients with any oHCM treatment 1 year prior to index were excluded.

Study Outcomes

- Medication adherence for SoC pharmacotherapies for HCM.
- Incidence rates of ADEs for SoC.

Statistical Methods

- Medication adherence was assessed by proportion of days covered (PDC) and with a threshold of 0.80 considered adherent.
- Incidence rates (IR) of ADEs were evaluated per 100 person-years using generalized linear models with Poisson distribution. Adherence as measured by PDC (mean ± SD; percent adherent) and ADEs (mean [95% CI]) were reported at 2 years.

Table 1: Patient selection criteria

Step	Description	n
1	≥1 medical claim diagnosis of HCM (ICD-10 diagnosis codes: I42.1, I42.2) from January 1, 2016–March 31, 2024	438,530
2	≥2 diagnoses of oHCM (I42.1) ≥30 days apart OR 1 diagnosis of HCM (I42.2) along with 1 diagnosis of oHCM (I42.1) ≥30 days apart	101,856
3	AND any time on or after index diagnosis date, treatment with a BB, CCB, disopyramide	76,349
4	AND with 12 months of activity prior to index diagnosis date	69,465
5	AND age ≥18 years at index diagnosis date	68,504
6	AND no other oHCM treatment (drugs or procedures) 12 months prior to index diagnosis date	21,291
7	AND no diagnosis of Fabry disease or amyloidosis at any time	20,947
8	AND have no procedures within 30 days from index treatment	20,732
9	AND do not receive combination treatment except BB+CCB	20,539
	Total cohort of patients with oHCM	**20,539**

BB+CCB, beta-blocker + calcium channel blocker; HCM, hypertrophic cardiomyopathy; oHCM, obstructive hypertrophic cardiomyopathy.

RESULTS

- Among 20,539 patients with oHCM (mean age 60.3 ± 13.5 years; 53.1% were female; 35.0% reside in southern USA), the majority were receiving BB (66.8%) at index diagnosis, followed by CCB (20.7%), BB+CCB (10.1%), and disopyramide (1.1%) (**Table 2, Table 3**).

Table 2: Patient demographics

Characteristic, n (%) unless otherwise specified	oHCM N=20,539
Age, years	
Mean (SD)	60.3 (13.5)
Median (Q1–Q3)	63 (53–72)
Min, max	18, 79
18–34	1286 (6.3)
35–44	1490 (7.3)
45–54	2988 (14.5)
55–64	5239 (25.5)
65+	9536 (46.4)
Gender	
Male	9632 (46.9)
Female	10,907 (53.1)
US region	
Northeast	5089 (24.8)
North Central	5289 (25.8)
South	7191 (35.0)
West	2909 (14.2)
Unknown	61 (0.3)
Index therapy group	
Monotherapy	18,465 (89.9)
BB Only	13,711 (66.8)
CCB Only	4245 (20.7)
Disopyramide only	216 (1.1)
Combination therapy (BB + CCB)	2074 (10.1)
Length from index diagnosis to end of study follow-up, months	
Mean (SD)	30.2 (25.3)
Median (Q1–Q3)	24 (9–46)
Min, max	0, 99

BB, beta-blocker; CCB, calcium channel blocker; max, maximum; min, minimum; Q, quartile.

Table 3: Clinical characteristics

Characteristic, n (%)	oHCM N=20,539
Any comorbidity	18,556 (90.3)
Hypertension	12,748 (62.1)
Renal failure	1902 (9.3)
Atrial fibrillation	3498 (17.0)
Atrial flutter	491 (2.4)
Ventricular fibrillation	127 (0.6)
Ventricular tachycardia	1098 (5.3)
Diabetes	4144 (20.2)
Valvular disease	6731 (32.8)
Stroke	654 (3.2)
Dyslipidemia	9466 (46.1)
Coronary artery disease	4338 (21.1)
Chronic pulmonary disease	3741 (18.2)
Congestive heart failure	5172 (25.2)
Liver disease	982 (4.8)
Symptoms (±3 months from index diagnosis)	
Any symptom	11,837 (57.6)
Fatigue	1491 (7.3)
Chest pain	4309 (21.0)
Syncope	1973 (9.6)
Dyspnea	5929 (28.9)
Heart failure	3838 (18.7)
Palpitations	2711 (13.2)

- At 2 years, mean PDC was 0.55 ± 0.33; 67% of patients had a PDC <0.80.
- Adherence was highest among patients receiving BB+CCB (0.58 ± 0.31), followed by BB (0.56 ± 0.33), CCB (0.49 ± 0.33), and disopyramide (0.33 ± 0.28) (**Figure 1**).

Figure 1: Medication adherence among patients with oHCM

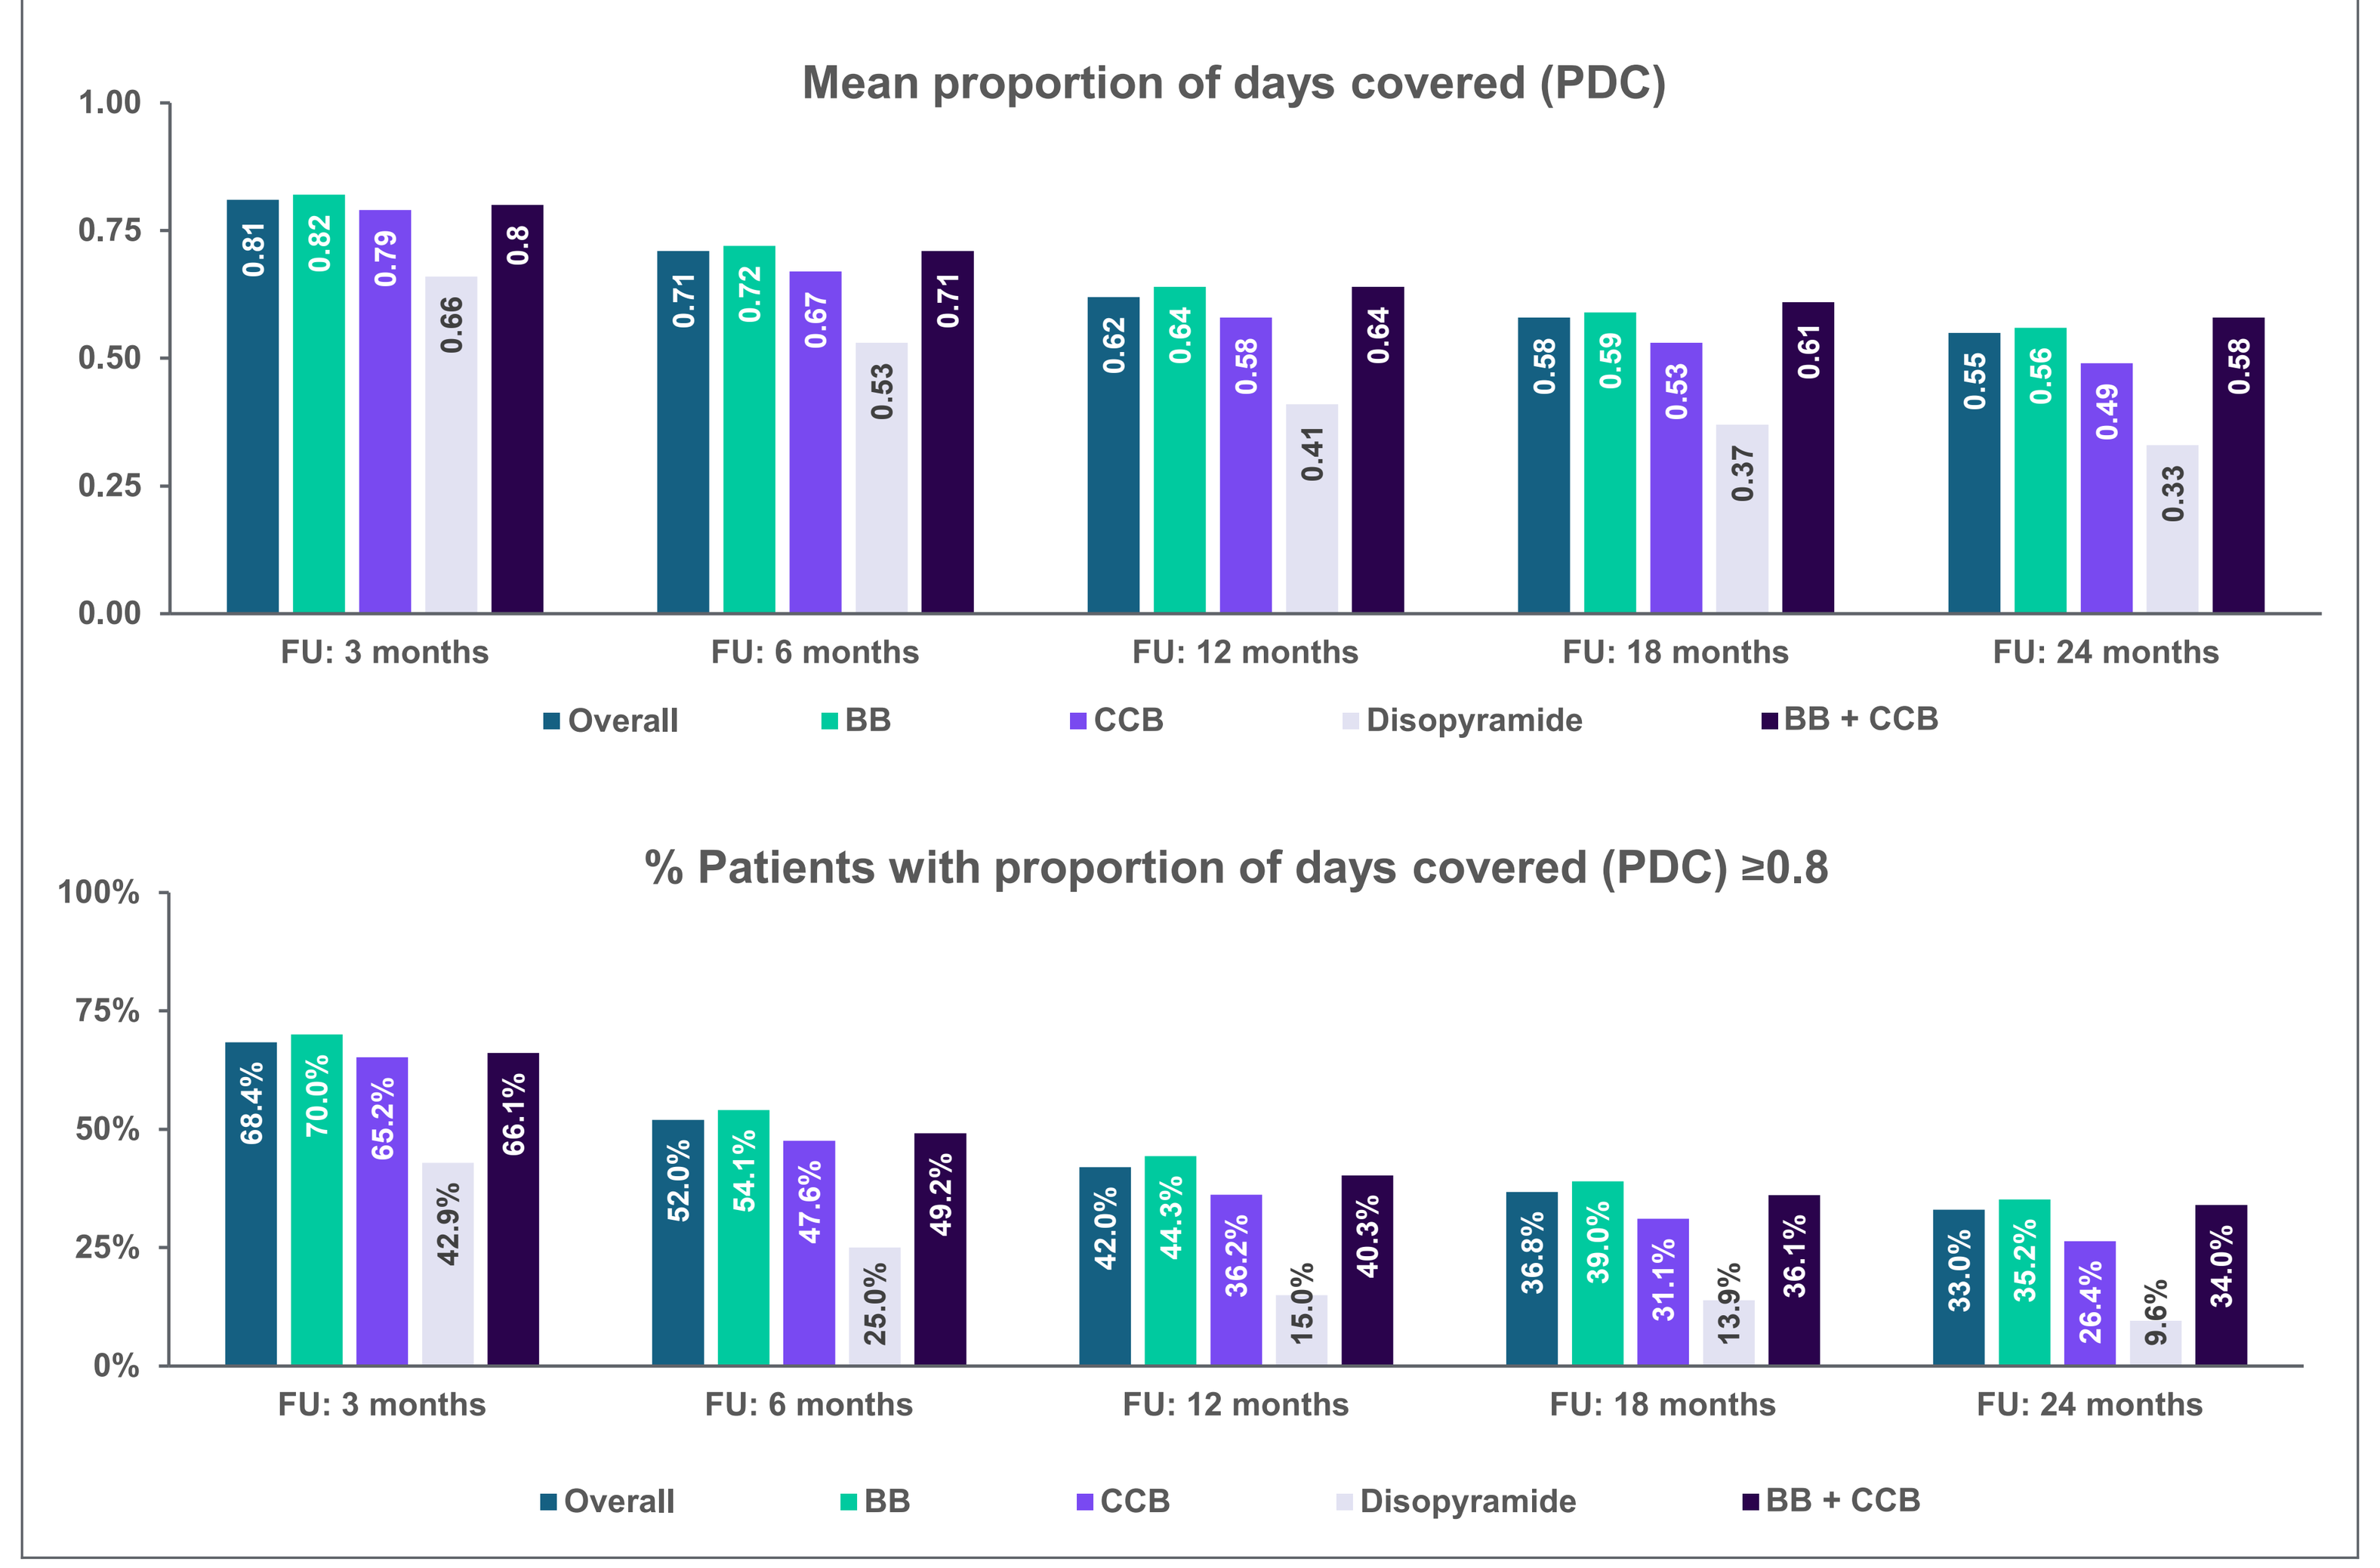
Mean proportion of days covered (PDC)
1.00
0.75
0.50
0.25
0.00
0.81
0.82
0.79
0.66
0.8
0.71
0.72
0.67
0.53
0.71
0.62
0.64
0.58
0.41
0.64
0.58
0.59
0.53
0.37
0.61
0.55
0.56
0.49
0.33
0.58
FU: 3 months
FU: 6 months
FU: 12 months
FU: 18 months
FU: 24 months
Overall
BB
CCB
Disopyramide
BB + CCB
% Patients with proportion of days covered (PDC) ≥0.8
100%
75%
50%
25%
0%
68.4%
70.0%
65.2%
42.9%
66.1%
52.0%
54.1%
47.6%
25.0%
49.2%
42.0%
44.3%
36.2%
15.0%
40.3%
36.8%
39.0%
31.1%
13.9%
36.1%
33.0%
35.2%
26.4%
9.6%
34.0%
FU: 3 months
FU: 6 months
FU: 12 months
FU: 18 months
FU: 24 months
Overall
BB
CCB
Disopyramide
BB + CCB

BB, beta-blocker; CCB, calcium channel blocker; FU, follow-up.

- Following oHCM diagnosis, 2449 (27.4%) patients developed an ADE after receiving a new pharmacotherapy (**Table 4**).
- The most common ADEs included shortness of breath (IR: 15.5 [14.8–16.2]), tiredness (IR: 6.7 [6.3–7.1]), dizziness (IR: 6.4 [6.0–6.8]), fatigue (IR: 5.5 [5.2–5.9]), and nausea (IR: 5.0 [4.7–5.4]) (**Table 4**).
- Other ADEs reported in <5% of patients included bradycardia, depression, diarrhea, edema, headache, hypotension, rash, constipation, and syncope (**Table 4)**.
- Incidence of a new major adverse cardiovascular events (MACE) following index treatment was 34.2%, with 18.8% having dyslipidemia, 10.5% coronary artery disease, 9.5% heart failure, and 7.8% atrial fibrillation (**Table 4**).

Limitations

- Real-world data in this study utilized ICD-9 and ICD-10 coding for disease identification and study outcomes, and may be subject to inconsistencies without patient-level genetic and anatomical confirmation.
- Due to the respective nature of claims analyses, causality of ADEs and MACE in this study cannot be inferred.

Table 4: Incidence rates of ADEs and MACE

Patients with oHCM (N=20,539)	Patients without ADE[a], n	Patients who developed ADE[b], n (%)	Follow-up time[c], n	Incidence rate (95% CI)[c]
Any ADE, including MACE	4337	1679 (38.7)	2665	63.0 (60.1–66.1)
Any ADE, excluding MACE	8928	2449 (27.4)	6872	35.6 (34.3–37.1)
Bradycardia	19,290	833 (4.3)	18,421	4.5 (4.2–4.8)
Depression	18,109	843 (4.7)	17,382	4.8 (4.5–5.2)
Diarrhea	19,739	527 (2.7)	19,024	2.8 (2.5–3.0)
Dizziness	18,442	1104 (6.0)	17,263	6.4 (6.0–6.8)
Edema	19,087	819 (4.3)	18,156	4.5 (4.2–4.8)
General ADE	20,509	21 (0.1)	20,214	0.1 (0.1–0.2)
Headache	18,952	721 (3.8)	18,165	4.0 (3.7–4.3)
Hypotension	19,334	657 (3.4)	18,868	3.5 (3.2–3.8)
Pruritus	20,288	175 (0.9)	19,811	0.9 (0.8–1.0)
Rash	20,167	292 (1.4)	19,560	1.5 (1.3–1.7)
Shortness of breath	14,573	2009 (13.8)	12,966	15.5 (14.8-16.2)
Tiredness	18,459	1140 (6.2)	17,136	6.7 (6.3–7.1)
Constipation	19,460	609 (3.1)	18,747	3.2 (3.0–3.5)
Nausea	18,818	902 (4.8)	17,909	5.0 (4.7–5.4)
Fatigue	18,722	968 (5.2)	17,467	5.5 (5.2–5.9)
Syncope	18,606	755 (4.1)	17,923	4.2 (3.9–4.5)
MACE	6879	2146 (31.2)	4707	45.6 (43.7–47.6)
Atrial fibrillation	16,087	1252 (7.8)	15,333	8.2 (7.7–8.6)
Atrial flutter	19,726	431 (2.2)	19,208	2.2 (2.0–2.5)
Coronary artery disease	15,511	1631 (10.5)	14,288	11.4 (10.9–12.0)
Dyslipidemia	10,916	2047 (18.8)	8303	24.7 (23.6–25.7)
Heart failure	16,344	1545 (9.5)	15,677	9.9 (9.4–10.4)
Stroke	19,688	386 (2.0)	19,204	2.0 (1.8–2.2)
Supraventricular tachycardia	19,580	521 (2.7)	18,844	2.8 (2.5–3.0)
Ventricular fibrillation	20,365	71 (0.3)	20,017	0.4 (0.3–0.4)
Ventricular tachycardia	19,114	643 (1.8)	18,178	3.5 (3.3–3.8)

[a] Prior to index treatment.
[b] Post index treatment.
[c] Per 100 person-years.
ADE, adverse drug effect; MACE, major adverse cardiac event; oHCM, obstructive hypertrophic cardiomyopathy.

CONCLUSIONS

- Most patients with symptomatic oHCM were nonadherent to standard pharmacotherapy, with more than 1 in 4 patients reporting ADEs.
- These findings highlight the urgent need for safe, effective, and less burdensome pharmacologic treatments for oHCM.

DISCLOSURES

RJR: Consultant fees / honoraria: Amgen Inc., AstraZeneca, Aventria, Boehringer Ingelheim Pharmaceuticals, Cytokinetics Incorporated, Jassen Pharmaceuticals, Johnson & Johnson, Roivant, Salix Pharmaceuticals, Serb Pharmaceuticals. **MB, SS:** Employees and shareholders of Cytokinetics, Incorporated. **EP:** Nothing to disclose. **NRD:** Contracted with CMS; research grants and consulting: Amgen, Arrowhead, AstraZeneca, Bayer, Boehringer Ingelheim, Bristol Myers Squibb, CSL Behring, CSL Vifor, Cytokinetics Incorporated, Merck, Milestone, Novartis, scPharmaceuticals, Verve Therapeutics.

ACKNOWLEDGMENTS

This study was funded by Cytokinetics, Incorporated. Editorial support for this presentation was provided by Engage Scientific Solutions and was funded by Cytokinetics, Incorporated. For information, contact Michael Butzner, DrPH, MPH, Cytokinetics, Inc., South San Francisco, CA, USA

mbutzner@cytokinetics.com

To obtain a PDF of this poster:
Scan the QR code
No personal information is stored

Cytokinetics®

Presented at AMCP | Houston, TX, USA | March 31–April 3, 2025

CYTOKINETICS® and the CYTOKINETICS and C-shaped logo are registered trademarks of Cytokinetics in the U.S. and certain other countries.